Abstract # 3059: Imaging of Solid Tumors Using 68Ga-FAP-2286

Thomas A. Hope, Rahul Raj Aggarwal, Brad Kline, Mallika Sachdev Dhawan, Robin Kate Kelley, Robert R. Flavell, Courtney Lawhn Heath, Yan Li, Sima P. Porten, Hope S. Rugo, Sue S. Yom, Robin Ippisch and Vadim S Koshkin

UCSF

Introduction

Fibroblast Activation Protein (FAP) is a transmembrane protein overexpressed on cancer associated fibroblasts (CAFs), and is abundantly present in many epithelial cancers, suggesting FAP is an attractive imaging and therapeutic target. FAP-2286 is a cyclic peptide that binds to FAP and is currently being evaluated as a radioligand therapy to treat patients with FAP-positive solid tumors. The role of 68Ga-FAP-2286 as a diagnostic agent is unknown. We present an interim analysis of the ability of 68Ga-FAP-2286 to detect metastatic disease across multiple cancer types.

Methods

This is a first in human Phase I/II study of 68Ga-FAP-2286 (NCT04621435) with a planned total enrollment of 80 patients across 3 cohorts: dosimetry cohort (n = 10), cohort with RECIST measurable disease (n = 40) and a cohort at risk for metastases without measurable disease (n = 30). By the cutoff date of February 12, 2022, 27 patients were enrolled (3 in cohort 1, 15 in cohort 2, and 9 in cohort 3). For each patient, the five largest lesions were included for analysis, and for each lesion, the maximum standardized uptake value (SUVmax) and the size (short axis for lymph nodes) from the 68Ga-FAP-2286 PET were documented. In patients who had an available FDG PET performed within 8 weeks of 68Ga-FAP-2286 PET, uptake on the two scans was compared.

Conclusion: 68Ga-FAP-2286 is a promising imaging agent across cancers, although its benefit is not seen equally. Bladder cancer had the highest absolute uptake and highest relative uptake compared to FDG PET; prostate cancer had the lowest uptake. Further work should be undertaken to define the settings where 68Ga-FAP-2286 PET may inform clinical decision making, and which patients may benefit from FAP-targeted radioligand therapy.

Case 1

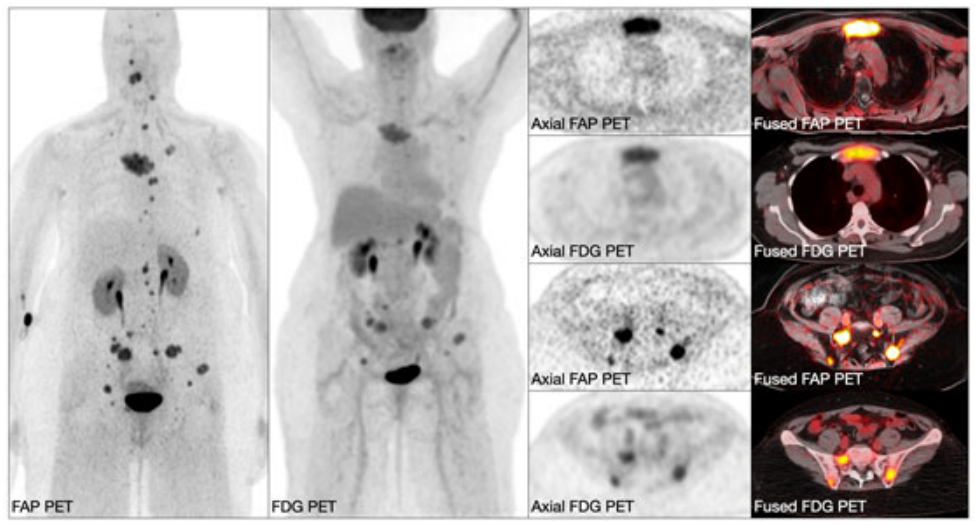
FAP PET
FDG PET
Axial FAP PET
Fused FAP PET
Axial FDG PET
Fused FDG PET
Axial FAP PET
Fused FAP PET
Axial FDG PET
Fused FDG PET

60-year-old woman with ductal adenocarcinoma of the breast. FAP PET demonstrated numerous osseous metastases, many of which were not visualized on the FDG PET performed one week earlier. On FDG PET, the dominant sternal metastasis had an SUVmax of 6.3, while FAP-2286 had an SUVmax of 23.2. In the pelvis, the left iliac metastasis had an SUVmax of 6.5 on FDG PET, while FAP PET had an SUVmax of 21.4.

