Abstract 559

Real-world Progression-Free Survival and Overall Survival for Patients With Advanced Ovarian Cancer Utilizing PARP Inhibitor Second-line Maintenance Therapy vs Active Surveillance

Robert Reid,[1] Junxin Shi,[2] Alisha Monnette,[2] Katrine L. Wallace[3]

[1]US Oncology, Medical Oncology, Virginia Cancer Specialists, Fairfax, VA, USA; [2]Real World Research, Ontada, The Woodlands, TX, USA; [3]Health Economics and Outcomes Research, Clovis Oncology, Boulder, CO, USA

SUMMARY

- **We report here one of the first studies to determine survival outcomes for use of poly(ADP-ribose) polymerase inhibitors (PARPi) as second-line maintenance therapy vs active surveillance in a real-world setting**
- **This study of PARPi therapy vs active surveillance confirms the efficacy benefits demonstrated in randomized studies of PARPis vs placebo, and provides evidence of the real-world effectiveness of PARPi maintenance therapy to improve survival in a population with advanced ovarian cancer**
- **Our results highlight the need for increased education and adherence to National Comprehensive Cancer Network guidelines outside of the setting of randomized controlled trials**

REFERENCES

1. American Cancer Society. 2022. https://www.cancer.org/content/dam/cancer-org/research/cancer-facts-and-statistics/annual-cancer-facts-and-figures/2022/2022-cancer-facts-and-figures.pdf. Accessed 01 August 2022.
2. Islami F, et al. *J Natl Cancer Inst*. 2021;113:1648-69.
3. Coleman RL, et al. *Lancet*. 2017;390:1949-61.
4. Pujade-Lauraine E, et al. *Lancet Oncol*. 2017;18:1274-84.
5. Mirza MR, et al. *N Engl J Med*. 2016;375:2154-64.
6. National Comprehensive Cancer Network. 2022. https://www.nccn.org/professionals/physician_gls/pdf/ovarian.pdf. Accessed 01 August 2022.
7. Garofalo D, et al. *J Clin Oncol*. 2019;37:5579.
8. Moss HA, et al. *Gynecol Oncol*. 2021;163:50-6.
9. Reid R, et al. *J Clin Pathways*. 2022;8:38-46.

ACKNOWLEDGEMENTS

This study is funded by Clovis Oncology, Inc. Editorial support was funded by Clovis Oncology, Inc., and provided by Kathleen Blake of Ashfield MedComms, an Inizio company.

Copyright © 2022 Robert.Reid@USOncology.com

INTRODUCTION

- Although the age-adjusted incidence of ovarian cancer is lower than other cancers among women, such as breast cancer (11.2 vs 127.5 per 100,000 in the United States), the mortality rate is much higher. The 5-year survival rate for patients with ovarian cancer in the United States is only 49% versus 90% for those with breast cancer[1]
 - Most patients are diagnosed with advanced stage disease and no screening tools currently exist for early detection of this cancer[2]
 - Given the high mortality rate, ensuring availability and access to life-prolonging therapies is essential
- In phase 3 trials, poly(ADP-ribose) polymerase inhibitors (PARPi) have been shown to extend the time between chemotherapy and relapse (ie, progression-free survival [PFS]) in patients with advanced recurrent ovarian cancer receiving therapy in the second-line (2L) and beyond.[3–5]
- On the basis of these studies, the National Comprehensive Cancer Network (NCCN) has recommended PARPis since 2017 as 2L+ maintenance treatment for patients with ovarian cancer (OC), with or without *BRCA1* or *BRCA2* (BRCA) alterations, who have not previously received a PARPi[6]
- Despite these recommendations, the use of PARPis as 2L maintenance therapy in community settings is estimated at only 33%–63%[7–9]
- Limited data exist on the benefit provided by PARPi use specifically as 2L maintenance therapy in real-world settings

OBJECTIVE

- The aim of this study was to estimate real-world PFS, overall survival (OS), and time to subsequent treatments for patients treated with 2L PARPi maintenance therapy vs active surveillance

METHODS

- From the iKnowMed electronic health record database of the US Oncology Network, which includes 1300 physicians operating in over 470 care sites in the US, adult females were included if they:
 - Were diagnosed with advanced epithelial OC,
 - Were documented to have International Federation of Gynecology and Obstetrics (FIGO) stage III or IV epithelial OC,
 - Had ≥2 visits within the database between 1/1/2016 and 12/1/2020, and
 - Received a 2L platinum-containing regimen for advanced OC
- This analysis excluded patients who previously received a PARPi and those who received treatment for other documented primary cancer diagnoses or multiple 2L maintenance therapies during the study observation period
- The data collected for the study included 1) use of any 2L maintenance therapy, and 2) 2L PARPi therapy or active surveillance following discontinuation of 2L platinum-based chemotherapy
- The study observation period was between January 1, 2016, and December 31, 2020. Patient follow-up for OS occurred within the study observation period. The minimum follow-up period was 3 months; the maximum follow-up period was 58 months
- Patients were followed longitudinally until March 31, 2021, the last patient record, or date of death, whichever occurred earliest
- A subset of patient charts from the PARPi and active surveillance groups was further reviewed to confirm eligibility for inclusion in the analyses of PFS, OS, time to first subsequent treatment (TFST), and time to second subsequent treatment (TSST) from initiation of 2L maintenance therapy
 - To enable unconfounded comparisons of OS, PFS, TFST, and TSST, we conducted a manual chart review to create a matched analysis population. All eligible patients from the PARPi group were selected, and then patients who underwent active surveillance were matched by age and Eastern Cooperative Oncology Group performance status (ECOG PS) to patients who received a PARPi
 - Kaplan-Meier survival methods and log-rank tests were used to estimate and compare OS, PFS, TFST, and TSST of the groups from initiation of 2L maintenance therapy to 24 months

