LIO-1: Lucitanib + Nivolumab in Patients With Advanced Solid Tumors—Updated Phase 1b Results and Initial Experience in Phase 2 Ovarian Cancer Cohort (NCT04042116; ENGOT-GYN3/AGO/LIO)

Abstract 5538

Erika Hamilton,[1] Camille Gunderson Jackson,[2] Ramez N. Eskander,[3] Floor J. Backes,[4] Vicky Makker,[5] Fernanda Musa,[6] Alexander Olawaiye,[7] Jill Alldredge,[8] Bhavana Pothuri,[9] Angeles Alvarez Secord,[10] Nicole Concin,[11,12] Antonio González-Martín,[13] Jowell Go,[14] Kenton Wride,[14] Denise Lepley,[14] Terri Cameron,[15] Manish R. Patel[16]

[1]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA; [2]Stephenson Cancer Center, University of Oklahoma Health Sciences Center, Oklahoma City, OK, USA; [3]UC San Diego Moores Cancer Center, La Jolla, CA, USA; [4]Ohio State University Wexner Medical Center and James Cancer Hospital, Columbus, OH, USA; [5]Memorial Sloan Kettering Cancer Center, Weill Cornell Medical Center, New York, NY, USA; [6]Swedish Cancer Institute, Seattle, WA, USA; [7]Magee-Womens Hospital of UPMC, Pittsburgh, PA, USA; [8]Anschutz Cancer Pavilion, Aurora, CO, USA; [9]Laura and Isaac Perlmutter Cancer Center at NYU Langone, New York, NY, USA; [10]Duke Cancer Institute, Duke University School of Medicine, Durham, NC, USA; [11]Ev. Kliniken Essen Mitte, Germany; [12]Medizinische Universität Innsbruck, Austria; [13]Clínica Universidad de Navarra, Madrid, Spain; [14]Clovis Oncology, Inc., Boulder, CO, USA; [15]Clovis Oncology UK Ltd., Cambridge, UK; [16]Florida Cancer Specialists/Sarah Cannon Research Institute, Sarasota, FL, USA.

CONCLUSIONS

- **Phase 1b cohort data defined the recommended phase 2 starting dose as lucitanib 6 mg orally once daily and nivolumab 480 mg intravenously every 28 days**
 - **Data suggest that the combination of lucitanib + nivolumab has promising antitumor activity (47.1% disease control rate) with a manageable safety profile across multiple tumor types**
- **The phase 2 study is ongoing; data from the initial 24 patients enrolled into stage 1 of the phase 2 ovarian cancer (OC) cohort are presented**
 - **In this heavily pretreated patient population, 1 confirmed partial response was seen**
 - **The OC cohort is closed to further enrollment, but ongoing patients continue to be followed**
 - **Safety is consistent with that reported in the phase 1b portion of the study**
- **An individualized safety-based lucitanib dose-titration strategy (including escalation from 6, to 8, to 10 mg once daily) appears feasible, as explored in the phase 2 OC cohort. Further investigation of this strategy is ongoing**

Poster Download
Also available at: https://clovisoncology.com/files/ASCO2021_Hamilton_Poster.pdf
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.
Corresponding author: *Erika Hamilton*; ehamilton@tnonc.com.

Poster Slides
Also available at: https://clovisoncology.com/files/ASCO2021_Hamilton_Slides.pdf
Please scan this QR code or visit the website to view the poster as slides.

Supplementary Material
Also available at: https://clovisoncology.com/files/ASCO2021_Hamilton_Poster_Supplement.pdf
Please scan this QR code or visit the website to view additional materials for this poster.

REFERENCES

1. Bello et al. *Cancer Res*. 2011;71:1396-405.
2. Mahoney et al. *Clin Ther*. 2015;37:764-82.
3. Hamilton et al. *Ann Oncol*. 2020;31(suppl 4):S480-1.

ACKNOWLEDGMENTS

The authors would like to thank Dr. Joyce Liu (Dana Farber Cancer Institute, Boston, MA, USA) and Dr. Richard Penson (Massachusetts General Hospital, Boston, MA, USA) for their contributions to the study. This research was sponsored by Clovis Oncology, Inc. Medical writing support was provided by Andrew Croskery (Clovis Oncology, Ltd., London, UK) and editorial support funded by Clovis Oncology was provided by Shelly Lim and Stephen Bublitz of Ashfield MedComms, an Ashfield Health company.

INTRODUCTION

- Lucitanib is an oral tyrosine kinase inhibitor that selectively inhibits VEGFR1–3, PDGFRα/β, and FGFR1–3[1]
- Nivolumab is a human monoclonal antibody that binds to programmed cell death receptor 1 (PD-1) and blocks its interaction with programmed cell death ligand 1 (PD-L1) and 2, releasing PD-1–mediated inhibition of the antitumor immune response[2]
- The LIO-1 study (NCT04042116; ENGOT-GYN3/AGO/LIO) is assessing the efficacy and safety of the combination of lucitanib + nivolumab in patients with advanced and metastatic solid tumors. Phase 1b of the study identified the recommended phase 2 dose of oral lucitanib as 6 mg once daily (QD) + nivolumab (480 mg intravenously [IV] every 28 days)[3]
- Phase 2 is evaluating the efficacy and safety of lucitanib + nivolumab in patients with advanced gynecologic malignancies (endometrial, cervical, ovarian, and clear-cell ovarian/endometrial cancers) through a Simon 2-stage study design, using individualized dose titration for lucitanib to optimize exposure and potential benefit of the combination
- Here, we present updated phase 1b results in patients with advanced solid tumors and describe the initial experience for stage 1 of the phase 2 ovarian cancer (OC) cohort

