Timing of Adverse Events During Maintenance Treatment With Rucaparib for Recurrent Ovarian Cancer in the Phase 3 ARIEL3 Study

Poster number: 821P

Andrew Dean,[1] Amit M. Oza,[2] Domenica Lorusso,[3] Carol Aghajanian,[4] Ana Oaknin,[5] Nicoletta Colombo,[6] Johanne I. Weberpals,[7] Andrew R. Clamp,[8] Giovanni Scambia,[3] Alexandra Leary,[9] Robert W. Holloway,[10] Margarita Amenedo Gancedo,[11] Peter C. Fong,[12] Jeffrey C. Goh,[13] David M. O'Malley,[14] Terri Cameron,[15] Lara Maloney,[16] Sandra Goble,[16] Robert L. Coleman,[17] Jonathan A. Ledermann[18]

[1]St John of God Subiaco Hospital, Australia; [2]Princess Margaret Cancer Centre, University Health Network, Toronto, Canada; [3]Fondazione Policlinico Universitario A. Gemelli IRCCS, Rome, Italy; [4]Memorial Sloan Kettering Cancer Center, New York, USA; [5]Vall d'Hebron University Hospital, Vall d'Hebron Institute of Oncology (VHIO), Barcelona, Spain; [6]European Institute of Oncology IRCCS and University of Milan-Bicocca, Italy; [7]Ottawa Hospital Research Institute, Canada; [8]The Christie NHS Foundation Trust and University of Manchester, UK; [9]Gustave Roussy Cancer Center, INSERM U981, and Groupe d'Investigateurs Nationaux pour l'Etude des Cancers Ovariens (GINECO), Villejuif, France; [10]Florida Hospital Cancer Institute, Orlando, USA; [11]Oncology Center of Galicia, La Coruña, Spain; [12]Auckland City Hospital and University of Auckland, New Zealand; [13]Cancer Care Services, Royal Brisbane and Women's Hospital, Herston, Australia, and University of Queensland, St Lucia, Australia; [14]The Ohio State University, James Cancer Center, Columbus, USA; [15]Clovis Oncology UK Ltd., Cambridge, UK; [16]Clovis Oncology, Inc., Boulder, USA; [17]US Oncology Research, The Woodlands, USA; [18]UCL Cancer Institute, University College London and UCL Hospitals, UK

HIGHLIGHTS

- **The overall safety profile of rucaparib maintenance treatment in patients with recurrent ovarian cancer from ARIEL3 remains consistent with previous reports,[1,2] with no new safety signals identified**
- **Prevalence of any-grade nausea declined progressively over the 24-month evaluation period**
- **Prevalence of any-grade anaemia/decreased haemoglobin peaked at month 4, decreasing to a plateau after month 8**
- **The first onset of frequently reported TEAEs generally occurred early in treatment (≤45 days). The median duration of the first event of frequently reported TEAEs was generally <60 days (Figure 3)**

Poster Download
Also available at: https://clovisoncology.com/files/ESMO2020_Dean_Poster.pdf
Copies of this e-poster obtained through Quick Response (QR) codes and/or web links are for personal use only and may not be reproduced without written permission of the authors.
Corresponding author: Andrew Dean; adean747@mac.com.

REFERENCES

1. Coleman et al. *Lancet.* 2017;390:1949-61.
2. Ledermann et al. *Lancet Oncol.* 2020;21:710-22.

ACKNOWLEDGEMENTS

This research was sponsored by Clovis Oncology, Inc. Medical writing support was provided by Andrew Croskery (Clovis Oncology, Ltd., London, UK) and editorial support funded by Clovis Oncology was provided by Frederique H. Evans of Ashfield Healthcare Communications.

PRESENTING AUTHOR DISCLOSURE

Andrew Dean has served in a consulting or advisory role for Precision Oncology Australia, Shire Pharmaceuticals, and Specialised Therapeutics Australia.

INTRODUCTION

- In the randomised, placebo-controlled, double-blind, phase 3 study, ARIEL3 (NCT01968213), the poly(ADP-ribose) polymerase inhibitor rucaparib significantly improved progression-free survival vs placebo as maintenance treatment for patients with platinum-sensitive, recurrent ovarian cancer who were in complete or partial response to platinum-based chemotherapy[1]
- Here we present an analysis of treatment-emergent adverse events (TEAEs) with 2 years of additional follow-up for patients continuing treatment in ARIEL3 compared with the previous safety report[2]
 - We also include new analyses of the prevalence of key TEAEs over time as well as an analysis of the median time to and median duration of first occurrence of TEAEs

METHODS

- Safety data were summarised for all patients who were randomised into ARIEL3 and who received at least one dose of study treatment
 - After treatment discontinuation, patients were followed for TEAEs for up to 28 days
- TEAEs were classified per MedDRA version 19.1 and graded by investigator assessment as per the National Cancer Institute Common Terminology Criteria for Adverse Events, version 4.03
- Additional safety analyses include:
 - Prevalence of nausea and anaemia/decreased haemoglobin over time
 - Median time to onset of first TEAE
 - Median duration of first TEAE, defined as the duration of first TEAE, regardless of grade; TEAEs overlapping by ≤2 days were considered as the same continuous event; TEAEs occurring after the first event were not included in the duration calculation
 - AE duration was calculated using Kaplan-Meier methodology, in which ongoing events without a known end date were censored at the date of the last dose plus 28 days
- The visit cutoff date for these safety analyses was 31 December 2019

RESULTS

Safety Overview

- The current analysis of TEAEs in ARIEL3 remains consistent with previous reports,[1,2] with no new safety signals identified
 - As of the previous safety data cutoff (31 Dec 2017),[2] 60/372 patients in the safety population were still receiving rucaparib and 5/189 patients were receiving placebo
 - As of the current analysis (31 Dec 2019), 33/372 and 1/189 patients were still receiving rucaparib or placebo, respectively
 - Median treatment duration was 8.3 months in the rucaparib arm and 5.5 months in the placebo arm at both the previous and current safety data cutoff dates
- All patients in the rucaparib arm and 182/189 (96.3%) patients in the placebo arm experienced ≥1 TEAE
 - Grade ≥3 TEAEs were reported by 231/372 (62.1%) patients in the rucaparib arm and 31/189 (16.4%) patients in the placebo arm (**Table 1**)
 - Grade 4 TEAEs were reported by 28/372 (7.5%) and 2/189 patients (1.1%) in the rucaparib and placebo arms, respectively. The most frequent grade 4 TEAEs in the rucaparib arm were neutropenia/decreased neutrophils and thrombocytopenia/decreased platelets (7/372 patients [1.9%] each)

Table 1. Summary of TEAEs

	Rucaparib (n=372)[a]	Placebo (n=189)
Treatment duration, median (range), mo	8.3 (0–67)	5.5 (0–68)
Patients with at least one TEAE	372 (100)	182 (96.3)
Patients with at least one grade ≥3 TEAE	231 (62.1)	31 (16.4)
Treatment interruption and/or dose reduction due to TEAE	271 (72.8)	20 (10.6)
Treatment interruption due to TEAE	248 (66.7)	19 (10.1)
Dose reduction due to TEAE	209 (56.2)	8 (4.2)
Discontinued due to TEAE[b]	64 (17.2)	3 (1.6)
Deaths due to TEAE[b]	6 (1.6)[c]	1 (0.5)[d]
Deaths due to disease progression	2 (0.5)[e]	1 (0.5)[f]

Data are n (%), except where indicated.
[a]Three patients randomised to the rucaparib arm did not receive a dose of rucaparib and were excluded from the safety population; [b]Excluding disease progression; [c]Myelodysplastic syndrome (n=2); acute myeloid leukaemia (n=1); cardiac arrest (n=1); haematophagic histiocytosis (n=1); unclassifiable high grade B-cell lymphoma (n=1); [d]Pulmonary embolism (n=1); [e]Malignant neoplasm progression (n=2); [f]Metastases to meninges (n=1).
TEAE, treatment-emergent adverse event.

