Rucaparib + Sacituzumab Govitecan: Initial Data From the Phase 1b/2 SEASTAR Study (NCT03992131)

Poster number: 547P

Timothy A. Yap,[1] Erika Hamilton,[2] Todd Bauer,[2] Ecaterina E. Dumbrava,[1] Rinath Jeselsohn,[3] Aaron Enke,[4] Sabrina Hurley,[4] Kevin Lin,[4] Jenn Habeck,[4] Kenton Wride,[4] Heidi Giordano,[4] Geoffrey Shapiro[3]

[1]The University of Texas MD Anderson Cancer Center, Houston, USA; [2]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, USA; [3]Dana-Farber Cancer Institute, Boston, USA; [4]Clovis Oncology, Inc., Boulder, USA

SUMMARY

- **Initial encouraging signs of antitumour activity were seen with the combination of rucaparib + sacituzumab govitecan in patients with advanced solid tumours (n=6), including patients with prior poly(ADP-ribose) polymerase inhibitor exposure and without a deleterious homologous recombination repair gene mutation (Figure 2)**
- **Despite early toxicities, all 6 patients continued treatment, with treatment-emergent adverse events effectively managed with dose modification and/or growth factor support**
- **Further evaluation of the combination is warranted**

Poster Download
Also available at: https://clovisoncology.com/files/ESMO2020_Yap_Poster.pdf
Copies of this e-poster obtained through Quick Response (QR) codes and/or web links are for personal use only and may not be reproduced without written permission of the authors.
Corresponding author: Timothy A. Yap; TYap@mdanderson.org.

Supplementary Material
Also available at:
https://clovisoncology.com/files/ESMO2020_Yap_Poster_Supplement.pdf
Please scan this QR code or visit the website to view additional materials for this poster.

REFERENCES

1. Delaney et al. *Clin Cancer Res.* 2000;6:2860-7.
2. Murai et al. *Mol Cancer Ther.* 2014;13:433-43.
3. Tahara et al. *Mol Cancer Ther.* 2014;13:1170-80.
4. Kummar et al. *Cancer Res.* 2011;71:5626-34.
5. Cardillo et al. *Clin Cancer Res.* 2017;23:3405-15.
6. Bardia et al. *N Engl J Med.* 2019;380:741-51.

ACKNOWLEDGEMENTS

This study was funded by Clovis Oncology, Inc. Medical writing and editorial support funded by Clovis Oncology were provided by Nathan Yardley and Frederique H. Evans of Ashfield Healthcare Communications.

PRESENTING AUTHOR DISCLOSURE

Timothy Yap reports research support paid to institution only (no personal fees) from Clovis Oncology, AstraZeneca, Bayer, Constellation, Cyteir, Eli Lilly, EMD Serono, Forbius/Formation Biologics, F-Star, GlaxoSmithKline, Genentech, ImmuneSensor, Ipsen, Jounce, Karyopharm, Kyowa, Novartis, Pfizer, Ribon Therapeutics, Regeneron, Sanofi, Seattle Genetics, Tesaro, and Vertex Pharmaceuticals; has served in a consulting or advisory role for Clovis Oncology, Almac, Aduro, AstraZeneca, Atrin, Axiom, Bayer, Calithera, Cybrexa, EMD Serono, F-Star, Guidepoint, Ignyta, I-Mab, Janssen, Kyn Therapeutics, Merck, Pfizer, Roche, Seattle Genetics, and Zai Labs.

INTRODUCTION

- Preclinically, poly(ADP-ribose) polymerase (PARP) inhibitors such as rucaparib and DNA topoisomerase I (TOP1) inhibitors such as irinotecan have shown highly synergistic antitumour effects, but clinical development has been hampered by overlapping toxicities[1-5]
 - Tumour-targeted delivery of a TOP1 inhibitor may reduce systemic toxicity of the combination
 - Sacituzumab govitecan is an antibody-drug conjugate composed of an anti-Trop-2 antibody coupled to the active metabolite of irinotecan (SN-38) via a proprietary hydrolysable linker[6]
- Here, we describe the safety, tolerability, and preliminary efficacy of rucaparib + sacituzumab govitecan from the SEASTAR study, an open-label, parallel-arm study of the PARP inhibitor rucaparib in combination with other anticancer agents

METHODS

- For the phase 1b portion, eligible patients had metastatic triple-negative breast cancer, urothelial cancer, or platinum-resistant ovarian cancer; patients with another solid tumour type with a deleterious mutation in a homologous recombination repair (HRR) gene (*BRCA1, BRCA2, PALB2, RAD51C*, or *RAD51D*) were also eligible
- Prior treatment with a PARP inhibitor was allowed; patients previously treated with irinotecan, topotecan, or any derivative in any formulation were excluded
- A 3+3 design was used, with a starting dose of rucaparib 300 mg orally twice daily + sacituzumab govitecan 6 mg/kg intravenously on days 1 and 8 of a 21-day cycle in cohort 1
- Dose-limiting toxicities (DLTs) were assessed during cycle 1 (see Supplementary Material)
- Haematopoietic growth factor support was allowed except during the DLT period
- The primary endpoints of phase 1b were the incidence of grade 3 or 4 treatment-emergent adverse events (TEAEs) and number of participants who experienced a DLT
 - A key secondary endpoint was investigator-assessed objective response per Response Evaluation Criteria In Solid Tumors, version 1.1 (RECIST)

RESULTS

- Six patients were enrolled in 2 dose cohorts; demographics, disease history, and on-study findings for each patient are shown in **Figure 1**
- Two of 3 patients in cohort 1 experienced dose-limiting grade 4 neutropenia during cycle 1
- In cohort 2, the rucaparib starting dose was reduced from 300 mg twice daily to 300 mg once daily; grade 3/4 neutropenia led to the delay of cycle 2 by 1–2 weeks in all 3 patients
- All 6 patients were able to continue treatment for at least 12 weeks, with TEAEs managed through treatment interruption, dose reduction, and/or growth factor support
 - One patient remains on treatment as of the data cutoff date (11 Aug 2020)
- Summaries of safety and efficacy data are shown in the **Table** and **Figure 2**, respectively
- All patients had a best response of stable disease or better
- Three patients had a confirmed partial response; all 3 patients had been previously treated with a PARP inhibitor (2 niraparib, 1 veliparib) until disease progression
 - Two responders did not have a known deleterious HRR gene mutation: 1 patient had metastatic, triple-negative breast cancer (**Figure 3; Supplementary Material**) and 1 patient had metastatic, high-grade serous ovarian cancer