METHODS

- The phase 1b dose-escalation portion of LIO-1 utilized a 4+3 dose-escalation design and enrolled patients with advanced solid tumors with no satisfactory treatment options
 - Lucitanib was evaluated at doses of 6, 8, and 10 mg QD in combination with a fixed dose of nivolumab at 480 mg IV every 28 days
 - Dose-limiting toxicities (DLTs) were assessed during the first 28 days of treatment
 - Tumor response was assessed by investigators per Response Evaluation Criteria In Solid Tumors, version 1.1 (RECIST)
- The primary endpoints were safety and DLTs to determine the recommended phase 2 dose of the combination of lucitanib + nivolumab, and confirmed best overall response rate (ORR) by RECIST per investigator assessment
 - A key secondary endpoint was disease control rate defined as confirmed complete response (CR) or partial response (PR), or stable disease (SD) ≥16 weeks
- The phase 2 OC cohort enrolled patients with recurrent high-grade epithelial ovarian, fallopian tube, or primary peritoneal cancer who have received ≥2 prior chemotherapy regimens (including ≥1 platinum doublet) or had disease progression ≤6 months after completing first-line platinum-based chemotherapy (up to 10 patients)
- Patients initiated treatment with lucitanib 6 mg QD + fixed-dose nivolumab
 - Those who met safety-based dose-titration criteria (**Table 1**) escalated to lucitanib 8 mg QD and then to 10 mg QD at subsequent cycles
 - Lucitanib dose could be escalated at start of cycles 2–5, provided dose-titration criteria had continued to be met since study start, and ≤2 cycles had elapsed between escalations

Table 1. Safety-Based Dose-Titration Criteria

• Blood pressure ≤150/90 mmHg	• No treatment-related diarrhea grade >1
• No treatment-related adverse events grade >2	• Not requiring any antihypertensive agents (or change to existing antihypertensive regimen if preexisting stable, well-controlled blood pressure at baseline)
• No proteinuria >1+ (or urinary protein >1.0 g/24 h)	

- Lucitanib treatment could be interrupted and/or the dose could be reduced in 2-mg decrements to a minimum of 2 mg in the event of toxicity. Nivolumab dose reductions were not permitted
- We present data from the entire phase 1b cohort as well as from 24 patients enrolled to stage 1 of the phase 2 OC cohort, with a data cutoff date of March 29, 2021

RESULTS

Phase 1b

Patients

- Overall, 17 patients were treated with oral lucitanib (6 mg QD [n=7], 8 mg QD [n=7], and 10 mg QD [n=3]) + nivolumab (480 mg IV every 28 days)
 - Median (range) age was 63.0 (45.0–77.0) years; 11 (64.7%) patients had Eastern Cooperative Oncology Group performance status (ECOG PS) 1

Efficacy

- Median (range) treatment duration was 3.6 (0.5–18.4+) months
- One patient with anal cancer had a confirmed CR and remained on study for approximately 10 months; 1 patient with cervical cancer had a confirmed PR and has been ongoing on study for >19 months as of the data cutoff date (**Figure 1**)
 - 10 patients had a best overall response of SD; 3 had progressive disease (PD); and 2 were not evaluable
 - As of the data cutoff date, 1 patient remains on treatment
- Overall disease control rate was 47.1%

Figure 1. Phase 1b: Time on Lucitanib Treatment and Best Overall Response to Lucitanib + Nivolumab