- In the rucaparib arm, the most frequently reported TEAEs of any grade were nausea (284/372 [76.3%]) and asthenia/fatigue (267/372 [71.8%]); the most frequently reported grade ≥3 TEAEs were anaemia/decreased haemoglobin (83/372 [22.3%]) and alanine aminotransferase (ALT)/aspartate aminotransferase (AST) increase (39/372 [10.5%]) (**Figure 1**)
- In the rucaparib arm, treatment-emergent myelodysplastic syndrome (MDS) occurred in 4 (1.1%) patients and treatment-emergent acute myeloid leukaemia (AML) occurred in 1 (0.3%) patient, representing 2 additional cases of treatment-emergent MDS than reported previously[2]
 - Of the patients who experienced MDS or AML, 3 had a germline BRCA-mutant carcinoma, 1 had a somatic BRCA-mutant carcinoma and 1 had a BRCA wild-type/low loss of heterozygosity carcinoma
- No patients in the placebo arm reported treatment-emergent MDS or AML

Figure 1. Most Frequent (≥20%) TEAEs of Any Grade in Either Treatment Arm

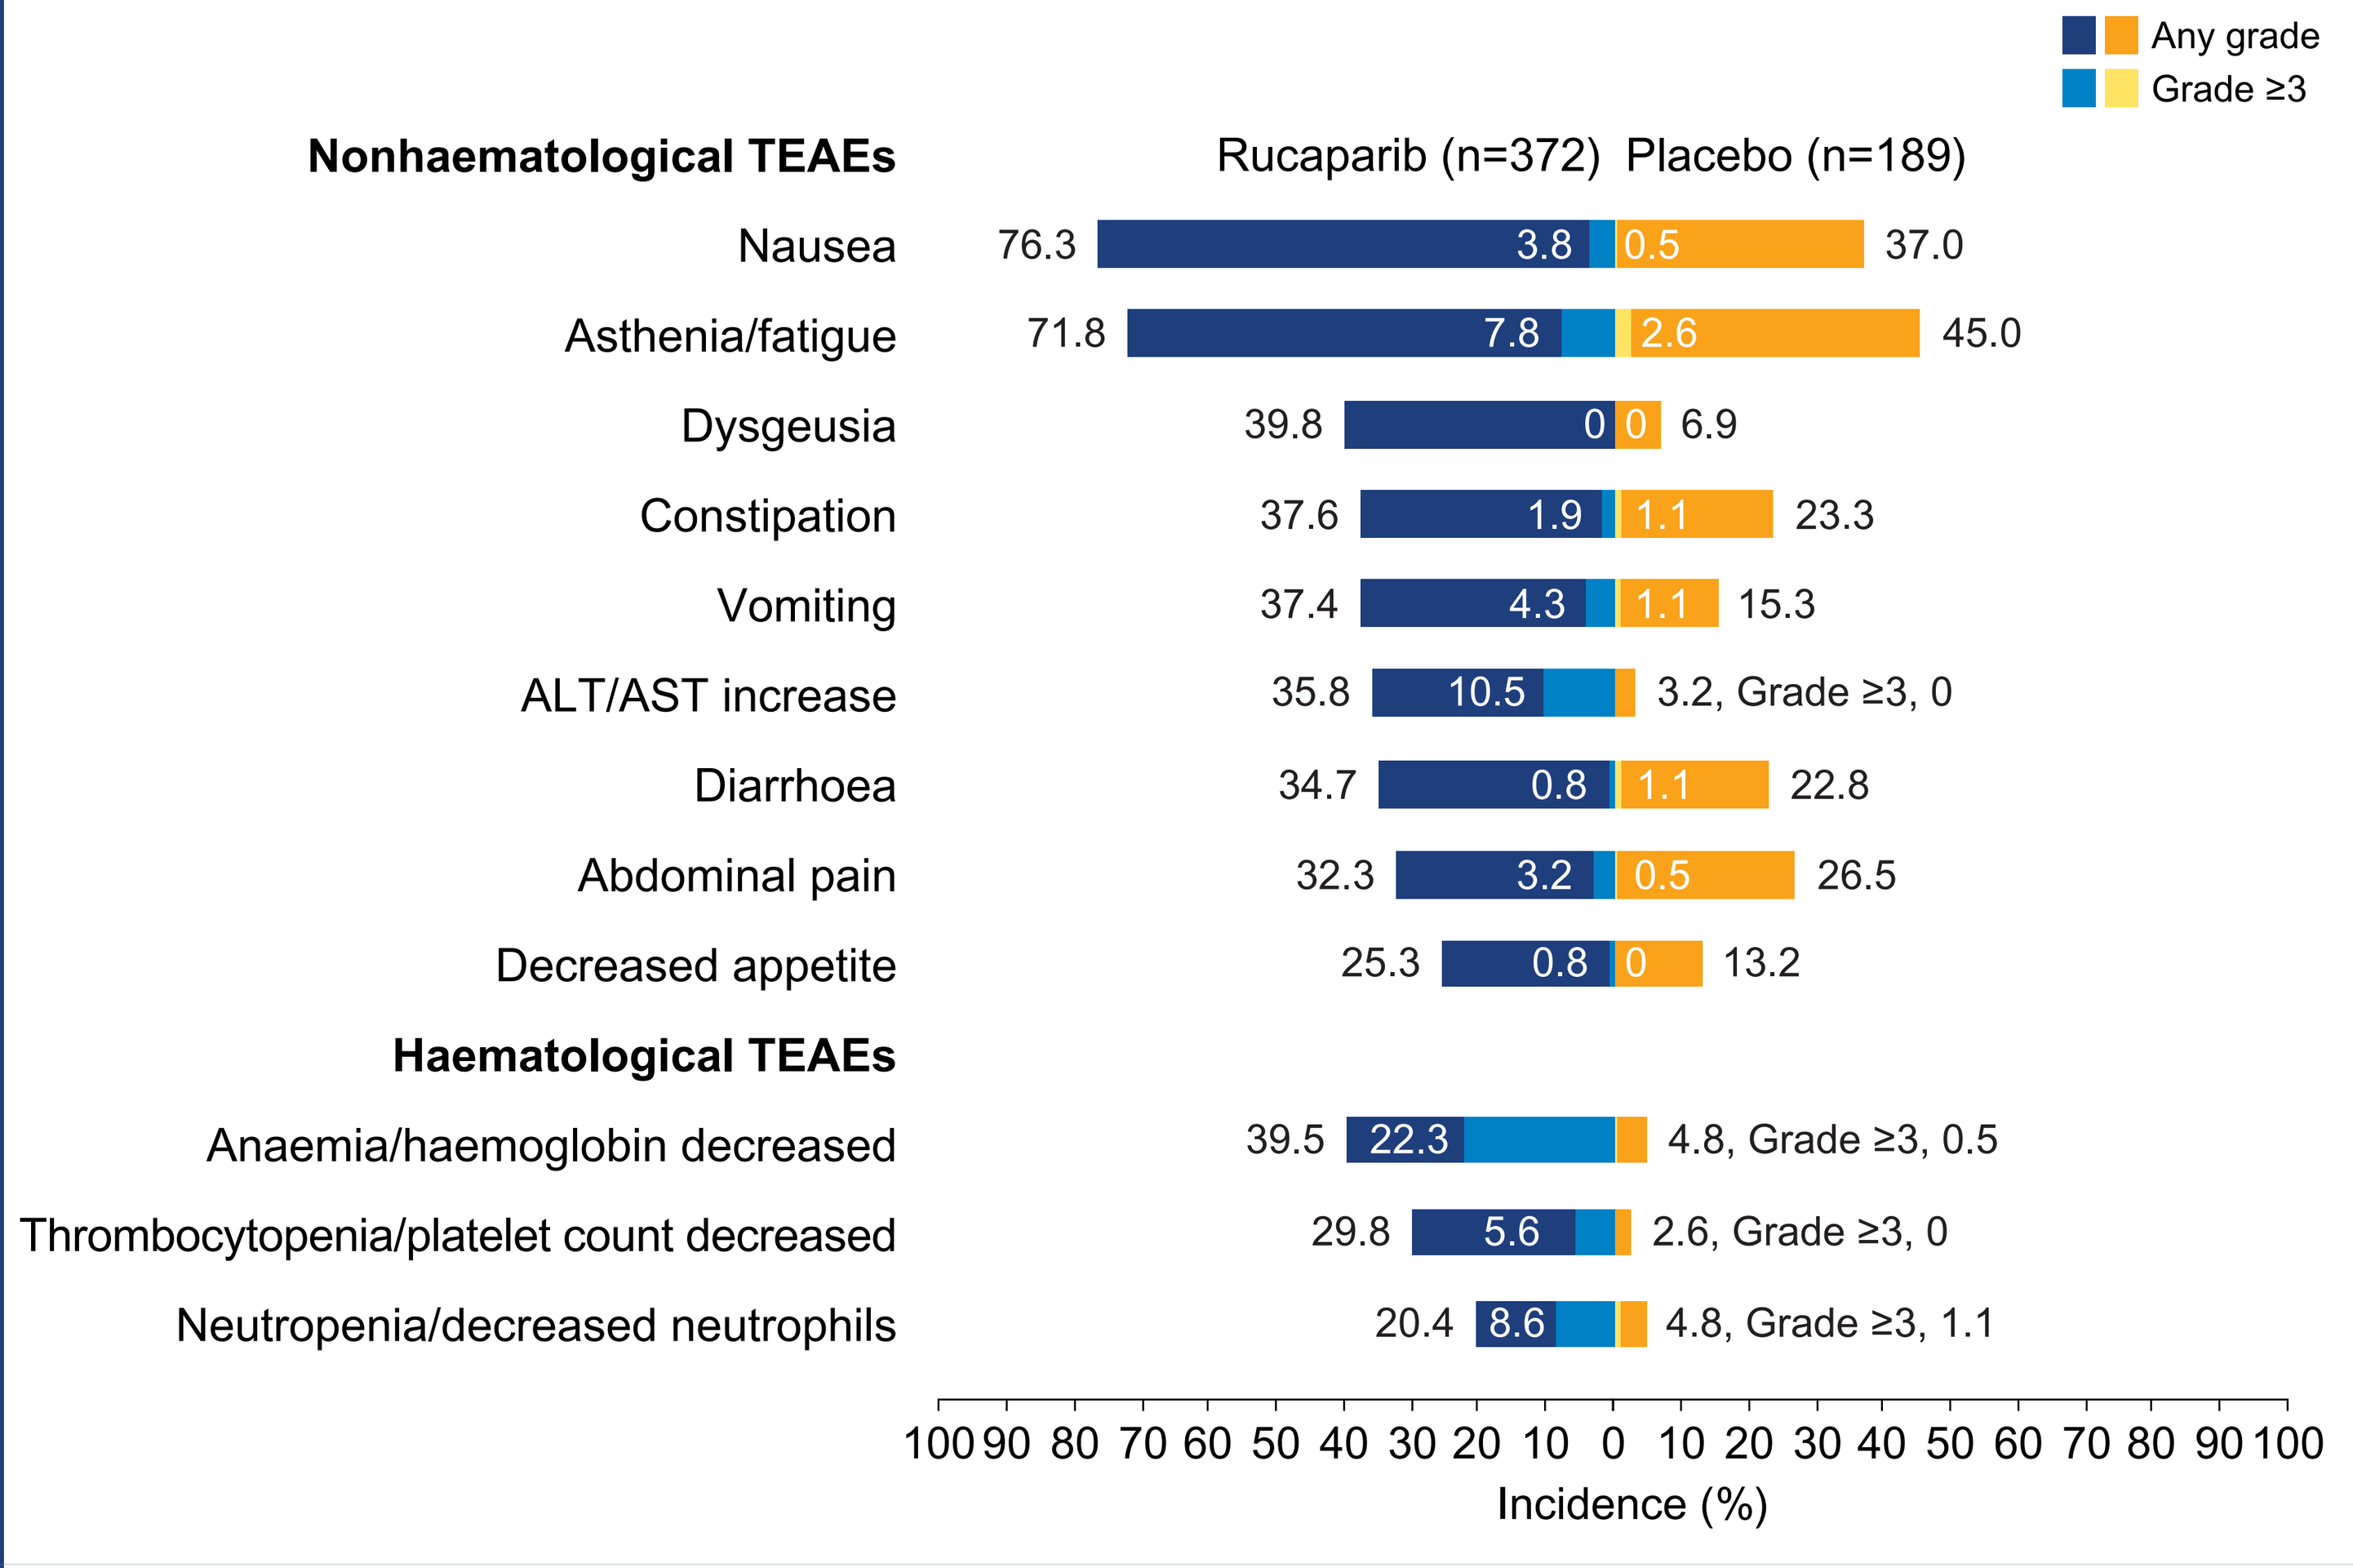
Any grade
Grade ≥3
Nonhaematological TEAEs
Rucaparib (n=372)
Placebo (n=189)
Nausea 76.3 3.8 0.5 37.0
Asthenia/fatigue 71.8 7.8 2.6 45.0
Dysgeusia 39.8 0 0 6.9
Constipation 37.6 1.9 1.1 23.3
Vomiting 37.4 4.3 1.1 15.3
ALT/AST increase 35.8 10.5 3.2, Grade ≥3, 0
Diarrhoea 34.7 0.8 1.1 22.8
Abdominal pain 32.3 3.2 0.5 26.5
Decreased appetite 25.3 0.8 0 13.2
Haematological TEAEs
Anaemia/haemoglobin decreased 39.5 22.3 4.8, Grade ≥3, 0.5
Thrombocytopenia/platelet count decreased 29.8 5.6 2.6, Grade ≥3, 0
Neutropenia/decreased neutrophils 20.4 8.6 4.8, Grade ≥3, 1.1
100 90 80 70 60 50 40 30 20 10 0 10 20 30 40 50 60 70 80 90 100
Incidence (%)

ALT, alanine aminotransferase; AST, aspartate aminotransferase; TEAE, treatment-emergent adverse event.

