Abstract 5537

Clinical and Molecular Characteristics of ARIEL3 Patients Who Derived Exceptional Benefit From Rucaparib Maintenance Treatment for High-Grade Ovarian Cancer (HGOC)

Tanya Kwan,[1] Amit M. Oza,[2] Domenica Lorusso,[3] Carol Aghajanian,[4] Ana Oaknin,[5] Andrew Dean,[6] Nicoletta Colombo,[7] Johanne I. Weberpals,[8] Andrew Clamp,[9] Giovanni Scambia,[10] Alexandra Leary,[11] Robert W. Holloway,[12] Margarita Amenedo Gancedo,[13] Peter Fong,[14] Jeffrey C. Goh,[15] David M. O'Malley,[16] Lara Maloney,[1] Sandra Goble,[1] Jonathan A. Ledermann,[17] Robert L. Coleman[18]

[1]Clovis Oncology, Inc., Boulder, CO, USA; [2]Princess Margaret Cancer Centre, University Health Network, Toronto, Canada; [3]Fondazione IRCCS, Istituto Nazionale dei Tumori, Milan, Italy*; [4]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [5]Vall d'Hebron Institute of Oncology (VHIO), Hospital Universitari Vall d'Hebron, Vall d'Hebron Barcelona Hospital Campus, Spain; [6]St John of God Subiaco Hospital, Australia; [7]University of Milan-Bicocca and European Institute of Oncology (IEO) IRCCS, Italy; [8]Ottawa Hospital Research Institute, Canada; [9]The Christie NHS Foundation Trust and University of Manchester, UK; [10]Fondazione Policlinico Universitario A. Gemelli IRCCS and Scientific Directorate, Rome, Italy; [11]Gustave Roussy Cancer Center, INSERM U981, and Groupe d'Investigateurs Nationaux pour l'Etude des Cancers Ovariens (GINECO), Villejuif, France; [12]Florida Hospital Cancer Institute, Orlando, FL, USA; [13]Oncology Center of Galicia, La Coruña, Spain; [14]Auckland City Hospital and University of Auckland, New Zealand; [15]Cancer Care Services, Royal Brisbane and Women's Hospital, Herston, and University of Queensland, St Lucia, Australia; [16]The Ohio State University, James Cancer Center, Columbus, OH, USA; [17]UCL Cancer Institute, University College London and UCL Hospitals, UK; [18]University of Texas MD Anderson Cancer Center, Houston, TX, USA†

*Affiliation where the work was conducted; current affiliation: Fondazione Policlinico Universitario A. Gemelli IRCCS and Scientific Directorate, Rome, Italy. †Affiliation where the work was conducted; current affiliation: US Oncology Research, The Woodlands, TX, USA.

SUMMARY

- **In ARIEL3, 21% of patients in the rucaparib-treated arm derived exceptional benefit (progression-free survival ≥2 years) versus only 2% of those in the placebo arm**
- **Exceptional benefit in ARIEL3 was more common in, but not exclusive to, patients with favorable clinical characteristics and known mechanisms of poly(ADP-ribose) polymerase inhibitor sensitivity, including *BRCA1*, *BRCA2*, *RAD51C*, and *RAD51D* mutations**
- **Our results suggest that rucaparib can deliver exceptional benefit to a diverse set of patients with high-grade ovarian cancer**

Poster Download
Also available at: https://clovisoncology.com/files/ASCO2021_Kwan_Poster.pdf
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.
Corresponding author: *Tanya Kwan; tkwan@clovisoncology.com.*

Poster Slides
Also available at: https://clovisoncology.com/files/ASCO2021_Kwan_Slides.pdf
Please scan this QR code or visit the website to view the poster as slides.

Supplementary Material
Also available at: https://clovisoncology.com/files/ASCO2021_Kwan_Poster_Supplement.pdf
Please scan this QR code or visit the website to view additional materials for this poster.

REFERENCES

1. Rubraca (rucaparib) tablets [prescribing information]. Boulder, CO: Clovis Oncology, Inc.; 2020.
2. Rubraca (rucaparib) tablets [summary of product characteristics]. Swords, Ireland: Clovis Oncology Ireland Ltd.; 2019.
3. Coleman et al. *Lancet*. 2017;390:1949-61.
4. Swisher et al. *Nat Commun*. 2021:12:2487.
5. Dean et al. *Ann Oncol*. 2020;31(suppl 4):abst 821P.
6. Morice et al. *Lancet Haematol*. 2021;8:e122-34.

ACKNOWLEDGMENTS

This study was funded by Clovis Oncology, Inc. Medical writing and editorial support funded by Clovis Oncology, Inc., were provided by Nathan Yardley and Stephen Bublitz of Ashfield MedComms, an Ashfield Health company.

ASCO Annual Meeting | June 4–8, 2021 | Virtual Meeting

INTRODUCTION

- Rucaparib is an oral, poly(ADP-ribose) polymerase (PARP) inhibitor approved in the United States and Europe for the maintenance treatment of patients with recurrent ovarian cancer who have responded to platinum-based chemotherapy[1,2]
- In ARIEL3 (NCT01968213), rucaparib maintenance treatment improved progression-free survival (PFS) across all predefined subgroups of patients with high-grade ovarian cancer[3]
 - Characterization of patients who derive durable benefit from PARP inhibitor maintenance treatment may provide insights into improving outcomes
- Here, we present an exploratory analysis of clinical and molecular characteristics in patients associated with exceptional benefit from rucaparib maintenance treatment

METHODS

- ARIEL3 study design and patient eligibility criteria have been described previously[3]
- Patients with recurrent, platinum-sensitive high-grade ovarian cancer who had responded to their last platinum-based regimen were randomized 2:1 to receive rucaparib 600 mg twice a day or placebo
- Molecular features (genomic alterations, *BRCA1* promoter methylation) and baseline clinical characteristics were compared within each treatment arm between the following subgroups:
 - Patients who derived exceptional benefit (PFS ≥2 years, defined as double the median PFS in the intent-to-treat population, which was 10.8 months,[3] rounded to the closest year)
 - Patients with disease progression on first scan (≈12 weeks; the short-term [ST] subgroup)
- Genomic analyses were performed using archival tumor tissues (full details can be found in the **Supplementary Material**)

