Abstract 5517

Subgroup Analysis of Rucaparib Versus Chemotherapy as Treatment for BRCA-mutated, Advanced, Relapsed Ovarian Carcinoma: Effect of Platinum Sensitivity in the Randomized, Phase 3 Study ARIEL4

Amit M. Oza,[1] Alla Lisyanskaya,[2] Alexander Fedenko,[3] Mikhail Dvorkin,[4] Andreia Cristina de Melo,[5] Yaroslav Shparyk,[6] Irina Rakhmatullina,[7] Igor Bondarenko,[8] Nicoletta Colombo,[9] Valentyn Svintsitskiy,[10] David Cibula,[11] Róbert Póka,[12] Ana Oaknin,[13] Tamar Safra,[14] Beata Mackowiak-Matejczyk,[15] Ling Ma,[16] Karen McLachlan,[17] Sandra Goble,[17] Rebecca Kristeleit[18]

[1]Princess Margaret Cancer Centre, University Health Network, Toronto, Canada; [2]Saint Petersburg City Oncological Dispensary, Russia*; [3]N.N. Blokhin Russian Cancer Research Center, Moscow, Russia†; [4]Omsk Region Clinical Oncologic Dispensary, Russia; [5]Instituto Nacional de Câncer, Hospital do Câncer II, Rio de Janeiro, Brazil; [6]Lviv Regional Oncology Dispensary, Ukraine; [7]Republic Clinical Oncology Dispensary of the Ministry of Healthcare of Republic of Bashkortostan, Ufa, Russia; [8]Dnipropetrovsk Medical Academy, Dnipro, Ukraine; [9]University of Milan-Bicocca and European Institute of Oncology (IEO) IRCCS, Italy; [10]National Cancer Institute of the Ministry of Health of Ukraine, Kyiv, Ukraine; [11]First Faculty of Medicine, Charles University and General University Hospital in Prague, Czech Republic; [12]Clinical Center, University of Debrecen, Hungary; [13]Vall d'Hebron Institute of Oncology (VHIO), Hospital Universitari Vall d'Hebron, Vall d'Hebron Barcelona Hospital Campus, Spain; [14]Sourasky Medical Center, Tel Aviv, Israel; [15]Bialostockie Centrum Onkologii im. Marii Sklodowskiej-Curie, Białystok, Poland; [16]Rocky Mountain Cancer Centers, Lakewood, CO, USA; [17]Clovis Oncology, Inc., Boulder, CO, USA; [18]University College London (UCL) Cancer Institute and UCL Hospitals, London, UK‡

*Affiliation where the work was conducted; current affiliation: Almazov National Medical Research Center, Saint Petersburg, Russia; †Affiliation where the work was conducted; current affiliation: P.A. Herzen Cancer Research Institute, Moscow, Russia; ‡Affiliation where the work was conducted; current affiliation: Guy's and St Thomas' NHS Foundation Trust, Great Maze Pond, London, UK

SUMMARY

- **Rucaparib-treated patients had comparable or longer progression-free survival vs chemotherapy across all platinum status subgroups (platinum resistant, partially platinum sensitive, fully platinum sensitive)**
 - **Importantly, comparable efficacy was observed with rucaparib against platinum-based chemotherapy in patients with fully platinum-sensitive disease; rucaparib also showed response rates similar to those of platinum-based chemotherapy in this subgroup of patients, with a trend toward a longer duration of objective response**
- **Safety profiles for rucaparib and chemotherapy across the platinum status subgroups were consistent with known safety profiles of these agents**
- **These results suggest that rucaparib is an effective treatment option for heavily pretreated patients regardless of their sensitivity to platinum-based therapy**

Poster Download
Also available at: https://clovisoncology.com/files/ASCO2021_Oza_Poster.pdf
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.
Corresponding author: *Amit M. Oza*; *amit.oza@uhn.ca*.

Poster Slides
Also available at: https://clovisoncology.com/files/ASCO2021_Oza_Slides.pdf
Please scan this QR code or visit the website to view the poster as slides.

Supplementary Material
Also available at: https://clovisoncology.com/files/ASCO2021_Oza_Poster_Supplement.pdf
Please scan this QR code or visit the website to view additional materials for this poster.

REFERENCES

1. Rubraca (rucaparib) tablets [prescribing information]. Boulder, CO: Clovis Oncology, Inc.; 2020.
2. Rubraca (rucaparib) tablets [summary of product characteristics]. Swords, Ireland: Clovis Oncology Ireland Ltd.; 2019.
3. Oza et al. *Gynecol Oncol*. 2017;147:267-75.
4. Kristeleit et al. *Int J Gynecol Cancer*. 2019;29:1396-404.
5. Kristeleit et al. Presented at 2021 Society of Gynecologic Oncology (SGO) Annual Meeting 2021; March 19, 2021; Virtual Meeting. Abst ID 11479.

ACKNOWLEDGMENTS

This study was funded by Clovis Oncology, Inc. Medical writing and editorial support funded by Clovis Oncology, Inc., were provided by Shelly Lim and Stephen Bublitz of Ashfield MedComms, an Ashfield Health company.

ASCO Annual Meeting | June 4–8, 2021 | Virtual Meeting

INTRODUCTION

- Rucaparib, a poly(ADP-ribose) polymerase (PARP) inhibitor, is approved in the United States and Europe as monotherapy treatment of patients with *BRCA1* or *BRCA2* (BRCA)-mutated, epithelial ovarian, fallopian tube, or primary peritoneal cancer who have received 2 or more prior lines of platinum-based chemotherapy or (in Europe) who are unable to tolerate further platinum-based chemotherapy[1,2]
 - Approval was based on data from 2 phase 1/2 studies[3,4]
- In the confirmatory phase 3 ARIEL4 study (NCT02855944), patients with advanced, relapsed ovarian carcinoma harboring a deleterious BRCA mutation and platinum-resistant, partially platinum-sensitive, or fully platinum-sensitive disease were randomized to receive either rucaparib or standard-of-care platinum- or nonplatinum-based chemotherapy
 - In the efficacy population, rucaparib significantly improved the primary endpoint of progression-free survival (PFS) vs chemotherapy[5] (**Figure 1**)
- Here we present results from a prespecified exploratory analysis investigating the effect of platinum sensitivity on the efficacy and safety of rucaparib vs chemotherapy in ARIEL4 patients
 - Analyses of other exploratory subgroups on the efficacy of rucaparib vs chemotherapy are also presented

Figure 1. Primary Endpoint – Investigator-assessed PFS: Efficacy Population[5]