Prevalence of TEAEs by Month and Grade

- Nausea, the most common nonhaematological TEAE, was predominantly grade 1, and the prevalence declined from a peak of 66.9% in month 1 of treatment to 22.2% in month 24 (**Figure 2A**)
- The prevalence of any-grade anaemia/decreased haemoglobin, the most common haematological TEAE, peaked at 31.4% during month 4, thereafter declining to a plateau of ~20% after month 8, with a corresponding decrease in prevalence of grade ≥2 anaemia/decreased haemoglobin over this time period (**Figure 2B**)

Figure 2A. Prevalence of Nausea by Month and Grade in the Treatment Arms

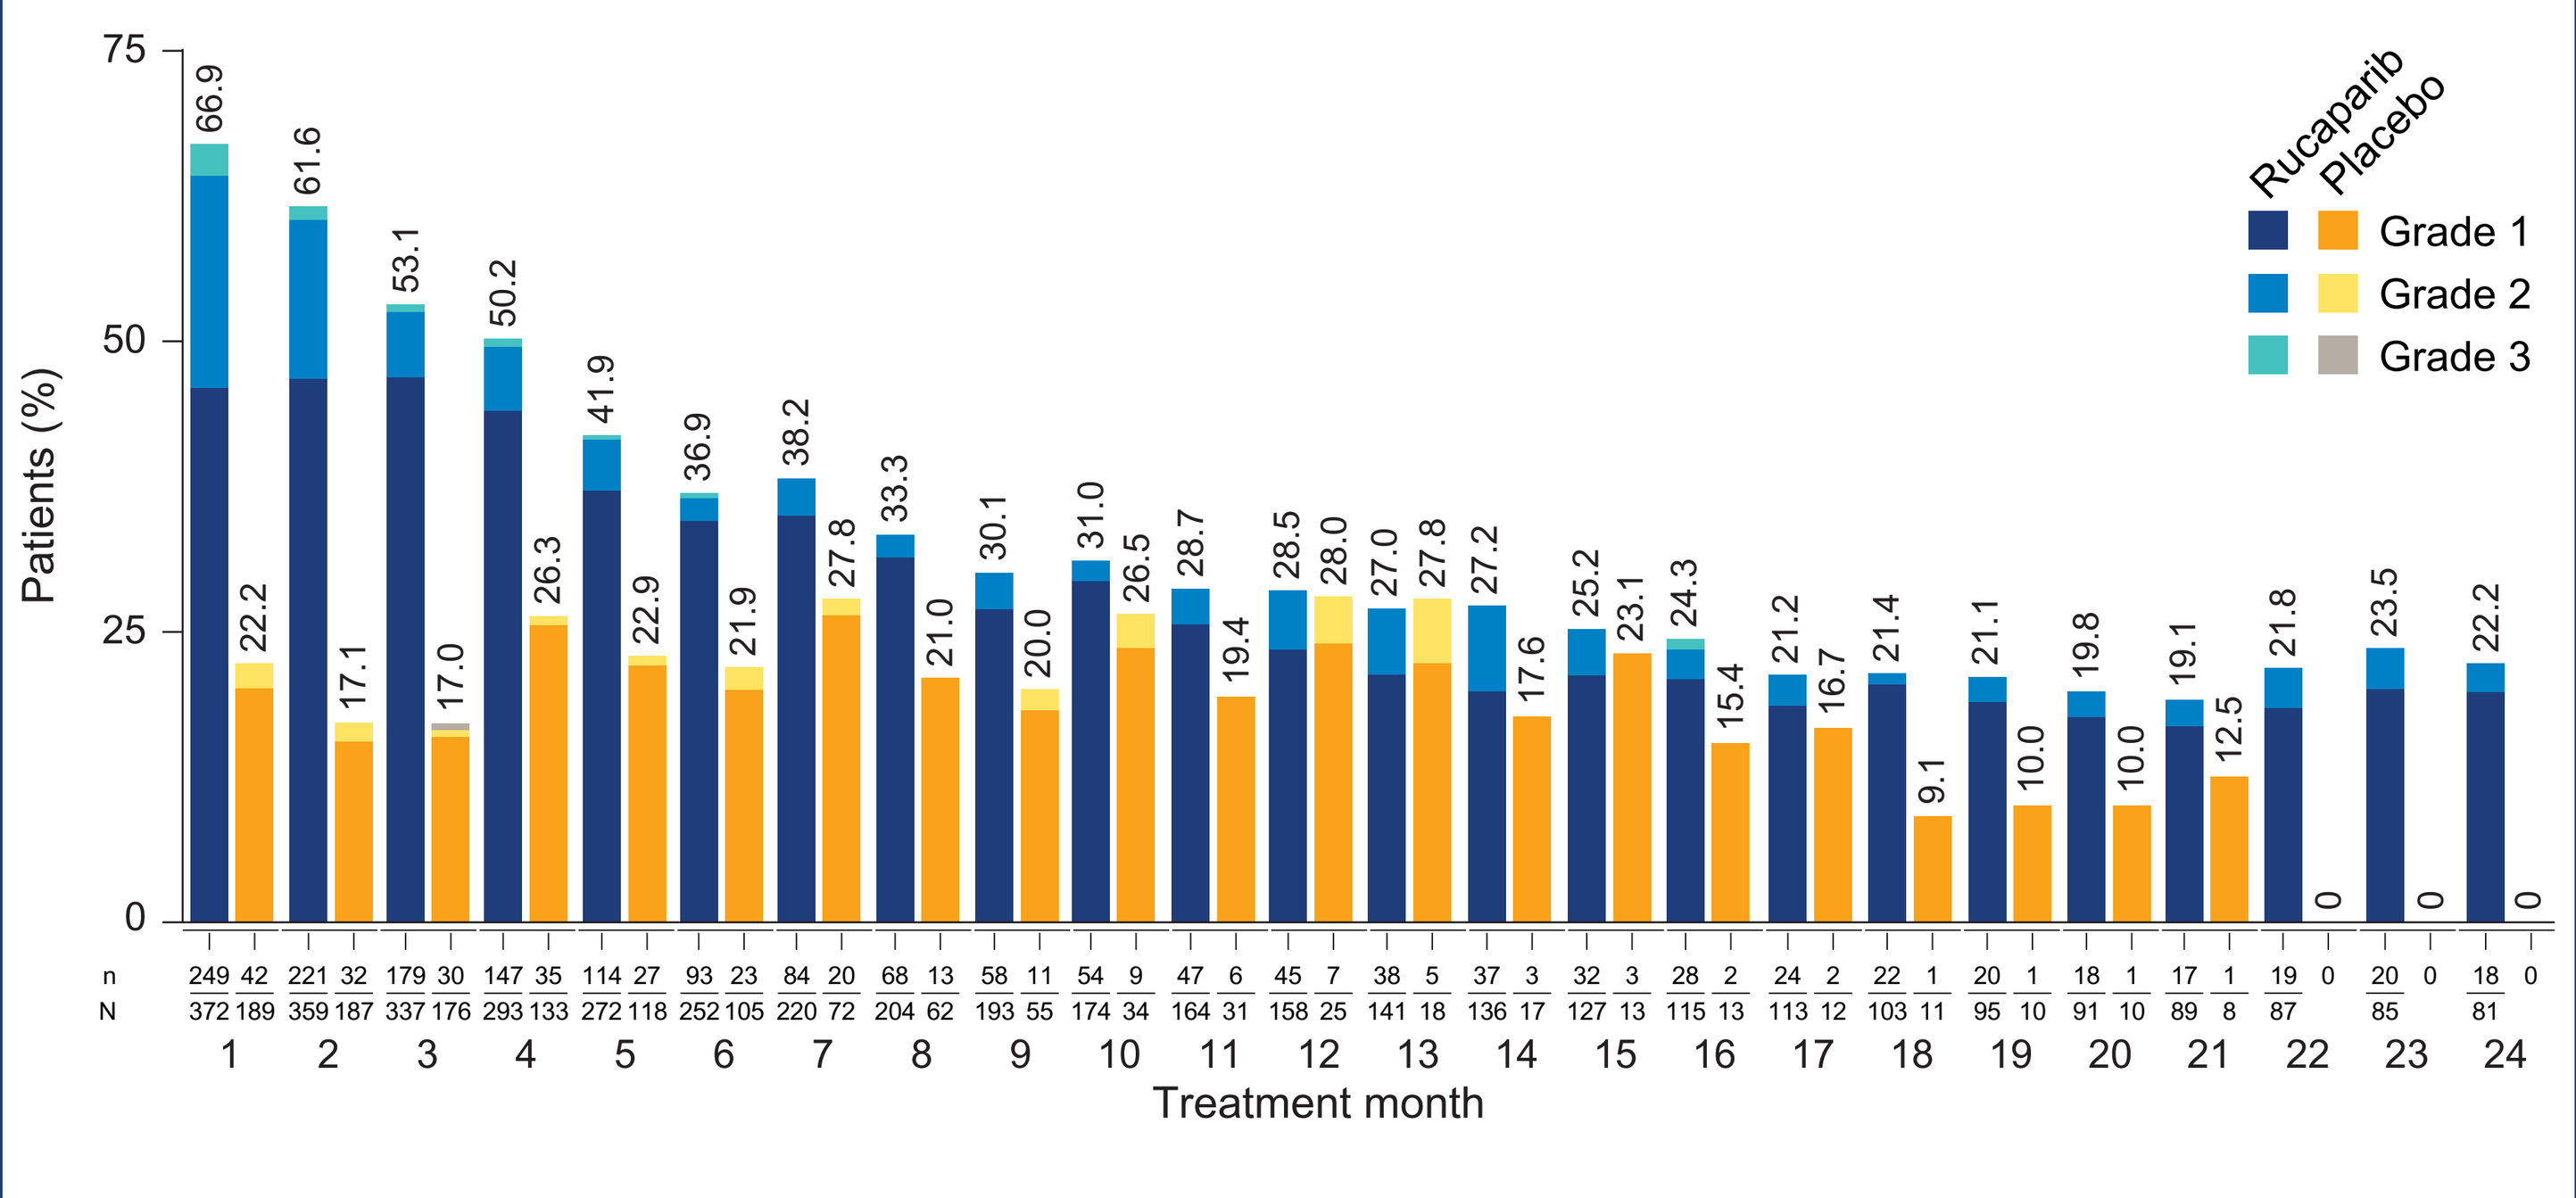
Rucaparib
Placebo
Grade 1
Grade 2
Grade 3
Patients (%)
0 25 50 75
66.9 22.2 61.6 17.1 53.1 17.0 50.2 26.3 41.9 22.9 36.9 21.9 38.2 27.8 33.3 21.0 30.1 20.0 31.0 26.5 28.7 19.4 28.5 28.0 27.0 27.8 27.2 17.6 25.2 23.1 24.3 15.4 21.2 16.7 21.4 9.1 21.1 10.0 19.8 10.0 19.1 12.5 21.8 0 23.5 0 22.2 0
n 249 42 221 32 179 30 147 35 114 27 93 23 84 20 68 13 58 11 54 9 47 6 45 7 38 5 37 3 32 3 28 2 24 2 22 1 20 1 18 1 17 1 19 0 20 0 18 0
N 372 189 359 187 337 176 293 133 272 118 252 105 220 72 204 62 193 55 174 34 164 31 158 25 141 18 136 17 127 13 115 13 113 12 103 11 95 10 91 10 89 8 87 85 81
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24
Treatment month