RESULTS

- As of the December 31, 2019, data cutoff date, with a median follow-up of 51.4 months, 33/375 (9%) and 1/189 (0.5%) patients were still receiving rucaparib or placebo, respectively
- Overall, 21.1% of rucaparib patients (79/375) and 2.1% of placebo patients (4/189) showed exceptional benefit, with a median (range) PFS of 41.9 (24.2–67.2) and 37.1 (27.4–66.0) months, respectively (**Figure 1**)
- A majority (68/79 [86.1%]) of rucaparib-arm exceptional benefit patients achieved longer PFS in ARIEL3 than their penultimate platinum-free interval (PPFI) (**Supplementary Figure 1**)
 - The median (range) PFS–PPFI difference was 21.3 (–77.3 to 56.1) months, indicating that most exceptional benefit patients derived more durable benefit (ie, longer PFS) from rucaparib maintenance therapy than from their penultimate platinum therapy
- Patients who progressed at their first scan on study were included in the ST subgroup: rucaparib, 64/375 (17.1%); placebo, 62/189 (32.8%)

Figure 1. PFS in ARIEL3 Patients Who Derived Exceptional Benefit From Rucaparib

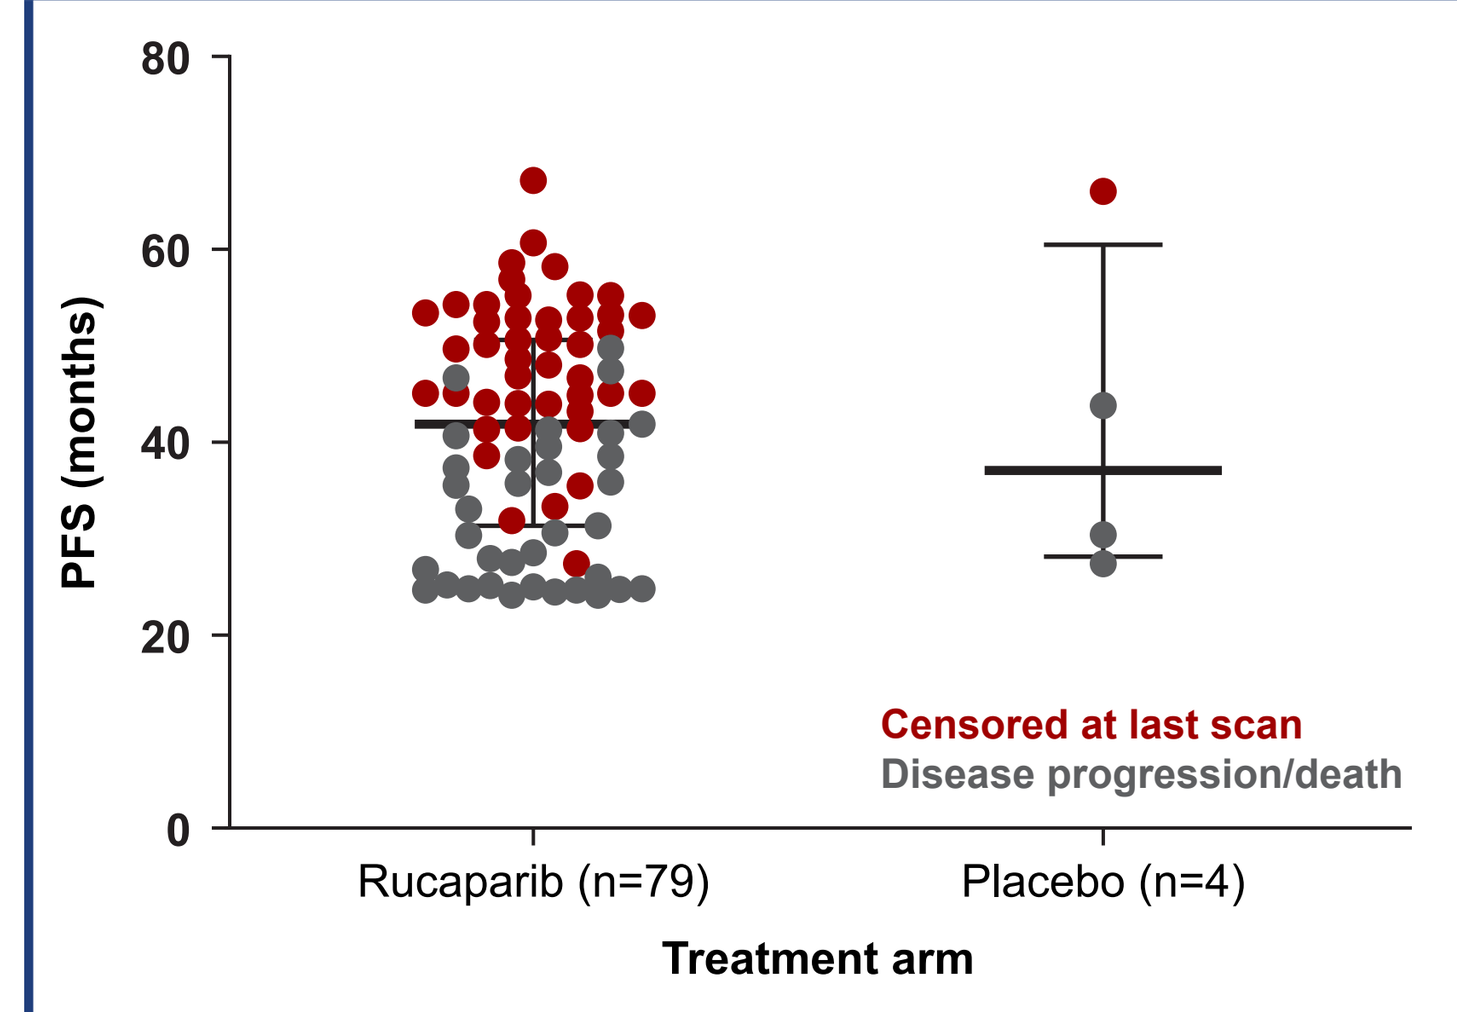
80
60
40
20
0
PFS (months)
Censored at last scan
Disease progression/death
Rucaparib (n=79)
Placebo (n=4)
Treatment arm

Patients who discontinued drug for reasons other than disease progression were censored for the PFS analysis. Horizontal line shows the median; error bars represent the interquartile range.
PFS, progression-free survival.