Figure 1. SEASTAR Patients Receiving Rucaparib + Sacituzumab Govitecan

COHORT 1 - starting dose: 300 mg BID rucaparib + 6 mg/kg IV sacituzumab govitecan

PATIENT 1

Demographics and disease history
- Asian, female, age 56 years
- Metastatic, granulosa cell ovarian cancer
- No HRR gene mutation detected
- Five prior anticancer regimens

Study profile

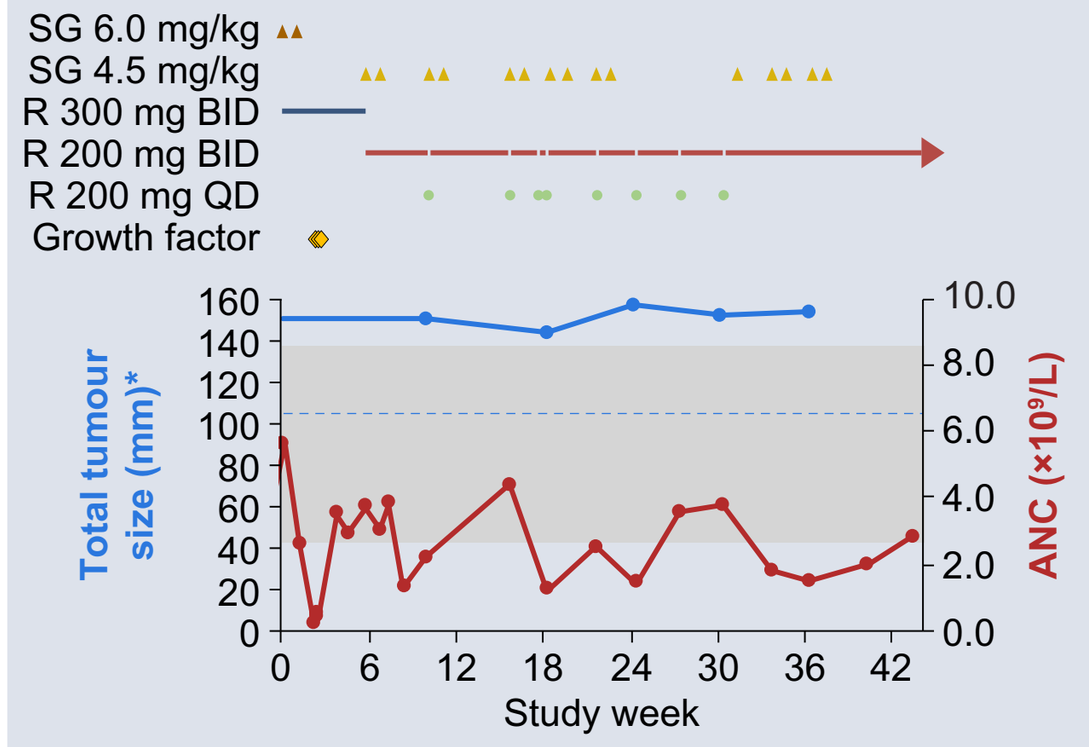
SG 6.0 mg/kg
SG 4.5 mg/kg
R 300 mg BID
R 200 mg BID
R 200 mg QD
Growth factor
Total tumour size (mm)*
ANC (×10⁹/L)
Study week

Investigator-assessed tumour response
- SD (duration: 36.3+ weeks)

Grade ≥2 TEAEs
- Grade 2: abdominal cramping, diarrhoea, fatigue, hair loss, intermittent hyponatraemia
- Grade 3: *Clostridium difficile* infection
- Grade 4: neutropenia (DLT)

PATIENT 2

Demographics and disease history
- White, female, age 60 years
- Metastatic, high-grade endometrial cancer
- *BRCA1* gene mutation
- Three prior anticancer regimens
 - One prior PARP inhibitor (1st-line maintenance with single-agent niraparib)

Study profile

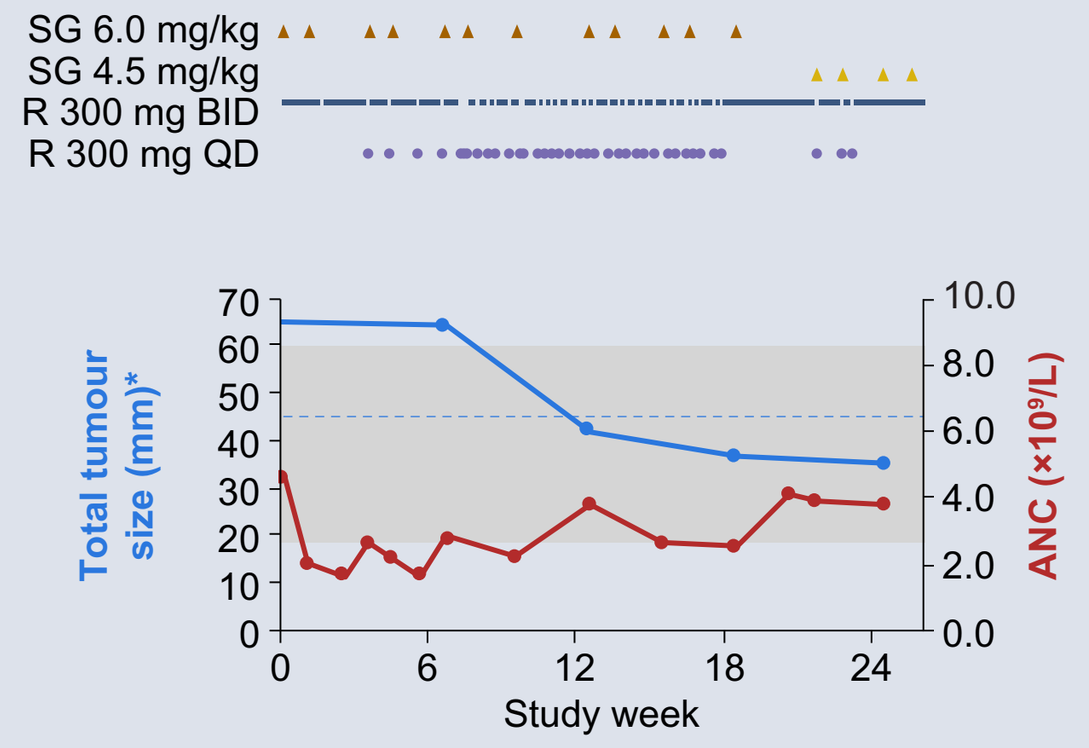
SG 6.0 mg/kg
SG 4.5 mg/kg
R 300 mg BID
R 300 mg QD
Total tumour size (mm)*
ANC (×10⁹/L)
Study week