Prevalence is defined as the proportion of patients experiencing a particular treatment-emergent adverse event during each month and was based on the number of patients treated for that month. A treated patient is defined as receiving ≥1 dose of rucaparib or placebo during the month.

Figure 2B. Prevalence of Anaemia/Decreased Haemoglobin by Month and Grade in the Treatment Arms

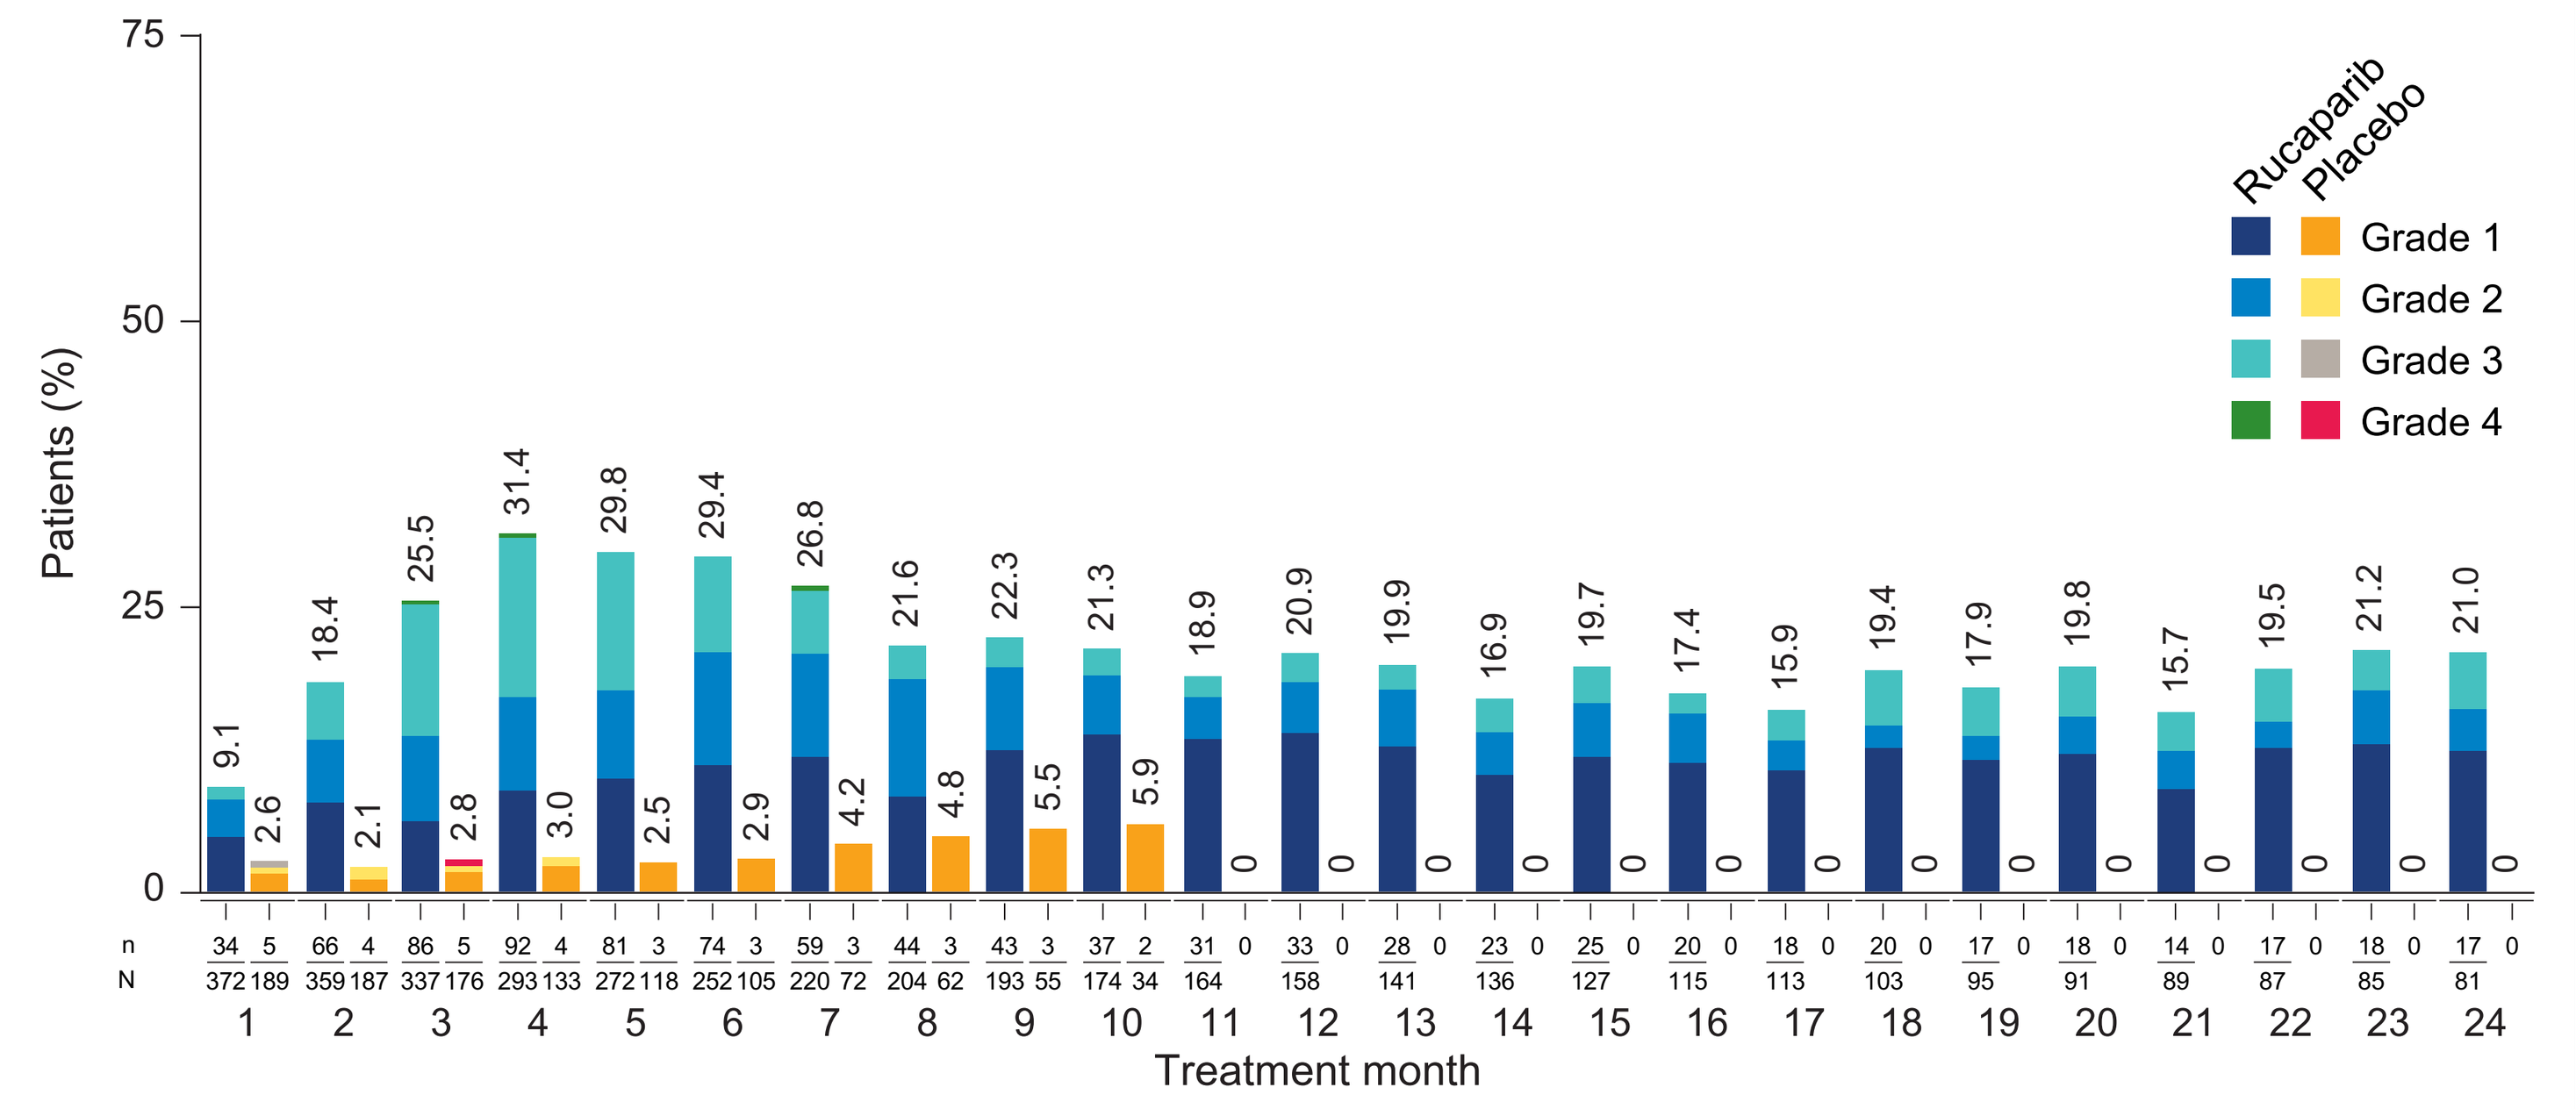
Rucaparib
Placebo
Grade 1
Grade 2
Grade 3
Grade 4
Patients (%)
0 25 50 75
9.1 2.6 18.4 2.1 25.5 2.8 31.4 3.0 29.8 2.5 29.4 2.9 26.8 4.2 21.6 4.8 22.3 5.5 21.3 5.9 18.9 0 20.9 0 19.9 0 16.9 0 19.7 0 17.4 0 15.9 0 19.4 0 17.9 0 19.8 0 15.7 0 19.5 0 21.2 0 21.0 0
n 34 5 66 4 86 5 92 4 81 3 74 3 59 3 44 3 43 3 37 2 31 0 33 0 28 0 23 0 25 0 20 0 18 0 20 0 17 0 18 0 14 0 17 0 18 0 17 0
N 372 189 359 187 337 176 293 133 272 118 252 105 220 72 204 62 193 55 174 34 164 158 141 136 127 115 113 103 95 91 89 87 85 81
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24
Treatment month

Prevalence is defined as the proportion of patients experiencing a particular treatment-emergent adverse event during each month and was based on the number of patients treated for that month. A treated patient is defined as receiving ≥1 dose of rucaparib or placebo during the month.

