Genomic Characteristics and Response to Rucaparib and Enzalutamide in the Phase 1b RAMP Study of Metastatic Castration-Resistant Prostate Cancer (mCRPC) Patients

Abstract 445

Arpit Rao,[1] Charles J. Ryan,[1] David Morris,[2] Vasileios Assikis,[3] Gautam Jha,[1] Adriel-John Ablaza,[4] Jenn Habeck,[4] Andrea Loehr,[4] Jim Xiao,[4] Esha A. Gangolli[4]

[1]University of Minnesota, Minneapolis, USA; [2]Urology Associates, P.C., Nashville, USA; [3]Piedmont Cancer Institute, P.C., Atlanta, USA; [4]Clovis Oncology, Inc., Boulder, USA

SUMMARY

- **Unselected patients with mCRPC who had progressed on androgen receptor (AR)-directed therapies reported declines in prostate-specific antigen (PSA) following treatment with the combination of rucaparib 600 mg twice daily and enzalutamide 160 mg once daily**
 - **These declines in PSA were observed even in the presence of *AR* alterations and the absence of DNA damage repair gene alterations**
- **Combination treatment with rucaparib and enzalutamide in patients with mCRPC had a safety profile consistent with that associated with each drug as monotherapy and had no clinically significant drug-drug interactions[1]**
- **These data support further study of the rucaparib and enzalutamide combination**
 - **The phase 3 CASPAR study (NCT04455750) is enrolling biomarker-unselected patients with mCRPC**

Poster Download
Also available at: https://clovisoncology.com/files/AACR2021_Rao_Poster.pdf
Copies of this e-poster obtained through Quick Response (QR) codes and/or web links are for personal use only and may not be reproduced without written permission of the authors.
Corresponding author: *Arpit Rao*; raoa@umn.edu.

Poster Slides
Also available at: https://clovisoncology.com/files/AACR2021_Rao_Slides.pdf
Please scan this QR code or visit the website to view the poster as slides.

REFERENCES

1. Rao et al. *J Clin Oncol*. 2021;39(suppl 6):Abst 79.
2. Rubraca (rucaparib) tablets [prescribing information]. Boulder, CO: Clovis Oncology, Inc.; 2020.
3. Xtandi (enzalutamide) tablets [prescribing information]. Northbrook, IL: Astellas Pharma US, Inc.; 2020.
4. Asim et al. *Nature Comm*. 2017;8(1):374.
5. Clarke et al. *Lancet Oncol*. 2018;19(7):975-86.

ACKNOWLEDGMENTS

This study was funded by Clovis Oncology, Inc. Medical writing and editorial support funded by Clovis Oncology, Inc., were provided by Shelly Lim and Frederique H. Evans of Ashfield MedComms, an Ashfield Health company.

INTRODUCTION

- Rucaparib, a poly(ADP-ribose) polymerase (PARP) inhibitor, is approved in the United States as monotherapy for patients with *BRCA1/2*-mutated mCRPC who have been treated with androgen receptor (AR)-directed therapy and a taxane[2]
- Enzalutamide is an AR-signaling inhibitor approved in the United States for the treatment of patients with CRPC or metastatic castration-sensitive prostate cancer[3]
- Synthetic lethality has been observed, independent of DNA damage repair (DDR) gene defects, when combining AR-directed therapy (eg, enzalutamide) and a PARP inhibitor for prostate cancer[4,5]
- Preliminary results from the phase 1b RAMP study (NCT04179396) investigating the combination of rucaparib and enzalutamide in unselected patients with mCRPC have shown promising signs of antitumor activity, and its safety profile was consistent with that of each drug when used as monotherapy[1]
 - Concomitant treatment with rucaparib and enzalutamide did not lead to significant changes in the pharmacokinetic (PK) profile for either agent or their metabolites (M324 and *N*-desmethyl enzalutamide)
- Here, we evaluated the association between treatment response and genomic alterations of known clinical significance in mCRPC among patients from the RAMP study