Case 2

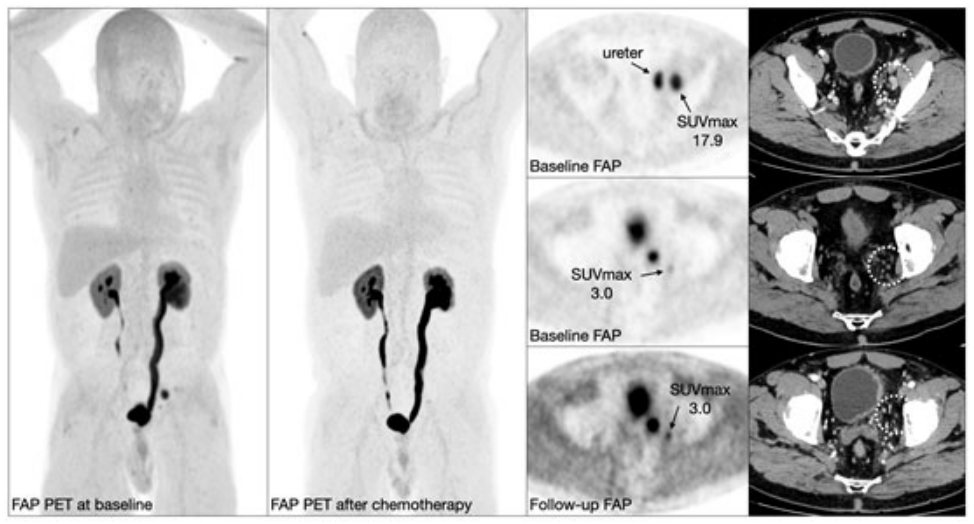
ureter
SUVmax
17.9
Baseline FAP
SUVmax
3.0
Baseline FAP
SUVmax
3.0
FAP PET at baseline
FAP PET after chemotherapy
Follow-up FAP

49-year-old man imaged at diagnosis of muscle invasive bladder cancer. Initial FAP-2286 PET demonstrated a 1.1 cm left pelvic side wall lymph node with an SUVmax of 17.9. Additionally, a 4 mm left pelvic side wall node was seen with an SUVmax of 3.0. After chemotherapy the nodes decreased in size and the larger node had residual uptake (SUVmax of 3.0). At time of cystectomy the residual node with FAP uptake was positive on histopathology.

Results

Of the 27 enrolled patients, 9 had bladder cancer, 5 sarcoma, 4 head and neck squamous cell cancer (HNSCC), 3 breast cancer, and 3 castration resistant prostate cancer (CRPC). Most patients (89%, 24/27) had tumors positive for uptake on 68Ga-FAP-2286 PET, including 30 lesions < 1.5 cm, and 17 less than 1.0 cm. 16 patients had a paired FDG PET. In these patients, the average SUVmax on 68Ga-FAP-2286 PET was 244% higher than on FDG PET. Only two patients had higher uptake on FDG PET than on 68Ga-FAP-2286 PET (HNSCC and DSRCT). The highest relative uptake was seen in 2 patients with breast cancer (both 3.4x higher on 68Ga-FAP-2286 PET – Case 1). The highest absolute uptake was seen in bladder cancer, with an average SUVmax of 16.6 (Case 2). The lowest uptake on 68Ga-FAP-2286 PET was CRPC with an average SUVmax of 7.0. Sarcoma had variable uptake with one patient having an SUVmax of 4.5 (Ewing's), while two patients had an SUVmax over 30 (both undifferentiated pleomorphic). Although sarcoma had high uptake on 68Ga-FAP-2286 PET, it was similar to FDG PET uptake across the 5 patients (ratio to FDG PET = 1.0).

This research was funded by an IIT from Clovis Oncology

University of California, San Francisco

thomas.hope@ucsf.edu

@thomashopemd