Poster number: 659P

Rucaparib Population Pharmacokinetics and Exposure-Response Analyses in Patients With Metastatic Castration-Resistant Prostate Cancer (mCRPC) in TRITON2

Simon Chowdhury,[1] Akash Patnaik,[2] David Campbell,[3] Jeremy Shapiro,[4] Alan H. Bryce,[5] Ray McDermott,[6] Brieuc Sautois,[7] Nicholas J. Vogelzang,[8] Richard Bambury,[9] Eric Voog,[10] Jingsong Zhang,[11] Michelle Green,[12] Jeri Beltman,[13] Darrin Despain,[13] Andrew D. Simmons,[13] Simon Watkins,[14] Tony Golsorkhi,[13] Jim Xiao,[13] Wassim Abida[15]

[1]Guy's Hospital and Sarah Cannon Research Institute, London, UK; [2]University of Chicago, USA; [3]Barwon Health, University Hospital Geelong, Australia; [4]Cabrini Hospital, Malvern, Australia; [5]Mayo Clinic, Phoenix, USA; [6]Adelaide and Meath Hospital (Incorporating the National Children's Hospital), Dublin, Ireland; [7]Liège University Hospital (CHU Sart Tilman), Belgium; [8]Comprehensive Cancer Centers of Nevada, Las Vegas, USA; [9]Cork University Hospital, Ireland; [10]Clinique Victor Hugo Centre Jean Bernard, Le Mans, France; [11]H. Lee Moffitt Cancer Center, Tampa, USA; [12]Certara Strategic Consulting, Menlo Park, USA; [13]Clovis Oncology, Inc., Boulder, USA; [14]Clovis Oncology UK, Ltd., Cambridge, UK; [15]Memorial Sloan Kettering Cancer Center, New York, USA

SUMMARY

- **Population pharmacokinetic (PK) analyses suggest that there is no difference in rucaparib PK in men with mCRPC and women with ovarian cancer**
- **Differences in the amount of rucaparib in the blood (measured by $AUC_{avg,ss}$) after administration of rucaparib 600 mg twice daily (BID) did not appear to impact the efficacy of treatment**
- **A higher maximum concentration of rucaparib in the blood (measured by $C_{max,avg,ss}$) was not associated with increased rates of most safety endpoints analysed, including haematological adverse events**
 - **Grade ≥2 creatinine increase was correlated with rucaparib exposure, consistent with reversible creatinine elevation observed in other rucaparib studies[1,2]; this may be due to inhibition of renal transporters (eg, MATE1 and MATE2-K) by rucaparib[3,4] rather than direct impact on renal function**

CLINICAL IMPLICATIONS

- **Rucaparib PK in men with mCRPC was comparable to that observed in women with ovarian cancer. These analyses support the use of rucaparib in men with mCRPC using a starting dose of 600 mg BID**

Poster Download
Also available at: https://clovisoncology.com/files/ESMO2020_Chowdhury_Poster.pdf
Copies of this e-poster obtained through Quick Response (QR) codes and/or web links are for personal use only and may not be reproduced without written permission of the authors.
Corresponding author: Simon Chowdhury; Simon.Chowdhury@gstt.nhs.uk.

REFERENCES

1. Kristeleit et al. *Int J Gynecol Cancer.* 2019;29:1396-404.
2. Coleman et al. *Lancet.* 2017;390:1949-61.
3. Rubraca (rucaparib) tablets [prescribing information]. Boulder, CO: Clovis Oncology, Inc.; 2020.
4. Zibetti Dal Molin et al. *Int J Gynecol Cancer.* 2020;30:89-93.
5. Xiao et al. *Clin Pharmacol Ther.* 2017;101(suppl 1):abst PII-144.
6. Liao et al. *Invest New Drugs.* 2020;38:765-75.
7. Abida et al. *J Clin Oncol.* 2020 [Epub ahead of print]. doi:10.1200/JCO.20.01035.
8. Shapiro et al. *Clin Pharmacol Drug Dev.* 2019;8:107-18.

ACKNOWLEDGEMENTS

This study was funded by Clovis Oncology, Inc. Medical writing and editorial support funded by Clovis Oncology, Inc., were provided by Nathan Yardley and Frederique H. Evans of Ashfield Healthcare Communications.

PRESENTING AUTHOR DISCLOSURE

Simon Chowdhury has served in a consulting or advisory role and/or on speakers bureaus for Clovis Oncology, Astellas, Bayer, BeiGene, Janssen, Pfizer and Sanofi; received honoraria from GlaxoSmithKline and Novartis; received research funding from Clovis Oncology, Johnson & Johnson, and Sanofi; received financial support for travel and accommodation from Clovis Oncology and Beigene; and holds stock in Curve.life.