Baseline Clinical Characteristics of Exceptional Benefit Patients

- Among patients who received rucaparib, those who experienced exceptional benefit had more favorable clinical prognostic factors at baseline compared with those in the ST subgroup, including no measurable disease at baseline, complete response to most recent line of platinum, and longer PPFI (**Table 1**)
- Age, Eastern Cooperative Oncology Group (ECOG) performance status, number of prior regimens of chemotherapy, and number of prior regimens of platinum-based therapy were not associated with exceptional benefit

Table 1. Baseline Characteristics in the Exceptional Benefit and ST Subgroups

	Rucaparib				Placebo			
Characteristic	Exceptional benefit (n=79)	ST subgroup (n=64)	*P* value	Odds ratio (95% CI)	Exceptional benefit (n=4)	ST subgroup (n=62)	*P* value	Odds ratio (95% CI)
Age, median (range), y	61 (42–79)	61 (39–78)	0.770	—	54 (48–62)	62.5 (36–84)	0.292	—
ECOG PS, n (%)			0.711				>0.99	
0	55 (69.6)	47 (73.4)		0.8 (0.4–1.8)	3 (75.0)	44 (71.0)		1.2 (0.2–16.7)
1	24 (30.4)	17 (26.6)		1.2 (0.6–2.5)	1 (25.0)	18 (29.0)		0.8 (0.1–5.8)
Prior chemotherapy regimens, n (%)			0.725				>0.99	
2	50 (63.3)	43 (67.2)		0.8 (0.4–1.7)	3 (75.0)	39 (62.9)		1.8 (0.2–23.9)
≥3	29 (36.7)	21 (32.8)		1.2 (0.6–2.4)	1 (25.0)	23 (37.1)		0.6 (0.0–4.0)
Prior platinum regimens, n (%)			0.725				>0.99	
2	52 (65.8)	44 (68.8)		0.9 (0.4–1.8)	3 (75.0)	40 (64.5)		1.7 (0.2–22.3)
≥3	27 (34.2)	20 (31.3)		1.1 (0.6–2.3)	1 (25.0)	22 (35.5)		0.6 (0.0–4.3)
No measurable disease, n (%)	58 (73.4)	26 (40.6)	**<0.001**	**4.0 (2.0–8.0)**	3 (75.0)	33 (53.2)	0.620	2.6 (0.4–35.3)
Complete response to latest platinum, n (%)	31 (39.2)	13 (20.3)	**0.018**	**2.5 (1.2–5.3)**	1 (25.0)	11 (17.7)	0.561	1.5 (0.1–11.2)
PPFI >12 mo, n (%)	55 (69.6)	30 (46.9)	**0.007**	**2.6 (1.3–5.2)**	4 (100)	29 (46.8)	0.114	NA

Bold denotes significant result (*P*<0.05). Statistical comparisons based on Fisher's exact test for all cases except age, which was compared with a 2-sided *t* test.
ECOG PS, Eastern Cooperative Oncology Group performance status; NA, not applicable; PPFI, penultimate platinum-free interval; ST, short-term.

Molecular Characteristics Associated With Exceptional Benefit

- *BRCA1* or *BRCA2* (BRCA) mutations were significantly enriched among rucaparib-treated patients who derived exceptional benefit compared to those in the ST subgroup; however, 33/79 (41.8%) of exceptional benefit patients had BRCA wild-type carcinomas (**Figure 2**, **Table 2**)
- Among patients with BRCA wild-type carcinomas, *RAD51C* and *RAD51D* alterations were significantly associated with exceptional benefit (**Figure 2**, **Table 2**, **Table 3**)
- Although present in both subgroups, low genome-wide loss of heterozygosity (LOH; <16%) was more prevalent in the ST subgroup, suggesting patients with no evidence of homologous recombination deficiency (HRD) are less likely to derive durable benefit from rucaparib maintenance (**Figure 2**, **Table 2**)
- Frequency of high archival *BRCA1* methylation (≥70%) was similar among patients who derived exceptional benefit and those in the ST subgroup (**Figure 2**, **Table 2**)
 - We previously demonstrated that although high methylation measured immediately prior to rucaparib treatment is associated with response to rucaparib, archival methylation is not, as the loss of methylation can occur during the course of platinum treatment[4]
- *RAD51C/D* mutations and high *BRCA1* methylation cases almost exclusively had high genome-wide LOH (≥16%), confirming that these alterations lead to HRD (**Figure 2**)

Figure 2. Baseline Characteristics and Genetic/Epigenetic Alteration Landscape of Rucaparib Exceptional Benefit and ST Subgroups

