ATLAS: A Phase 2, Open-Label Study of Rucaparib in Patients with Locally Advanced or Metastatic Urothelial Carcinoma

Petros Grivas,[1] Min Yuen Teo,[2] Nicholas Vogelzang,[3] Ajjai Alva,[4] Yousef Zakharia,[5] Nabil Adra,[6] Alexandra Drakaki,[7] Arif Hussain,[8] Rafael Morales-Barrera,[9] Andrea Necchi,[10] Alejo Rodriguez-Vida,[11] Susan Feyerabend,[12] Sumati Gupta,[13] Debra H. Josephs,[14] Yohann Loriot,[15] Sandy Srinivas,[16] Kenton Wride,[17] Daleen Thomas,[17] Rachel L. Dusek,[17] Andrew D. Simmons,[17] Dale Nepert,[17] Simon Chowdhury[18]

[1]University of Washington, Seattle, WA; [2]Memorial Sloan Kettering Cancer Center, New York, NY; [3]Comprehensive Cancer Centers of Nevada, Las Vegas, NV; [4]University of Michigan Comprehensive Cancer Center, Ann Arbor, MI; [5]University of Iowa and Holden Comprehensive Cancer Center, Iowa City, IA; [6]Indiana University Simon Cancer Center, Indianapolis, IN; [7]University of California Department of Hematology/Oncology, Los Angeles, CA; [8]University of Maryland Greenebaum Cancer Center, Baltimore, MD; [9]Vall d'Hebron Institute of Oncology, Vall d' Hebron University Hospital, Universitat Autònoma de Barcelona, Barcelona, Spain; [10]IRCCS Istituto Nazionale dei Tumori, Milan, Italy; [11]Hospital del Mar, Barcelona, Spain; [12]Studienpraxis Urologie, Nürtingen, Germany; [13]Huntsman Cancer Institute, University of Utah, Salt Lake City, UT; [14]Guy's and St. Thomas' Hospital, London, UK; [15]Gustave Roussy Cancer Campus, Villejuif, France; [16]Stanford University School of Medicine, Stanford, CA; [17]Clovis Oncology, Inc., Boulder, CO; [18]Guy's Hospital and Sarah Cannon Research Institute, London, UK

Abstract CT179

INTRODUCTION

- There are limited treatment options for patients with metastatic urothelial carcinoma who have progressed following treatment with platinum (cisplatin or carboplatin)–based chemotherapy and/or immune checkpoint inhibitor therapy
- Poly(ADP-ribose) polymerase (PARP) inhibitors are approved in ovarian and breast cancer; however, there are limited data on the activity of PARP inhibitors in urothelial cancer
- Nonclinical studies have shown that the PARP inhibitor rucaparib has antitumor activity through a mechanism called synthetic lethality in tumors with homologous recombination (HR) deficiency (HRD) (**Figure 1**)[1-3]
 - Rucaparib also decreased tumor growth in mouse xenograft models of human cancer with or without deficiencies in *BRCA*[4]
- In multiple tumor types, the presence of HRD and/or genome-wide loss of heterozygosity (LOH; a specific type of DNA damage indicative of HRD) are associated with response to PARP inhibitors, such as rucaparib[5-7]
- To explore the potential utility of rucaparib in urothelial cancer, The Cancer Genome Atlas[8] muscle-invasive bladder cancer dataset was investigated for genomic evidence of HRD
- Results from the analysis indicated that many urothelial tumors have high genomic LOH (**Figure 2**)
- In the recurrent ovarian cancer setting, both LOH high and LOH low patients benefited from rucaparib treatment[6]
- These data suggest that the PARP inhibitor rucaparib may have activity in patients with locally advanced, unresectable or metastatic urothelial cancer, regardless of tumor HRD status