RESULTS

- A total of 1154 patients met initial inclusion criteria (**Table 1**)
 - Of these patients, 315 (27%) subsequently had 2L PARPi maintenance therapy and 584 (51%) did not receive 2L maintenance therapy (ie, underwent "active surveillance")
 - 255 patients (22%) either received multiple 2L maintenance regimens or an "other" maintenance therapy of either bevacizumab or a maintenance chemotherapy
 - Of patients who received a PARPi or active surveillance, 143 patient charts were manually reviewed (85 [59%] PARPi, 58 [41%] active surveillance)
 - Among patients who received a PARPi, 21 (25%) received rucaparib, 32 (38%) received olaparib, and 32 (38%) received niraparib
- Mean (± standard deviation) age at advanced OC diagnosis was 65.2 (±11.6) years
- Median follow-up time from discontinuation of 2L platinum chemotherapy until the end of observation was 26.8 months (range 0.4–99.6)
- No statistically significant differences were observed between most baseline characteristics
 - Greater proportions of patients were diagnosed at a later stage in the PARPi group vs the active surveillance group (*P*=0.0224) (**Table 2**)
- PFS from initiation of 2L maintenance therapy to 24 months was significantly longer with PARPi maintenance therapy vs active surveillance: 21.5% (95% confidence interval [CI], 11.7–33.3) vs 10.2% (95% CI, 3.8–20.2); log-rank *P*=0.0004 (**Figure 1**)
- OS at 24 months was significantly longer with PARPi maintenance therapy vs active surveillance: 61.6% (95% CI, 45.5–74.2) vs 46.7% (95% CI, 31.6–60.5); log-rank *P*=0.0364 (**Figure 2**)
- Patients in the PARPi group had a significantly longer median TFST than did patients in the active surveillance group: 10.8 months (95% CI, 9.0–13.9) vs 2.2 months (95% CI, 1.4–5.4); log rank *P*<0.0001 (**Figure 3**)
- Patients in the PARPi group had a significantly longer median TSST than did patients in the active surveillance group: 17.9 months (95% CI, 15.2–23.4) vs 9.7 months (95% CI, 7.2–15.2); log rank *P*<0.0108 (**Figure 4**)

Table 1. Attrition Table

	Patients, n (%)
Met inclusion criteria of age 18+, diagnosed with advanced epithelial OC (stage III or IV), at least 2 visits following diagnosis between 1/1/2016 and 12/1/2020, and received 2L treatment with platinum-containing regimen between 1/1/2016 and 12/31/2020	**1154 (100)**
Received any 2L maintenance therapy	570 (49.4)
PARPi maintenance therapy	315 (27.3)
"Other"[a] or multiple 2L regimens	255 (22.1)
Did not receive 2L maintenance therapy (underwent active surveillance)	584 (50.6)
Patient chart manual review cohort (analysis population):	**143 (100)**
PARPi	85 (59.4)
Active surveillance	58 (40.6)

[a]Includes bevacizumab and chemotherapy maintenance. Patients with multiple regimens were subsequently excluded from the analysis. 2L, second-line; OC, ovarian cancer; PARPi, poly(ADP-ribose) polymerase inhibitor.

Figure 1. Progression-Free Survival From Initiation of Second-line Maintenance Therapy