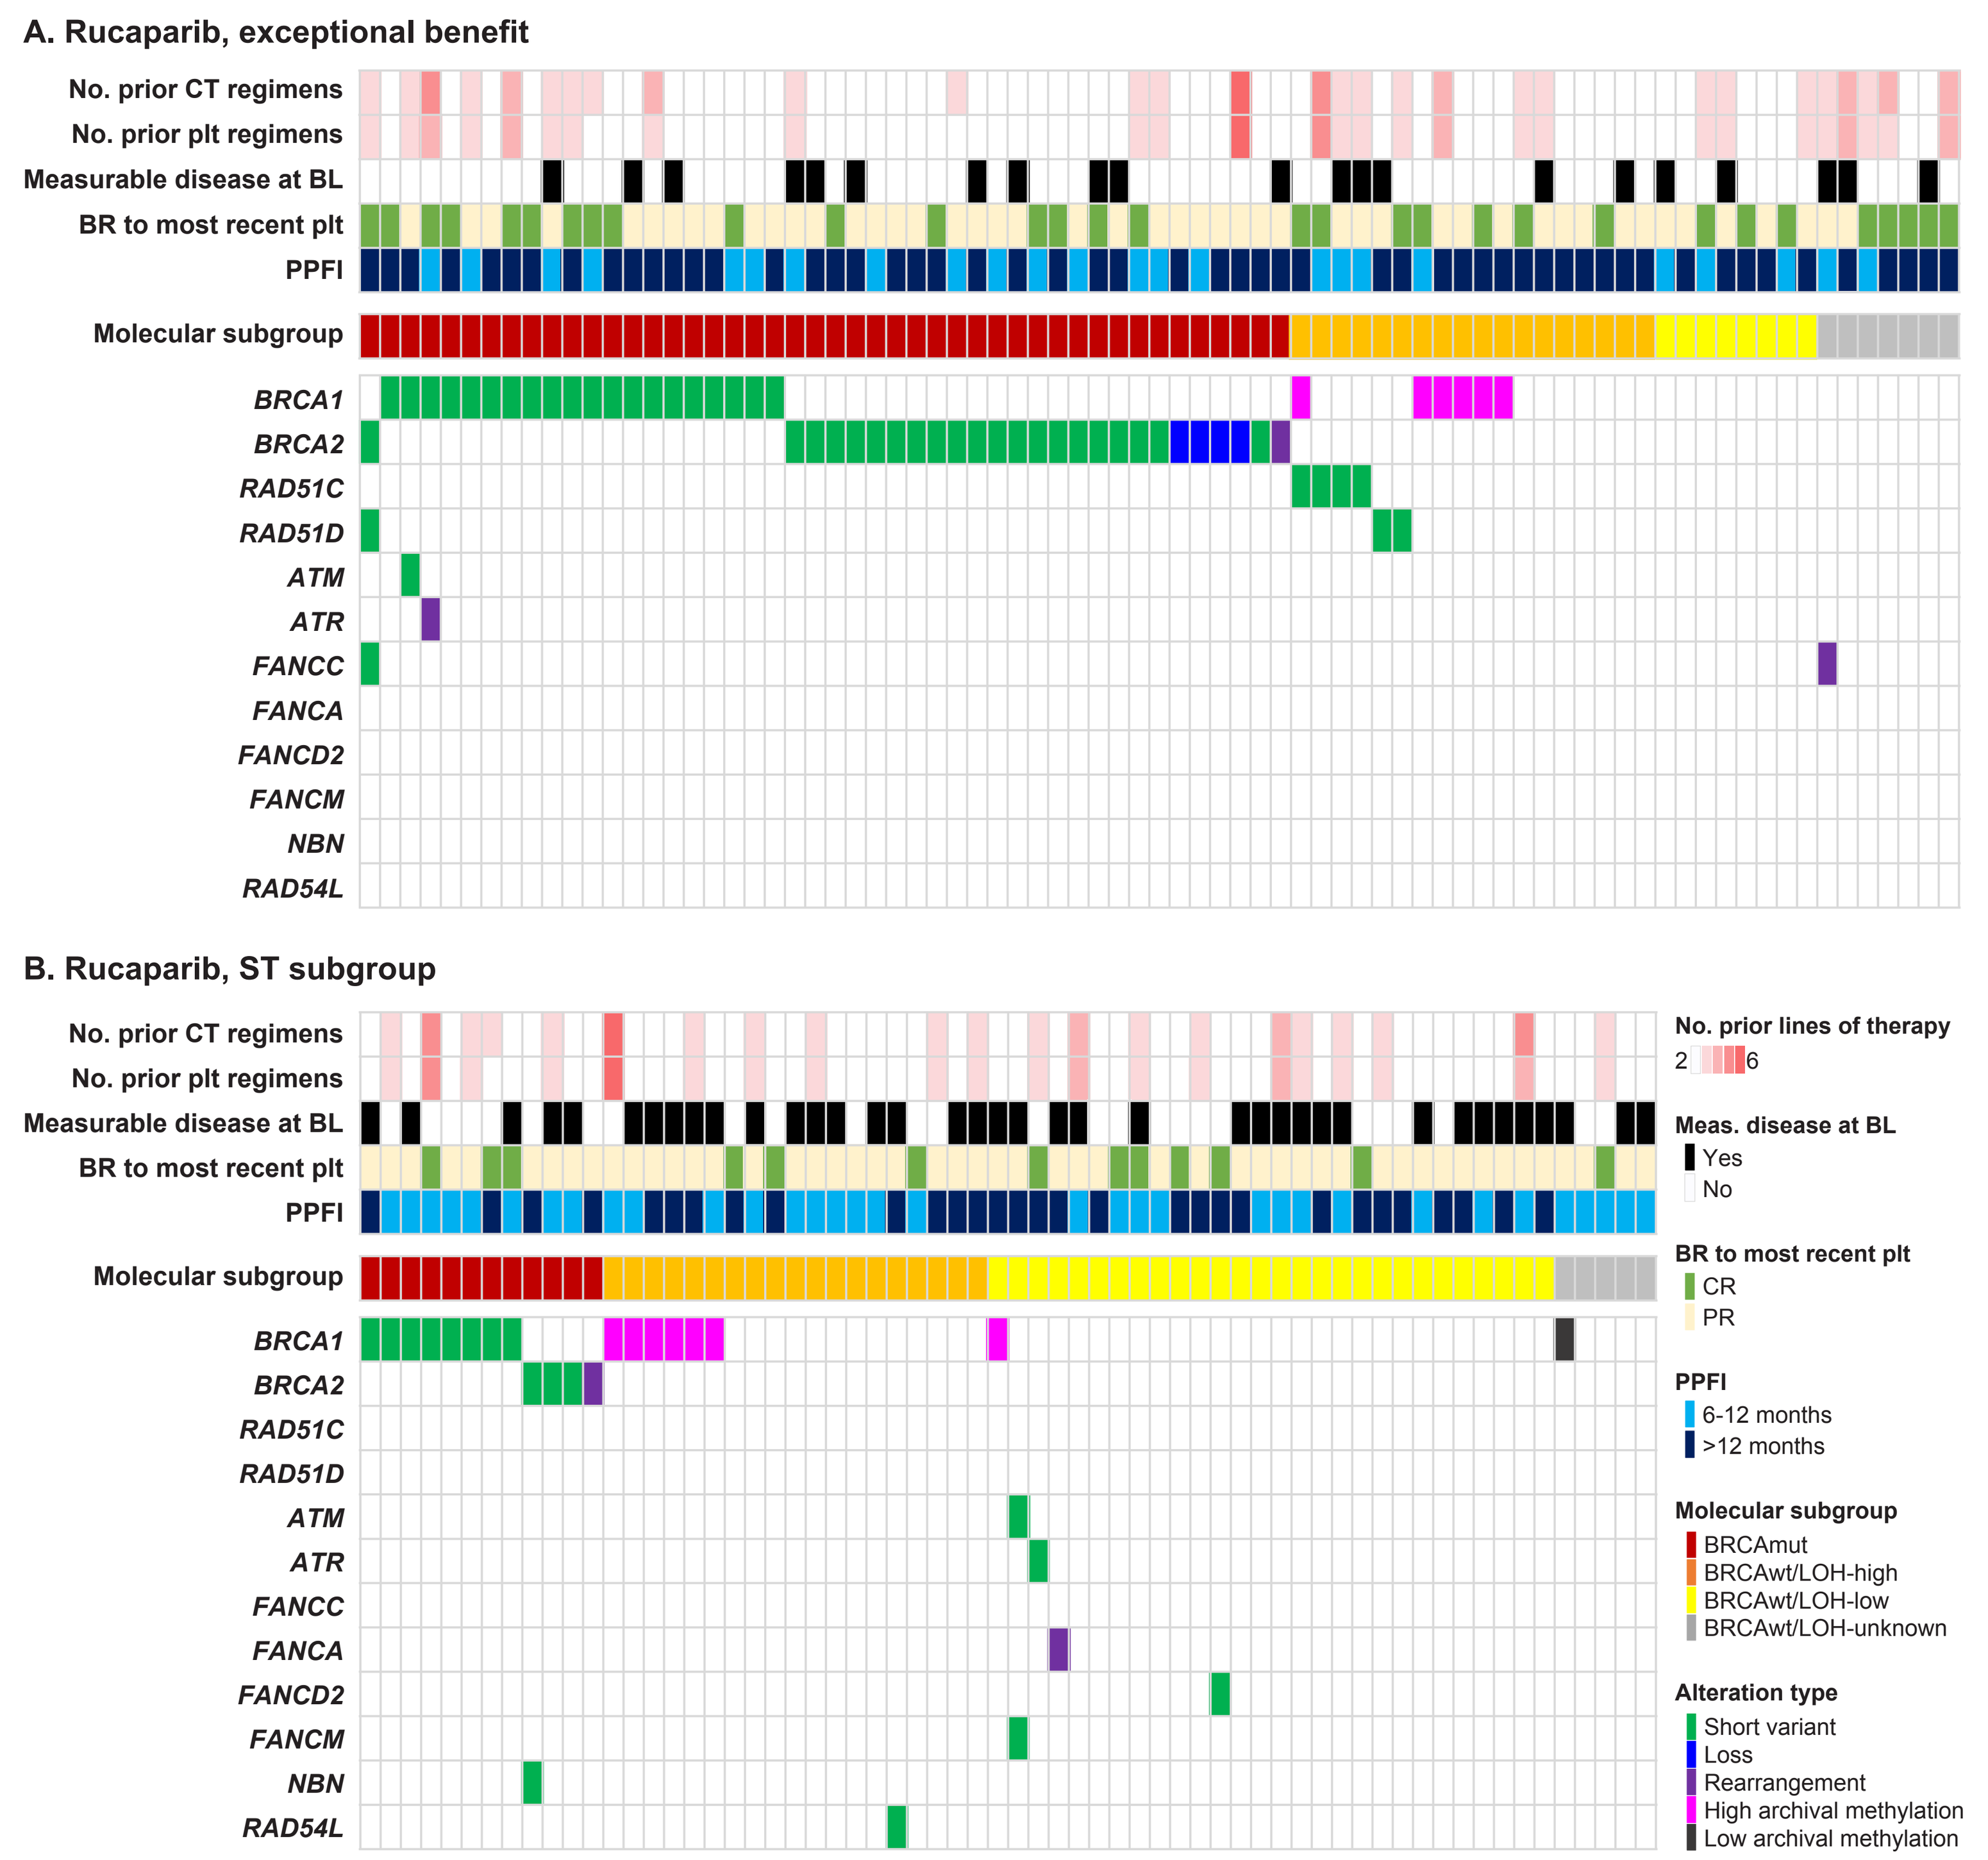
A. Rucaparib, exceptional benefit
No. prior CT regimens
No. prior plt regimens
Measurable disease at BL
BR to most recent plt
PPFI
Molecular subgroup
BRCA1
BRCA2
RAD51C
RAD51D
ATM
ATR
FANCC
FANCA
FANCD2
FANCM
NBN
RAD54L
B. Rucaparib, ST subgroup
No. prior CT regimens
No. prior plt regimens
Measurable disease at BL
BR to most recent plt
PPFI
Molecular subgroup
BRCA1
BRCA2
RAD51C
RAD51D
ATM
ATR
FANCC
FANCA
FANCD2
FANCM
NBN
RAD54L
No. prior lines of therapy
2
6
Meas. disease at BL
Yes
No
BR to most recent plt
CR
PR
PPFI
6-12 months
>12 months
Molecular subgroup
BRCAmut
BRCAwt/LOH-high
BRCAwt/LOH-low
BRCAwt/LOH-unknown
Alteration type
Short variant
Loss
Rearrangement
High archival methylation
Low archival methylation