INTRODUCTION

- Rucaparib is a poly(ADP-ribose) polymerase (PARP) inhibitor that received accelerated approval in the United States for the treatment of adult patients with a deleterious *BRCA1* or *BRCA2* (BRCA) alteration (germline and/or somatic)-associated mCRPC who have been treated with androgen receptor (AR)-directed therapy and a taxane-based chemotherapy
- A population pharmacokinetics (PK) model to describe rucaparib PK was developed based on data from women (n=454) with ovarian cancer (OC) treated with rucaparib[5]; however, rucaparib PK in men have not been previously established
 - Rucaparib PK is described by a 2-compartmental model with sequential zero- and first-order absorption and elimination by both urinary excretion and hepatic metabolism[5,6]
- Here, we report rucaparib PK in men with mCRPC and investigate exposure and safety/efficacy correlations using data from the phase 2 study, TRITON2 (NCT02952534)

METHODS

- TRITON2 enrolled men with mCRPC associated with a deleterious germline or somatic alteration in *BRCA1*, *BRCA2*, or another prespecified DNA damage repair gene, and disease progression following 1 to 2 lines of next-generation AR-directed therapy for prostate cancer and 1 prior taxane-based chemotherapy for castration-resistant disease[7]
- Patients in TRITON2 received a starting dose of rucaparib 600 mg twice daily (BID)
- Sparse plasma PK data were analysed with a population PK model developed using PK data from women with OC[5]
- Exposure-efficacy and exposure-safety analyses were conducted with selected efficacy and safety endpoints; covariate effects were also explored (**Table 1**)

Table 1. Overall Exposure-Efficacy and Exposure-Safety Analyses

	Exposure-efficacy analyses	Exposure-safety analyses
Analysis population	• Patients with a BRCA alteration who received ≥1 rucaparib dose and had ≥1 evaluable PK sample	• Patients with a BRCA or non-BRCA gene alteration who received ≥1 rucaparib dose and had ≥1 evaluable PK sample
Endpoints	• Investigator-assessed confirmed ORR (per modified RECIST/PCWG3 criteria) • Duration of response • Maximum percentage reduction from baseline in sum of diameters of target lesions • Confirmed PSA response (≥50% decrease) • Confirmed PSA response (≥90% decrease) • Maximum percentage reduction from baseline in PSA • Duration of PSA response • Time to PSA progression	Laboratory variables[a]: • Haematological – grade ≥3 neutrophil decrease, grade ≥3 leucocyte decrease, grade ≥3 lymphocyte decrease, grade ≥3 platelet decrease, and grade ≥3 haemoglobin decrease; maximum haemoglobin reduction • Hepatic – grade ≥3 ALT increase • Renal – grade ≥2 creatinine increase Adverse events[a]: • Grade ≥3 fatigue/asthenia
PK exposure measure[b]	• Steady-state AUC normalised by actual dose for each patient up to the event of interest ($AUC_{avg,ss}$)	• Steady-state C_{max} normalised by actual dose for each patient up to the event of interest ($C_{max,avg,ss}$)
Model	• Linear logistic regression for binary endpoints • Linear regression for continuous endpoints • Cox regression for time-to-event endpoints	• Linear logistic regression for binary endpoints • Linear regression for continuous endpoints
Covariates[c]	• Age • ECOG PS • Germline BRCA alteration status • Prior chemotherapy[d] • Presence of measurable disease at baseline • Baseline PSA • Baseline albumin • Baseline LDH • Baseline ALP	• Age • ECOG PS • BRCA vs non-BRCA gene alteration • Presence of measurable disease at baseline • Investigator-assessed number of bone lesions[e] • Baseline albumin • Baseline haemoglobin

[a]All laboratory parameters and adverse events were based on the worst grade experienced using standard Common Terminology Criteria for Adverse Events, version 5.0, grading; exposure-safety relationships were evaluated for binary endpoints with at least 10 events and at least 5% incidence. [b]A published population PK model[5] was used to estimate $AUC_{avg,ss}$ and $C_{max,avg,ss}$. [c]Clinical covariates were tested in multivariate models. [d]Includes docetaxel or cabazitaxel. [e]Reported as high (≥10 lesions) or low (<10 lesions).
ALP, alkaline phosphatase; ALT, alanine aminotransferase; AUC, area under the concentration-time curve; BRCA, *BRCA1* or *BRCA2*; C_{max}, maximum concentration; ECOG PS, Eastern Cooperative Oncology Group performance status; LDH, lactate dehydrogenase; ORR, objective response rate; PCWG3, Prostate Cancer Clinical Trials Working Group 3; PK, pharmacokinetic; PSA, prostate-specific antigen; RECIST, Response Evaluation Criteria In Solid Tumors, version 1.1.

RESULTS

- The PK analysis population included 199 men with mCRPC from TRITON2 (visit cutoff date, 13 Sep 2019)
- The observed PK data from patients in TRITON2 were consistent with the simulated data using the existing population PK model developed from patients with OC (**Figure 1**), suggesting no difference in rucaparib PK by tumor type or sex
- Model estimated individual steady-state exposures (post hoc) were used for subsequent exposure-response analyses (**Table 2**)