Investigator-assessed tumour response
- Confirmed PR (duration: 12.0 weeks)

Grade ≥2 TEAEs
- Grade 2: diarrhoea, eye infection

PATIENT 3

Demographics and disease history
- White, male, age 63 years
- Metastatic, transitional cell urothelial carcinoma
- *BRCA2* gene mutation
- Three prior anticancer regimens

Study profile

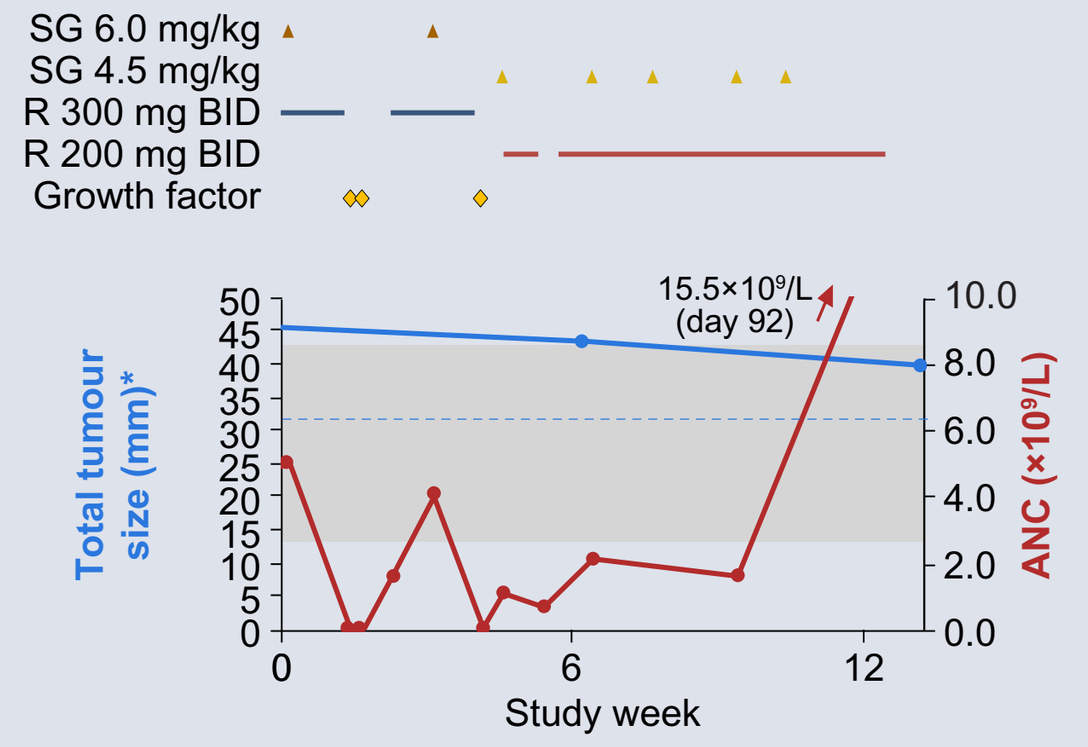
SG 6.0 mg/kg
SG 4.5 mg/kg
R 300 mg BID
R 200 mg BID
Growth factor
15.5×10⁹/L (day 92)
Total tumour size (mm)*
ANC (×10⁹/L)
Study week

Investigator-assessed tumour response
- SD (duration: 13.3 weeks)

Grade ≥2 TEAEs
- Grade 2: flu-like symptoms, generalised muscle weakness, hypophosphataemia, low bicarbonate
- Grade 3: anaemia, cholecystitis
- Grade 4: leukopenia, neutropenia (DLT)

COHORT 2 - starting dose: 300 mg QD rucaparib + 6 mg/kg IV sacituzumab govitecan

PATIENT 4

Demographics and disease history
- White, female, age 69 years
- Metastatic, high-grade serous ovarian cancer
- No HRR gene mutation detected
- Three prior anticancer regimens
 - One prior PARP inhibitor (2nd-line treatment with single-agent niraparib)

Study profile

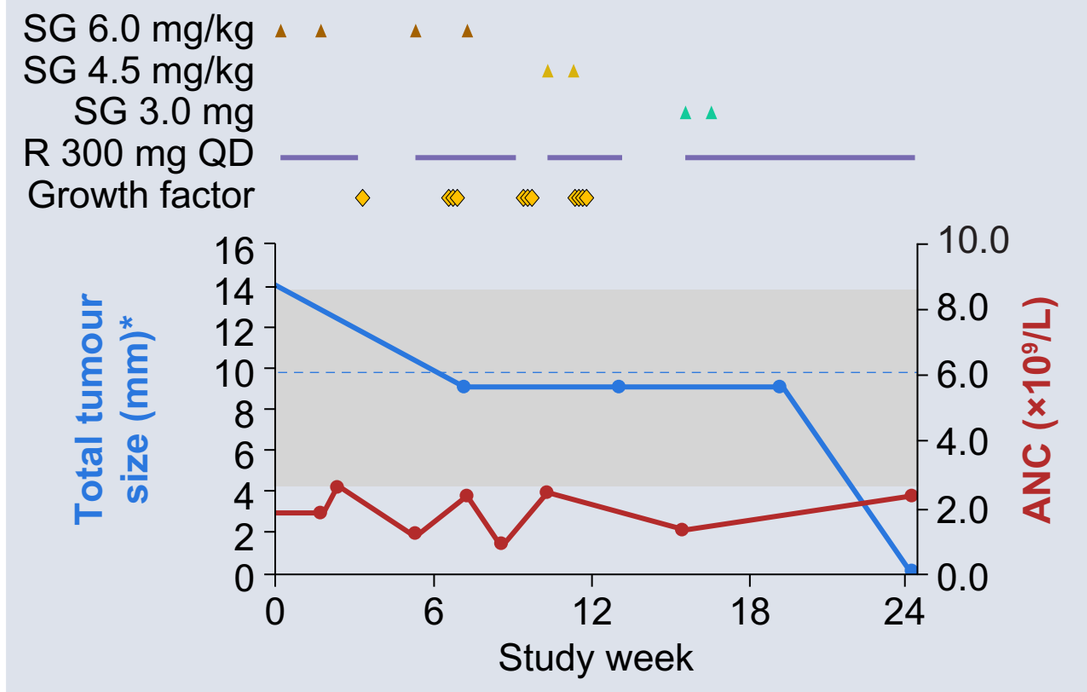
SG 6.0 mg/kg
SG 4.5 mg/kg
SG 3.0 mg
R 300 mg QD
Growth factor
Total tumour size (mm)*
ANC (×10⁹/L)
Study week