Time to Onset and Duration of First TEAE in the Rucaparib Arm

- For the majority of frequently reported (≥20%) any-grade TEAEs, median time to first onset was ≤45 days, except for anaemia/decreased haemoglobin, abdominal pain and neutropenia/decreased neutrophils which had median first onset times >45 days (**Figure 3A**)
 - Median time to first onset of grade ≥3 anaemia/decreased haemoglobin and ALT/AST increase was 85 and 16 days, respectively (**Figure 3C**)
 - Median time to onset of MDS was 844 days (95% CI, 365–1611); and of AML was 421 days
- For the frequently reported any-grade TEAEs, the median duration of the first event was generally <60 days, with the exception of asthenia/fatigue, decreased appetite and dysgeusia, for which it was ~90–160 days (**Figure 3B**)
 - Median duration of the first event of grade ≥3 anaemia/decreased haemoglobin and ALT/AST increase was <14 days (**Figure 3D**), which was shorter than the duration of the corresponding any-grade TEAEs (**Figure 3B**)

Figure 3. Median Time to First Onset and Median Duration of First TEAE of Most Frequently Reported Any-Grade (≥20%; A, B) and Grade ≥3 TEAEs (≥10%; C, D) in the Rucaparib Arm

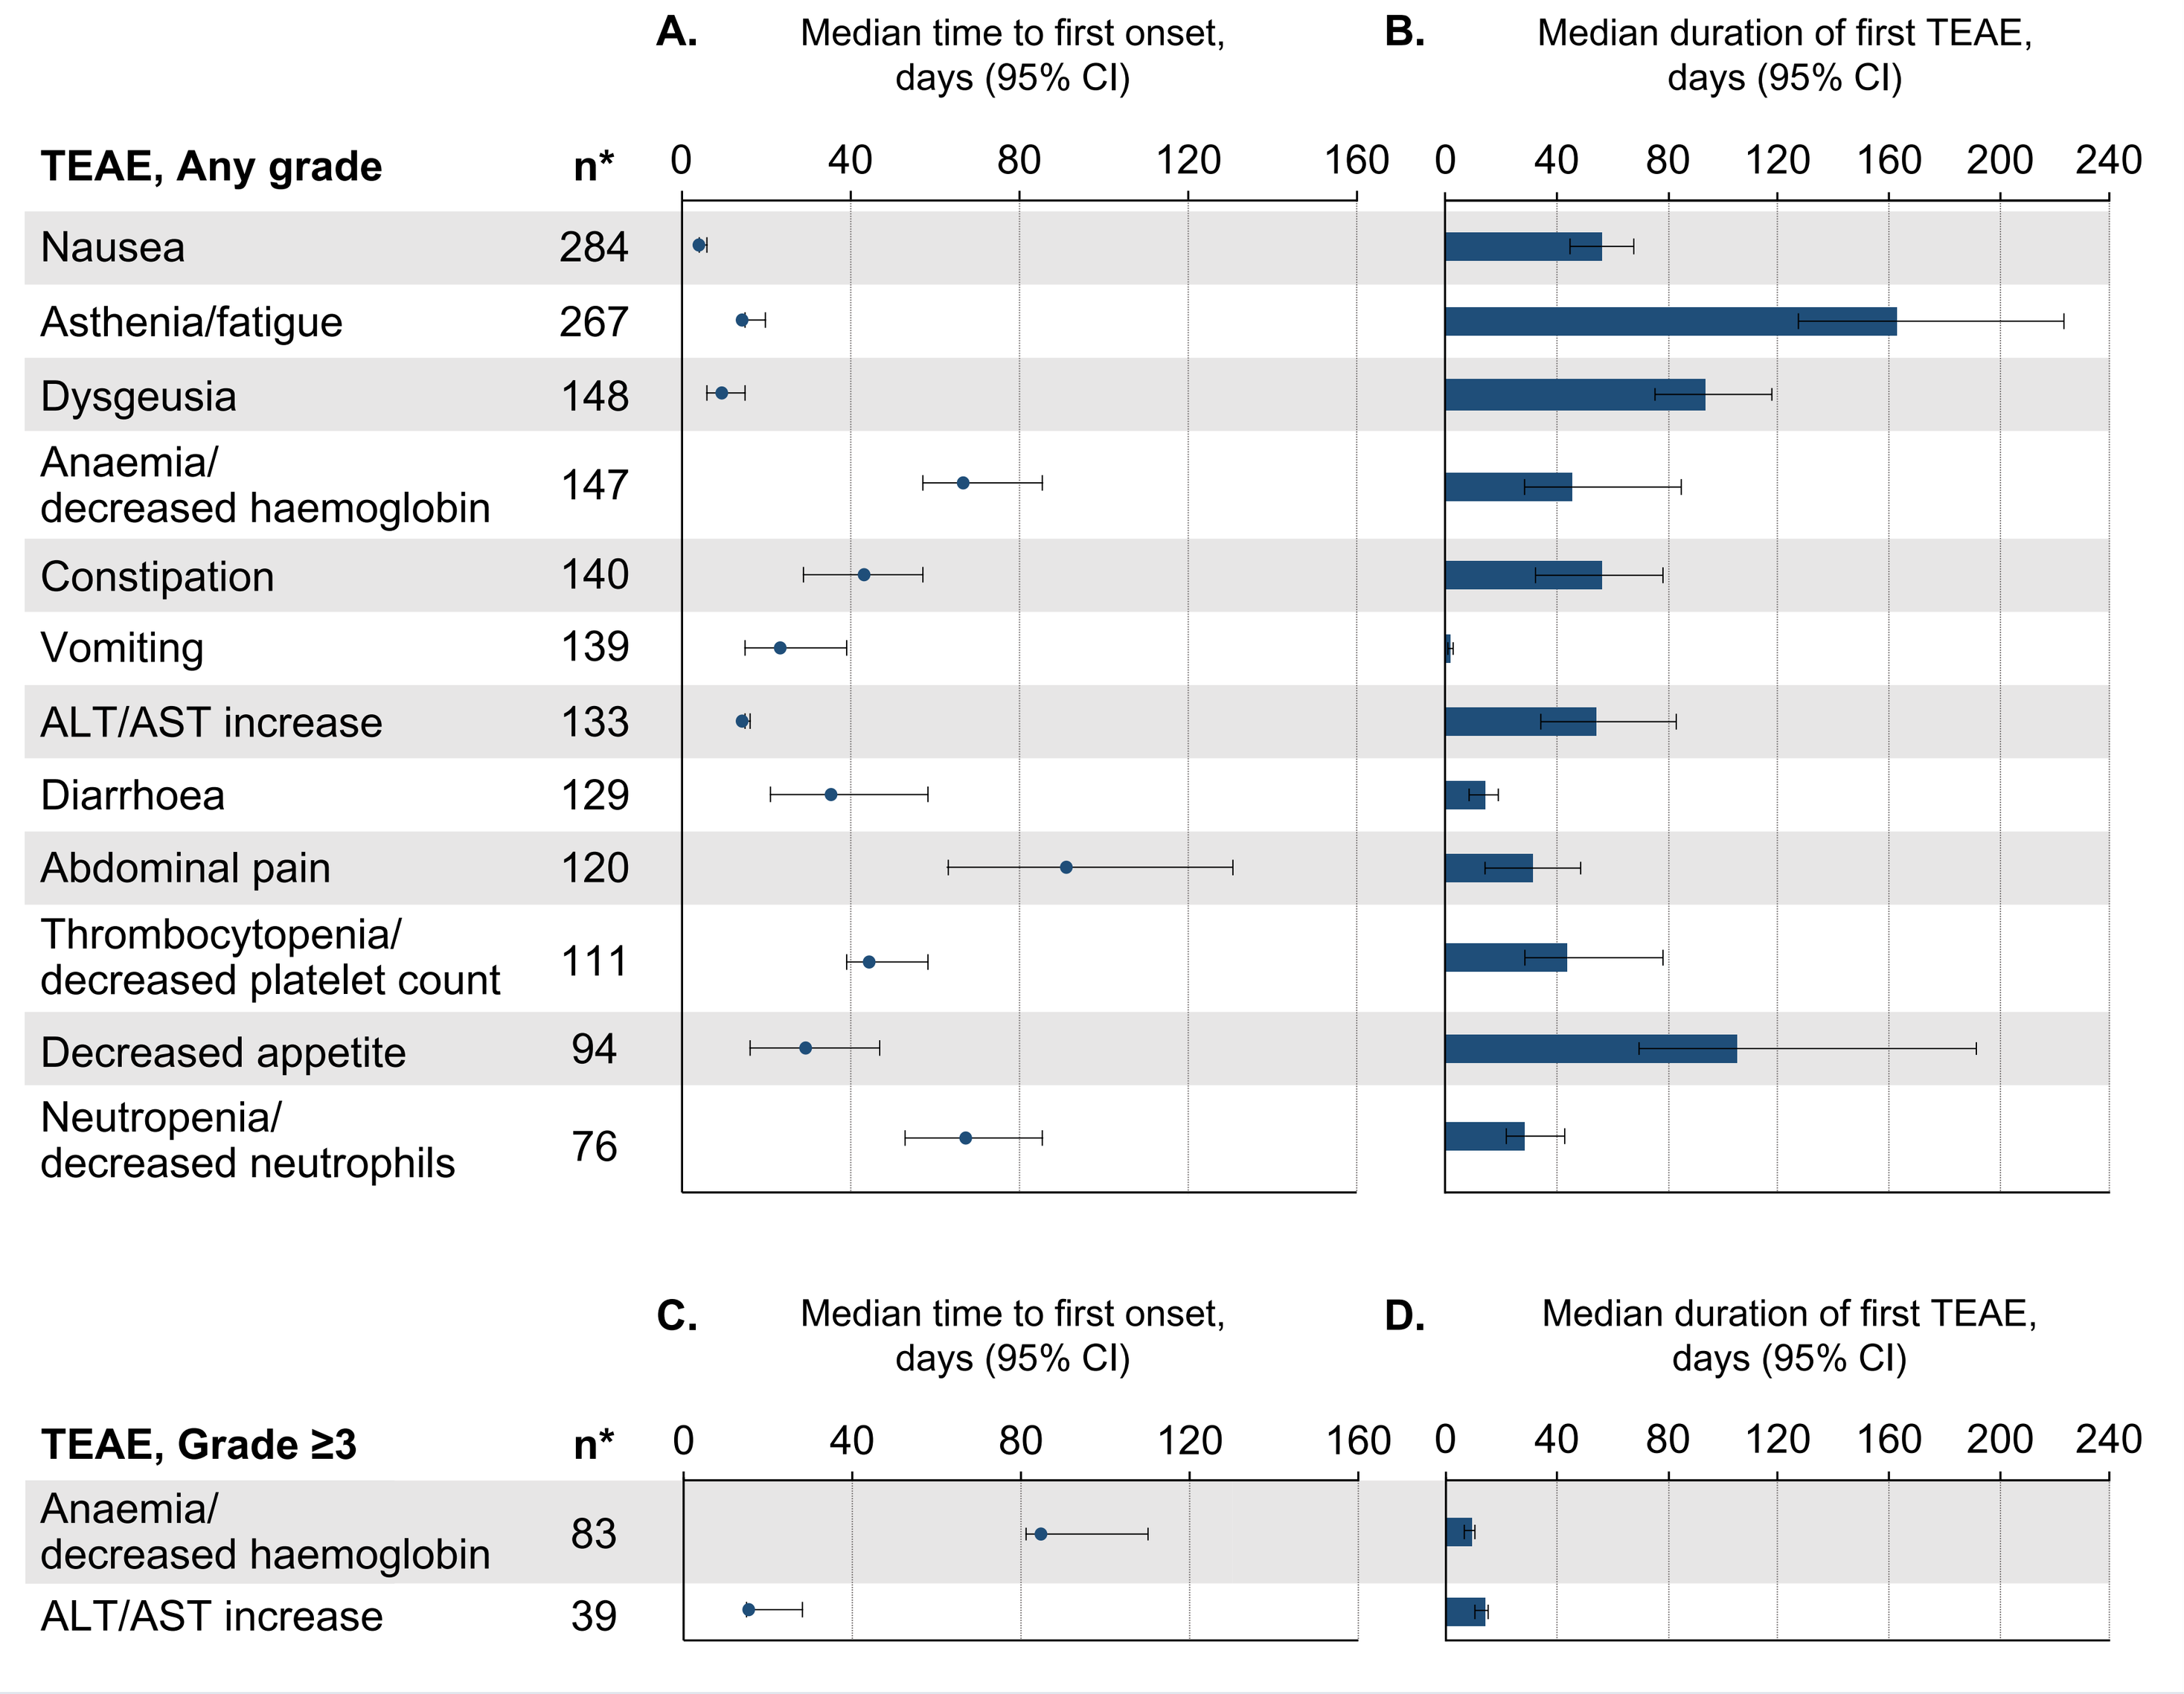
A. Median time to first onset, days (95% CI)
B. Median duration of first TEAE, days (95% CI)
TEAE, Any grade
n*
0 40 80 120 160
0 40 80 120 160 200 240
Nausea 284
Asthenia/fatigue 267
Dysgeusia 148
Anaemia/decreased haemoglobin 147
Constipation 140
Vomiting 139
ALT/AST increase 133
Diarrhoea 129
Abdominal pain 120
Thrombocytopenia/decreased platelet count 111
Decreased appetite 94
Neutropenia/decreased neutrophils 76
C. Median time to first onset, days (95% CI)
D. Median duration of first TEAE, days (95% CI)
TEAE, Grade ≥3
n*
0 40 80 120 160
0 40 80 120 160 200 240
Anaemia/decreased haemoglobin 83
ALT/AST increase 39

*Number of patients experiencing a TEAE with or without a known end date.
ALT, alanine aminotransferase; AST, aspartate aminotransferase; TEAE, treatment-emergent adverse event.

ESMO Congress | 19–21 September 2020 | Virtual Meeting