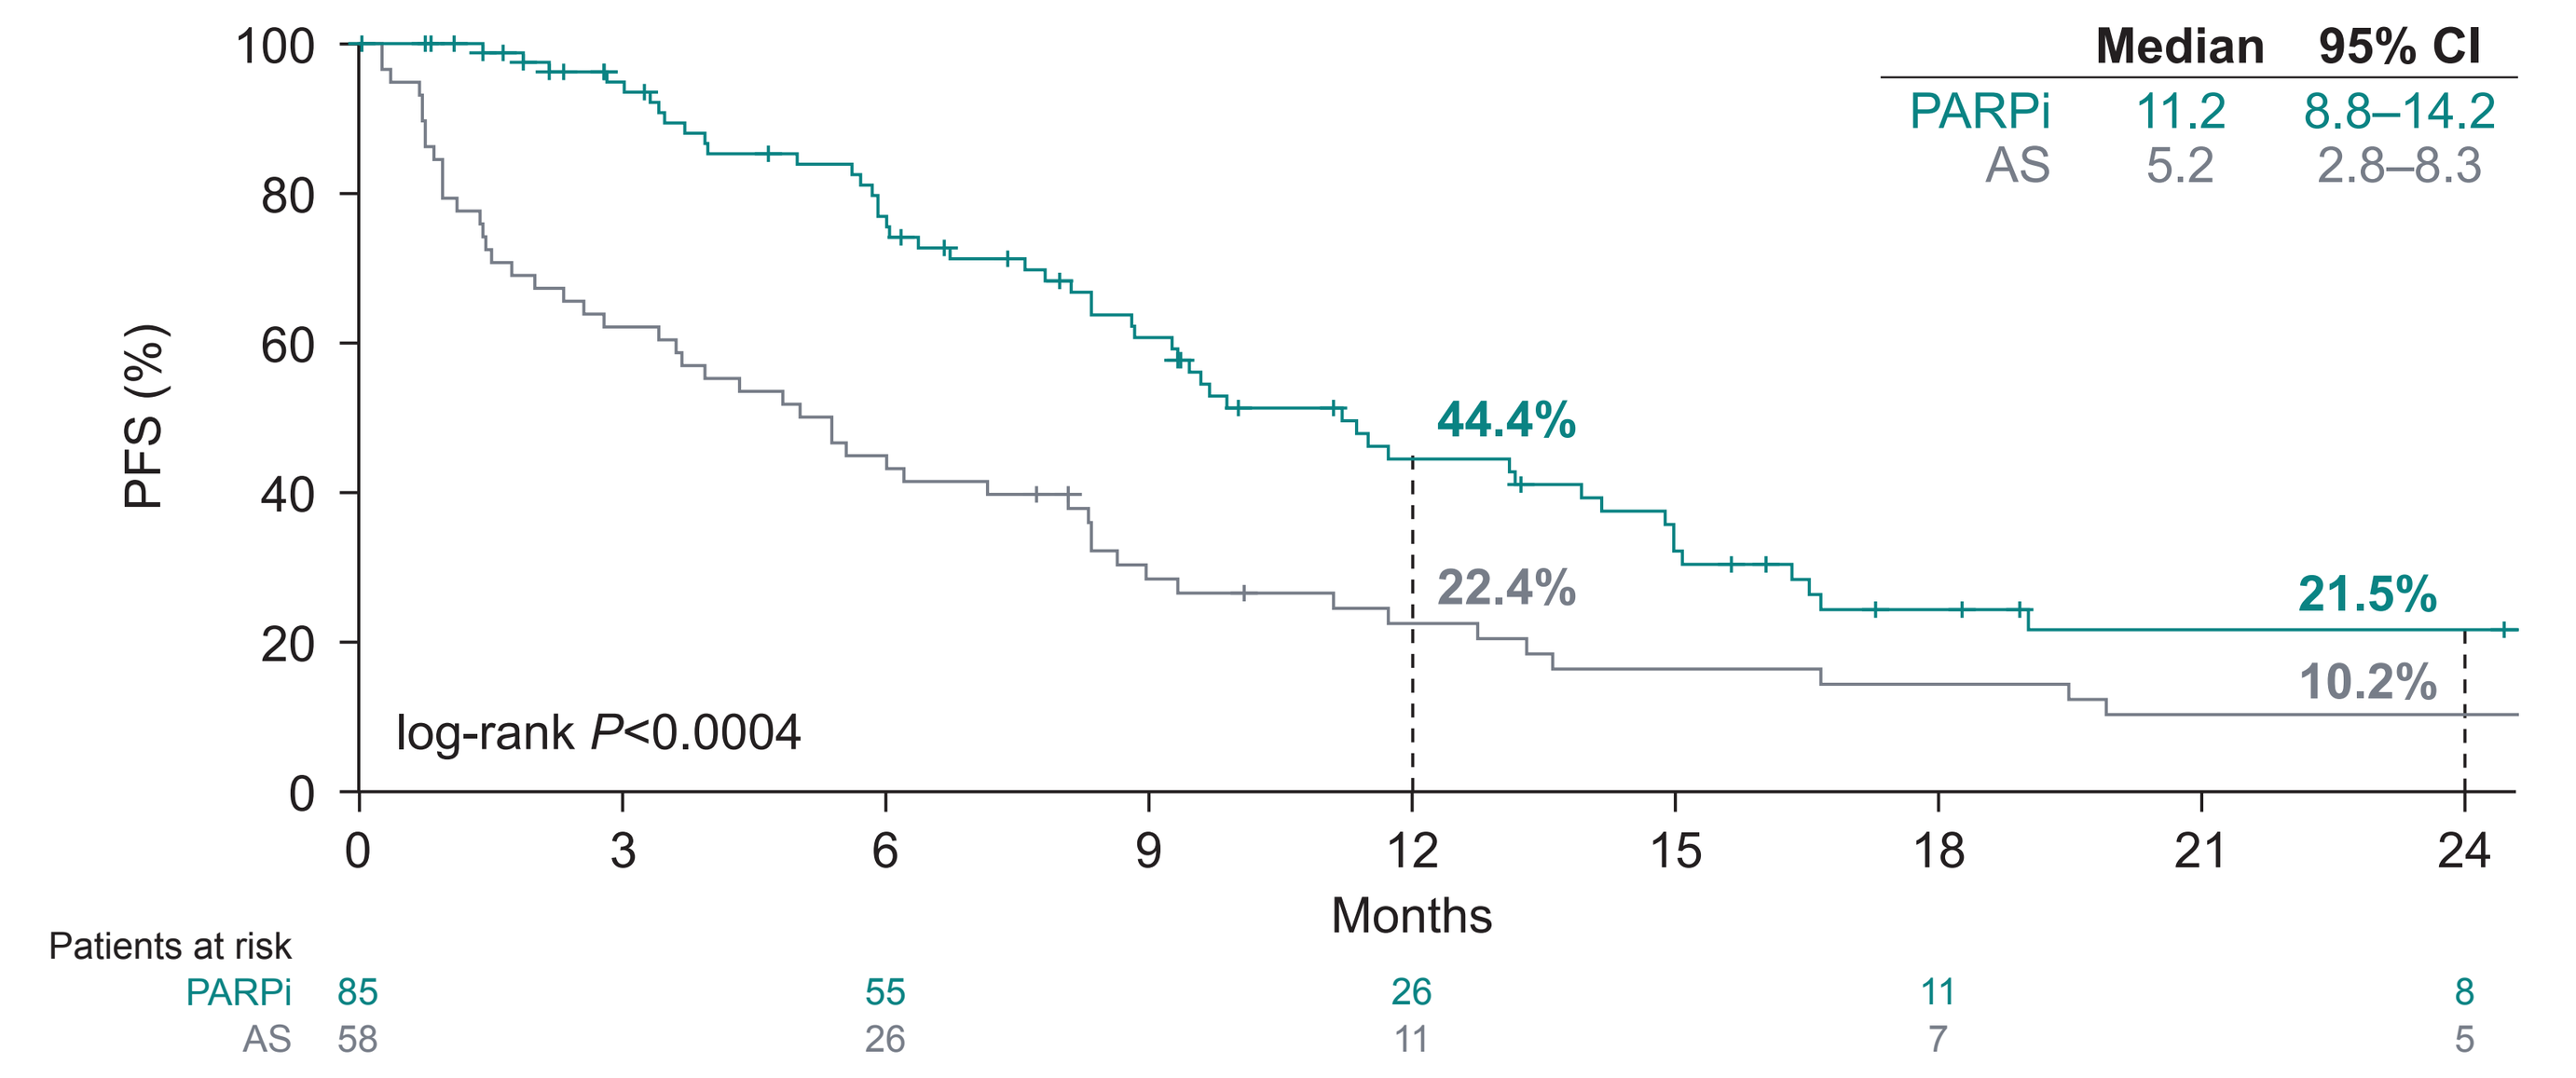
Median 95% CI
PARPi 11.2 8.8–14.2
AS 5.2 2.8–8.3
PFS (%)
44.4%
22.4%
21.5%
10.2%
log-rank P<0.0004
Months
Patients at risk
PARPi 85 55 26 11 8
AS 58 26 11 7 5

A log-rank test was used to compare PFS of the groups from initiation of 2L maintenance therapy to 24 months. 2L, second-line; AS, active surveillance; CI confidence interval; PARPi, poly(ADP-ribose) polymerase inhibitor; PFS, progression-free survival.