Investigator-assessed tumour response
- Confirmed PR (duration: 17.1 weeks)

Grade ≥2 TEAEs
- Grade 2: constipation, depression, diarrhoea, dyspnoea, fatigue, GERD, mucositis, neuropathy, sinusitis, swelling in left axilla
- Grade 3: leukopenia, thrombocytopenia
- Grade 4: neutropenia

PATIENT 5

Demographics and disease history
- White, female, age 57 years
- Metastatic, triple-negative breast cancer (invasive ductal carcinoma)
- No HRR gene mutation detected
- Eight prior anticancer regimens
 - One prior PARP inhibitor (8th-line treatment with veliparib + dinaciclib)

Study profile

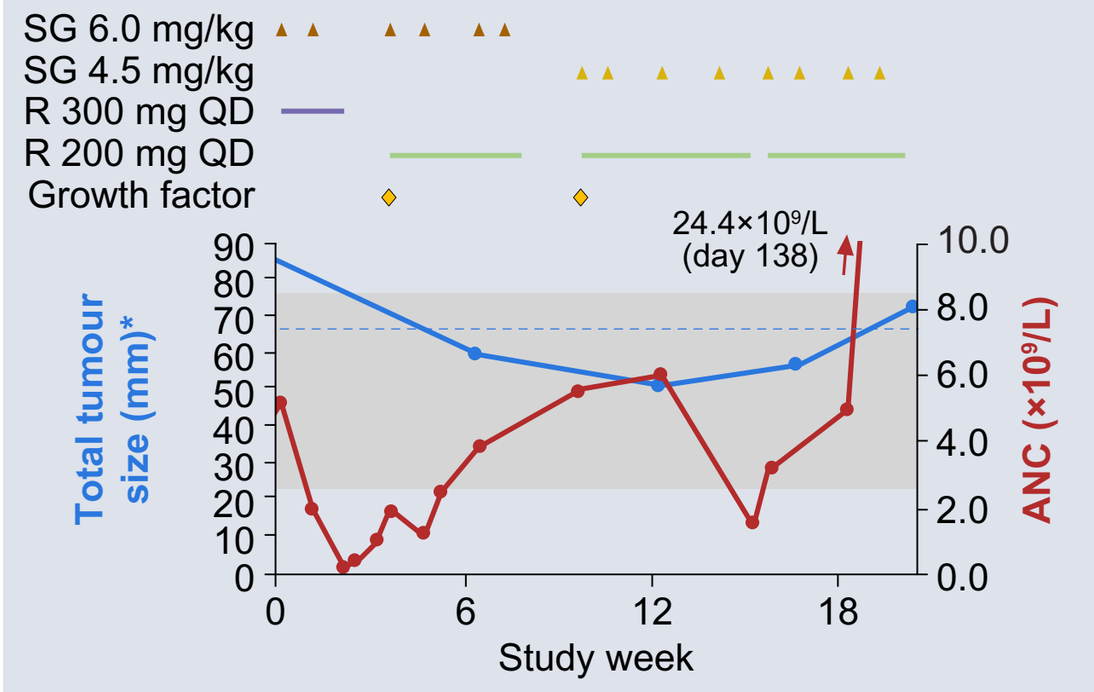
SG 6.0 mg/kg
SG 4.5 mg/kg
R 300 mg QD
R 200 mg QD
Growth factor
24.4×10⁹/L (day 138)
Total tumour size (mm)*
ANC (×10⁹/L)
Study week

Investigator-assessed tumour response
- Confirmed PR (duration: 14.0 weeks)

Grade ≥2 TEAEs
- Grade 2: anaemia, anxiety, back pain, bilirubin increased, diarrhoea, fatigue, mucositis oral, nausea, thrombocytopenia
- Grade 3: dyspnoea, febrile neutropenia
- Grade 4: neutropenia, leukopenia

PATIENT 6

Demographics and disease history
- White, female, age 50 years
- Metastatic, triple-negative breast cancer (invasive ductal carcinoma)
- Not tested for HRR gene mutation
- Six prior anticancer regimens

Study profile

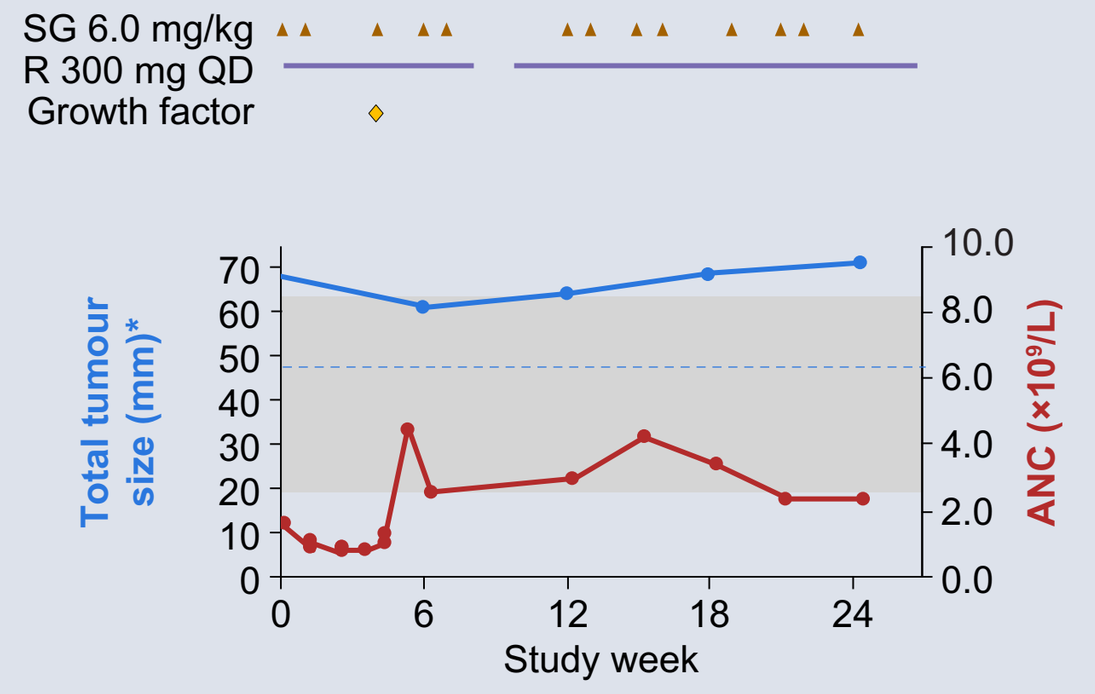
SG 6.0 mg/kg
R 300 mg QD
Growth factor
Total tumour size (mm)*
ANC (×10⁹/L)
Study week