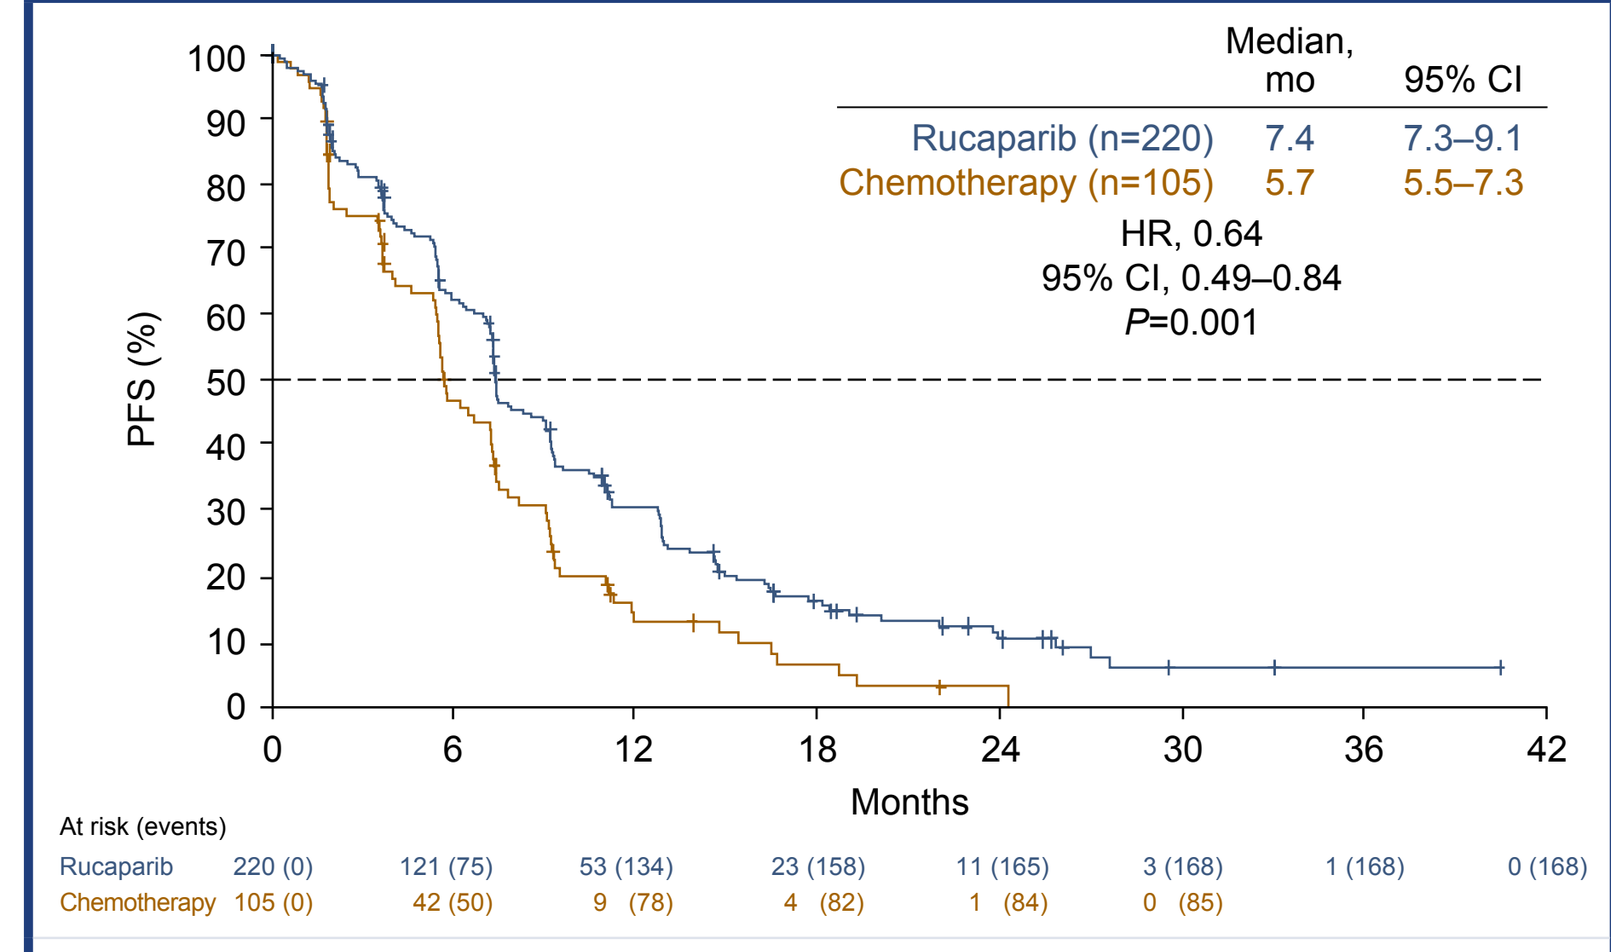

Median, mo
95% CI
Rucaparib (n=220) 7.4 7.3–9.1
Chemotherapy (n=105) 5.7 5.5–7.3
HR, 0.64
95% CI, 0.49–0.84
P=0.001
PFS (%)
Months
At risk (events)
Rucaparib 220 (0) 121 (75) 53 (134) 23 (158) 11 (165) 3 (168) 1 (168) 0 (168)
Chemotherapy 105 (0) 42 (50) 9 (78) 4 (82) 1 (84) 0 (85)

Visit cutoff: September 30, 2020. HR and associated *P* value calculated using a stratified Cox proportional hazards model.
HR, hazard ratio; PFS, progression-free survival.

METHODS

- Eligible patients were randomized 2:1 to oral rucaparib 600 mg twice daily or chemotherapy and stratified according to platinum-sensitivity status based on most recent platinum-based chemotherapy (**Figure 2**)
- Following radiologic disease progression per Response Evaluation Criteria In Solid Tumors version 1.1 (RECIST), patients could crossover from chemotherapy to rucaparib
- Disease/tumor assessments were performed every 8 weeks until 18 months on study, then every 16 weeks thereafter until disease progression or death
- The efficacy population consisted of all randomized patients with a confirmed BRCA mutation, excluding those with a BRCA reversion mutation
 - Among all randomized patients, BRCA reversion mutations were identified in 9.5% (n=17) of patients with platinum-resistant disease, 6.3% (n=6) of patients with partially platinum-sensitive disease, and in no patients with fully platinum-sensitive disease