Figure 2. Overall Survival From Initiation of Second-line Maintenance Therapy

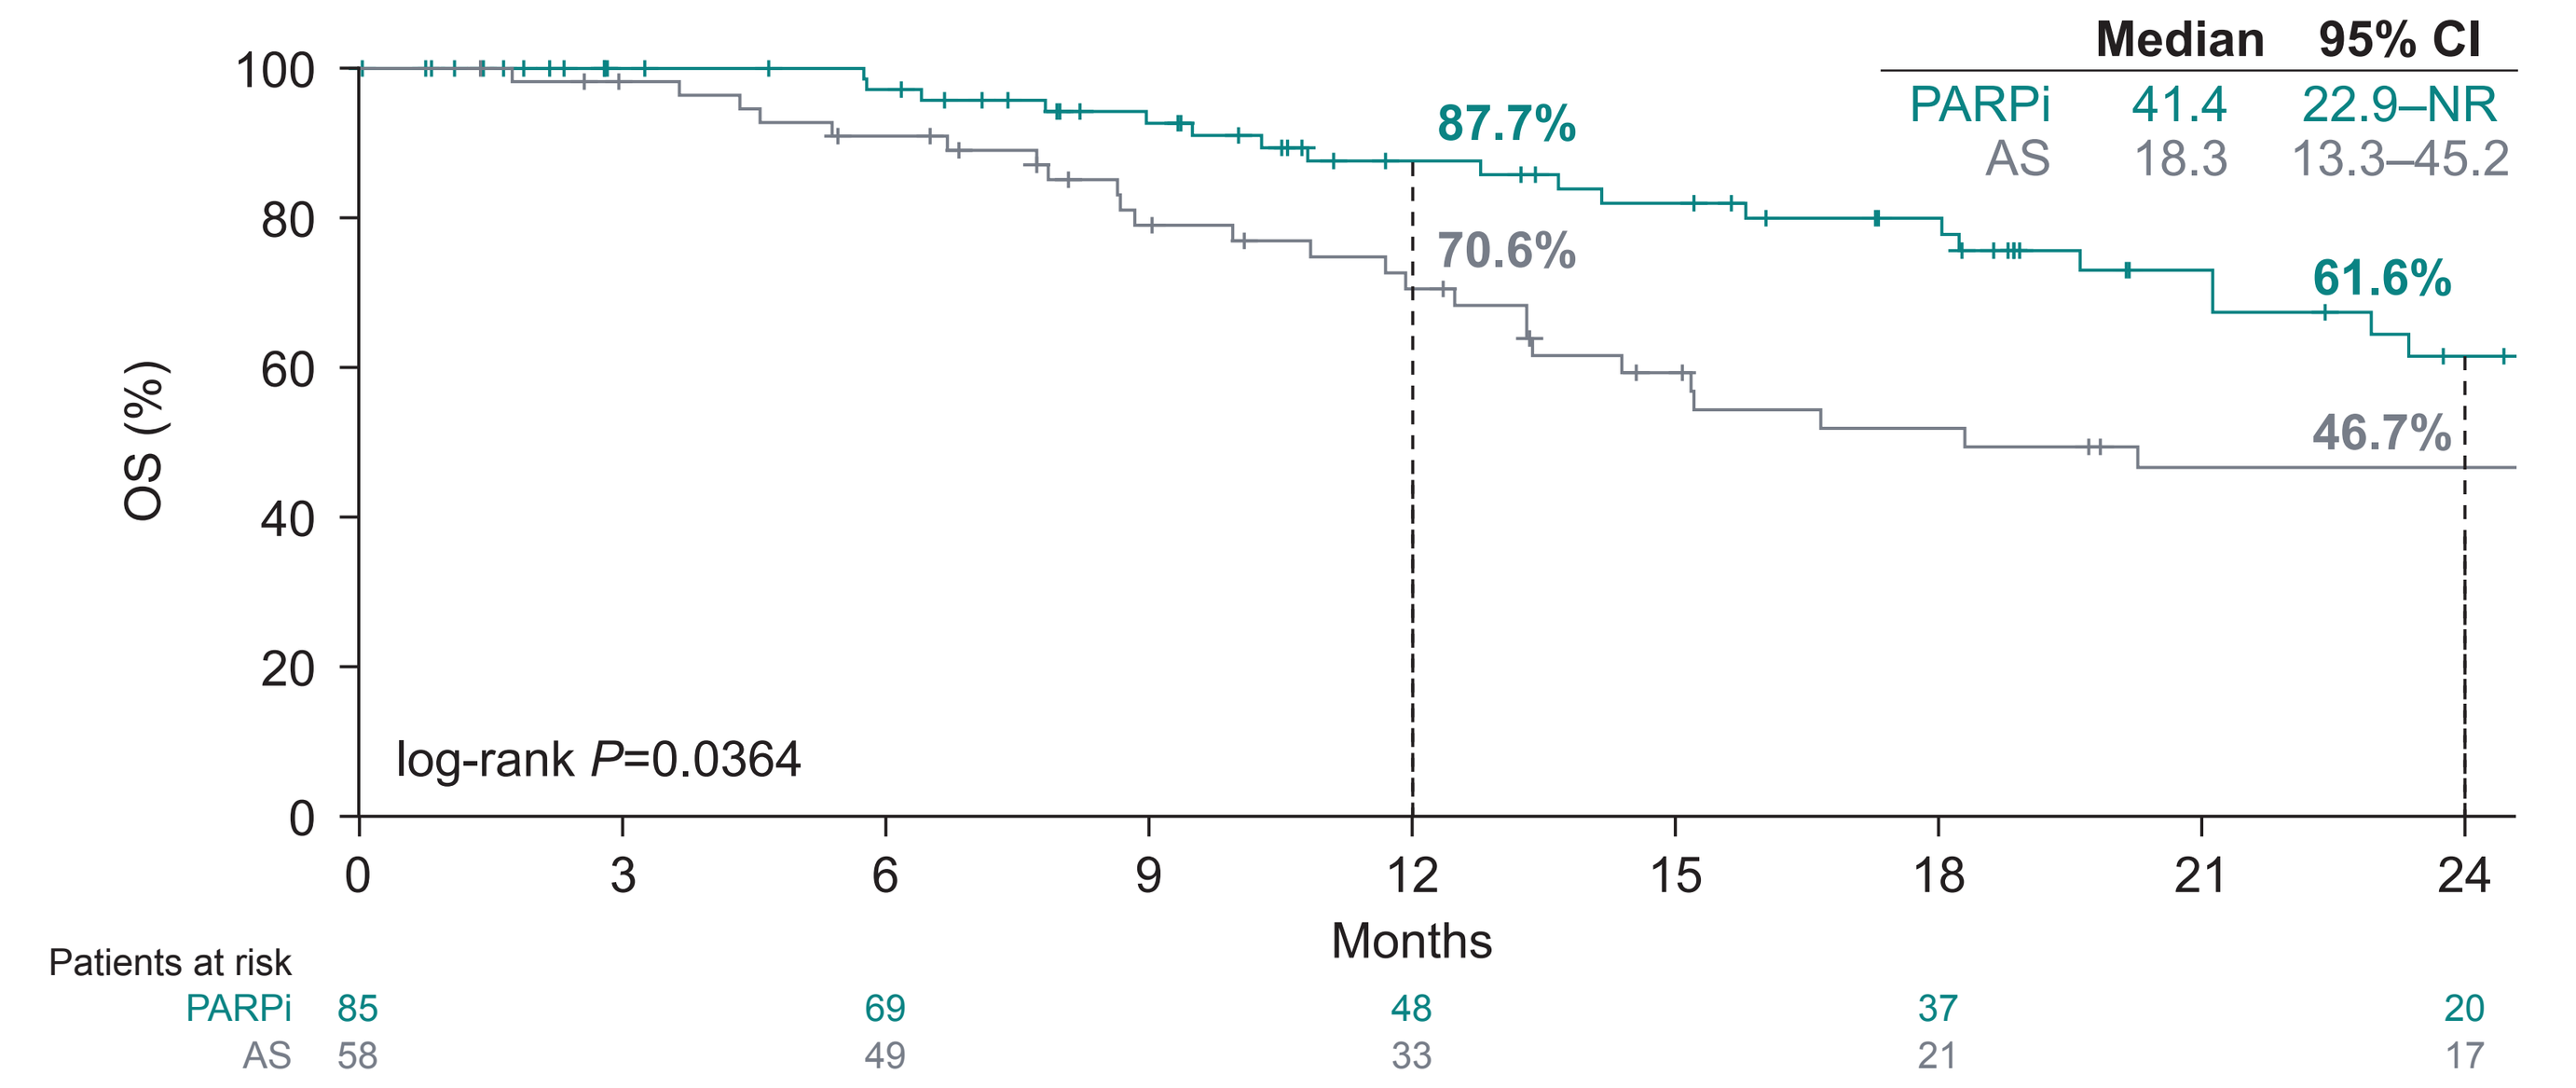
Median 95% CI
PARPi 41.4 22.9–NR
AS 18.3 13.3–45.2
OS (%)
87.7%
70.6%
61.6%
46.7%
log-rank P=0.0364
Months
Patients at risk
PARPi 85 69 48 37 20
AS 58 49 33 21 17