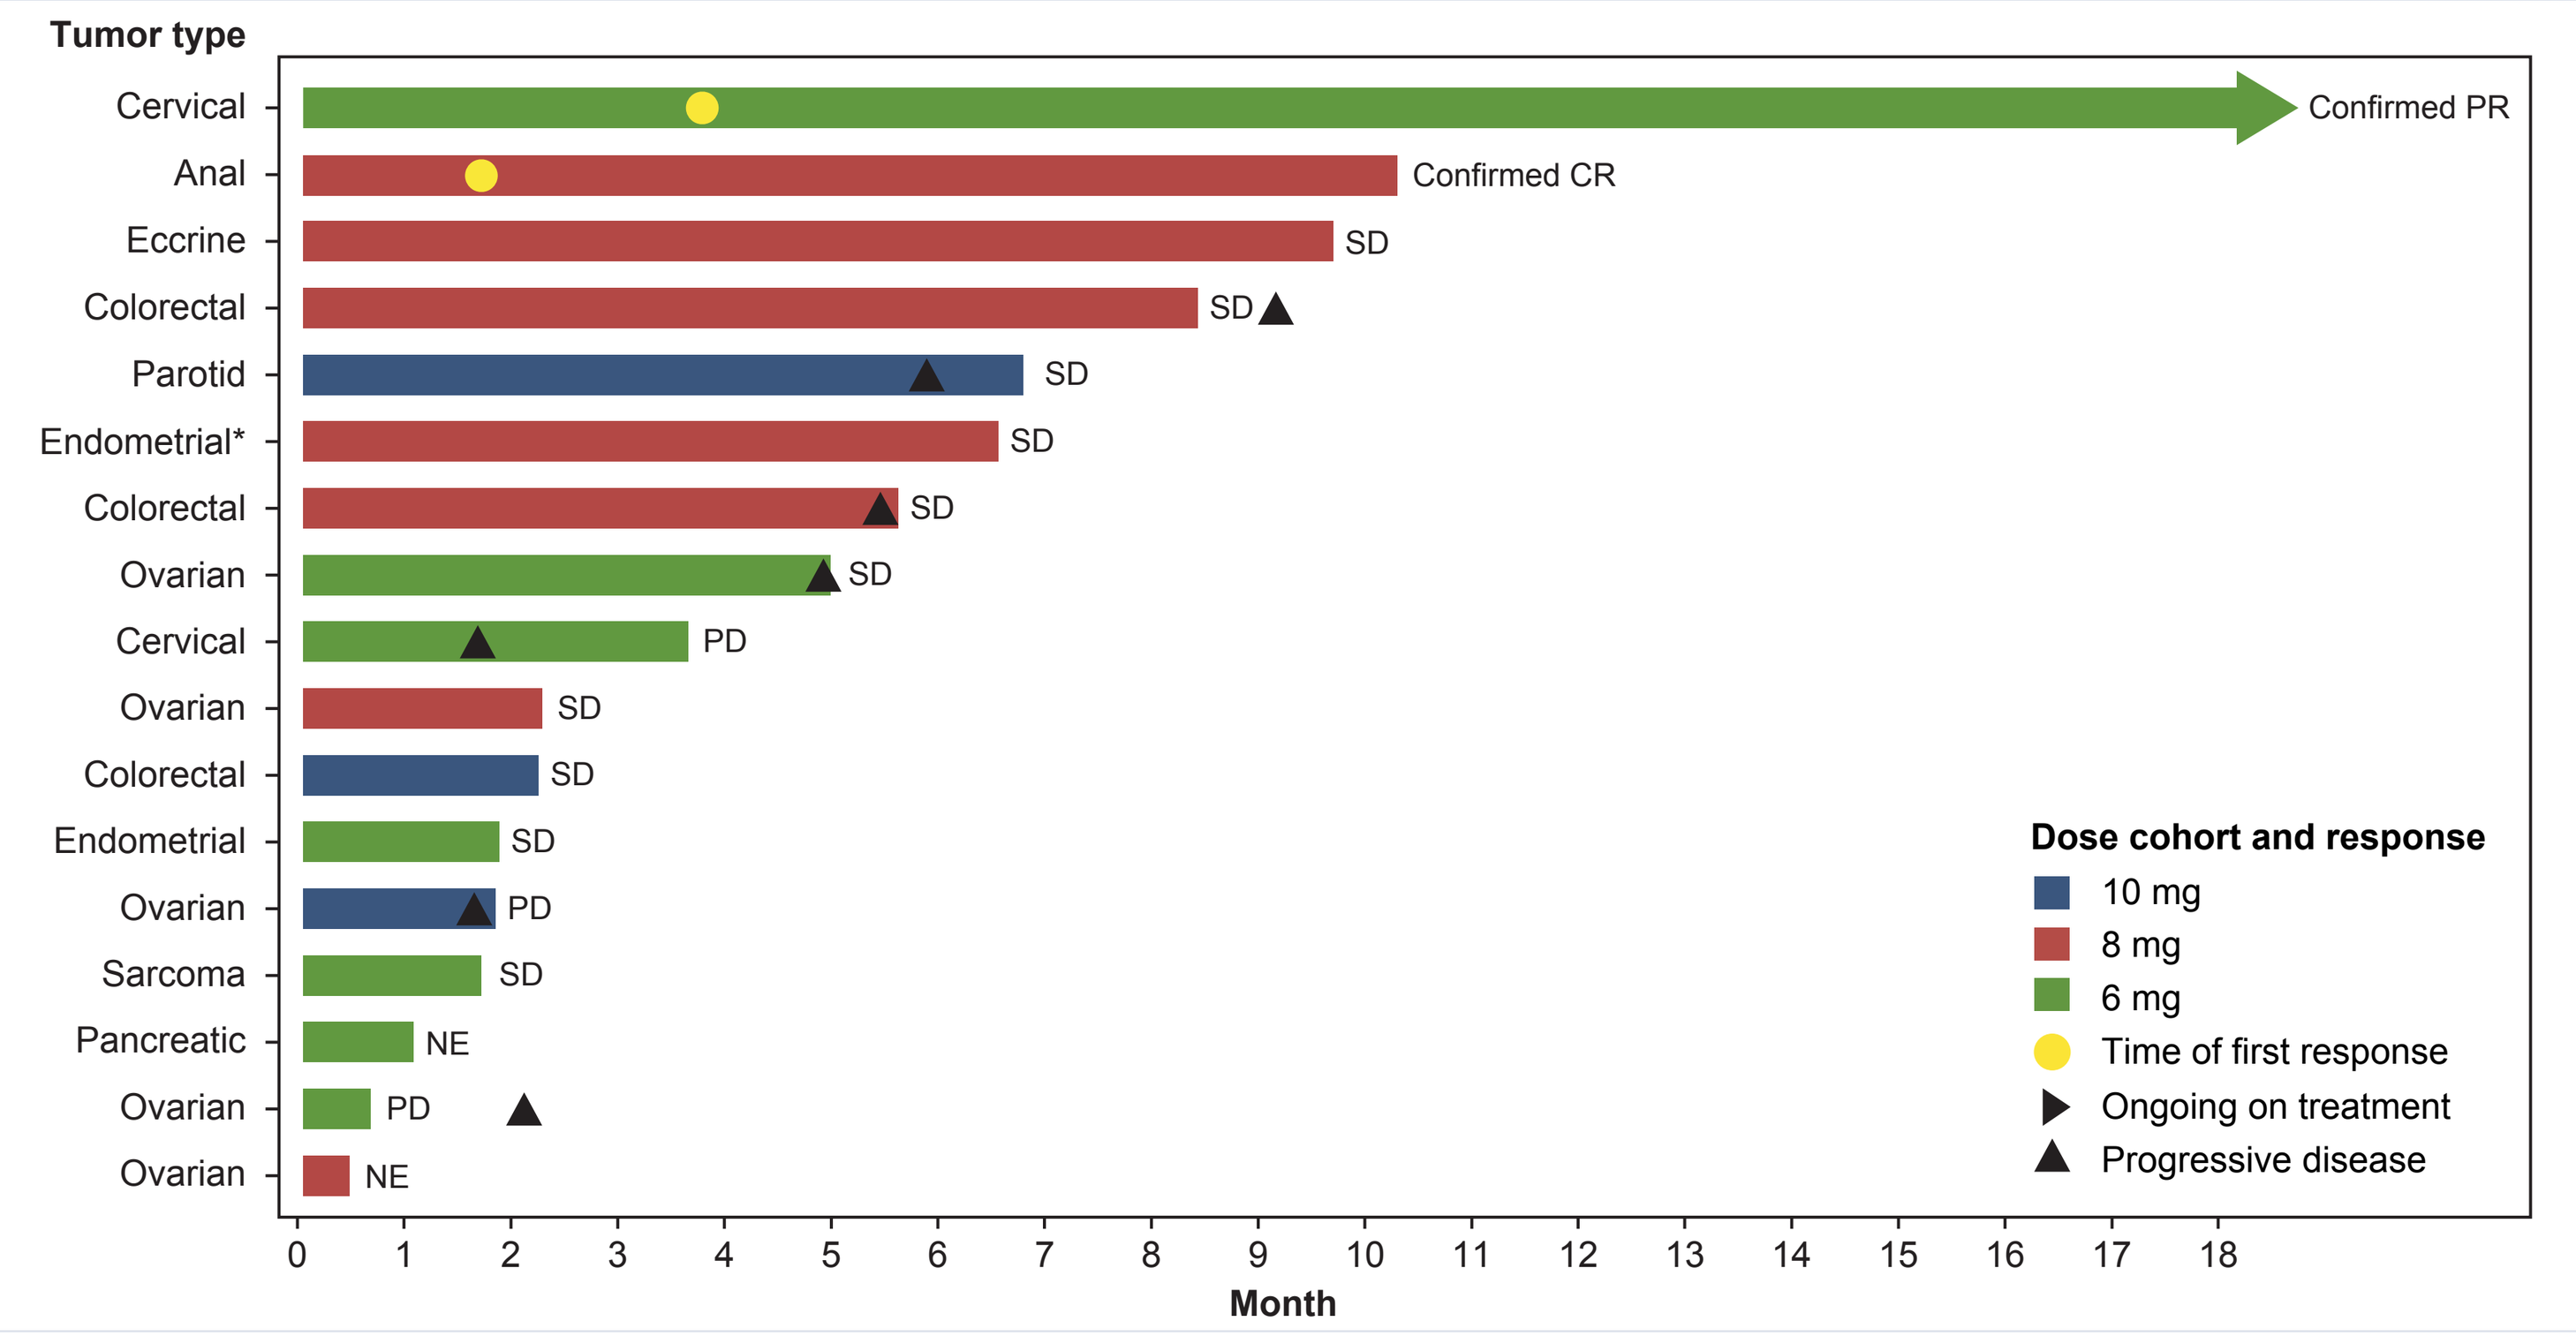
Tumor type
Cervical
Anal
Eccrine
Colorectal
Parotid
Endometrial*
Colorectal
Ovarian
Cervical
Ovarian
Colorectal
Endometrial
Ovarian
Sarcoma
Pancreatic
Ovarian
Ovarian
Confirmed PR
Confirmed CR
SD
PD
NE
Month
Dose cohort and response
10 mg
8 mg
6 mg
Time of first response
Ongoing on treatment
Progressive disease