Figure 2. ARIEL4 Study Population

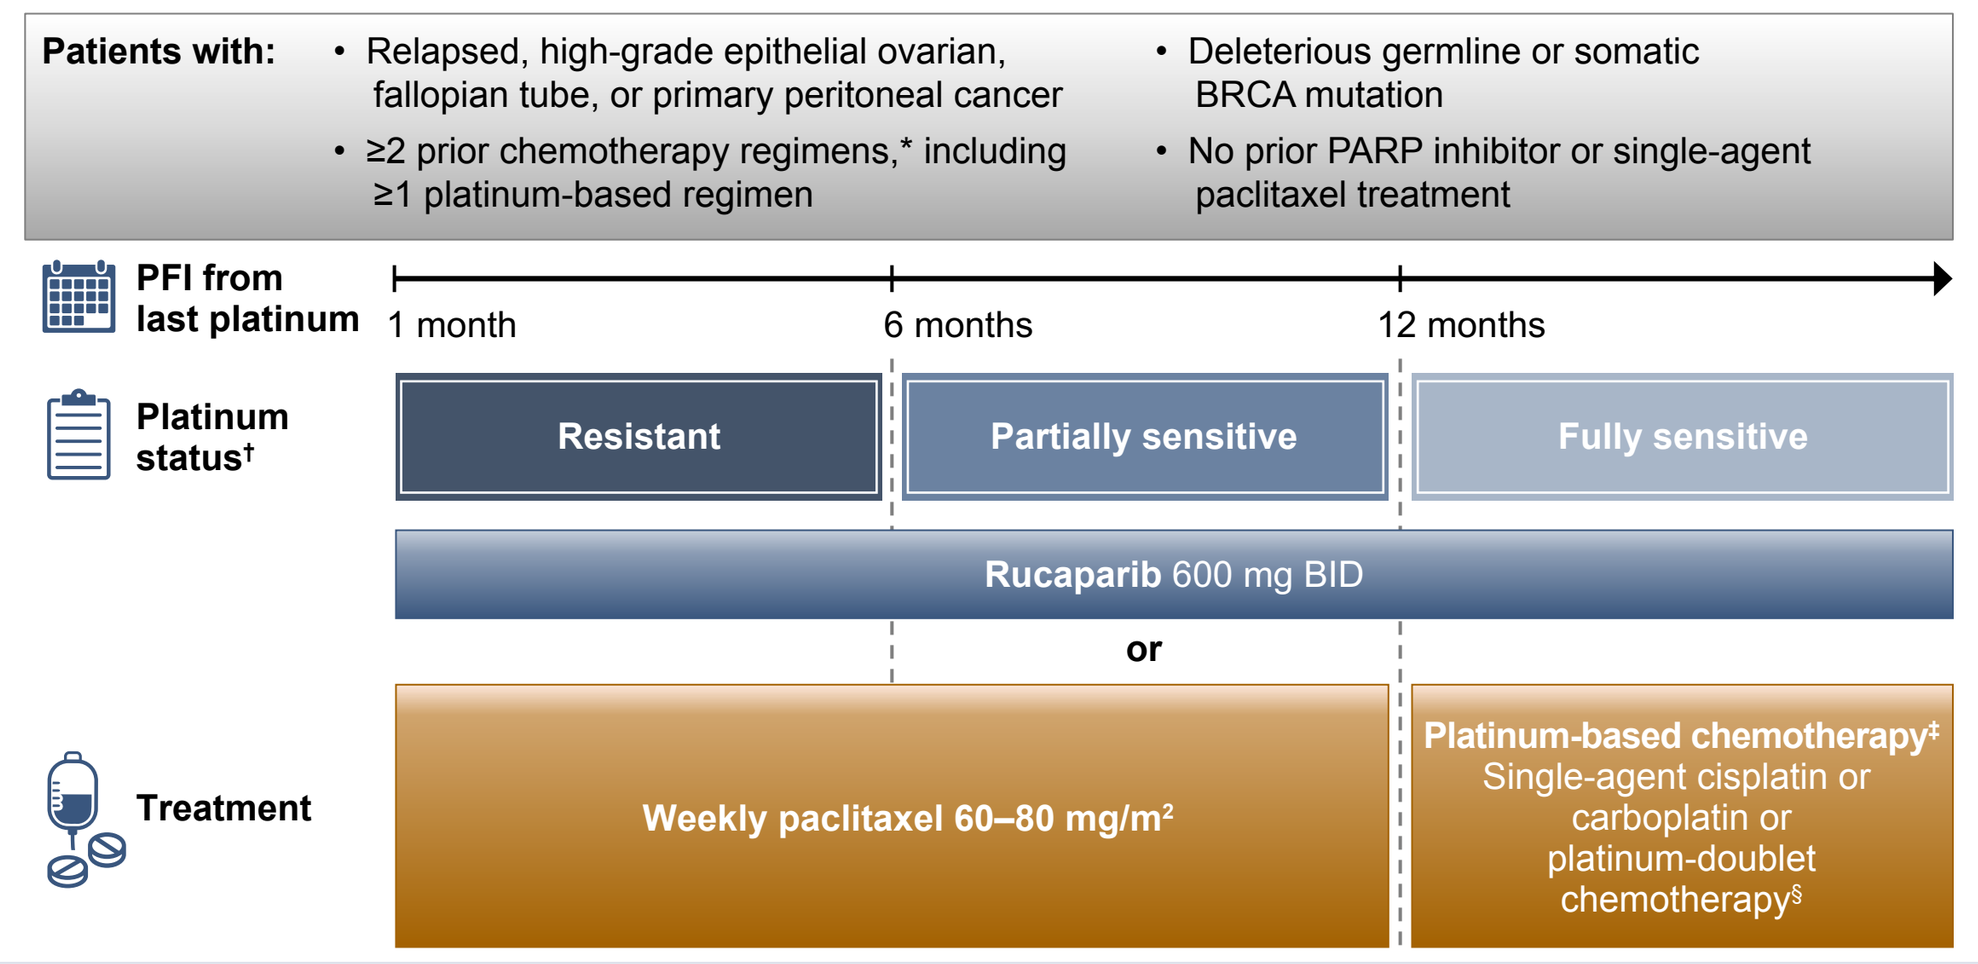

Patients with:
• Relapsed, high-grade epithelial ovarian, fallopian tube, or primary peritoneal cancer
• ≥2 prior chemotherapy regimens,* including ≥1 platinum-based regimen
• Deleterious germline or somatic BRCA mutation
• No prior PARP inhibitor or single-agent paclitaxel treatment
PFI from last platinum
1 month
6 months
12 months
Platinum status†
Resistant
Partially sensitive
Fully sensitive
Rucaparib 600 mg BID
or
Treatment
Weekly paclitaxel 60–80 mg/m²
Platinum-based chemotherapy‡
Single-agent cisplatin or carboplatin or platinum-doublet chemotherapy§

*With treatment-free interval ≥6 months following first chemotherapy received. †Randomization stratification factor. ‡At investigator's discretion. §Carboplatin and paclitaxel, carboplatin and gemcitabine, or cisplatin and gemcitabine.
BRCA, *BRCA1* or *BRCA2*; PARP, poly(ADP-ribose) polymerase; PFI, progression-free interval.

- Efficacy endpoints were prespecified and explored in subgroups of the efficacy population based on platinum-sensitivity status
 - Investigator-assessed PFS was evaluated per RECIST for rucaparib vs chemotherapy
 - Investigator-assessed objective response rate was evaluated per RECIST for rucaparib vs chemotherapy; response rates were also evaluated per RECIST and/or Gynecologic Cancer InterGroup (GCIG) cancer antigen-125 (CA-125) response criteria
 - The durations of confirmed complete or partial responses were assessed from the date of first response until first date of documented disease progression
- PFS was also explored in subgroups of the efficacy population based on baseline patient, genomic, and disease characteristics
- PFS was analyzed using the stratified Cox proportional hazards methodology
 - The stratified HR was used to estimate the HR between the treatment groups
- The safety population consisted of all patients who received at least 1 dose of randomized treatment

RESULTS

- The visit cutoff date was September 30, 2020

Patient Demographics

- In the efficacy population (n=325), 161 (49.5%) patients had platinum-resistant disease, 90 (27.7%) had partially platinum-sensitive disease, and 74 (22.8%) had fully platinum-sensitive disease (**Table 1**)
 - Among patients in the chemotherapy group (n=105), 79 (75.2%) were assigned to receive weekly paclitaxel, 10 (9.5%) were to receive platinum monotherapy, and 16 (15.2%) were to receive platinum-doublet therapy

Table 1. Patient Demographics and Baseline Characteristics in the Efficacy Population

	Platinum resistant		Partially platinum sensitive		Fully platinum sensitive	
Characteristic	Rucaparib (n=110)	Chemotherapy (n=51)	Rucaparib (n=62)	Chemotherapy (n=28)	Rucaparib (n=48)	Chemotherapy (n=26)
Age, median (range), y	55.0 (38–81)	58.0 (42–85)	58.0 (42–74)	64.0 (45–73)	60.0 (41–75)	57.0 (38–77)
ECOG PS, n (%)						
0	58 (52.7)	33 (64.7)	31 (50.0)	15 (53.6)	30 (62.5)	18 (69.2)
1	52 (47.3)	18 (35.3)	31 (50.0)	13 (46.4)	18 (37.5)	8 (30.8)
BRCA germline status, n (%)						
Germline	94 (85.5)	41 (80.4)	53 (85.5)	26 (92.9)	40 (83.3)	21 (80.8)
Somatic	16 (14.5)	9 (17.6)	9 (14.5)	2 (7.1)	8 (16.7)	5 (19.2)
Not available	0	1 (2.0)	0	0	0	0
Prior chemotherapy regimens, n (%)						
2	60 (54.5)	26 (51.0)	36 (58.1)	17 (60.7)	34 (70.8)	19 (73.1)
3–5	43 (39.1)	23 (45.1)	26 (41.9)	11 (39.3)	11 (22.9)	7 (26.9)
6–9	7 (6.4)	2 (3.9)	0	0	3 (6.3)	0
Prior platinum-based regimens, n (%)						
1	2 (1.8)	1 (2.0)	9 (14.5)	4 (14.3)	1 (2.1)	1 (3.8)
2	75 (68.2)	32 (62.7)	37 (59.7)	16 (57.1)	36 (75.0)	20 (76.9)
3–6	33 (30.0)	18 (35.3)	16 (25.8)	8 (28.6)	11 (22.9)	5 (19.2)
Prior nonplatinum regimens immediately before randomization, n (%)						
0	87 (79.1)	40 (78.4)	41 (66.1)	21 (75.0)	42 (87.5)	23 (88.5)
1–4	23 (20.9)	11 (21.6)	21 (33.9)	7 (25.0)	6 (12.5)	3 (11.5)

BRCA, *BRCA1* or *BRCA2*; ECOG PS, Eastern Cooperative Oncology Group performance status.