Figure 1. Schematic of Rucaparib-Induced Cytotoxicity in Cells with HRD

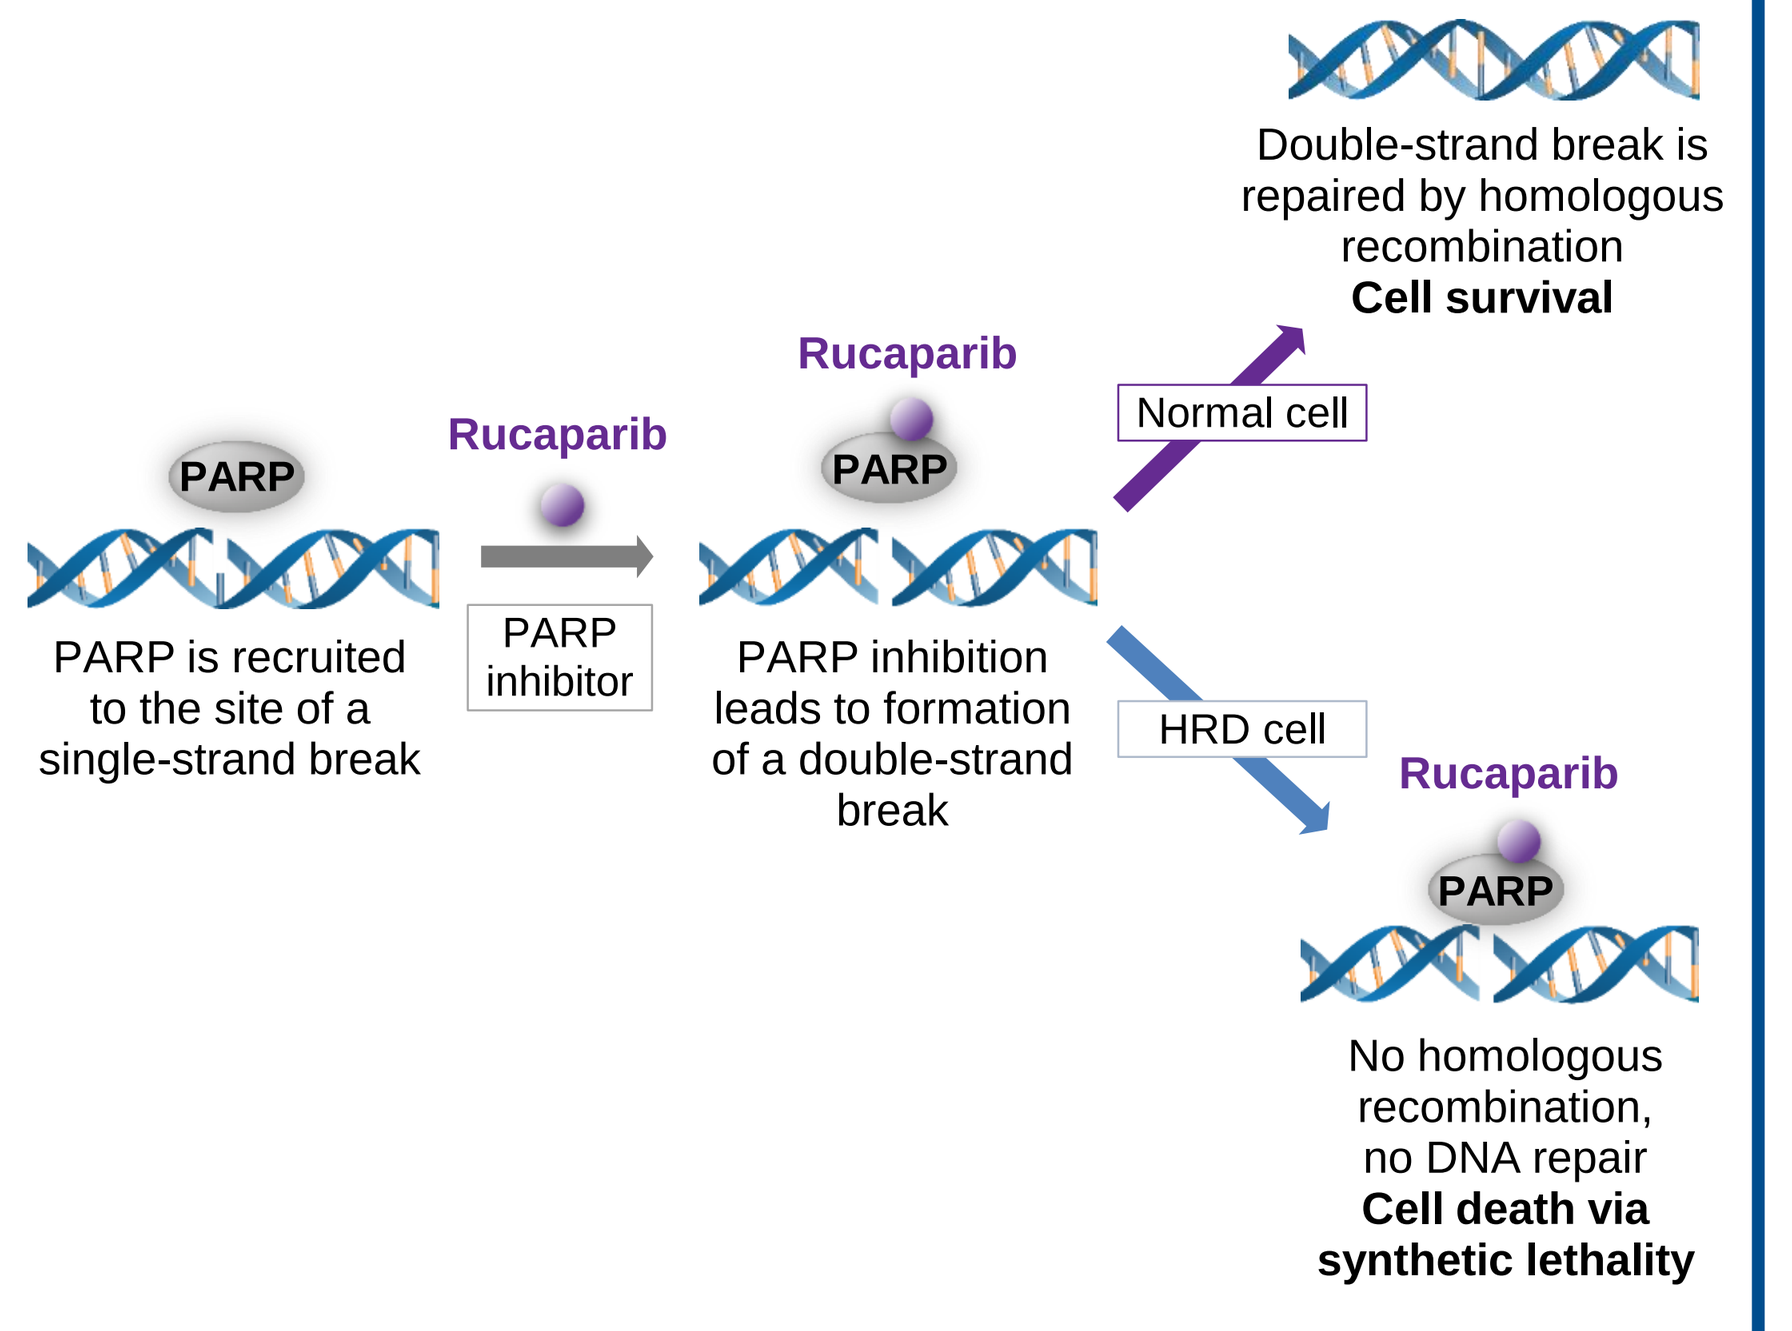
PARP
PARP is recruited to the site of a single-strand break
Rucaparib
PARP inhibitor
Rucaparib
PARP
PARP inhibition leads to formation of a double-strand break
Normal cell
Double-strand break is repaired by homologous recombination
Cell survival
HRD cell
Rucaparib
PARP
No homologous recombination, no DNA repair
Cell death via synthetic lethality