Data cutoff: March 29, 2021. Patients could remain on nivolumab following discontinuation of lucitanib. *Endometrial stromal sarcoma. CR, complete response; NE, not evaluable; PD, progressive disease; PR, partial response; SD, stable disease.

Safety

- In the lucitanib 6-mg group, 1 DLT was observed: grade 3 proteinuria that improved to grade 1 but remained ongoing after study discontinuation. No DLTs were reported in the 8- or 10-mg cohorts
- Within this small group of patients, no differences in treatment-emergent adverse event (TEAE) frequencies between dose levels were apparent; see **Supplementary Material** for more detail

Phase 2 OC Cohort

Patients

- Baseline characteristics are presented in **Table 2**

Table 2. Phase 2 OC Cohort: Baseline Characteristics

	n=24
Age, median (range), y	67.5 (42.0–84.0)
ECOG PS, n (%)	
0	16 (66.7)
1	8 (33.3)
Type of ovarian primary cancer, n (%)	
Epithelial	15 (62.5)
Fallopian tube	4 (16.7)
Primary peritoneal	3 (12.5)
Low-grade granulosa cell	2 (8.3)
Histological classification, n (%)	
Serous	19 (79.2)
Endometrioid	2 (8.3)
Mixed/other	3 (12.5)
Prior anticancer regimens, n (%)	
1	5 (20.8)
2	2 (8.3)
≥3	17 (70.8)
Primary platinum resistance, n (%)	7 (29.2)
Time to progression from last platinum, n (%)	
<1 month	10 (41.7)
≥1 to <6 months	10 (41.7)
≥6 months	4 (16.7)
Prior bevacizumab, n (%)	13 (54.2)
Prior PARP inhibitor, n (%)	11 (45.8)

Data cutoff: March 29, 2021.
ECOG PS, Eastern Cooperative Oncology Group performance status; OC, ovarian cancer; PARP, poly(ADP-ribose) polymerase.

Efficacy

- The efficacy population comprised 22 patients with high-grade OC; 2 patients with low-grade granulosa cell carcinoma (LGGC) were excluded from the efficacy analysis
- Of 19/22 patients who completed ≥1 cycle, 9 escalated to the 8-mg lucitanib dose. Of these, 3 patients subsequently escalated to 10 mg
- As of the data cutoff date, 1 patient with a confirmed PR has been ongoing on study for >4 months (**Figure 2**, **Figure 3**)
 - 5 patients experienced an initial PR but subsequently discontinued study treatment due to PD (n=3) or a TEAE (n=2) without a confirmatory scan; 8 additional patients had a best overall response of SD, 6 patients had PD, and 2 patients were not evaluable
 - 6 (27.3%) patients are continuing to receive treatment with lucitanib + nivolumab
- Both patients with LGGC had a best overall response of SD as of the data cutoff date. One patient discontinued after 6 cycles due to PD, while the other remains on study

Figure 2. Phase 2 OC Cohort: Time on Lucitanib Treatment and Best Overall Response to Lucitanib + Nivolumab