Efficacy

- PFS in each of the platinum status subgroups are shown in **Figure 3**

Figure 3. Investigator-Assessed PFS in the Efficacy Population

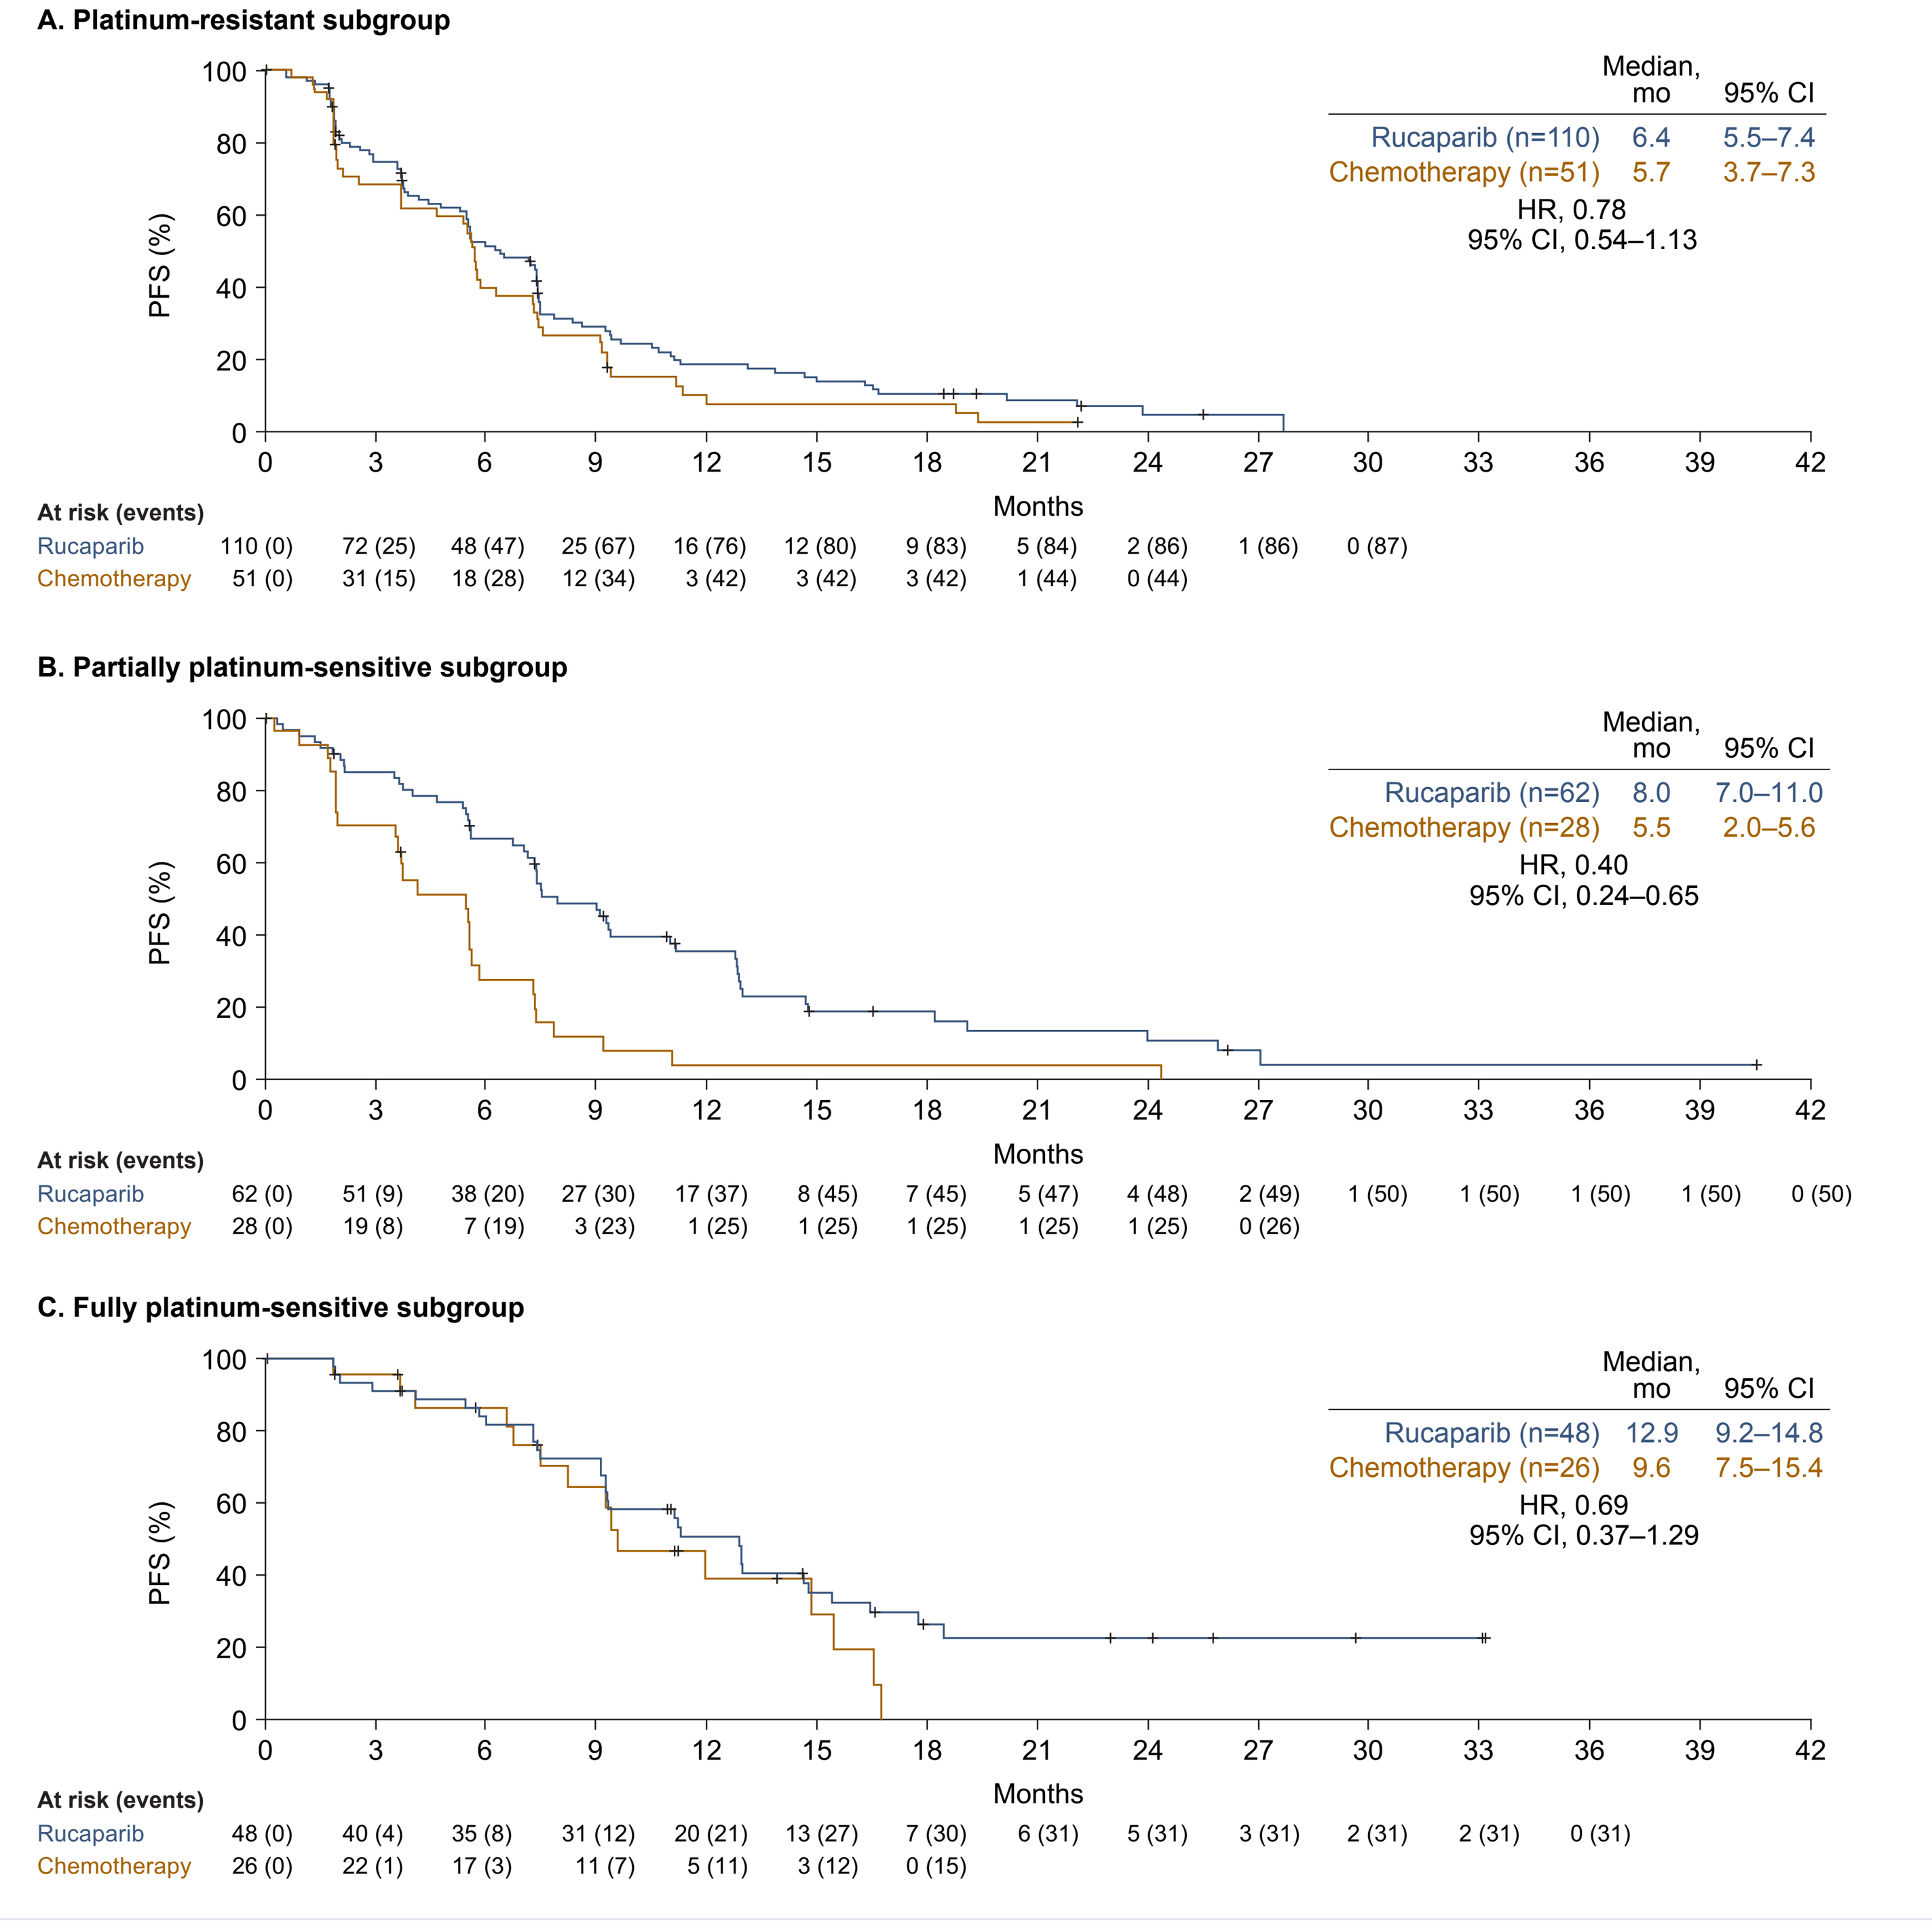

A. Platinum-resistant subgroup
Median, mo
95% CI
Rucaparib (n=110) 6.4 5.5–7.4
Chemotherapy (n=51) 5.7 3.7–7.3
HR, 0.78
95% CI, 0.54–1.13
PFS (%)
Months
At risk (events)
Rucaparib 110 (0) 72 (25) 48 (47) 25 (67) 16 (76) 12 (80) 9 (83) 5 (84) 2 (86) 1 (86) 0 (87)
Chemotherapy 51 (0) 31 (15) 18 (28) 12 (34) 3 (42) 3 (42) 3 (42) 1 (44) 0 (44)
B. Partially platinum-sensitive subgroup
Median, mo
95% CI
Rucaparib (n=62) 8.0 7.0–11.0
Chemotherapy (n=28) 5.5 2.0–5.6
HR, 0.40
95% CI, 0.24–0.65
At risk (events)
Rucaparib 62 (0) 51 (9) 38 (20) 27 (30) 17 (37) 8 (45) 7 (45) 5 (47) 4 (48) 2 (49) 1 (50) 1 (50) 1 (50) 1 (50) 0 (50)
Chemotherapy 28 (0) 19 (8) 7 (19) 3 (23) 1 (25) 1 (25) 1 (25) 1 (25) 1 (25) 0 (26)
C. Fully platinum-sensitive subgroup
Median, mo
95% CI
Rucaparib (n=48) 12.9 9.2–14.8
Chemotherapy (n=26) 9.6 7.5–15.4
HR, 0.69
95% CI, 0.37–1.29
At risk (events)
Rucaparib 48 (0) 40 (4) 35 (8) 31 (12) 20 (21) 13 (27) 7 (30) 6 (31) 5 (31) 3 (31) 2 (31) 2 (31) 0 (31)
Chemotherapy 26 (0) 22 (1) 17 (3) 11 (7) 5 (11) 3 (12) 0 (15)

HR and associated *P* value were calculated using a stratified Cox proportional hazards model.
HR, hazard ratio; PFS, progression-free survival.

- Analysis of other subgroups based on baseline characteristics showed that there was a trend for improved PFS in all subgroups treated with rucaparib compared with those treated with chemotherapy (**Figure 4**)

