Subgroup Analysis of Rucaparib Versus Chemotherapy as Treatment for BRCA-mutated, Advanced, Relapsed Ovarian Carcinoma: Effect of Platinum Sensitivity in the Randomized, Phase 3 Study ARIEL4

Amit M. Oza, Alla Lisyanskaya, Alexander Fedenko, Mikhail Dvorkin, Andreia Cristina de Melo, Yaroslav Shparyk, Irina Rakhmatullina, Igor Bondarenko, Nicoletta Colombo, Valentyn Svintsitskiy, David Cibula, Róbert Póka, Ana Oaknin, Tamar Safra, Beata Mackowiak-Matejczyk, Ling Ma, Karen McLachlan, Sandra Goble, Rebecca Kristeleit

ASCO Annual Meeting | June 4–8, 2021 | Virtual Meeting
Abstract 5517

Supplementary Material

Supplementary Figure. TEAEs per 100 Patient-Years of Treatment in the ARIEL4 Safety Population

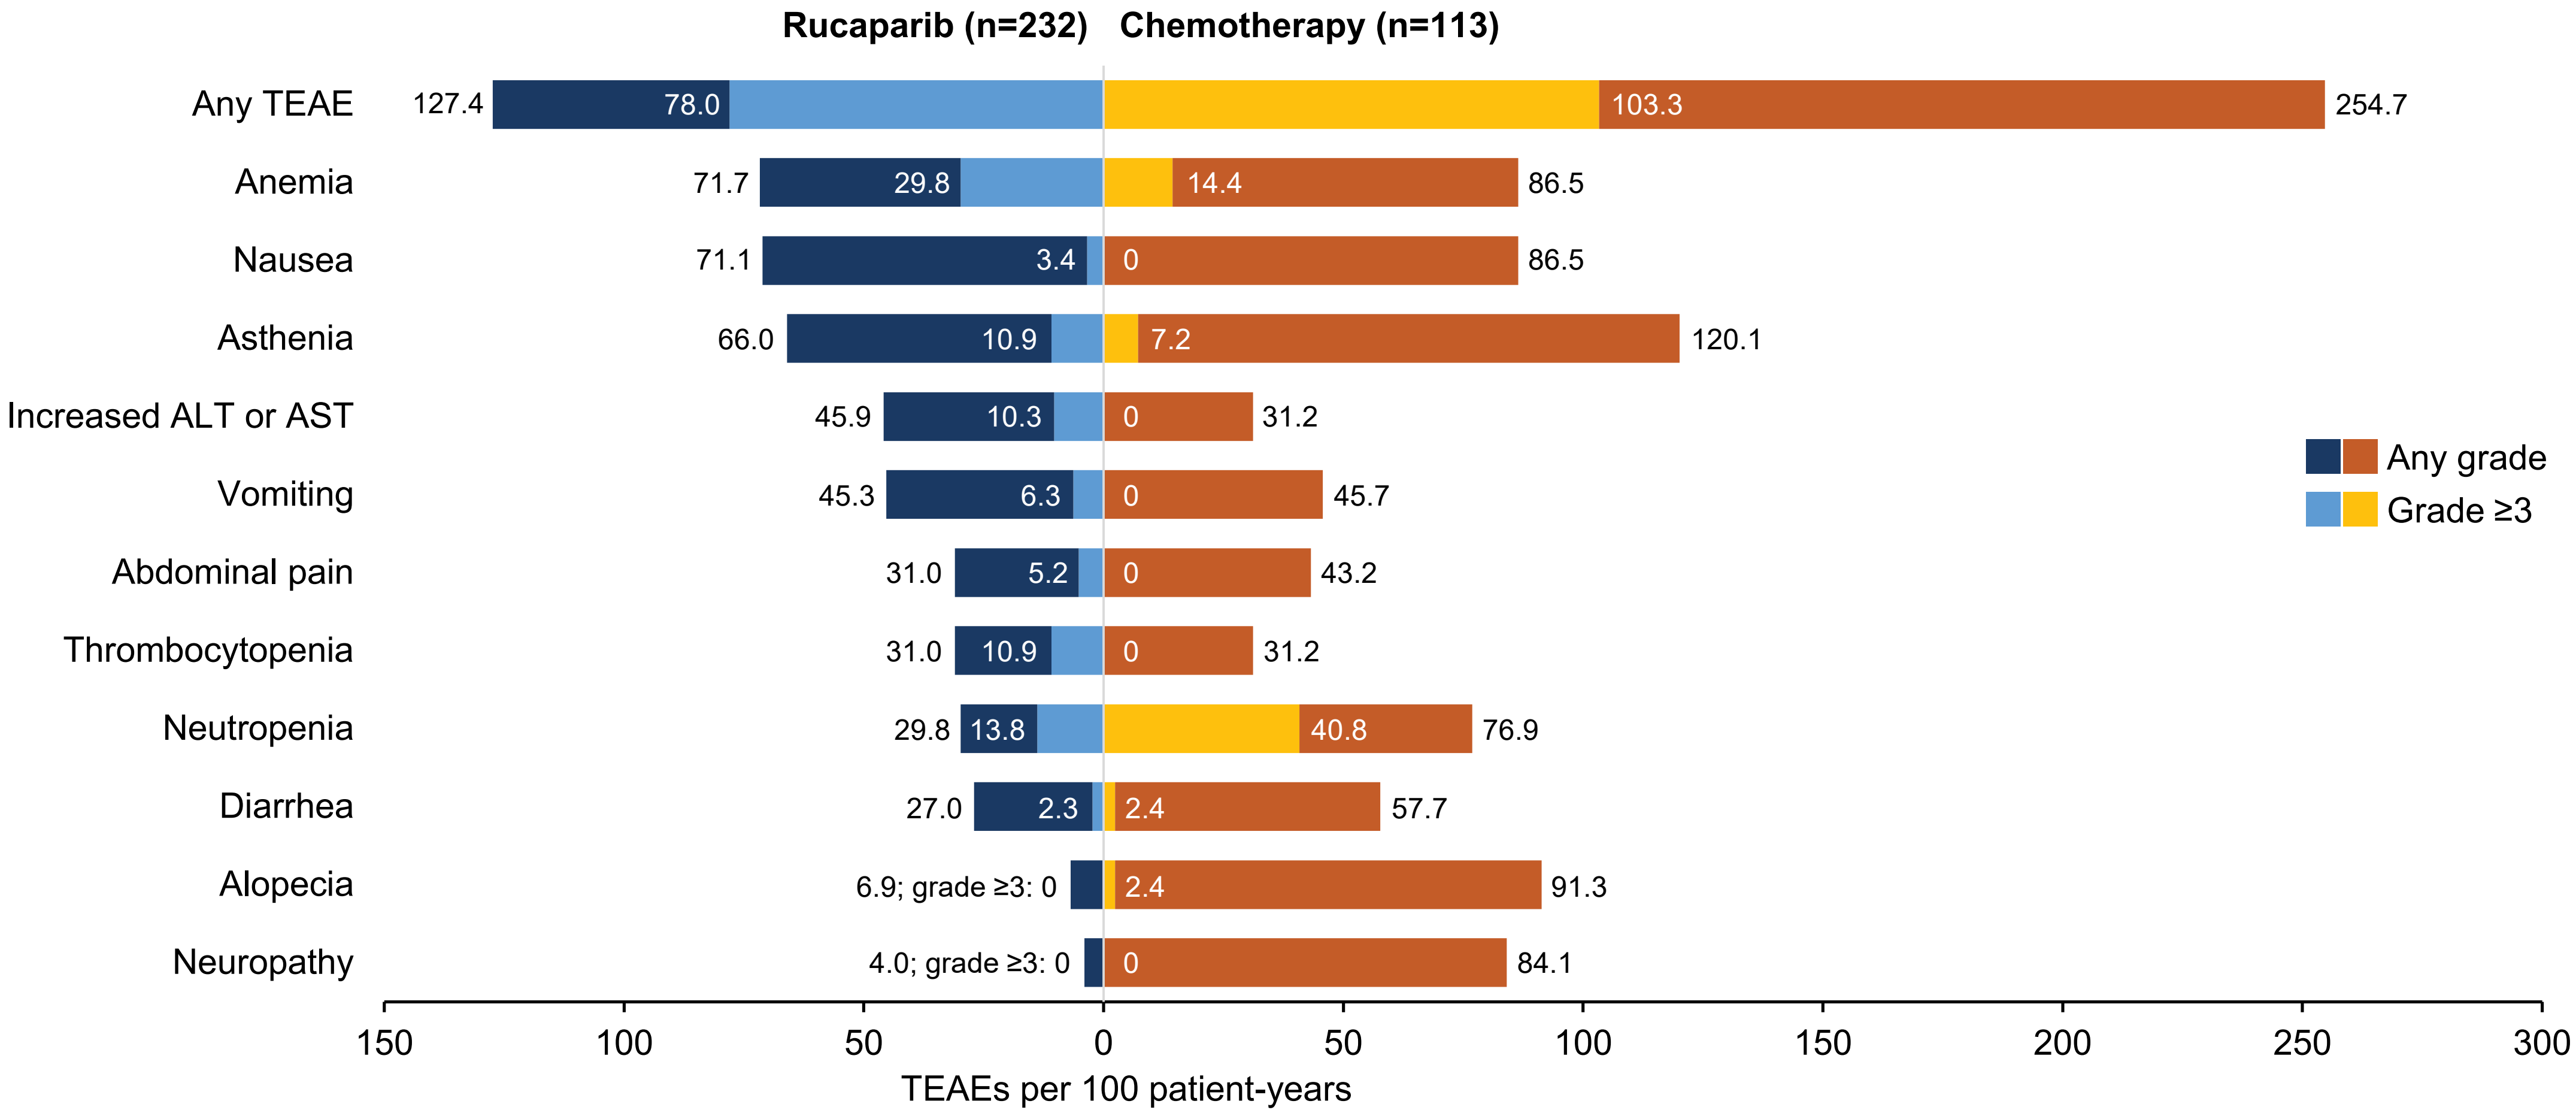
Rucaparib (n=232)
Chemotherapy (n=113)
Any TEAE 127.4 78.0 103.3 254.7
Anemia 71.7 29.8 14.4 86.5
Nausea 71.1 3.4 0 86.5
Asthenia 66.0 10.9 7.2 120.1
Increased ALT or AST 45.9 10.3 0 31.2
Vomiting 45.3 6.3 0 45.7
Abdominal pain 31.0 5.2 0 43.2
Thrombocytopenia 31.0 10.9 0 31.2
Neutropenia 29.8 13.8 40.8 76.9
Diarrhea 27.0 2.3 2.4 57.7
Alopecia 6.9; grade ≥3: 0 2.4 91.3
Neuropathy 4.0; grade ≥3: 0 0 84.1
Any grade
Grade ≥3
150 100 50 0 50 100 150 200 250 300
TEAEs per 100 patient-years

Most common TEAEs of any grade (≥20% in the overall safety population for either treatment group) are listed. Data are sorted based on decreasing rates of any-grade TEAEs per 100 patient-years of treatment in the rucaparib group. Asthenia includes fatigue; anemia includes decreased hemoglobin; thrombocytopenia includes decreased platelet count; neuropathy includes neurotoxicity, paraesthesia, peripheral neuropathy, peripheral motor neuropathy, peripheral sensory neuropathy, and polyneuropathy.

ALT, alanine aminotransferase; AST, aspartate aminotransferase; TEAE, treatment-emergent adverse event.