A log-rank test was used to compare OS of the groups from initiation of 2L maintenance therapy to 24 months. 2L, second-line; AS, active surveillance; CI confidence interval; NR, not reported; PARPi, poly(ADP-ribose) polymerase inhibitor; OS, overall survival.

Table 2. Characteristics of the Analysis Population

Baseline Characteristics	PARPi (n=85)	AS (n=58)	*P*-value
Age at index, years			
Mean (SD)	65.5 (11.1)	64.8 (12.5)	0.6452
FIGO stage of initial OC diagnosis, n (%)			0.0224
Stage I	1 (1.2)	1 (1.7)	
Stage II	3 (3.5)	7 (12.1)	
Stage III	68 (80.0)	34 (58.6)	
Undocumented	13 (15.3)	16 (27.6)	
Race, n (%)			0.4345
White	52 (61.2)	31 (53.4)	
Black	3 (3.5)	2 (3.4)	
Other race/multiple race	21 (24.7)	20 (34.5)	
Unknown	9 (10.6)	5 (8.6)	
Practice region, n (%)			0.1581
Midwest	25 (29.4)	10 (17.2)	
South	15 (17.6)	16 (27.6)	
West	45 (52.9)	32 (55.2)	
Body mass index, kg/m²			0.6787
Patients with available data	80	55	
Mean (SD)	28.1 (6.1)	29.0 (7.2)	
ECOG PS, n (%)			0.1600
0	11 (12.9)	4 (6.9)	
1	36 (42.4)	27 (46.6)	
2	3 (3.5)	6 (10.3)	
3	0	1 (1.7)	
Not documented	35 (41.2)	20 (34.5)	

AS, active surveillance; ECOG PS, Eastern Cooperative Oncology Group performance status; FIGO, International Federation of Gynecology and Obstetrics; OC, ovarian cancer; PARPi, poly(ADP-ribose) polymerase inhibitor; SD, standard deviation.