Figure 4. Investigator-Assessed PFS in Subgroups of the Efficacy Population

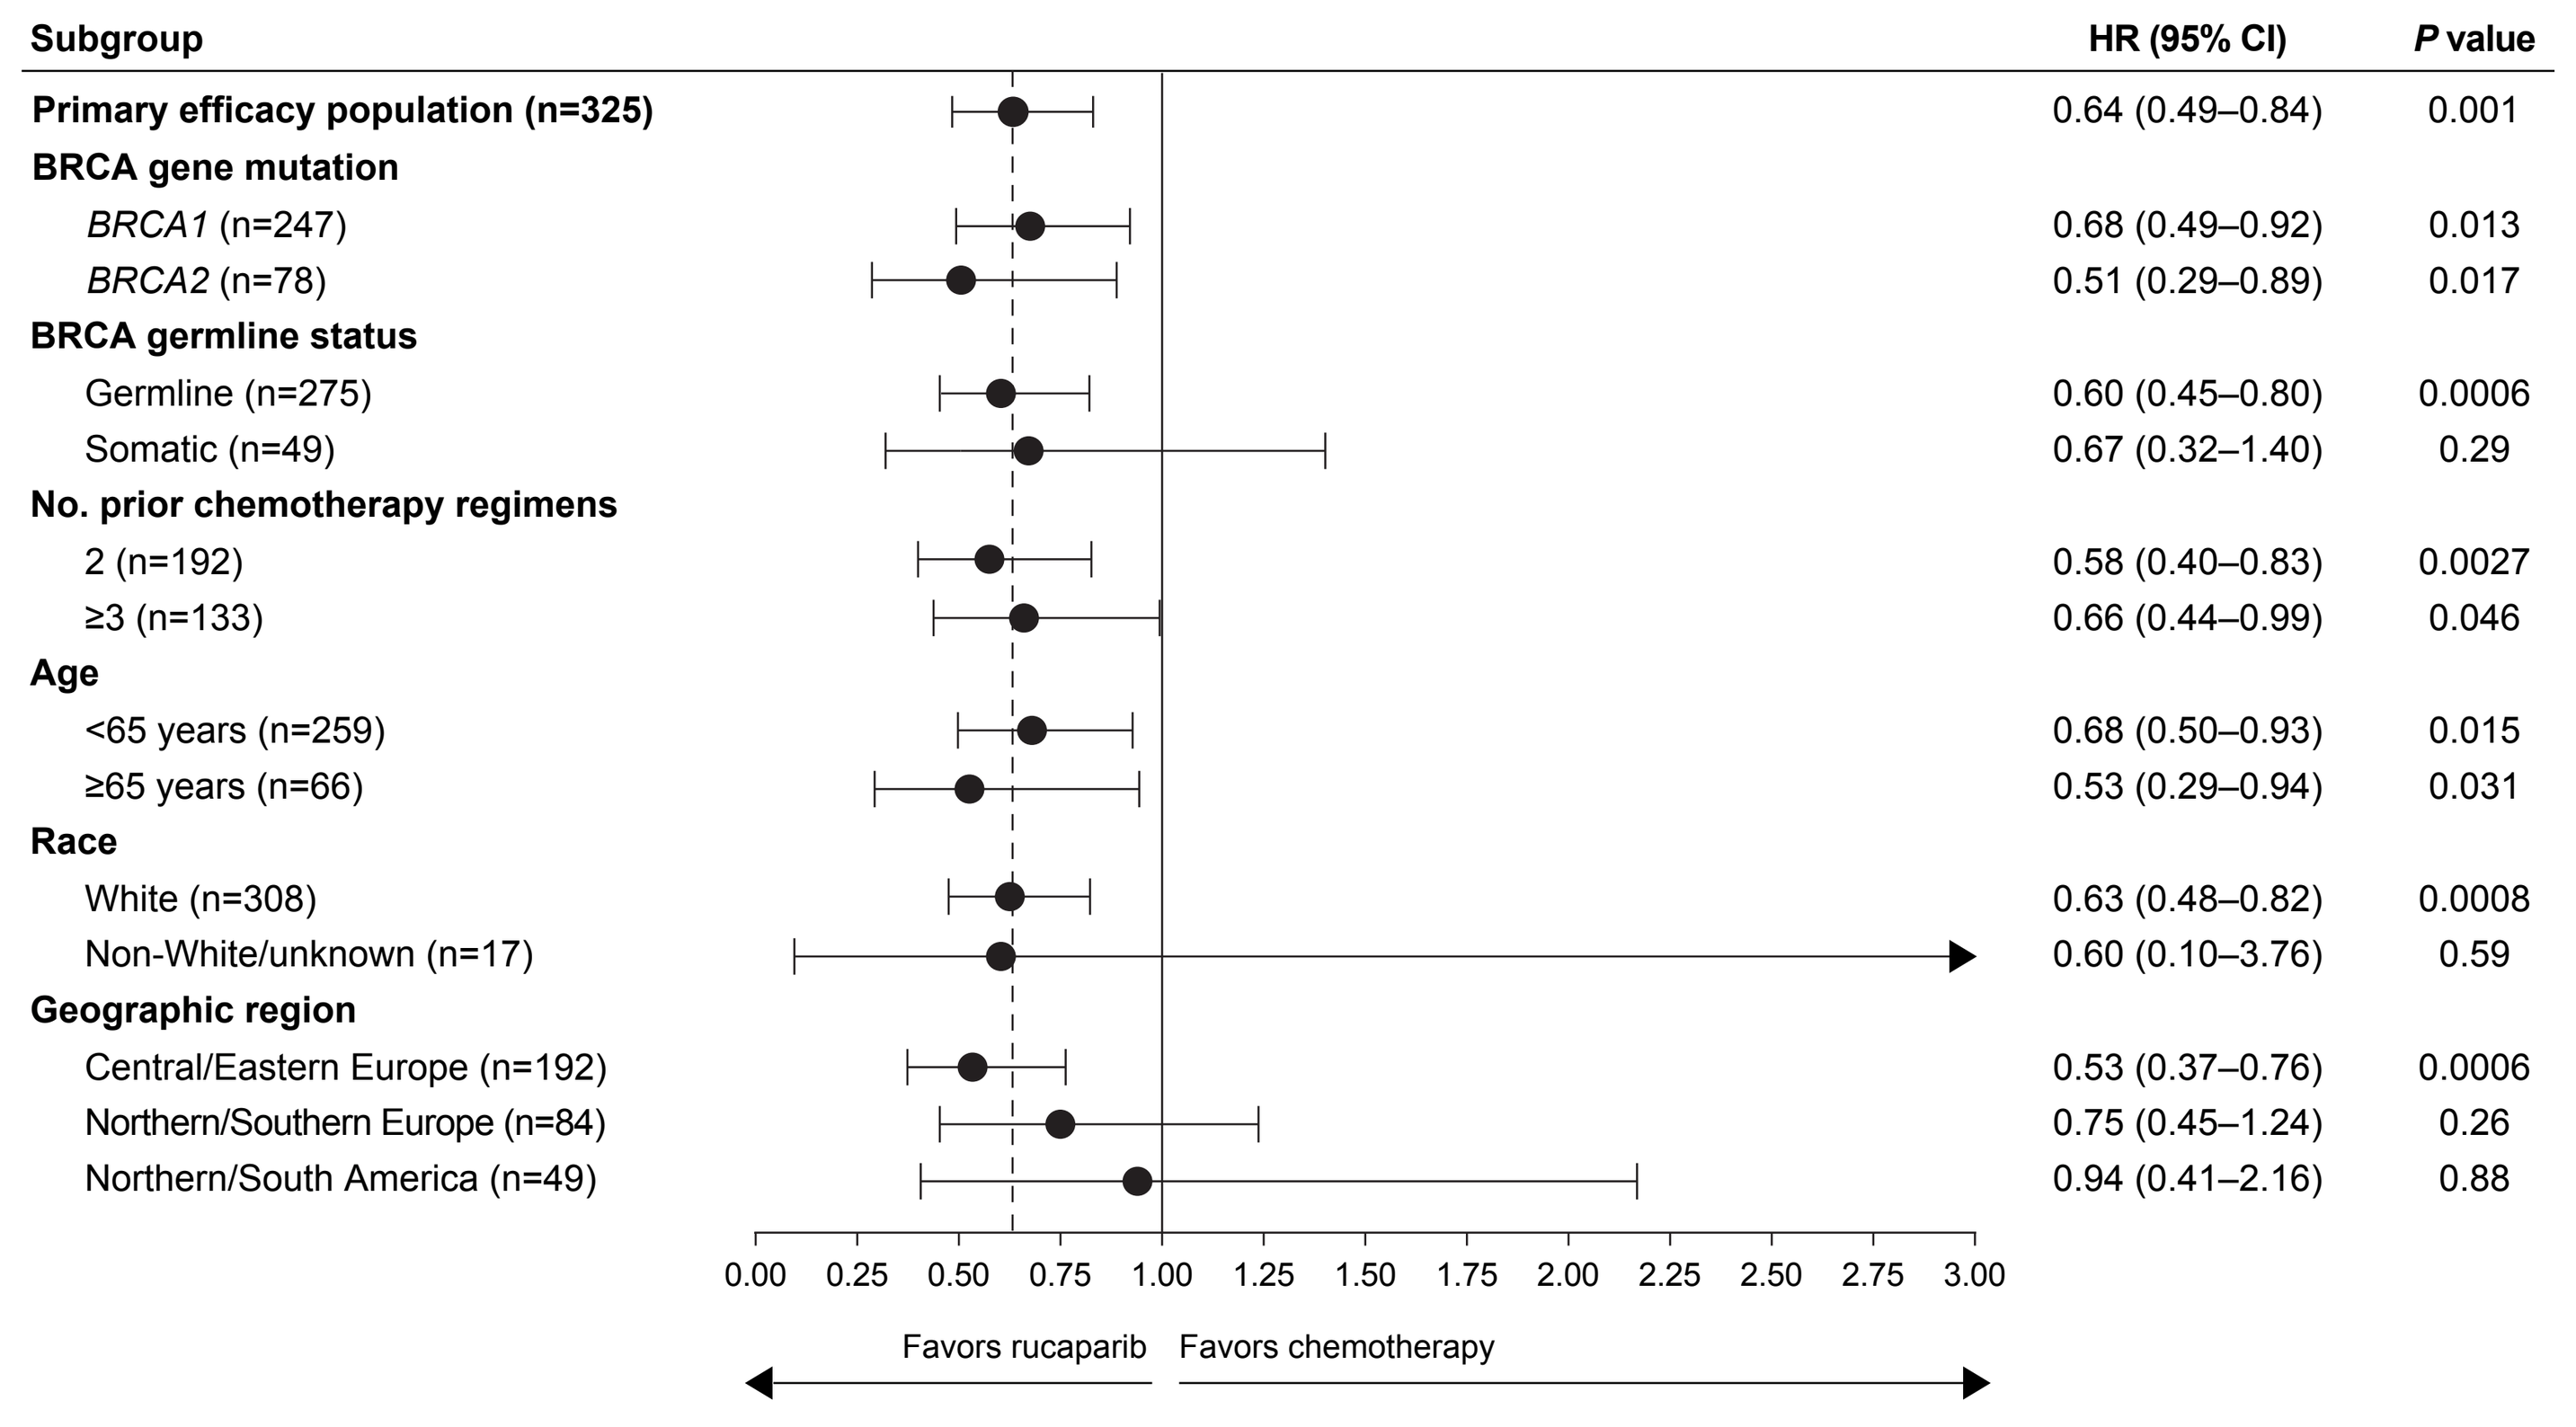

Subgroup
HR (95% CI)
P value
Primary efficacy population (n=325) 0.64 (0.49–0.84) 0.001
BRCA gene mutation
BRCA1 (n=247) 0.68 (0.49–0.92) 0.013
BRCA2 (n=78) 0.51 (0.29–0.89) 0.017
BRCA germline status
Germline (n=275) 0.60 (0.45–0.80) 0.0006
Somatic (n=49) 0.67 (0.32–1.40) 0.29
No. prior chemotherapy regimens
2 (n=192) 0.58 (0.40–0.83) 0.0027
≥3 (n=133) 0.66 (0.44–0.99) 0.046
Age
<65 years (n=259) 0.68 (0.50–0.93) 0.015
≥65 years (n=66) 0.53 (0.29–0.94) 0.031
Race
White (n=308) 0.63 (0.48–0.82) 0.0008
Non-White/unknown (n=17) 0.60 (0.10–3.76) 0.59
Geographic region
Central/Eastern Europe (n=192) 0.53 (0.37–0.76) 0.0006
Northern/Southern Europe (n=84) 0.75 (0.45–1.24) 0.26
Northern/South America (n=49) 0.94 (0.41–2.16) 0.88
Favors rucaparib
Favors chemotherapy

HRs estimated with a Cox proportional hazards model. BRCA germline status was not available for 1 patient.
BRCA, *BRCA1* or *BRCA2*; HR, hazard ratio; PFS, progression-free survival.