Investigator-assessed tumour response
- SD (duration: 24.3 weeks)

Grade ≥2 TEAEs
- Grade 3: neutropenia

Sacituzumab govitecan: ▲ 6 mg/kg; ▲ 4.5 mg/kg; ▲ 3 mg/kg
Rucaparib: — 300 mg BID; — 200 mg BID; — 300 mg QD; — 200 mg QD
◆ Growth factor support; ► Ongoing on study
Total tumour size; Threshold for 30% decrease in total tumour size; Absolute neutrophil count; Normal range for neutrophil count (2.6 to 8.5×10⁹/L)

Data cutoff date: 11 Aug 2020.
*Evaluated per RECIST.
ANC, absolute neutrophil count; BID, twice daily; DLT, dose-limiting toxicity; GERD, gastro-oesophageal reflux disease; HRR, homologous recombination repair; IV, intravenous; LDTL, longest diameter of target lesions; PARP, poly(ADP-ribose) polymerase; PR, partial response; QD, once daily; R, rucaparib; RECIST, Response Evaluation Criteria In Solid Tumors, version 1.1; SD, stable disease; SG, sacituzumab govitecan; TEAE, treatment-emergent adverse event.

Table. Most Common TEAEs (Reported in ≥3 Patients)

	Any grade n (%)	Grade ≥3 n (%)
Any TEAE	6 (100)	5 (83.3)
Neutropenia/ANC decreased	6 (100)	5 (83.3)
Diarrhoea	5 (83.3)	0
ALT/AST increased	4 (66.7)	0
Asthenia/fatigue	4 (66.7)	0
Dyspnoea	3 (50.0)	1 (16.7)
Hyponatraemia	3 (50.0)	0
Nausea	3 (50.0)	0
Thrombocytopenia/platelet count decreased	3 (50.0)	1 (16.7)
Vomiting	3 (50.0)	0

Data cutoff date: 11 Aug 2020.
ALT, alanine aminotransferase; ANC, absolute neutrophil count; AST, aspartate aminotransferase; TEAE, treatment-emergent adverse event.