HRD, homologous recombination deficiency; PARP, poly(ADP-ribose) polymerase.

Figure 2. Genomic LOH in Bladder, Ovarian, and AML Tumors

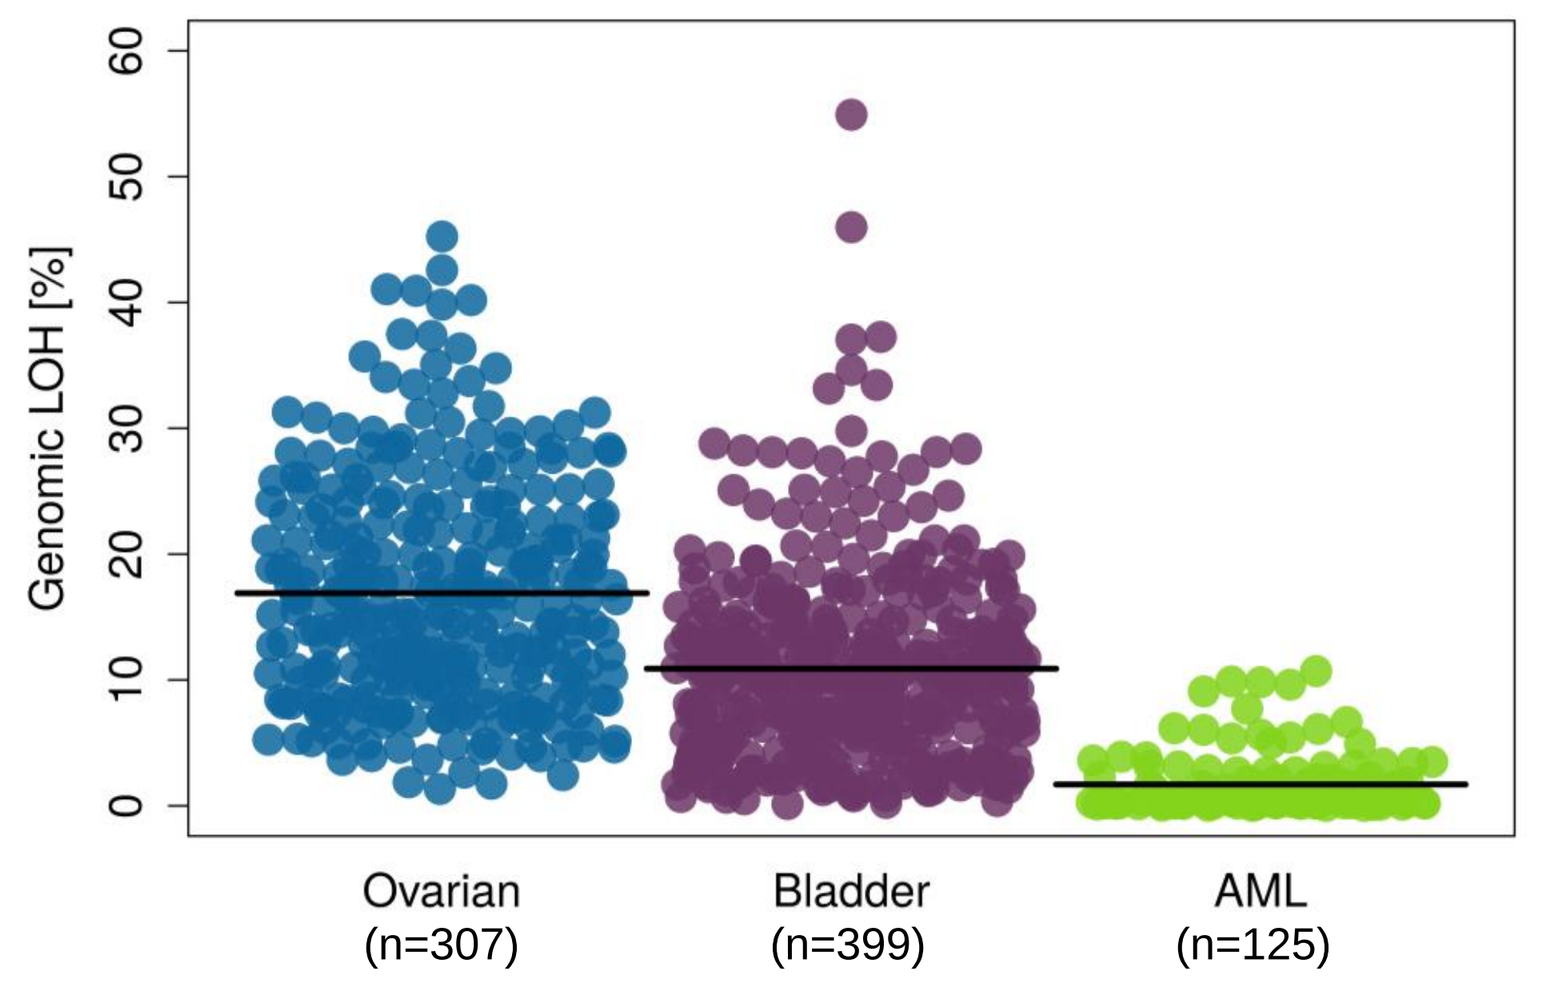
Genomic LOH [%]
0
10
20
30
40
50
60
Ovarian (n=307)
Bladder (n=399)
AML (n=125)