Figure 1. Visual Predictive Check of PK Model

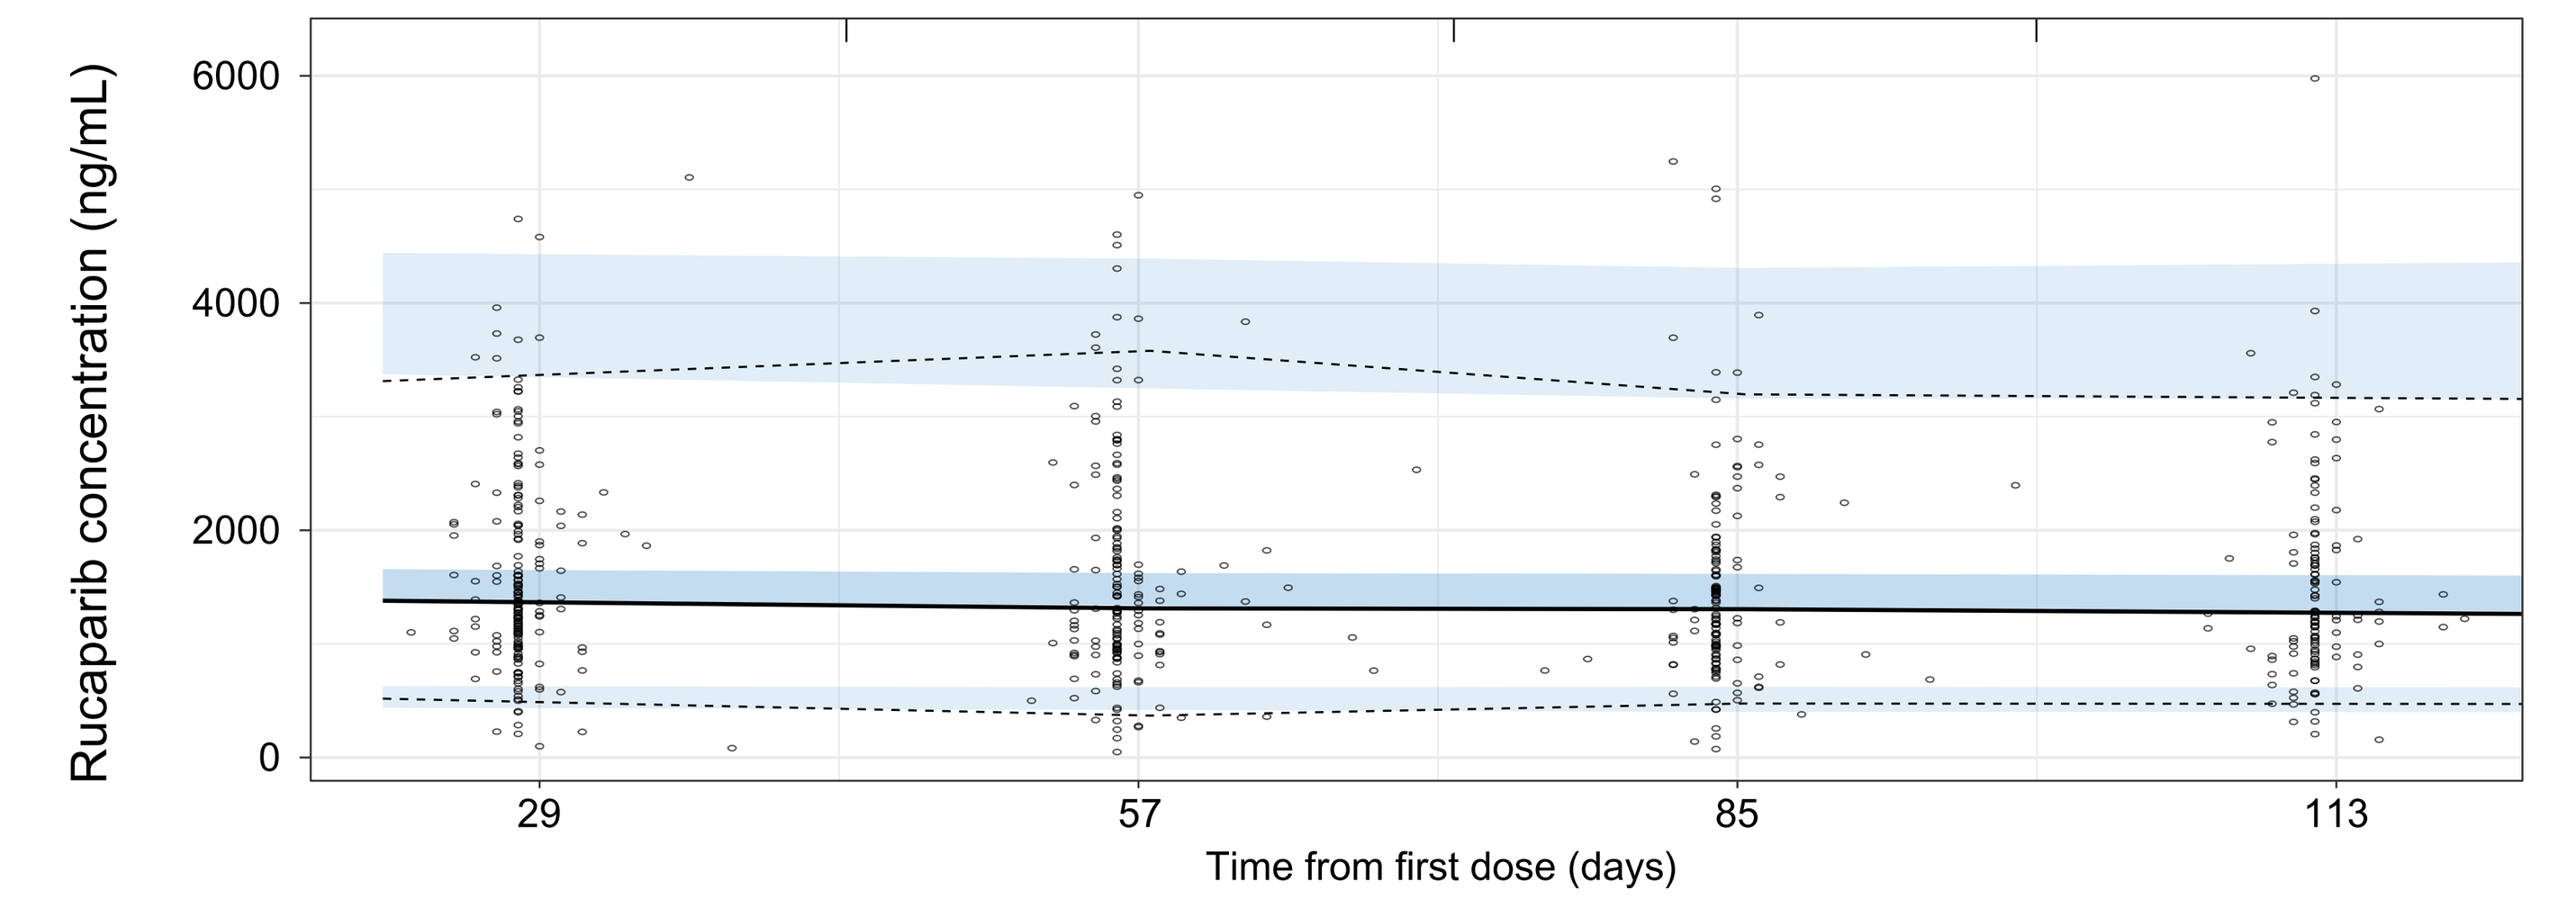
Rucaparib concentration (ng/mL)
6000
4000
2000
0
29
57
85
113
Time from first dose (days)