- Confirmed response rates in the platinum status subgroups are shown in **Table 2**
- Rucaparib-treated patients had a longer median duration of objective response vs chemotherapy-treated patients across platinum status subgroups: platinum resistant, 9.4 vs 7.2 months (HR 0.60, 95% CI 0.28–1.30); partially platinum sensitive, 9.2 vs 3.7 months (HR 0.49, 95% CI 0.18–1.28); and fully platinum sensitive, 10.8 vs 7.6 months (HR 0.62, 95% CI 0.26–1.51)

Table 2. Response Rates in Patients in the Platinum Status Subgroups

	Platinum resistant		Partially platinum sensitive		Fully platinum sensitive	
Characteristic	Rucaparib (n=110)	Chemotherapy (n=51)	Rucaparib (n=62)	Chemotherapy (n=28)	Rucaparib (n=48)	Chemotherapy (n=26)
Confirmed RECIST ORR, n/N (%) [95% CI][a]	**25/107 (23.4) [15.7–32.5]**	**13/48 (27.1) [15.3–41.8]**	**32/60 (53.3) [40.0–66.3]**	**5/25 (20.0) [6.8–40.7]**	**28/44 (63.6) [47.8–77.6]**	**13/23 (56.5) [34.5–76.8]**
Complete response	4/107 (3.7)	1/48 (2.1)	2/60 (3.3)	0/25 (0)	4/44 (9.1)	1/23 (4.3)
Partial response	21/107 (19.6)	12/48 (25.0)	30/60 (50.0)	5/25 (20.0)	24/44 (54.5)	12/23 (52.2)
Stable disease	48/107 (44.9)	20/48 (41.7)	19/60 (31.7)	13/25 (52.0)	10/44 (22.7)	5/23 (21.7)
Progressive disease	18/107 (16.8)	9/48 (18.8)	4/60 (6.7)	5/25 (20.0)	3/44 (6.8)	1/23 (4.3)
Not evaluable	16/107 (15.0)	6/48 (12.5)	5/60 (8.3)	2/25 (8.0)	3/44 (6.8)	4/23 (17.4)
Confirmed RECIST and/or CA-125 response, n/N (%) [95% CI][b]	**39/109 (35.8) [26.8–45.5]**	**16/49 (32.7) [19.9–47.5]**	**39/62 (62.9) [49.7–74.8]**	**9/27 (33.3) [16.5–54.0]**	**32/46 (69.6) [54.2–82.3]**	**19/25 (76.0) [54.9–90.6]**

[a]Patients with measurable disease at baseline. [b]Patients with measurable disease at baseline and/or evaluable by CA-125.
CA-125, cancer antigen 125; ORR, objective response rate; RECIST, Response Evaluation Criteria In Solid Tumors version 1.1.

Safety

- Across all platinum status subgroups, patients had longer median treatment duration with rucaparib vs chemotherapy: platinum resistant, 5.5 vs 4.4 months; partially platinum sensitive, 7.6 vs 4.6 months; and fully platinum sensitive, 12.0 vs 3.4 months
- An analysis of rates of the most common treatment-emergent adverse events adjusted for length of treatment exposure was performed for all platinum status subgroups (**Figure 5**) and for the overall safety population (**Supplementary Figure**)