Scatter plot of genomic LOH of ovarian cancer, muscle-invasive bladder cancer, and AML samples evaluable for this analysis from the TCGA[8] database. Each dot represents an individual tumor sample; horizontal lines represent mean percentage genomic LOH. Data analyses for ovarian cancer and AML, which are associated with high and low percentages of genomic LOH, respectively, are shown for comparison.
AML, acute myeloid leukemia; LOH, loss of heterozygosity; TCGA, The Cancer Genome Atlas.

ATLAS TRIAL OVERVIEW

- ATLAS (CO-338-085; EudraCT 2017-004166-10; NCT03397394) is an international, open-label, phase 2 trial evaluating single-agent rucaparib (600 mg twice daily) as treatment for locally advanced, unresectable or metastatic urothelial carcinoma previously treated with 1 or 2 anticancer systemic regimens (**Figure 3**)[9]
 - **Eligible patients are not required to have tumors associated with HRD, because rucaparib may potentially benefit patients with HR-proficient or HR-deficient tumors**
- Approximately 200 patients will be enrolled
 - Two interim analyses are planned after data are available for 60 and 120 patients
- **Primary objective:** to evaluate objective response in the intent-to-treat and molecularly defined HRD-positive populations
- **Secondary objectives:** evaluation of duration of response, progression-free survival, overall survival, safety and tolerability, and steady-state pharmacokinetics of rucaparib
- The trial has >90% power to reject the null hypothesis (P=0.10) at a one-sided 5% significance level if the true response rate for rucaparib is 20%

AACR Annual Meeting | March 29–April 3, 2019 | Atlanta, GA

Figure 3. ATLAS Trial Schema

Screening

Screening assessments within 28 days of first dose of rucaparib

Key eligibility criteria
- ≥18 years of age
- Locally advanced unresectable UC or mUC
 - Patients with or without tumors associated with HRD*
- Measurable disease per RECIST version 1.1
- Confirmed radiographic progression after 1 or 2 prior treatments (eg, platinum-based chemotherapy, immune checkpoint inhibitor, and/or clinical trial) for advanced/metastatic disease
- ECOG PS 0 or 1
- No prior PARP inhibitor treatment
- Adequate organ function
- Mandatory tumor tissue collected ≤28 days before the first dose of rucaparib or archival tumor tissue collected ≤6 months before rucaparib treatment, with no intervening antitumor therapy

Treatment

28-day cycle

Rucaparib 600 mg BID (N≈200)

ATLAS — RUCAPARIB IN PATIENTS WITH LOCALLY ADVANCED OR METASTATIC UROTHELIAL CARCINOMA

- Radiographic disease progression, unacceptable toxicity, or other reason for discontinuation
- Tumor assessments and blood sample collection for circulating tumor DNA every 8 weeks for up to 18 months, then every 12 weeks thereafter
- Patients receiving clinical benefit may continue treatment beyond progression

Post-treatment

End of treatment visit

28-day follow-up visit

Long-term follow-up
- Tumor assessments every 8–12 weeks for patients who discontinue for reason other than progression
- All patients to be followed every 12 weeks for survival and subsequent therapies

Planned analyses

Primary endpoints
- Investigator-assessed objective response† will be evaluated in all patients
 - Investigator-assessed objective response will also be evaluated in patients with tumors that are HRD positive*

Secondary endpoints
- Duration of response
- Progression-free survival
- Overall survival
- Safety
- Pharmacokinetics

*Tumor HRD status determined by percentage of genomic loss of heterozygosity in a tumor sample before efficacy analysis. †RECIST[10] criteria will be used to document radiographic response.
BID, twice daily; ECOG PS, Eastern Cooperative Oncology Group Performance Status; HRD, homologous recombination deficiency; mUC, metastatic urothelial carcinoma; PARP, poly(ADP-ribose) polymerase; RECIST, Response Evaluation Criteria In Solid Tumors; UC, urothelial carcinoma.