Data points represent the individual observed data. The solid black line represents the median of the observed data. The dashed black lines represent the 5th and 95th percentiles of the observed data. The shaded regions encompass 90% of the simulated values (n=1000) of the predicted medians (darker blue), 5th (lighter blue), and 95th (lighter blue) percentiles.
PK, pharmacokinetic.

Table 2. Exposure Summary in the Exposure-Efficacy and Exposure-Safety Analyses[a]

Variable	Exposure-efficacy analysis	Exposure-safety analysis
Patients (n)	115	209
PKs (n)[b]	113	199
$C_{max,avg,ss}$		
Mean ng/mL (SD)	1782 (750)	1844 (807)
[min, max]	[396, 4586]	[396, 5343]
$AUC_{avg,ss}$		
Mean ng/mL × h (SD)	39009 (17506)	40453 (18887)
[min, max]	[7929, 105073]	[7929, 123158]

[a]Overall exposure estimates over the full duration of treatment. [b]Number of patients with ≥1 evaluable PK sample.
$AUC_{avg,ss}$, steady-state area under the concentration-time curve normalised by actual dose for each patient up to the end of treatment; $C_{max,avg,ss}$, steady-state maximum concentration normalised by actual dose for each patient up to the end of treatment; PK, pharmacokinetic; SD, standard deviation.

Exposure-Efficacy Relationships

- Exposure-efficacy analyses in 113 men with mCRPC and a BRCA alteration suggested no statistically significant and clinically plausible exposure-efficacy relationships between the steady-state area under the concentration-time curve normalised by actual dose for each patient up to the event of interest ($AUC_{avg,ss}$) and investigator-assessed objective response rate (ORR) or other efficacy measures (**Table 3, Figure 2**)
- Covariate analysis suggested that patients with higher baseline prostate-specific antigen (PSA; as a continuous variable) had lower investigator-assessed ORR, and patients with measurable disease at baseline had greater PSA response than patients with nonmeasurable disease (**Table 3**)

Table 3. Exposure-Efficacy Relationships With Covariates in Men With mCRPC and a BRCA Alteration

Efficacy endpoint	n[a]	*P* value for exposure: No covariate	*P* value for exposure: Adjusted for covariates[b]	Significant covariates (*P*<0.05)
Investigator-assessed confirmed ORR[c]	64	0.632	0.448	Baseline PSA[d]
Duration of response[c]	33	0.838	0.833	Germline BRCA status[e]
Maximum percentage reduction from baseline in sum of diameters of target lesions	64	0.019[f]	NA	None identified
Confirmed PSA response (≥50% decrease)	113	0.721	0.047	Baseline albumin[g]
Confirmed PSA response (≥90% decrease)	113	0.559	0.610	Measurable disease at baseline[h]
Maximum percentage reduction from baseline in PSA	112	<0.001[f]	<0.001[f]	Measurable disease at baseline[h]
Duration of PSA response	62	0.527	0.335	Germline BRCA status[e]
Time to PSA progression	113	0.486	0.047	Germline BRCA status[e]