BL, baseline; BR, best response; BRCA, *BRCA1* or *BRCA2*; CR, complete response; CT, chemotherapy; LOH, loss of heterozygosity; mut, mutant; plt, platinum; PPFI, penultimate platinum-free interval; PR, partial response; ST, short-term; wt, wild-type.

Table 2. Genetic and Epigenetic Alterations in the Rucaparib Exceptional Benefit and ST Subgroups

Alteration	Exceptional benefit (n=79)	ST subgroup (n=64)	*P* value	Odds ratio (95% CI)
BRCAmut	46 (58.2)	12 (18.8)	**<0.001**	**6.0 (2.8–13.3)**
BRCAwt + *RAD51C/D*	6 (7.6)	0	**0.033**	NA
BRCAwt + other HRR gene	1 (1.3)	5 (7.8)	0.090	0.2 (0.0–1.2)
BRCAwt + LOH-high	18 (22.8)	19 (29.7)	0.443	0.7 (0.3–1.5)
BRCAwt + LOH-low	8 (10.1)	28 (43.8)	**<0.001**	**0.14 (0.06–0.35)**
BRCAwt + high *BRCA1* methylation	6/25 (24.0)	7/47 (14.9)	0.353	1.8 (0.5–6.0)

Bold denotes significant result (*P*<0.05). Statistical comparisons based on Fisher's exact test for all cases. Data are n (%) or n/N (%).
BRCA, *BRCA1* or *BRCA2*; HRR, homologous recombination repair; LOH, loss of heterozygosity; mut, mutated; NA, not applicable; ST, short-term; wt, wild-type.
Data for the placebo arm are available in Supplementary Table 1.

Table 3. Non-BRCA HRR Gene Mutations Detected in the Rucaparib Exceptional Benefit and ST Subgroups

Patient #	Gene	PFS (months)	Mutation	Germline/somatic status	Zygosity
Exceptional benefit					
1	*RAD51C*	46.7+	Splice site 572-1G>A	Somatic	Homozygous
2	*RAD51C*	38.6+	Splice site 706-2A>G	Germline	Homozygous
3	*RAD51C*	35.5+	Splice site 706-2A>G	NA	Homozygous
4	*RAD51C*	27.4+	R193*	Germline	Homozygous
5	*RAD51D*	54.3+	R120*	Germline	Homozygous
6	*RAD51D*	50.2+	R74*	Somatic	Homozygous
7	*FANCC*	24.2	Truncating rearrangement	NA	NA
ST subgroup					
8	*ATM*	2.9	R2832C	Germline	NA
	FANCM		L691fs*5	NA	NA
9[a]	*FANCA*	9.0	Duplication rearrangement	NA	NA
10	*FANCD2*	2.6	W1450*	NA	Heterozygous
11	*RAD54L*	2.7	H676fs*19	NA	Heterozygous
12	*ATR*	2.7	A1266fs*8	NA	Heterozygous

[a]This patient received rucaparib for 2 weeks then discontinued treatment but was included in the ST subgroup as they had disease progression on their first scan, which was performed at 9 months after the first dose of rucaparib (protocol deviation).
BRCA, *BRCA1* or *BRCA2*; HRR, homologous recombination repair; NA, not available; PFS, progression-free survival; ST, short-term.

BRCA Mutations

- Patients with BRCA mutations appeared to derive exceptional benefit from rucaparib regardless of which BRCA gene was mutated (*BRCA1* versus *BRCA2*), mutation origin (germline versus somatic) or variant type (short variant versus rearrangement/loss) (**Figure 3A-C**, **Table 4**)
- Similar trends were observed in the placebo-arm patients, but a low number of exceptional benefit cases hinders meaningful statistical analysis (**Figure 3D-F**, **Supplementary Table 2**)

Figure 3. Frequency and Types of BRCA Mutations in the Exceptional Benefit and ST Subgroups