BIOMARKER ASSESSMENT

- The ATLAS trial is enrolling biomarker unselected patients, with the goal of identifying and optimizing predictive biomarkers of rucaparib sensitivity and resistance (**Figure 4**)
 - Collecting these samples also allows for the comprehensive characterization of tumor evolution

Figure 4. ATLAS Biomarker Assessments Schedule

Screening	Treatment	End of treatment
Archival tumor tissue, if available + Mandatory fresh tumor tissue + Blood	Blood; Blood sample collection (▲) for ctDNA on day 1 of cycles 1–3, then Q8W for up to 18 months, followed by Q12W until end of treatment	Blood + Optional fresh tumor tissue

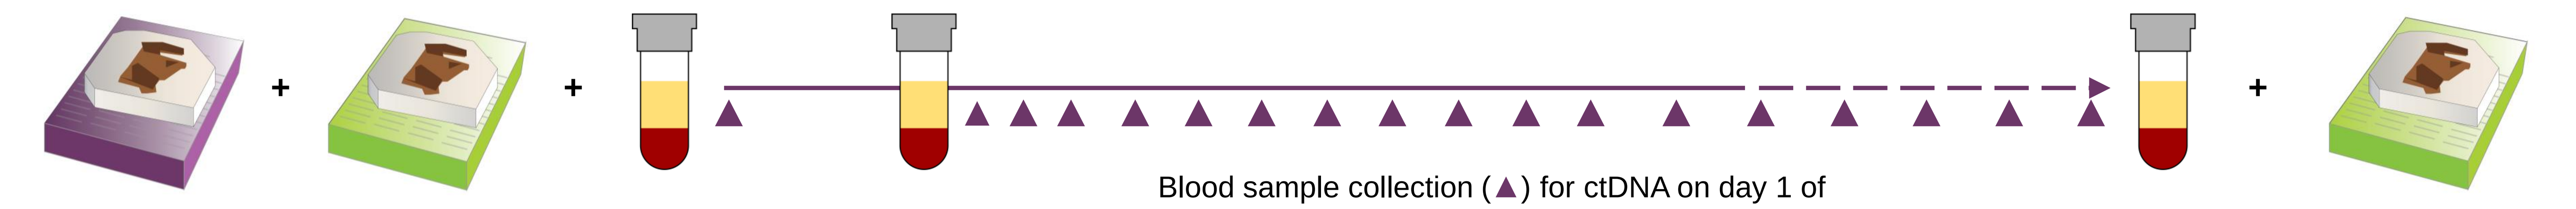

Next-generation sequencing

Biomarker assessments will explore:
- Effects of prior therapy on tumor and subsequent response to rucaparib
- Correlation of tumor HRD status (determined by percent genomic LOH) with response to rucaparib
- Other tumor biomarkers associated with response and/or resistance to rucaparib
- Changes over time in serially sampled ctDNA that may be associated with response and/or resistance to rucaparib
- Mutant allele frequencies and alterations in genes associated with DNA repair and oncogenic signaling pathways
- Additional genomic, transcriptional, and protein biomarkers will also be evaluated, including TMB, MSI, and PD-L1 expression

ctDNA, circulating tumor DNA; HRD, homologous recombination deficiency; LOH, loss of heterozygosity; MSI, microsatellite instability; PD-L1, programmed cell death ligand 1; Q8W, every 8 weeks; Q12W, every 12 weeks; TMB, tumor mutational burden.