[a]Number of patients evaluable for the endpoint and evaluable for PK with available rucaparib exposure estimates. [b]The model includes significant covariates. [c]Per modified RECIST/PCWG3 criteria. [d]Patients with higher baseline PSA levels had lower investigator-assessed confirmed ORR. [e]Patients with germline BRCA alteration status had higher relative risk for shorter duration of response, shorter duration of PSA response, and shorter time to PSA progression. [f]Although *P*<0.05, decreases in sum of diameters of target lesions and decreases in PSA were smaller at higher exposures; this was not considered clinically plausible. [g]Patients with lower baseline albumin had higher confirmed PSA response (≥50% decrease) rates. [h]Patients with measurable disease at baseline had higher confirmed PSA response (≥90% decrease) and greater maximum PSA reduction from baseline.
BRCA, *BRCA1* or *BRCA2*; mCRPC, metastatic castration-resistant prostate cancer; ORR, objective response rate; PCWG3, Prostate Cancer Clinical Trials Working Group 3; PK, pharmacokinetics; PSA, prostate-specific antigen; RECIST, Response Evaluation Criteria In Solid Tumors, version 1.1.

Figure 2. Exposure-Efficacy Relationships for (A) Investigator-assessed Confirmed ORR and (B) Confirmed PSA Response

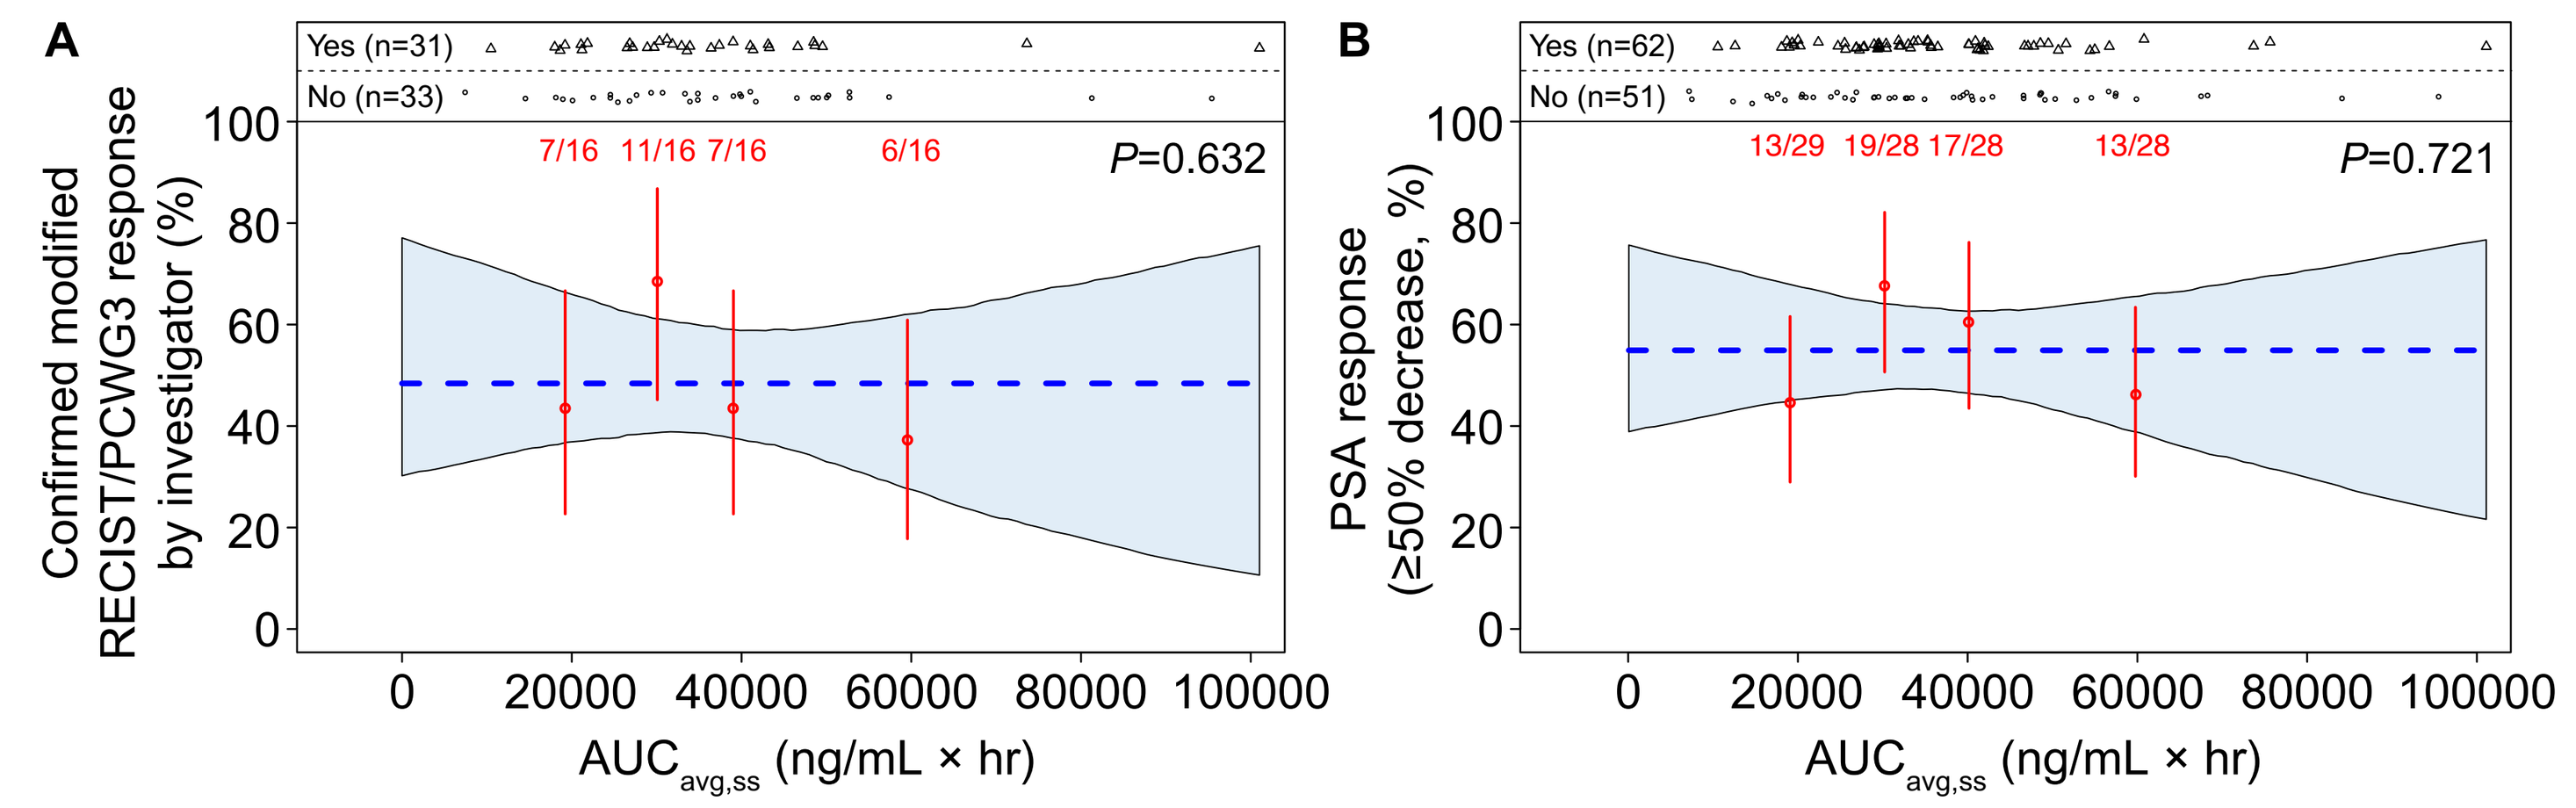
A
Yes (n=31)
No (n=33)
7/16 11/16 7/16 6/16
P=0.632
Confirmed modified RECIST/PCWG3 response by investigator (%)
0 20000 40000 60000 80000 100000
AUCavg,ss (ng/mL × hr)
B
Yes (n=62)
No (n=51)
13/29 19/28 17/28 13/28
P=0.721
PSA response (≥50% decrease, %)
0 20000 40000 60000 80000 100000
AUCavg,ss (ng/mL × hr)