Figure 3. Time to First Subsequent Treatment From Initiation of Second-line Maintenance Therapy

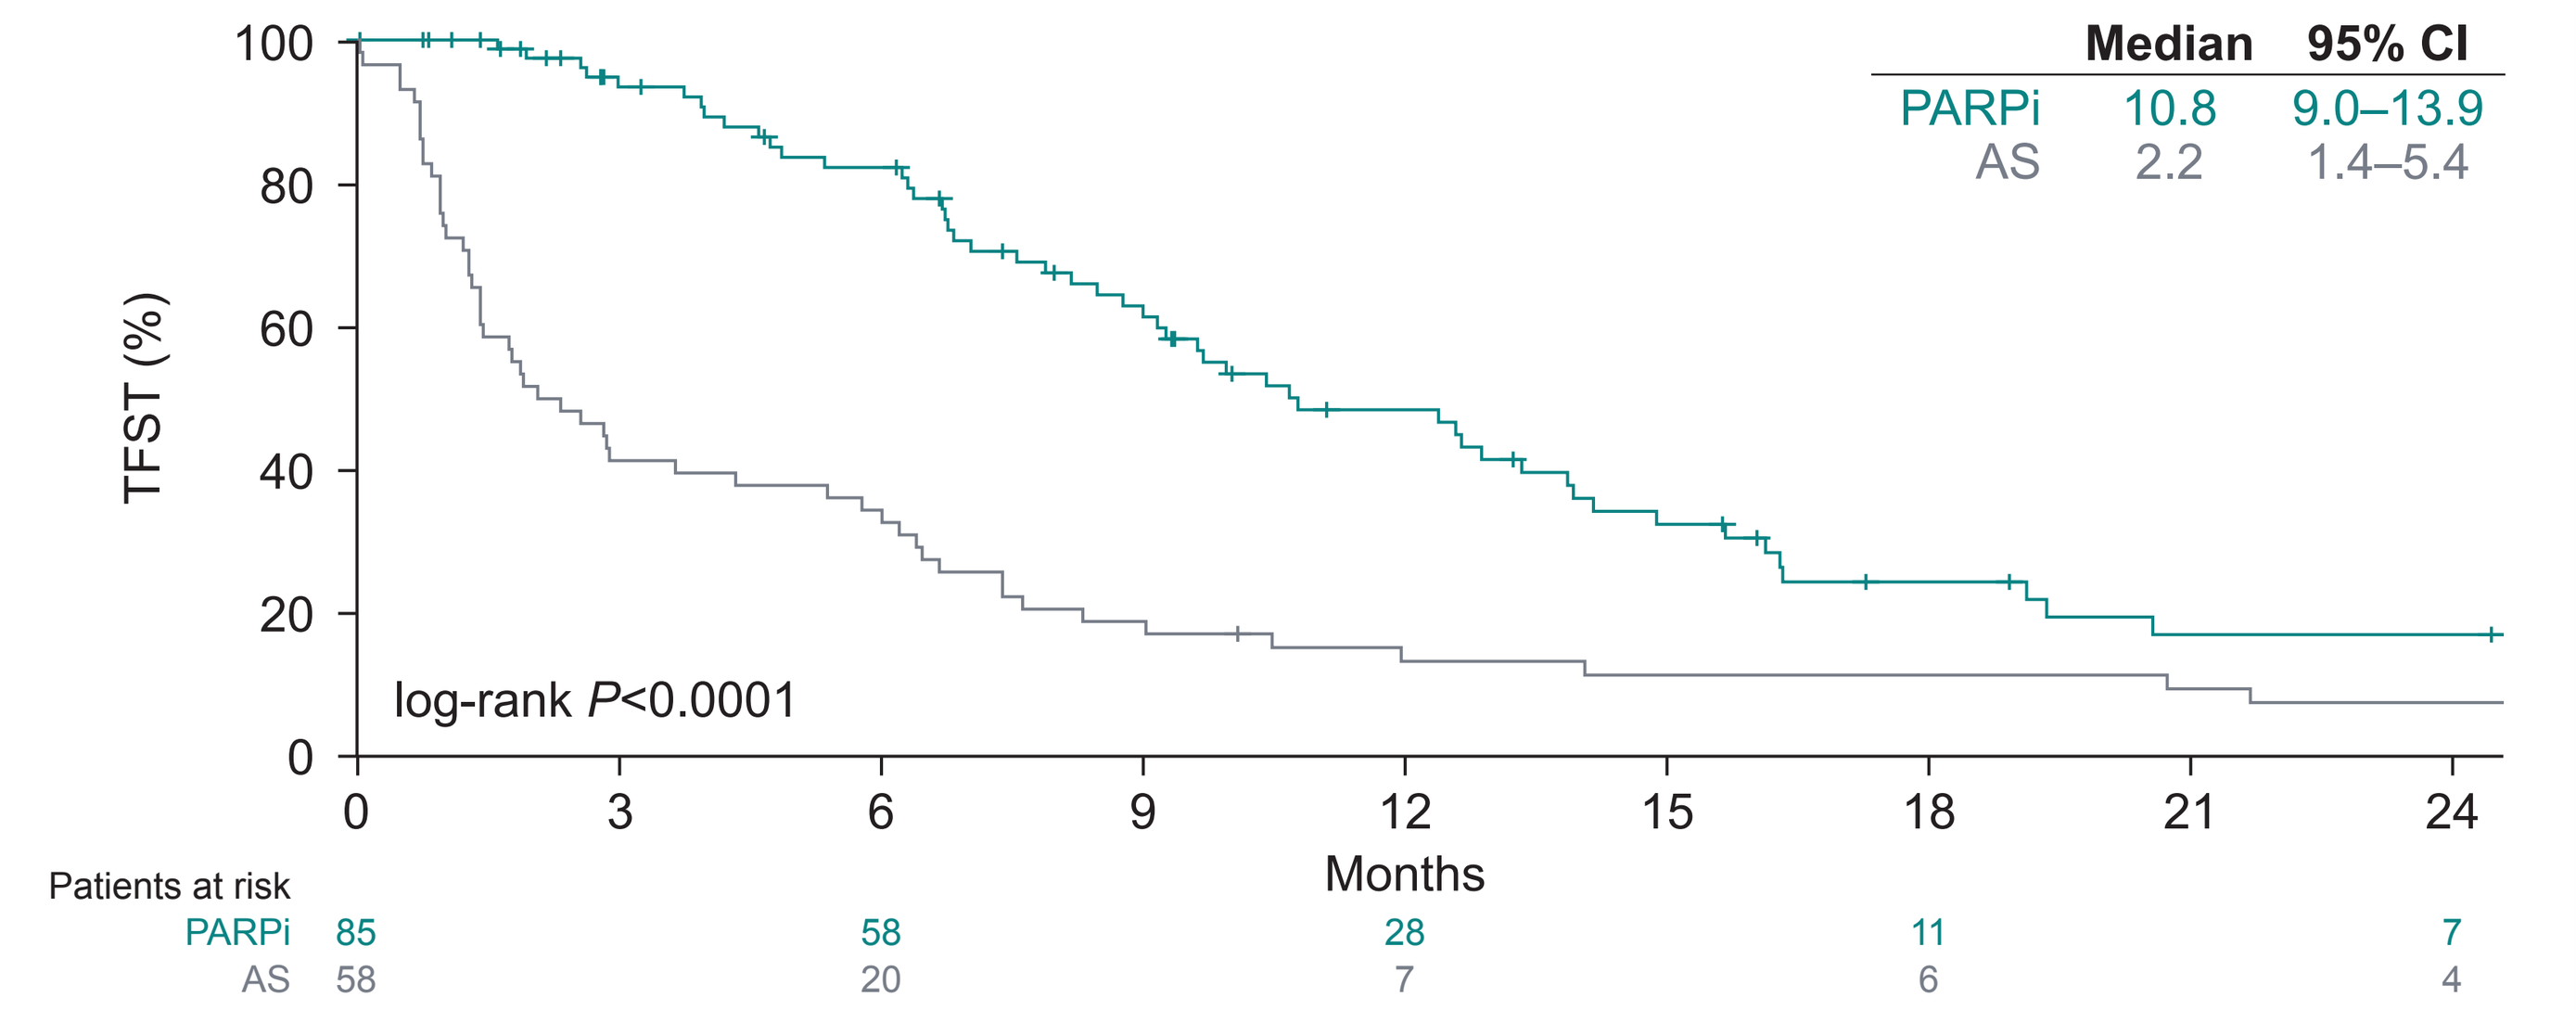
Median 95% CI
PARPi 10.8 9.0–13.9
AS 2.2 1.4–5.4
TFST (%)
log-rank P<0.0001
Months
Patients at risk
PARPi 85 58 28 11 7
AS 58 20 7 6 4