Figure 5. TEAEs per 100 Patient-Years of Treatment by Platinum Status

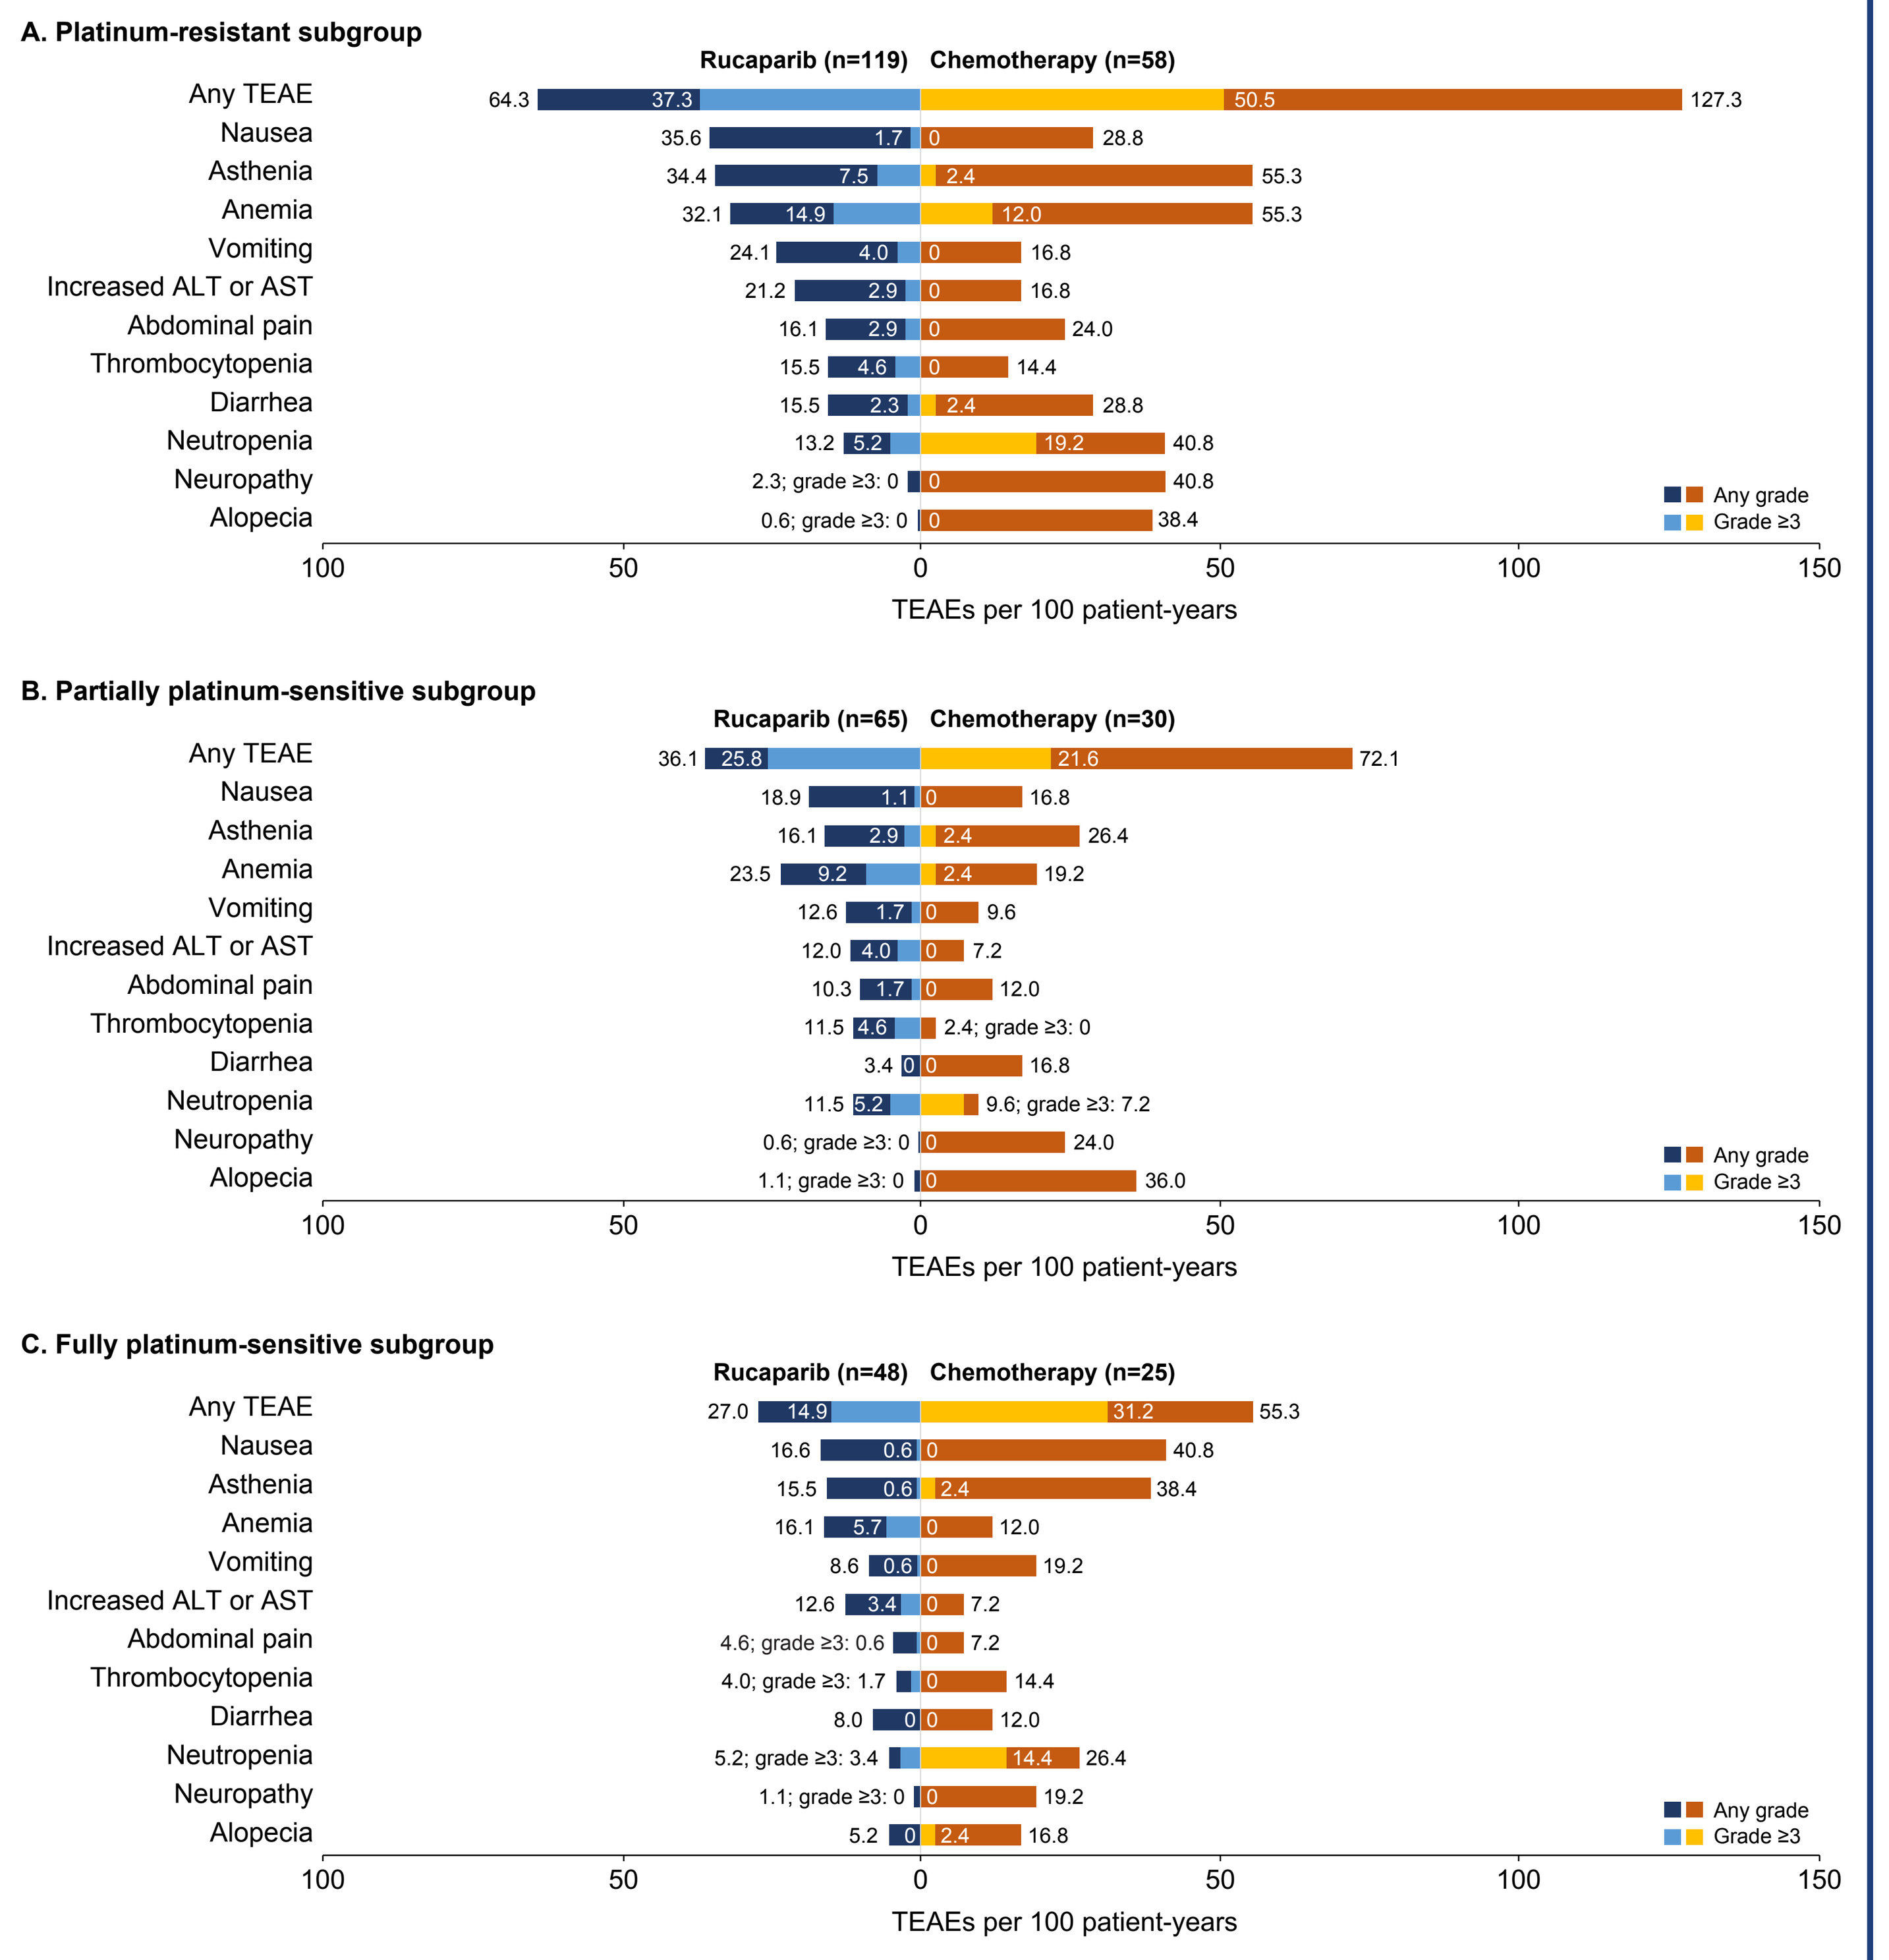

A. Platinum-resistant subgroup
Rucaparib (n=119)
Chemotherapy (n=58)
Any TEAE 64.3 37.3 50.5 127.3
Nausea 35.6 1.7 0 28.8
Asthenia 34.4 7.5 2.4 55.3
Anemia 32.1 14.9 12.0 55.3
Vomiting 24.1 4.0 0 16.8
Increased ALT or AST 21.2 2.9 0 16.8
Abdominal pain 16.1 2.9 0 24.0
Thrombocytopenia 15.5 4.6 0 14.4
Diarrhea 15.5 2.3 2.4 28.8
Neutropenia 13.2 5.2 19.2 40.8
Neuropathy 2.3; grade ≥3: 0 0 40.8
Alopecia 0.6; grade ≥3: 0 0 38.4
Any grade
Grade ≥3
TEAEs per 100 patient-years
B. Partially platinum-sensitive subgroup
Rucaparib (n=65)
Chemotherapy (n=30)
Any TEAE 36.1 25.8 21.6 72.1
Nausea 18.9 1.1 0 16.8
Asthenia 16.1 2.9 2.4 26.4
Anemia 23.5 9.2 2.4 19.2
Vomiting 12.6 1.7 0 9.6
Increased ALT or AST 12.0 4.0 0 7.2
Abdominal pain 10.3 1.7 0 12.0
Thrombocytopenia 11.5 4.6 2.4; grade ≥3: 0
Diarrhea 3.4 0 0 16.8
Neutropenia 11.5 5.2 9.6; grade ≥3: 7.2
Neuropathy 0.6; grade ≥3: 0 0 24.0
Alopecia 1.1; grade ≥3: 0 0 36.0
C. Fully platinum-sensitive subgroup
Rucaparib (n=48)