Figure 2. Duration of Treatment

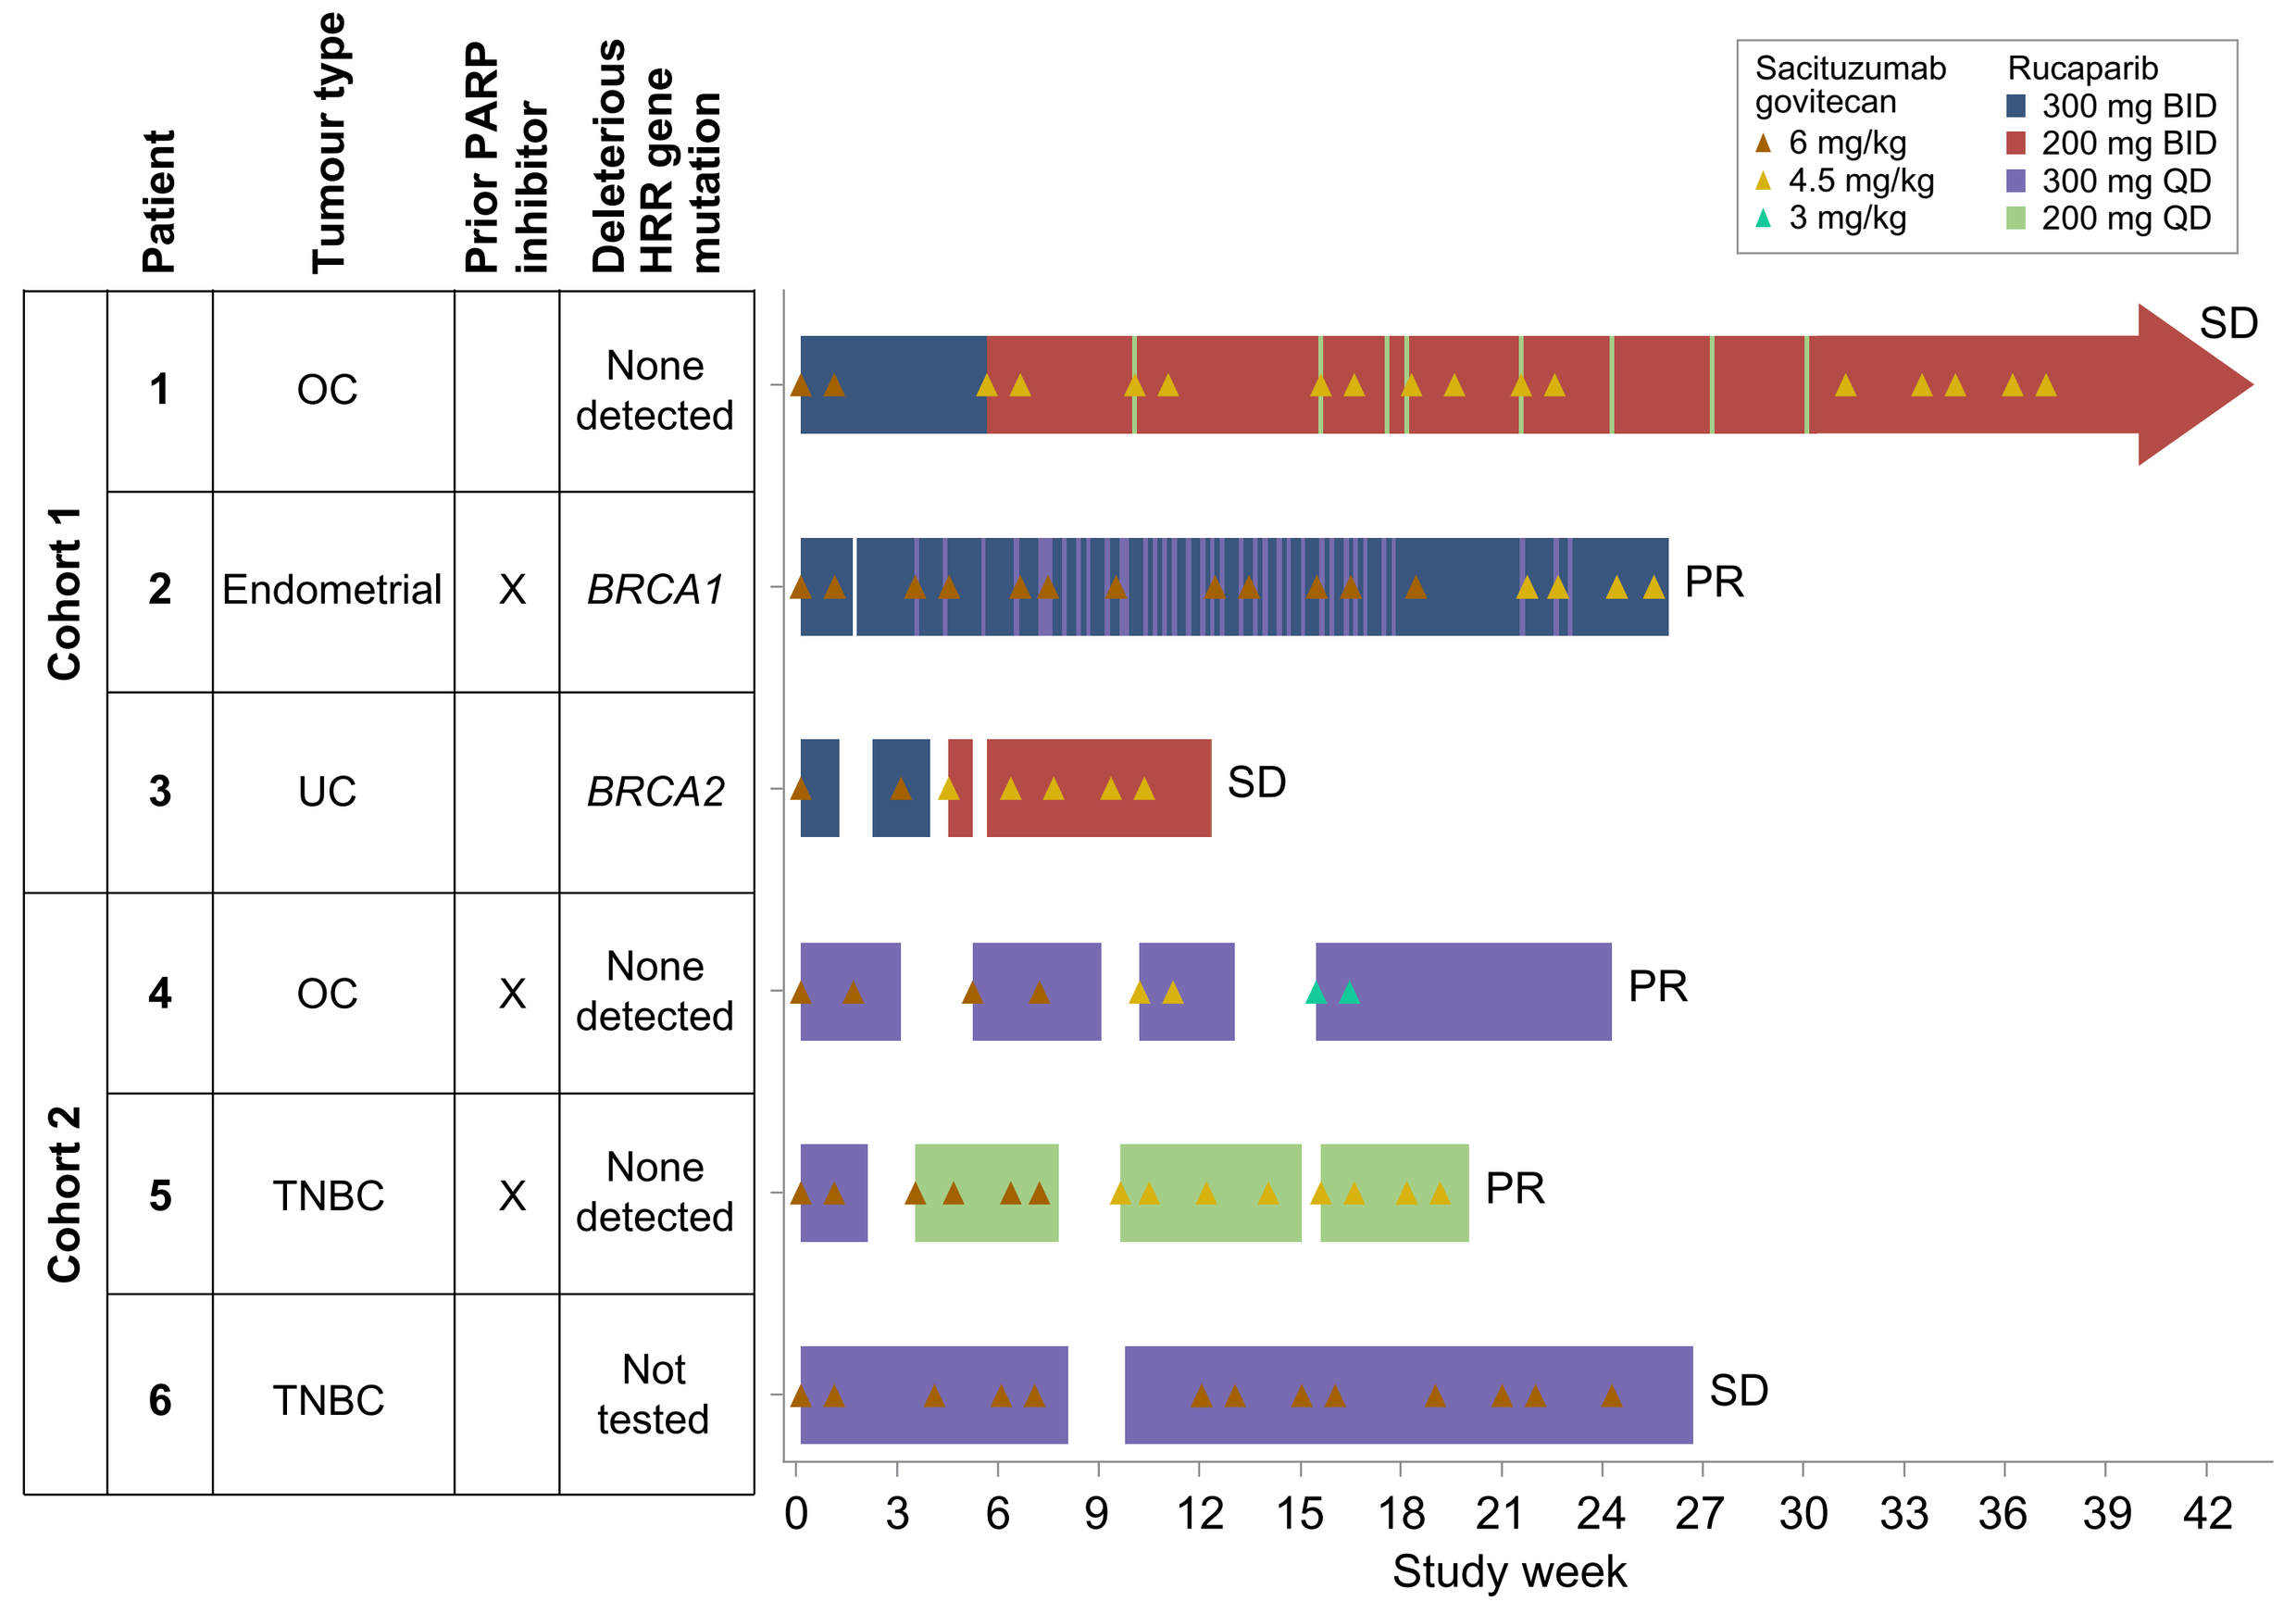
Patient
Tumour type
Prior PARP inhibitor
Deleterious HRR gene mutation
Sacituzumab govitecan
6 mg/kg
4.5 mg/kg
3 mg/kg
Rucaparib
300 mg BID
200 mg BID
300 mg QD
200 mg QD
Cohort 1
1 OC None detected SD
2 Endometrial X BRCA1 PR
3 UC BRCA2 SD
Cohort 2
4 OC X None detected PR
5 TNBC X None detected PR
6 TNBC Not tested SD
Study week