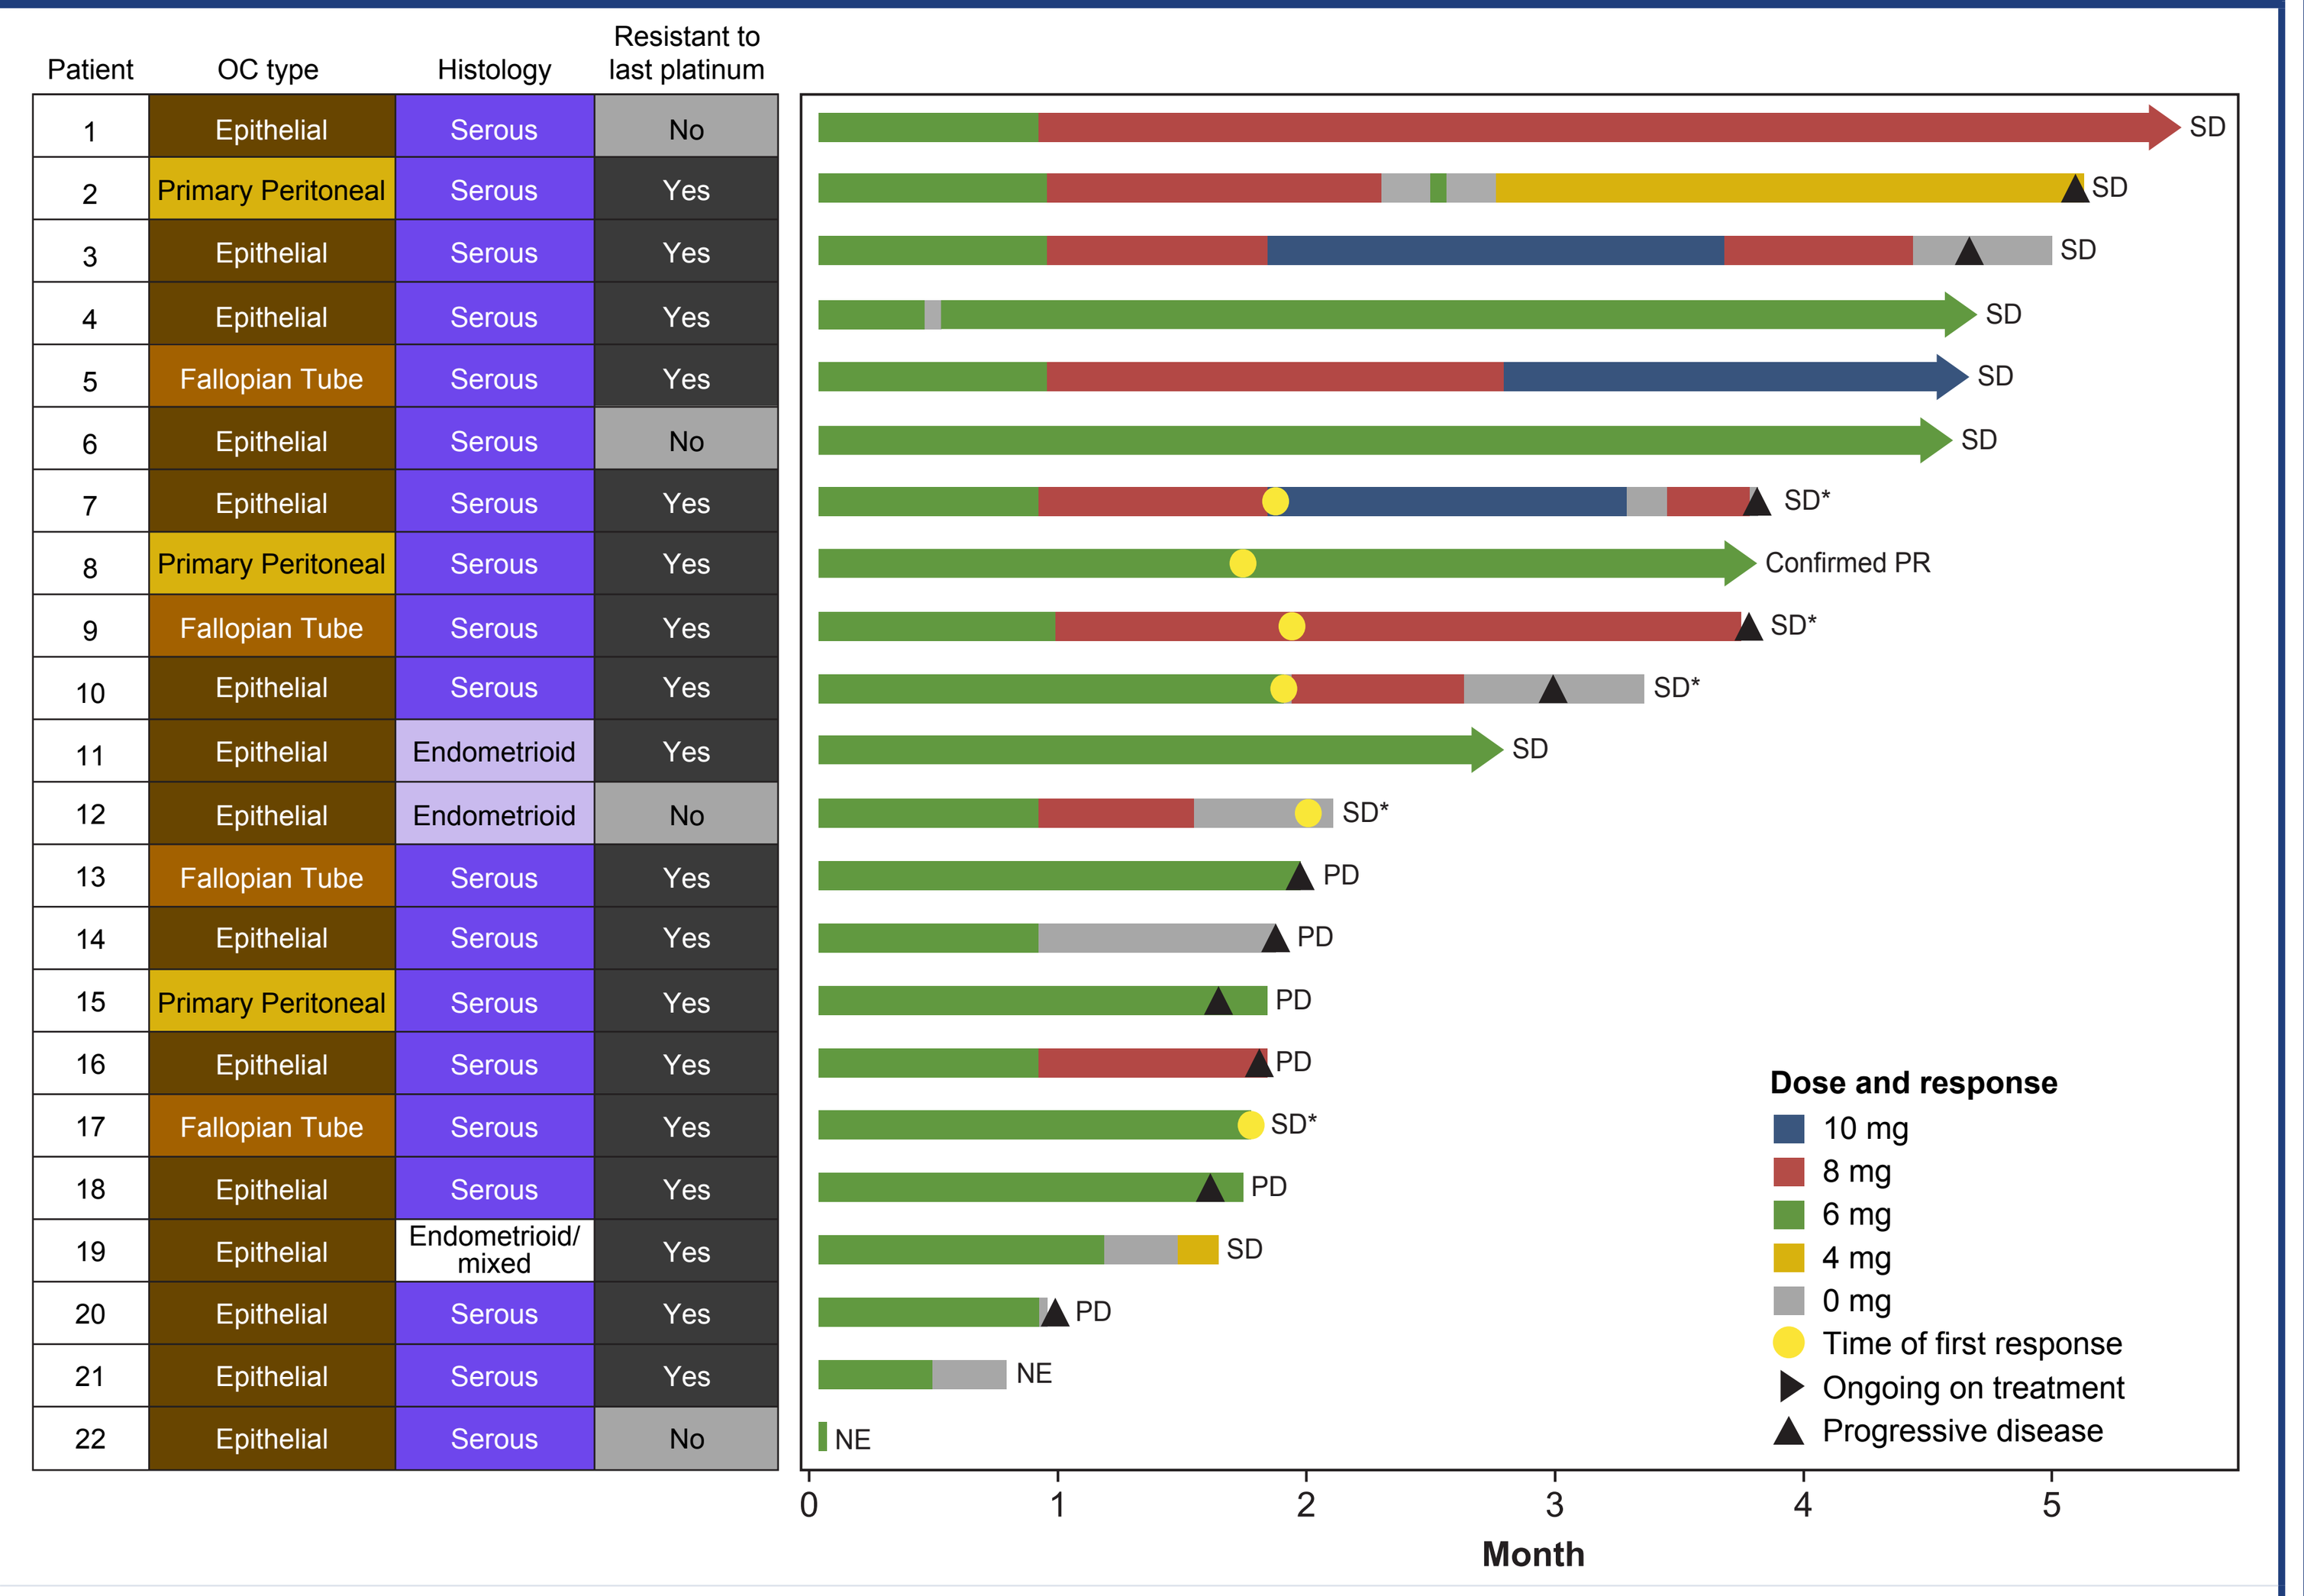
Patient
OC type
Histology
Resistant to last platinum
1 Epithelial Serous No
2 Primary Peritoneal Serous Yes
3 Epithelial Serous Yes
4 Epithelial Serous Yes
5 Fallopian Tube Serous Yes
6 Epithelial Serous No
7 Epithelial Serous Yes
8 Primary Peritoneal Serous Yes
9 Fallopian Tube Serous Yes
10 Epithelial Serous Yes
11 Epithelial Endometrioid Yes
12 Epithelial Endometrioid No
13 Fallopian Tube Serous Yes
14 Epithelial Serous Yes
15 Primary Peritoneal Serous Yes
16 Epithelial Serous Yes
17 Fallopian Tube Serous Yes
18 Epithelial Serous Yes
19 Epithelial Endometrioid/mixed Yes
20 Epithelial Serous Yes
21 Epithelial Serous Yes
22 Epithelial Serous No
SD
SD*
Confirmed PR
PD
NE
Month
Dose and response
10 mg
8 mg
6 mg
4 mg
0 mg
Time of first response
Ongoing on treatment
Progressive disease