Chemotherapy (n=25)
Any TEAE 27.0 14.9 31.2 55.3
Nausea 16.6 0.6 0 40.8
Asthenia 15.5 0.6 2.4 38.4
Anemia 16.1 5.7 0 12.0
Vomiting 8.6 0.6 0 19.2
Increased ALT or AST 12.6 3.4 0 7.2
Abdominal pain 4.6; grade ≥3: 0.6 0 7.2
Thrombocytopenia 4.0; grade ≥3: 1.7 0 14.4
Diarrhea 8.0 0 0 12.0
Neutropenia 5.2; grade ≥3: 3.4 14.4 26.4
Neuropathy 1.1; grade ≥3: 0 0 19.2
Alopecia 5.2 0 2.4 16.8

Most common TEAEs of any grade (≥20% in the overall safety population for either treatment group) are listed. Data are sorted based on decreasing rates of any-grade TEAEs per 100 patient-years of treatment for patients treated with rucaparib in the platinum-resistant subgroup. Asthenia includes fatigue; anemia includes decreased hemoglobin; thrombocytopenia includes decreased platelet count; neutropenia includes decreased neutrophil count; and neuropathy includes neurotoxicity, paresthesia, peripheral neuropathy, peripheral motor neuropathy, peripheral sensory neuropathy, polyneuropathy.
ALT, alanine aminotransferase; AST, aspartate aminotransferase; TEAE, treatment-emergent adverse event.