Tumor Tissue Collection and Analyses

- Tumor tissue samples to be collected:
 - Archival (required, if available)
 - Screening (mandatory)
 - Treatment discontinuation (optional)
- Tumor tissue samples will be analyzed by Foundation Medicine (Cambridge, MA) using the DX1 next-generation sequencing (NGS) assay. The readouts of this assay include:
 - Alterations in a panel of 310 cancer-related genes
 - Genomic LOH
 - Tumor mutational burden (TMB)
 - Microsatellite instability (MSI)
- Tumors will also be evaluated for expression of the immune regulatory protein PD-L1 using the 22C3 (Agilent) and SP142 (Ventana) immunohistochemistry assays at HistoGeneX (Wilrijk, Belgium)

Blood Sample Collection and Analyses

- Blood samples to be collected (mandatory):
 - At screening
 - Day 1 of cycles 1–3, then every 8 weeks for up to 18 months, followed by every 12 weeks until the end of treatment
 - At treatment discontinuation
- Circulating tumor DNA extracted from blood will be profiled by Foundation Medicine using the FoundationACT™ NGS assay, which analyzes a panel of 67 cancer-related genes

Results from NGS testing of tumor samples will be made available to the investigators

TRIAL SUMMARY

- The international, open-label, phase 2 trial ATLAS aims to assess the safety and efficacy of single-agent rucaparib in patients with locally advanced, unresectable or metastatic urothelial carcinoma, irrespective of tumor HRD status, who have progressed after 1 or 2 prior anticancer treatments for advanced/metastatic disease
- The translational research aspects of the ATLAS trial will assess the association between biomarkers and response and resistance to rucaparib
- The target enrollment is 200 patients at ≈65 sites in 6 countries
- There are 41 sites currently enrolling in the United States (**Figure 5**)