Data cutoff: March 29, 2021. Patients could remain on nivolumab following discontinuation of lucitanib. *Unconfirmed PR. NE, not evaluable; OC, ovarian cancer; PD, progressive disease; PR, partial response; SD, stable disease.

Figure 3. Phase 2 OC Cohort: Best Change in Sum of Target Lesions From Baseline

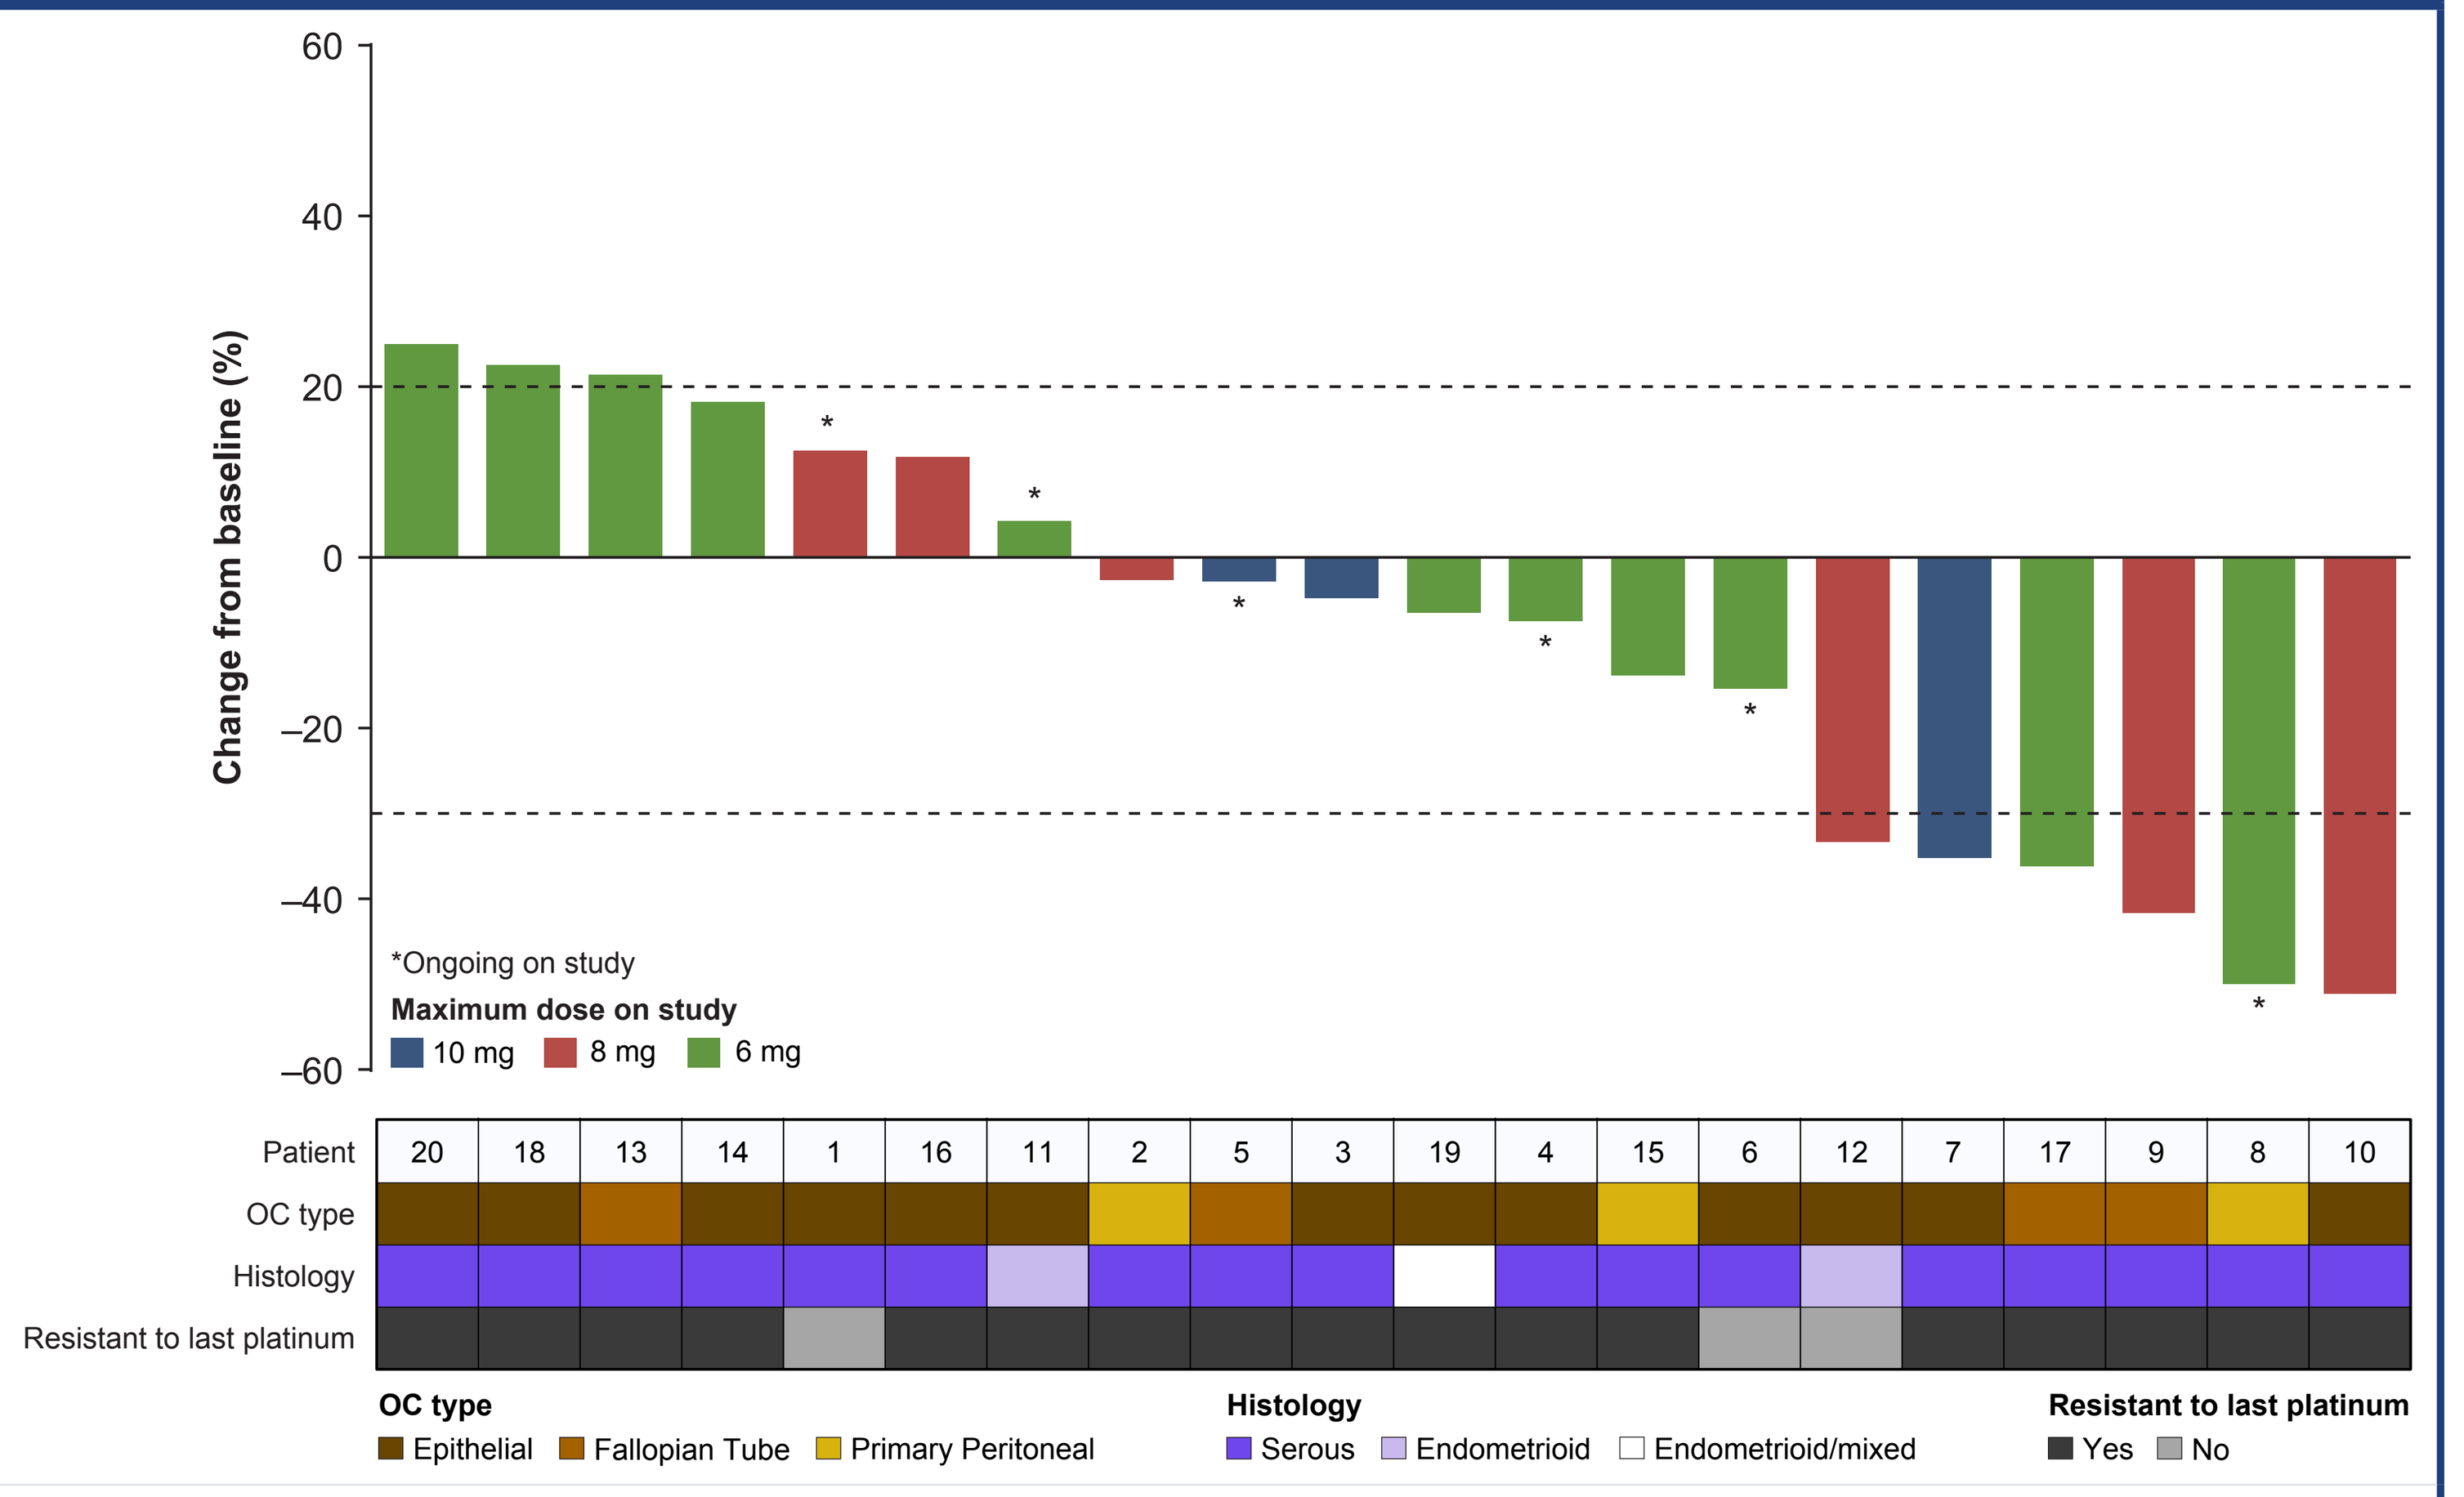
Change from baseline (%)
*Ongoing on study
Maximum dose on study
10 mg
8 mg
6 mg
Patient
20 18 13 14 1 16 11 2 5 3 19 4 15 6 12 7 17 9 8 10
OC type
Histology
Resistant to last platinum
OC type
Epithelial
Fallopian Tube
Primary Peritoneal
Histology
Serous
Endometrioid
Endometrioid/mixed
Resistant to last platinum
Yes
No

Data cutoff: March 29, 2021. OC, ovarian cancer.

Safety

- The phase 2 OC safety population comprised 24 patients who received lucitanib + nivolumab
- Treatment is ongoing with median (range) treatment duration of 2.7 (0.1–6.7+) months
- The most frequent any-grade TEAEs were consistent with the phase 1b experience: fatigue (n=13, 54.2%), hypertension (n=12, 50.0%), nausea (n=8, 33.3%), diarrhea (n=7, 29.2%), headache (n=7, 29.2%), and proteinuria (n=7, 29.2%); see **Supplementary Material** for more detail
 - 10 (41.7%) patients reported a grade ≥3 TEAE, with the most frequently reported being hypertension (n=5, 20.8%)
 - Hypertension was generally manageable with use of antihypertensive medication
 - 3 (12.5%) patients reported a TEAE leading to discontinuation of study treatment (hypertension and angina pectoris; large intestine perforation; diarrhea and fatigue)
 - 5 (20.8%) patients reported serious TEAEs considered related to lucitanib, nivolumab, or the combination; hypertension was the only serious TEAE considered related to study treatment that occurred in >1 patient

ASCO Annual Meeting | June 4–8, 2021 | Virtual Meeting