Data cutoff date: 11 Aug 2020.
BID, twice daily; HRR, homologous recombination repair; OC, ovarian cancer; PARP, poly(ADP-ribose) polymerase; PR, partial response; QD, once daily; RECIST, Response Evaluation Criteria In Solid Tumors, version 1.1; SD, stable disease; TNBC, triple-negative breast cancer; UC, urothelial cancer.

Figure 3. Representative Images of Confirmed Partial Response Observed in Patient with Metastatic TNBC (Patient 5)

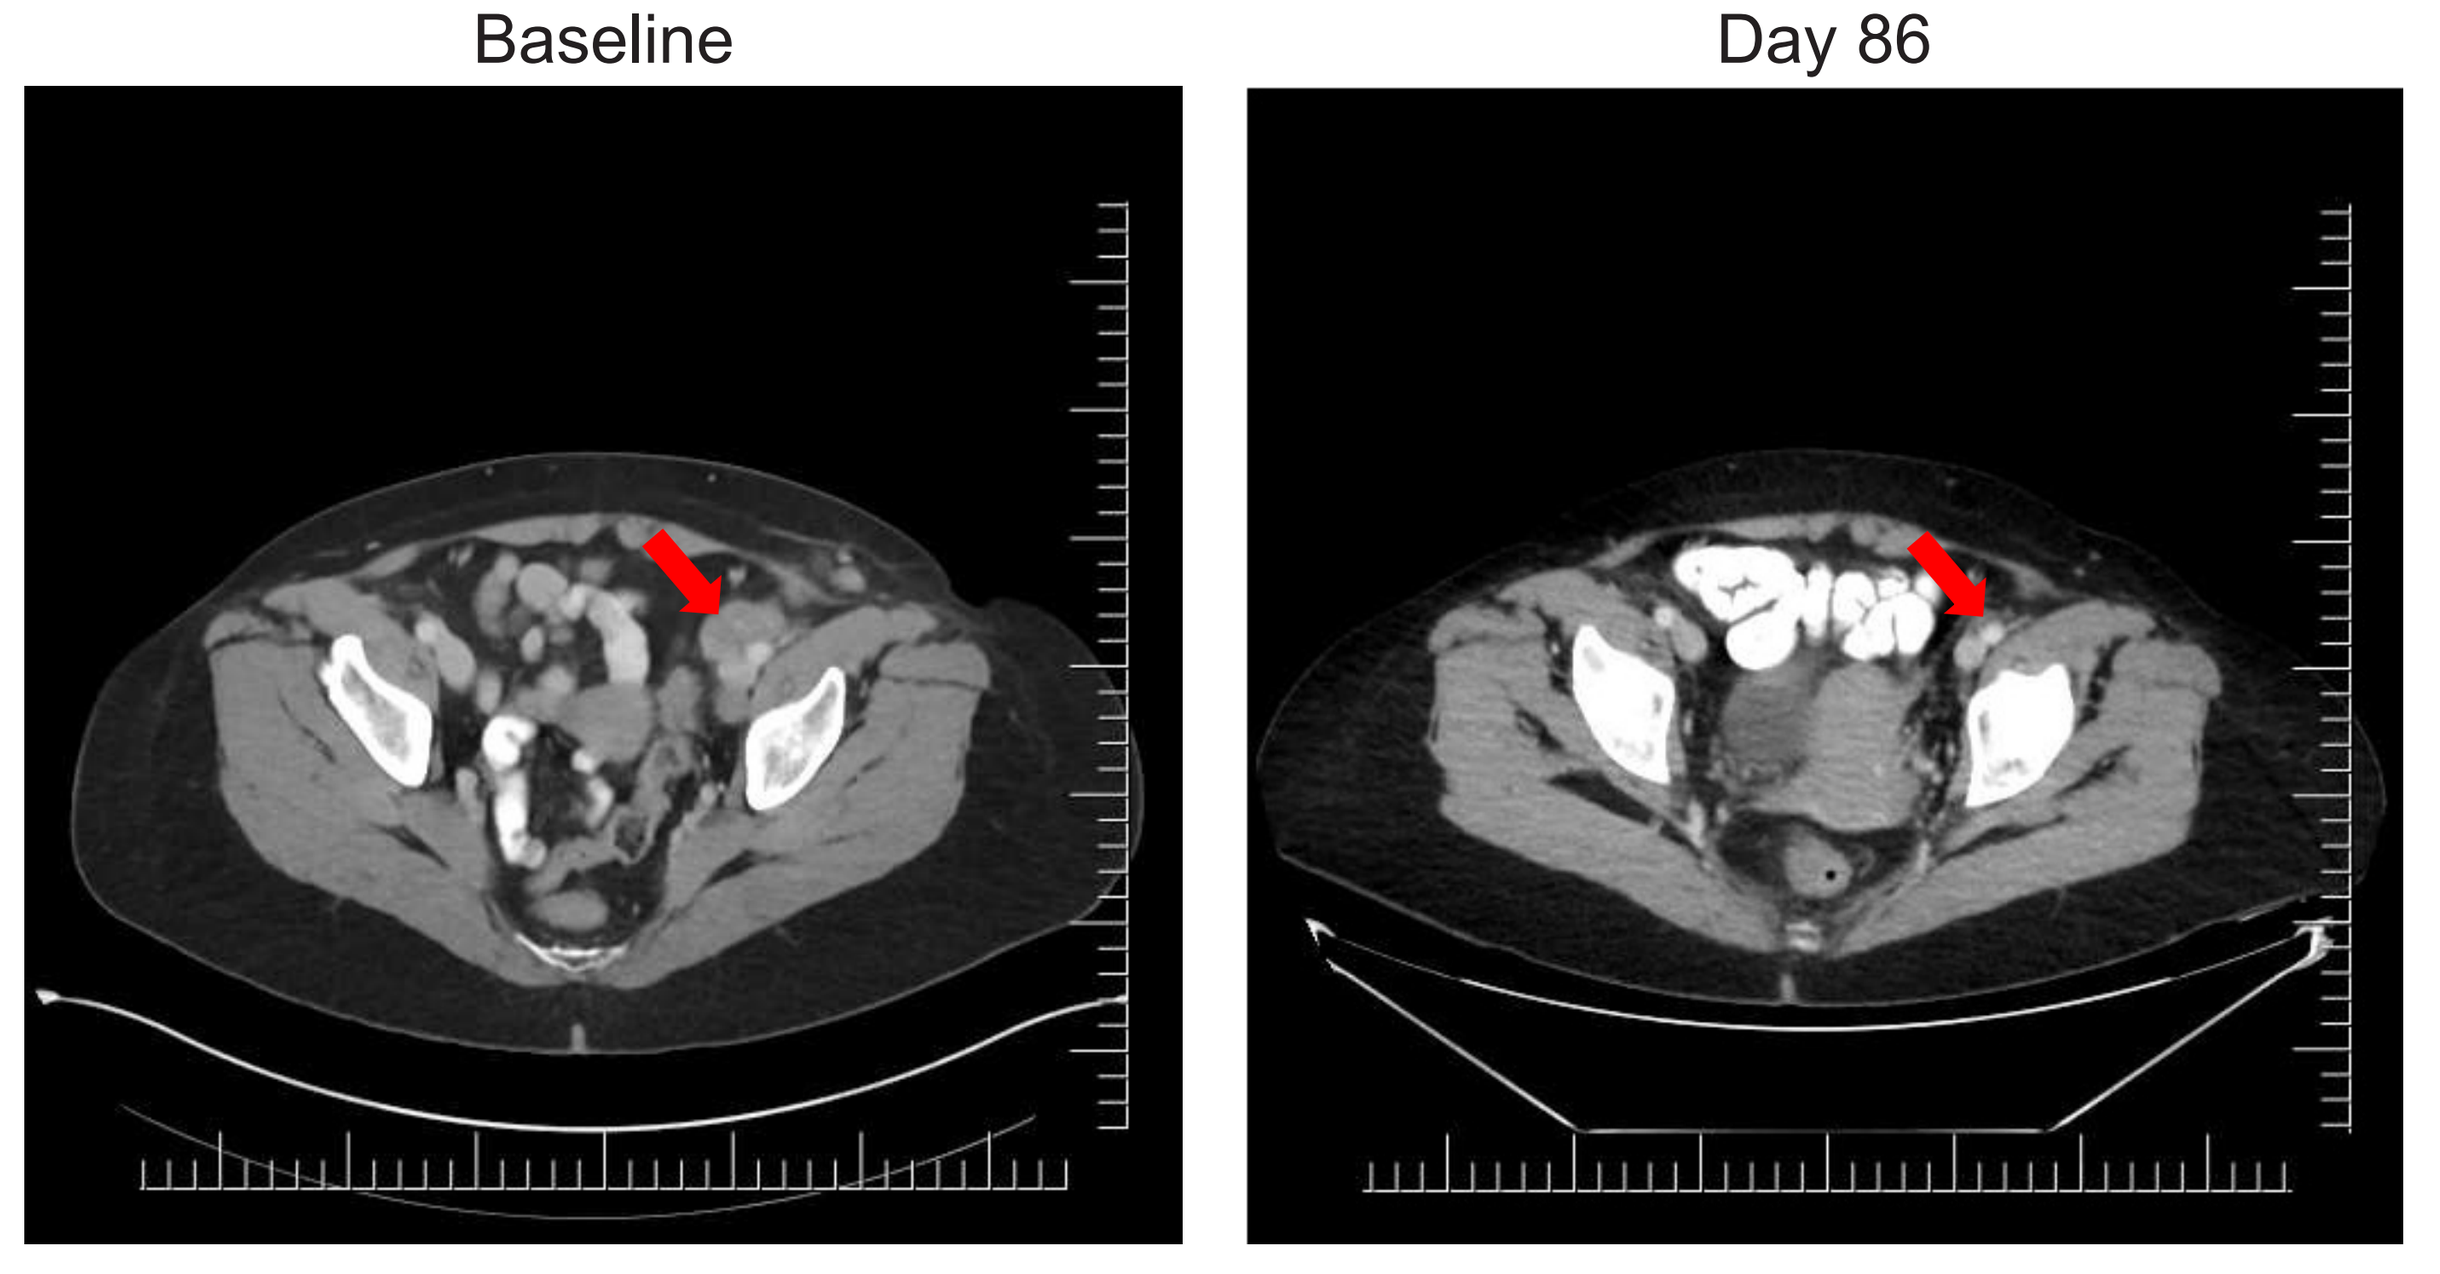
Baseline
Day 86

TNBC, triple-negative breast cancer.

ESMO Congress | 19–21 September 2020 | Virtual Meeting