Figure 5. US Sites Participating in ATLAS

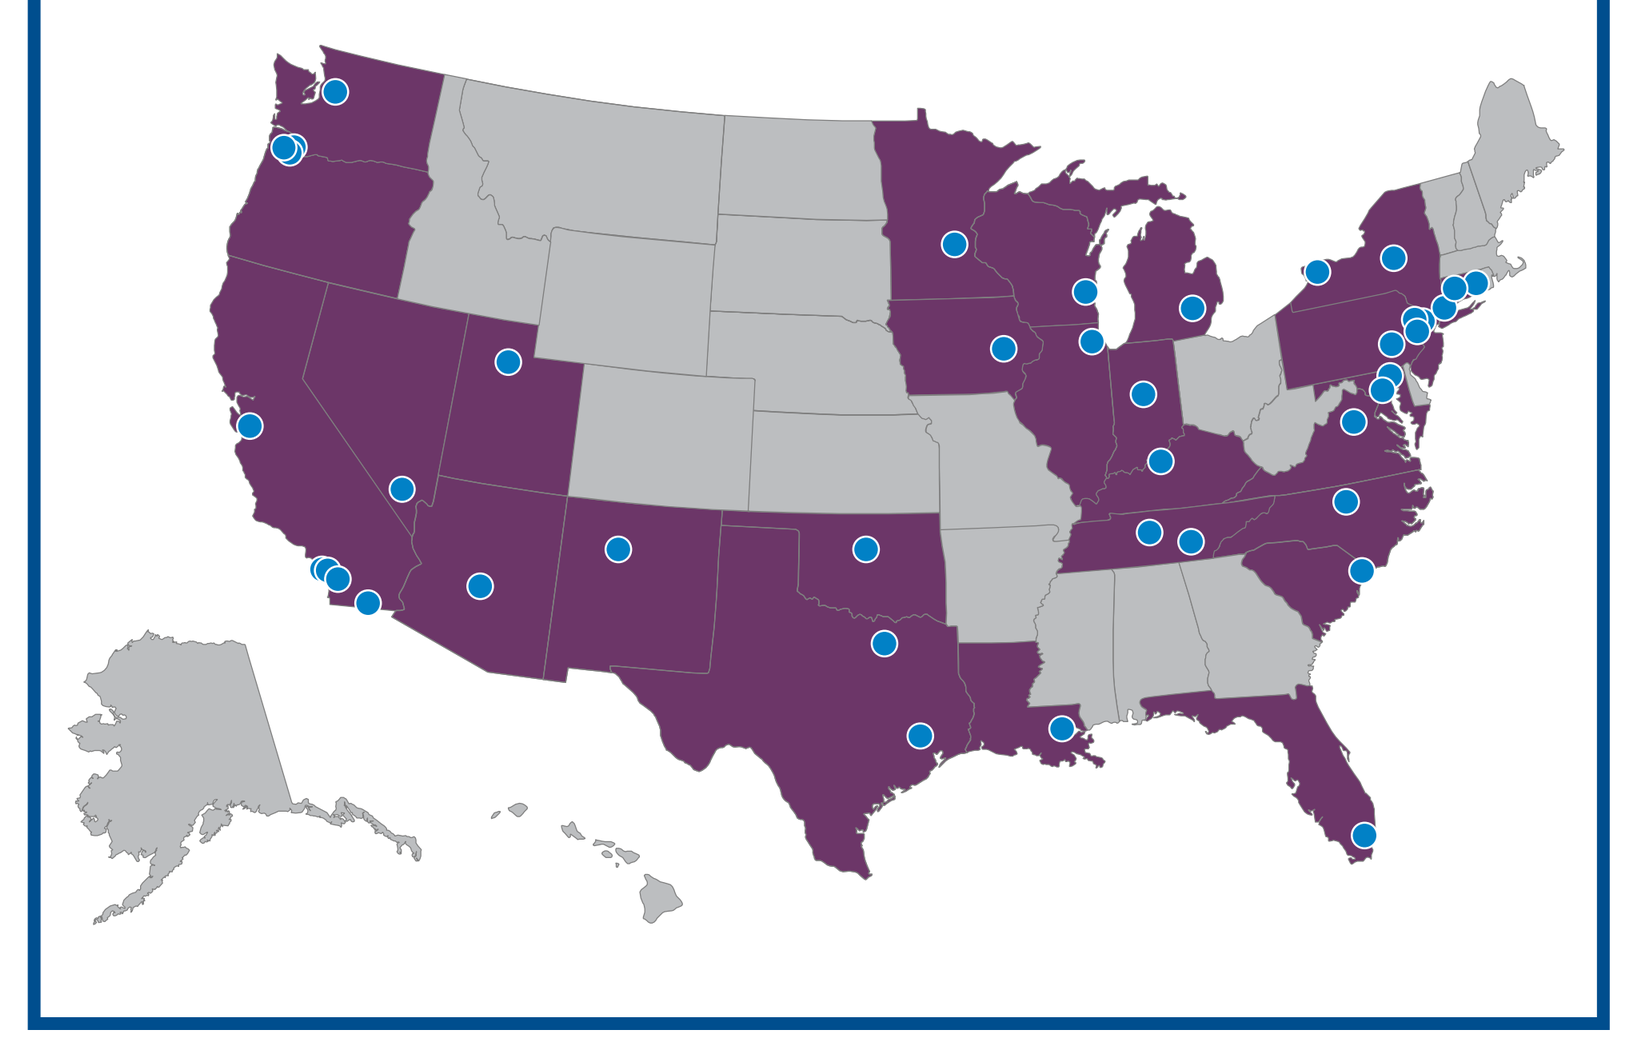

REFERENCES

1. Drew et al. *J Natl Cancer Inst.* 2011;103:334-46.
2. Nguyen et al. *Cancer Res*. 2017;77(13 suppl):abst 2476.
3. Robillard et al. *Cancer Res*. 2017;77(13 suppl):abst 2475.
4. Rubraca (rucaparib) tablets [prescribing information]. Boulder, CO: Clovis Oncology, Inc.; 2018.
5. Swisher et al. *Lancet Oncol*. 2017;18:75-87.
6. Coleman et al. *Lancet.* 2017;390:1949-61.
7. Abida et al. *Ann Oncol*. 2018;29:abst 793PD.
8. The Cancer Genome Atlas. https://cancergenome.nih.gov/. Accessed January 17, 2018.
9. Grivas et al. *J Clin Oncol.* 2018;36:abst TPS4592.
10. Eisenhauer et al. *Eur J Cancer*. 2009;45:228-47.

ACKNOWLEDGMENTS

This trial is funded by Clovis Oncology, Inc. The authors would like to thank Neeraj Agarwal (Huntsman Cancer Institute, University of Utah) for his contributions to the design and conduct of the ATLAS trial. Medical writing and editorial support funded by Clovis Oncology was provided by Ritu Pathak and Shannon Davis of Ashfield Healthcare Communications. The ATLAS trial design was originally presented at the ASCO Annual Meeting, June 1–5, 2018, Chicago, IL.[9]

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission from the authors.
Corresponding author: Petros Grivas; email: pgrivas@uw.edu