Yes/no represents the number of patients with/without the event being evaluated. Red points represent the mean exposure and event rates in patients stratified by exposure quartile. Vertical red bars represent the 5th to 95th percentile CIs of the event rate. Event numbers (patients with event/total patients) are displayed above each vertical bar. The shaded blue region represents 5th to 95th percentile CIs of the predicted event rate. The horizontal blue dashed line shows the average response rate when the *P* value is >0.05. Regression analyses were conducted separately for investigator-assessed ORR and PSA response.
$AUC_{avg,ss}$, steady-state area under the concentration-time curve normalised by actual dose for each patient up to the event of interest; ORR, objective response rate; PCWG3, Prostate Cancer Clinical Trials Working Group 3; PSA, prostate-specific antigen; RECIST, Response Evaluation Criteria In Solid Tumors, version 1.1.

Exposure-Safety Relationships

- In the PK analysis population (n=199), the ratio of actual to nominal cumulative dose (based on 600 mg BID starting dose) was 0.9, suggesting most patients received rucaparib 600 mg BID throughout treatment
- Statistically significant correlations with the steady-state maximum concentration normalised by actual dose for each patient up to the event of interest ($C_{max,avg,ss}$) were observed for grade ≥2 creatinine elevation (**Table 4; Figure 3**)
- No significant exposure-safety relationship was observed between haematological safety endpoints without covariates
 - Covariate analysis suggested that patients with lower baseline haemoglobin (as a continuous variable) had smaller decreases in haemoglobin levels after rucaparib exposure but had a higher incidence of grade ≥3 haemoglobin decrease (**Table 4**)
 - Patients with higher baseline albumin and patients with a BRCA alteration had larger decreases in haemoglobin than patients with lower baseline albumin and patients without a BRCA alteration, respectively (**Table 4**)