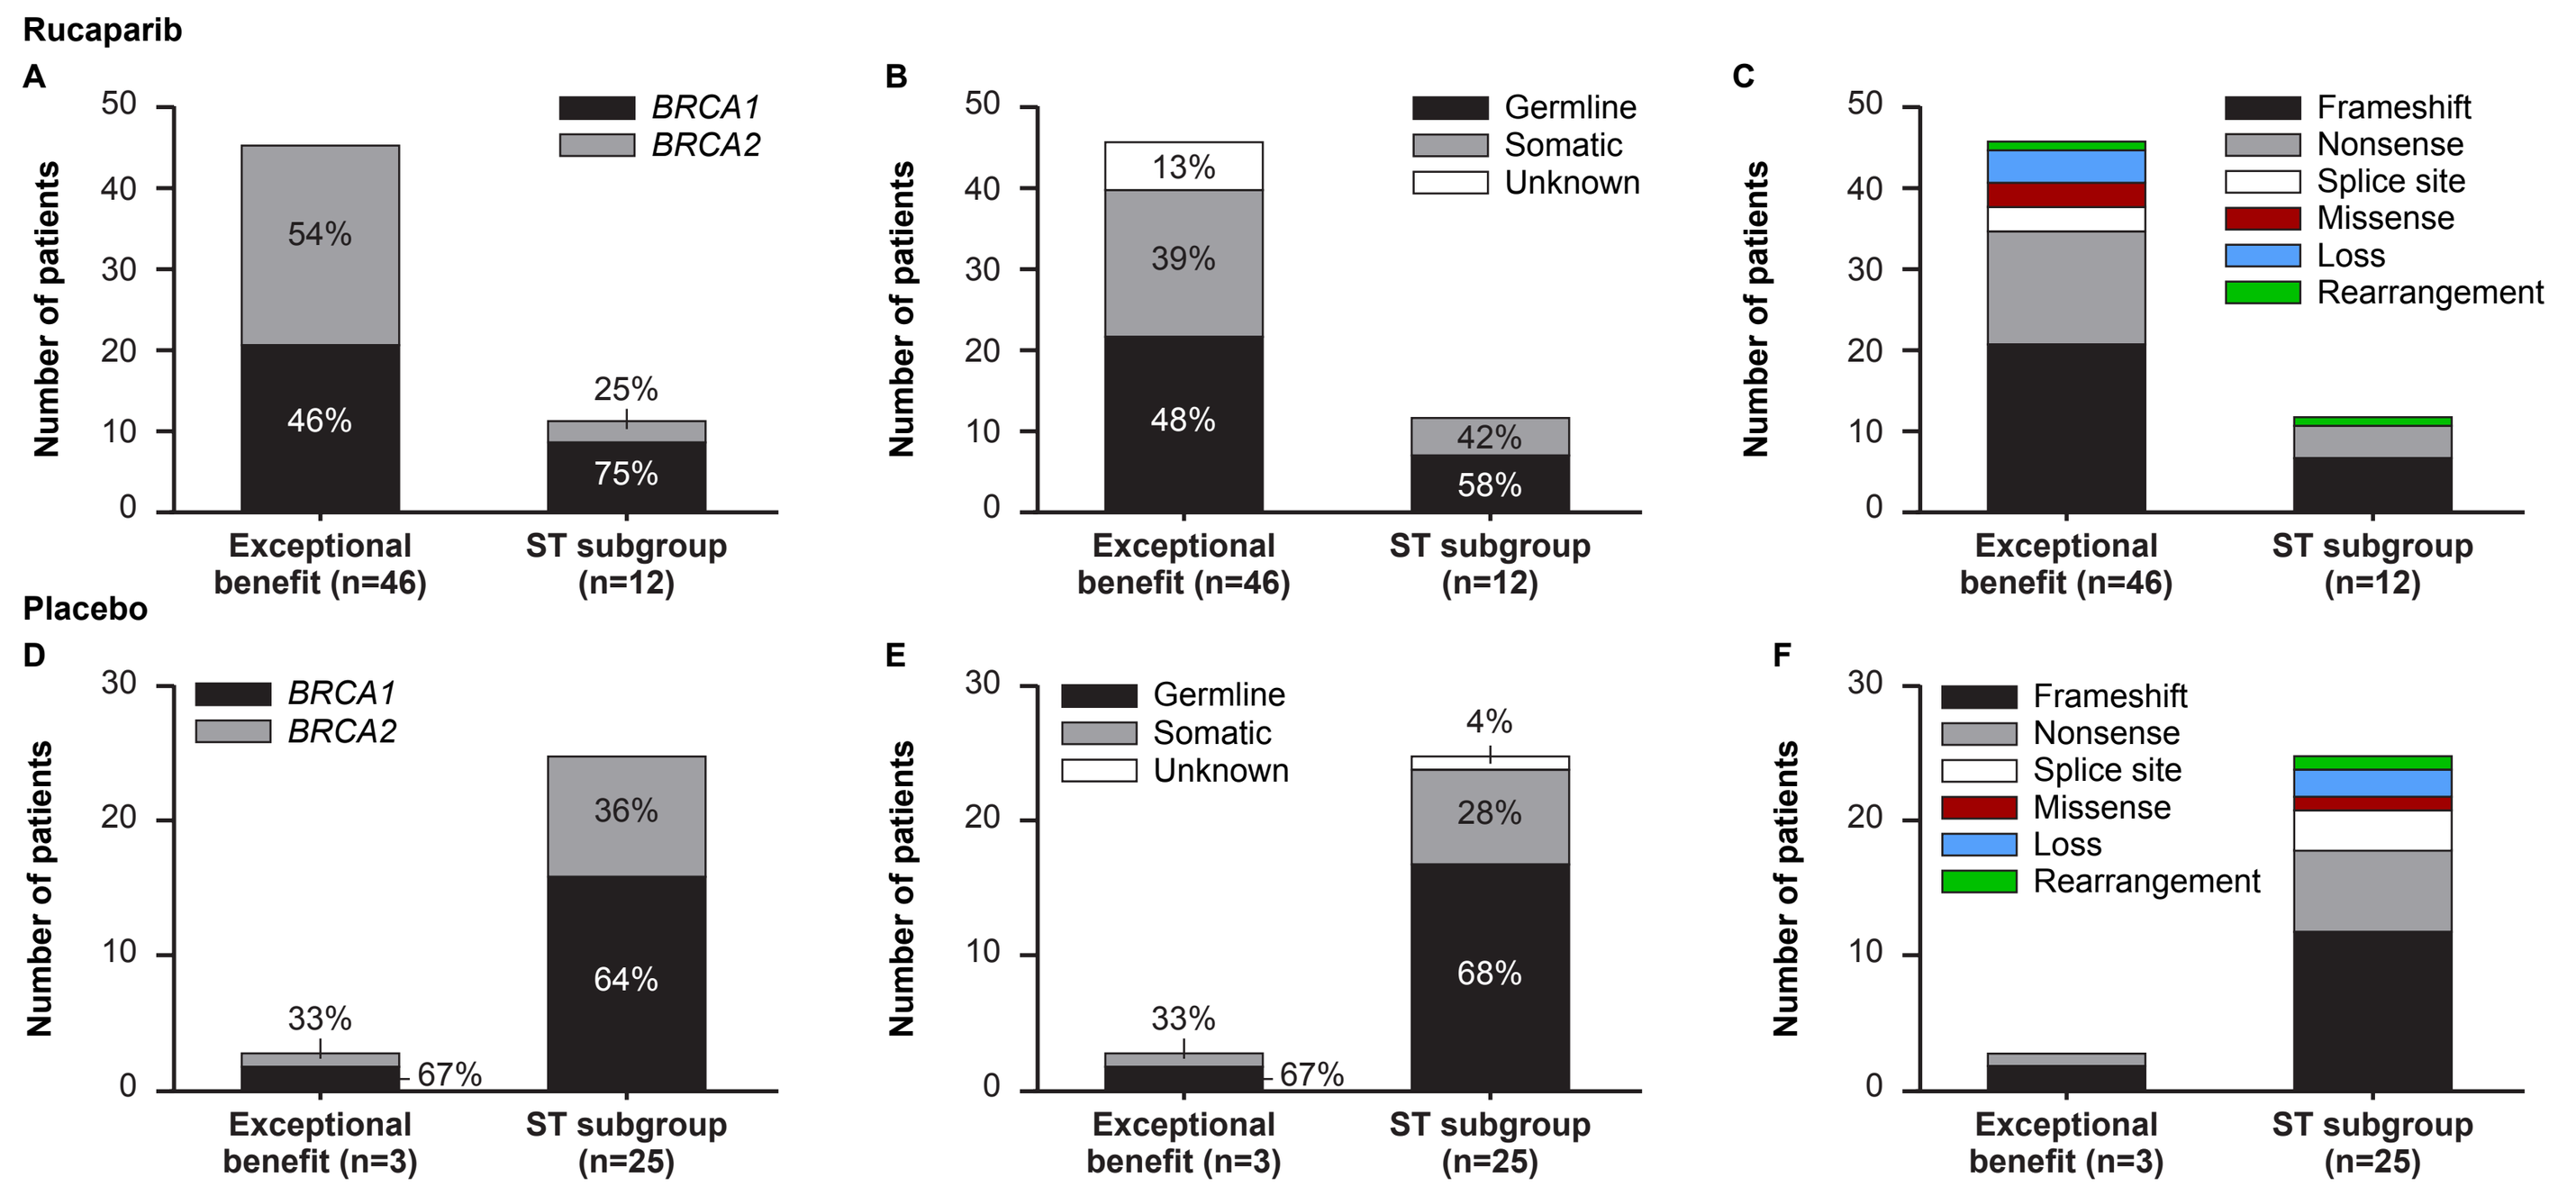
Rucaparib
A
BRCA1
BRCA2
54%
46%
25%
75%
Number of patients
Exceptional benefit (n=46)
ST subgroup (n=12)
B
Germline
Somatic
Unknown
13%
39%
48%
42%
58%
C
Frameshift
Nonsense
Splice site
Missense
Loss
Rearrangement
Placebo
D
BRCA1
BRCA2
33%
67%
36%
64%
Exceptional benefit (n=3)
ST subgroup (n=25)
E
Germline
Somatic
Unknown
33%
67%
4%
28%
68%
F
Frameshift
Nonsense
Splice site
Missense
Loss
Rearrangement