A log-rank test was used to compare TFST of the groups from initiation of 2L maintenance therapy to 24 months. 2L, second-line; AS, active surveillance; PARPi, poly(ADP-ribose) polymerase inhibitor; TFST, time to first subsequent treatment.

Figure 4. Time to Second Subsequent Treatment From Initiation of Second-line Maintenance Therapy

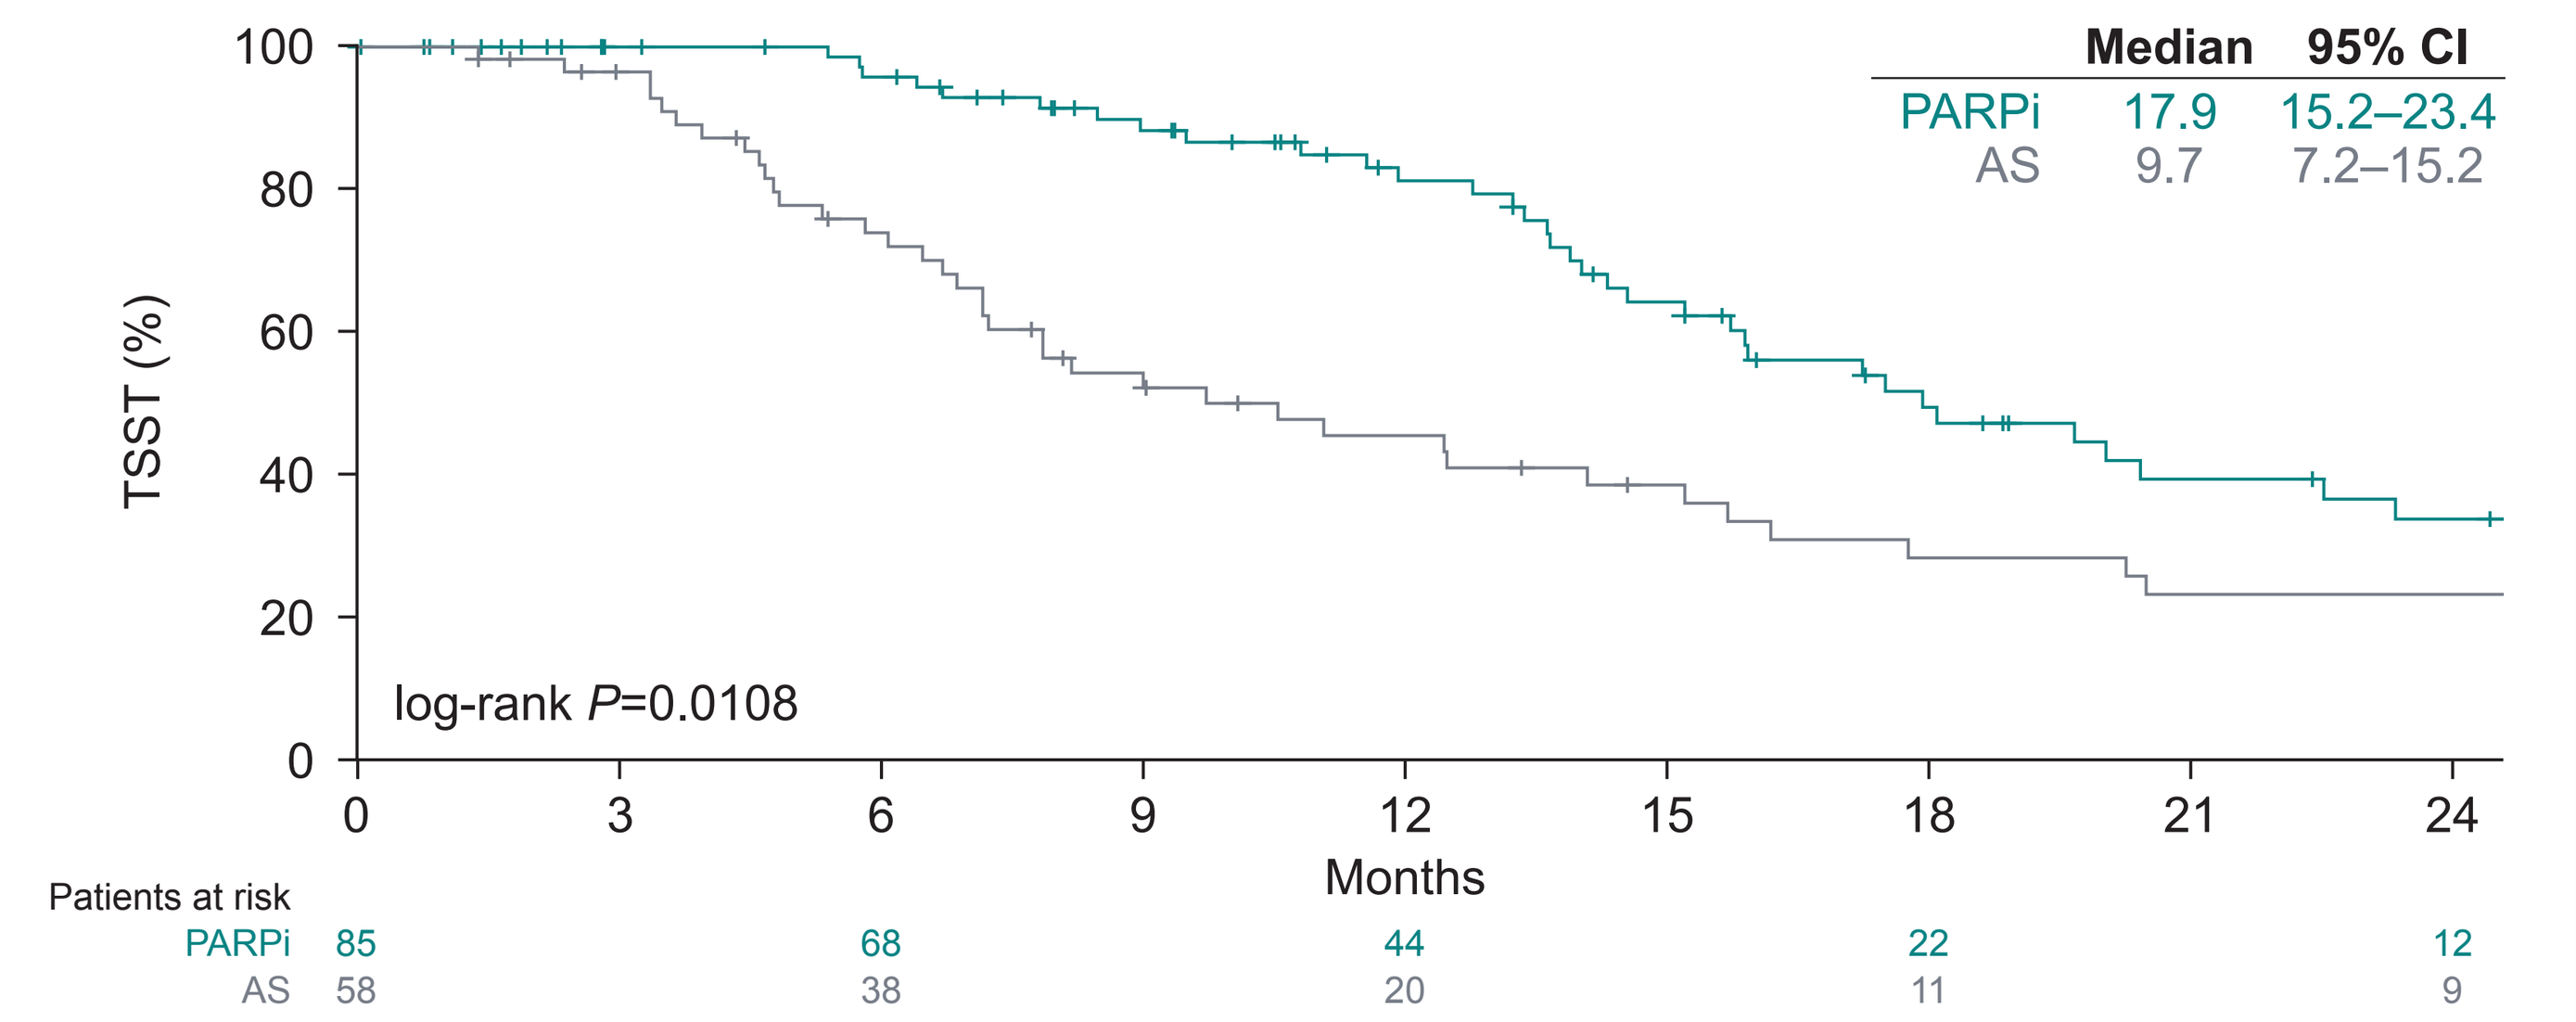
Median 95% CI
PARPi 17.9 15.2–23.4
AS 9.7 7.2–15.2
TSST (%)
log-rank P=0.0108
Months
Patients at risk
PARPi 85 68 44 22 12
AS 58 38 20 11 9

A log-rank test was used to compare TSST of the groups from initiation of 2L maintenance therapy to 24 months. 2L, second-line; AS, active surveillance; PARPi, poly(ADP-ribose) polymerase inhibitor; TSST, time to second subsequent treatment.

IGCS 2022 Annual Global Meeting | Sept 29 – Oct 1 | New York City, New York