Table 4. Exposure-Safety Relationships With Covariates in Men with a BRCA or non-BRCA Alteration

Safety endpoint (n=199)[a]	*P* value for exposure: No covariate	*P* value for exposure: Adjusted for covariates[b]	Significant covariates (*P*<0.05)
Neutrophil count decrease, grade ≥3	0.449	NA	None identified
Leucocyte count decrease, grade ≥3	0.200	NA	None identified
Lymphocyte count decrease, grade ≥3	0.121	NA	None identified
Platelet count decrease, grade ≥3	0.301	NA	None identified
Haemoglobin decrease, grade ≥3	0.062	0.081	Age <65,[c] age ≥75,[c] baseline haemoglobin[d]
Haemoglobin, CFB	0.167	0.005	Non-BRCA, baseline albumin, baseline haemoglobin[e]
ALT increase, grade ≥3	0.571	NA	None identified
Creatinine increase, grade ≥2	0.004	NA	None identified
Fatigue/asthenia, grade ≥3	0.525	0.336	Age <65, age ≥75[f]

[a]Number of patients evaluable for the endpoint and evaluable for PK with available rucaparib exposure estimates. [b]The model includes significant covariates. [c]Patients aged <65 years and those aged ≥75 years had a larger number of grade ≥3 haemoglobin decrease events than patients aged 65–74 years. [d]Patients with lower baseline haemoglobin levels had higher incidence of grade ≥3 haemoglobin decrease. [e]Patients with non-BRCA gene alterations, those with lower baseline albumin levels, and those with lower baseline haemoglobin levels had a smaller decrease from baseline haemoglobin levels. [f]Patients aged ≥75 years were more likely to develop grade ≥3 fatigue/asthenia than younger patients.
ALT, alanine aminotransferase; BRCA, *BRCA1* or *BRCA2*; CFB, change from baseline; ECOG PS, Eastern Cooperative Oncology Group performance status; NA, not applicable; PK, pharmacokinetics.