All data are shown among patients with BRCA mutations. (A,D) *BRCA1* and *BRCA2* mutations. (B,E) Germline and somatic BRCA mutations. (C,F) BRCA mutation types. Statistical comparisons related to rucaparib-arm graphs are shown in **Table 4**. All placebo-arm statistical comparisons were not significant (**Supplementary Table 2**).
BRCA, *BRCA1* or *BRCA2*; ST, short-term.

Table 4. Frequency and Types of BRCA Mutations in the Rucaparib Exceptional Benefit and ST Subgroups

	BRCAmut exceptional benefit (n=46)	BRCAmut ST subgroup (n=12)	*P* value	Odds ratio (95% CI)
Gene			0.106[a]	
BRCA1	21 (45.7)	9 (75.0)		0.3 (0.1–1.1)
BRCA2	25 (54.3)	3 (25.0)		3.6 (0.9–13.2)
Germline/somatic status			0.408[b]	
Germline	22 (47.8)	7 (58.3)		0.7 (0.2–2.4)
Somatic	18 (39.1)	5 (41.7)		0.9 (0.2–3.1)
Unknown	6 (13.0)	0		NA
Mutation type			>0.99[a]	
Short variant	41 (89.1)	11 (91.7)		0.7 (0.1–5.3)
Rearrangement/loss	5 (10.9)	1 (8.3)		1.3 (0.2–17.1)

Data are n (%).
[a]Significance based on Fisher's exact test. [b]Significance based on chi-square test.
BRCA, *BRCA1* or *BRCA2*; mut, mutated; NA, not applicable; ST, short-term.
Data for the placebo arm are available in Supplementary Table 2.

Safety

- Among rucaparib-treated patients, incidence rates of the most common treatment-emergent adverse events (TEAEs) were generally consistent between the exceptional benefit subgroup and the overall ARIEL3 patient population[5]
- There was a higher incidence in certain safety parameters in the exceptional benefit subgroup as compared to the overall population, which can be attributed to the length of time patients remained on treatment
 - Grade ≥3 TEAEs (74.7% vs 62.1%)
 - Treatment interruption and/or dose reduction due to a TEAE (83.5% vs 72.8%)
 - Any-grade abdominal pain (50.6% vs 32.3%)
- Most rucaparib-treated patients in the exceptional benefit subgroup (57/79; 72.2%) had ≥1 dose reduction; 33/79 patients (41.8%) had ≥2 dose reductions; median dose intensity was 0.83
- As of December 19, 2020, 18 myelodysplastic syndrome (MDS)/acute myeloid leukemia (AML) cases have been reported in the overall ARIEL3 patient population (14 rucaparib- and 4 placebo-treated patients); 9 cases were reported in rucaparib-treated patients in the exceptional benefit subgroup (3 during treatment, 6 during long-term follow-up)
 - Prior reports have suggested that PARP inhibitor exposure may be associated with an increased risk of MDS/AML, but longer time on PARP inhibitor treatment has not been associated with greater risk for MDS/AML[6]