Figure 3. Exposure-Safety Relationships for Grade ≥2 Creatinine Increase

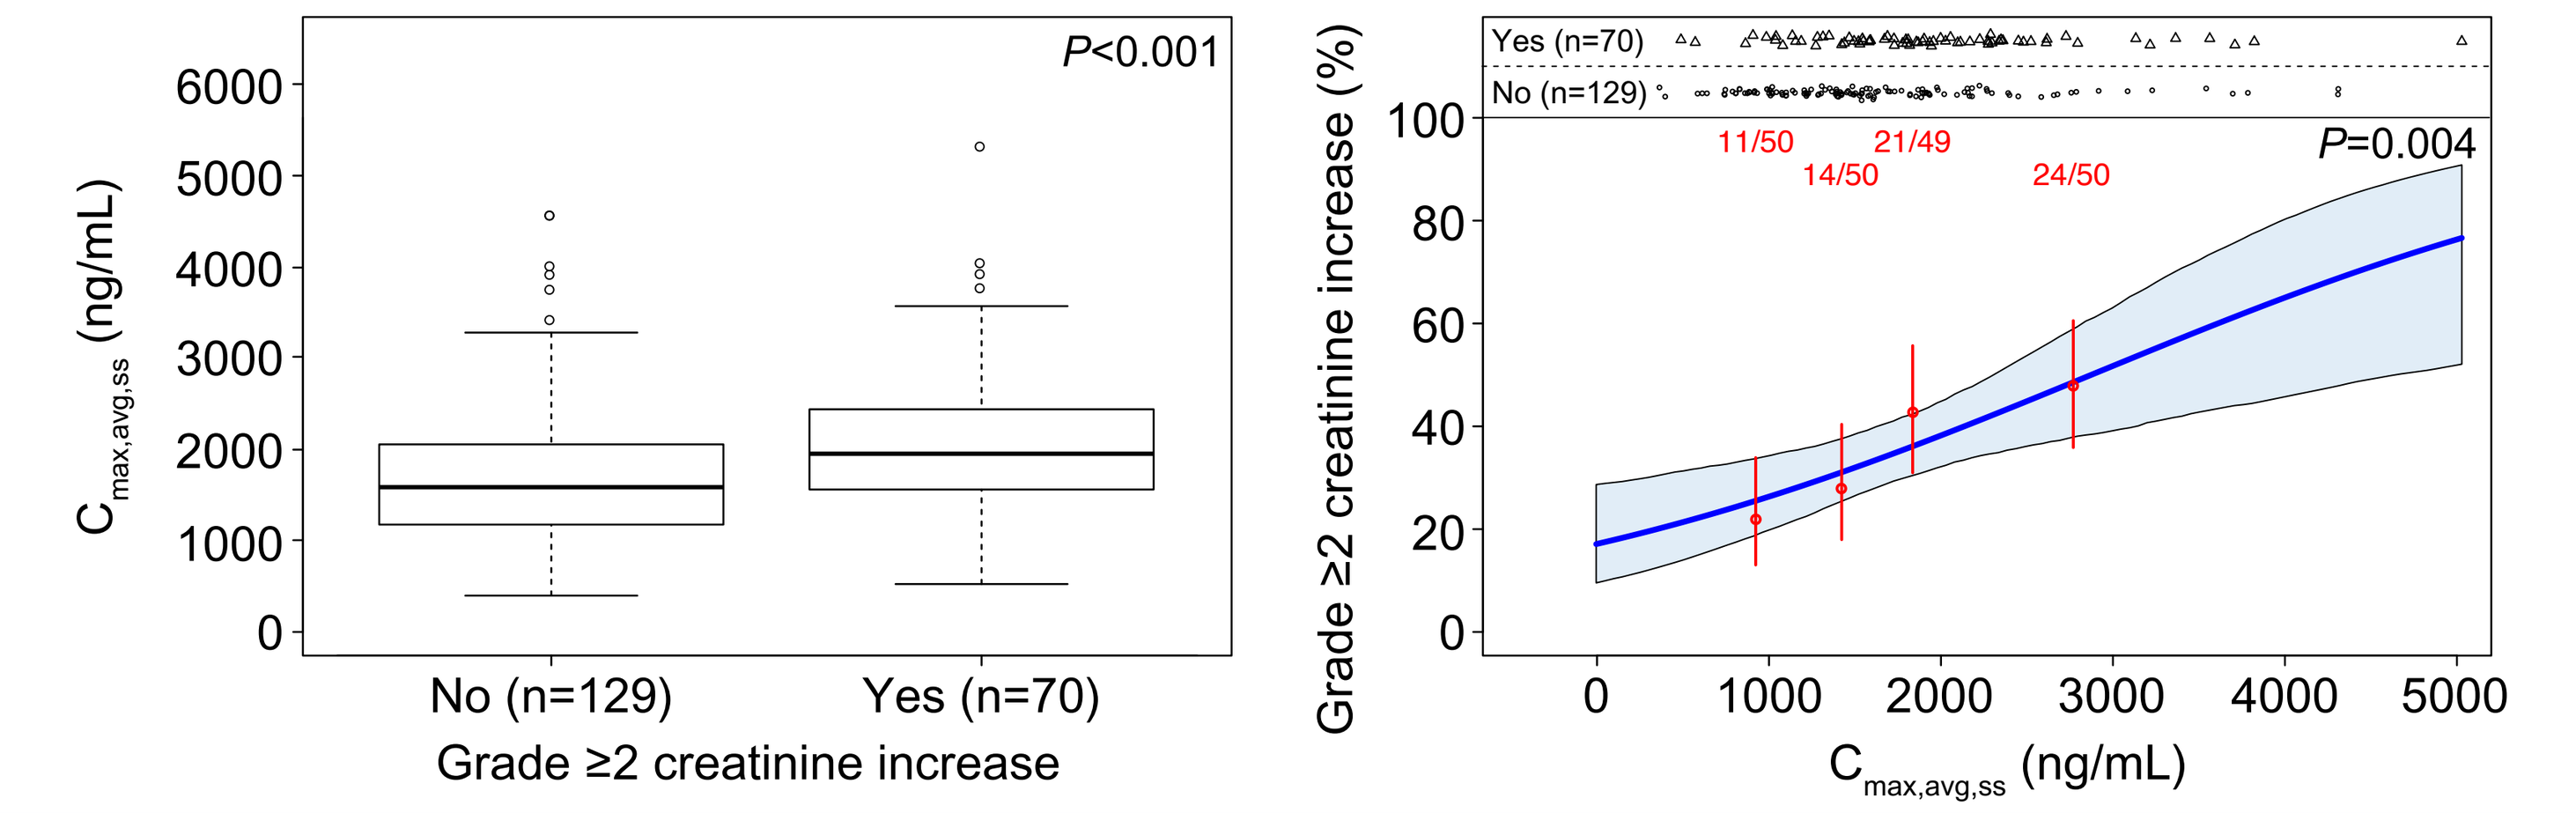
P<0.001
Cmax,avg,ss (ng/mL)
0 1000 2000 3000 4000 5000 6000
No (n=129)
Yes (n=70)
Grade ≥2 creatinine increase
Yes (n=70)
No (n=129)
11/50 14/50 21/49 24/50
P=0.004
Grade ≥2 creatinine increase (%)
0 1000 2000 3000 4000 5000
Cmax,avg,ss (ng/mL)

Yes/no represents the number of patients with/without the event being evaluated. Box plots show the steady-state maximum concentration of rucaparib, stratified by grade ≥2 creatinine increase with *P* value per t test. Red points represent the mean exposure and event rates in patients stratified by exposure quartile. Vertical red bars represent the 5th to 95th percentile CIs of the event rate. Event numbers (patients with event/total patients) are displayed above each vertical bar. The solid blue line is the logistic regression model fit (with *P* value) for the incidence of grade ≥2 creatinine increase. The shaded blue region represents 5th to 95th percentile CIs of the predicted event rate.
$C_{max,avg,ss}$, steady-state maximum concentration normalised by actual dose for each patient up to the event of interest.

ESMO Congress | 19–21 September 2020 | Virtual Meeting