METHODS

- Enrolled patients had a histologically or cytologically confirmed adenocarcinoma or poorly differentiated carcinoma of the prostate after receiving 0–2 lines of AR-directed therapy (eg, enzalutamide, abiraterone) and ≤2 lines of chemotherapy for mCRPC; prior PARP inhibitor treatment was not allowed
- Patients received rucaparib monotherapy (600 mg twice daily) during a 1-week run-in period, followed by rucaparib (600 mg twice daily) + enzalutamide (160 mg once daily) in continuous 28-day cycles
- Dose-limiting toxicities were assessed during the first 2 cycles of combination treatment, and trough PK was evaluated for rucaparib and enzalutamide and their respective metabolites
- Primary endpoints were PK and safety for the combination[1]
- Secondary endpoints were the change from baseline in prostate-specific antigen (PSA) levels and the objective response rate per modified Response Evaluation Criteria In Solid Tumors version 1.1 (RECIST) and Prostate Cancer Clinical Trials Working Group 3
 - A confirmed PSA response was defined as a ≥50% reduction in PSA from baseline confirmed by a consecutive measurement ≥3 weeks later
- An exploratory objective was to evaluate the association between genomic alterations and clinical outcomes from treatment
 - Next-generation sequencing was performed on formalin-fixed paraffin-embedded tumor tissues or circulating cell-free DNA extracted from plasma samples to detect DDR gene alterations (eg, *BRCA1/2*) or genomic signatures associated with resistance to treatment
 - Sequencing was performed in Clinical Laboratory Improvement Amendments-approved facilities using the FoundationOne®CDx and FoundationOne®Liquid CDx laboratory-developed tests (Foundation Medicine, Inc., Cambridge, MA), which covered 324 and 70 cancer-related genes, respectively

RESULTS

- As of December 2, 2020, 8 patients had received treatment (**Table 1**)
 - Seven (87.5%) out of 8 patients had at least 1 prior AR-directed therapy

Table 1. Patient Demographics and Baseline Characteristics

Characteristic	Patients (N=8)
Age, median (range), y	68.5 (56.0–79.0)
Race, n (%)	
White	7 (87.5)
Asian	1 (12.5)
ECOG PS, n (%)	
0	5 (62.5)
1	3 (37.5)
Baseline PSA, median (range), ng/mL	20.0 (2.5–155.0)
Gleason score ≥8, n (%)[a]	7 (87.5)
Number of prior mCRPC therapies, n (%)[b]	
0	1 (12.5)
1	1 (12.5)
2	4 (50.0)
≥3	2 (25.0)
Number of prior AR-directed therapies, n (%)	
0	1 (12.5)
1	6 (75.0)
2	1 (12.5)

Visit cutoff date: December 2, 2020.
[a]One patient had an unknown Gleason score. [b]Defined based on class of agent and not disease status at time of treatment; does not include luteinizing hormone–releasing hormone analogues, first-generation antiandrogens, or hormones.
AR, androgen receptor; ECOG PS, Eastern Cooperative Oncology Group performance status; mCRPC, metastatic castration-resistant prostate cancer; PSA, prostate-specific antigen.

Genomic Characteristics

- Genomic data were available for 6 (75%) of 8 treated patients, all of whom had received prior AR-directed therapy (**Figure 1**)
 - *AR* alterations, including rearrangements, amplifications, and single nucleotide variants, were observed in 4 (67%) patients (**Table 2**)
 - Mutations in *TP53* were detected in 4 (67%) patients and *PTEN* in 2 (33%) patients
 - No *BRCA1*, *BRCA2*, or *PALB2* alterations were detected
 - The only DDR gene alteration found was a subclonal (<1% allele frequency) alteration of *CHEK2* in 1 (17%) patient (**Figure 2**)

Figure 1. PSA Change From Baseline Over Time in Patients Treated With Rucaparib and Enzalutamide

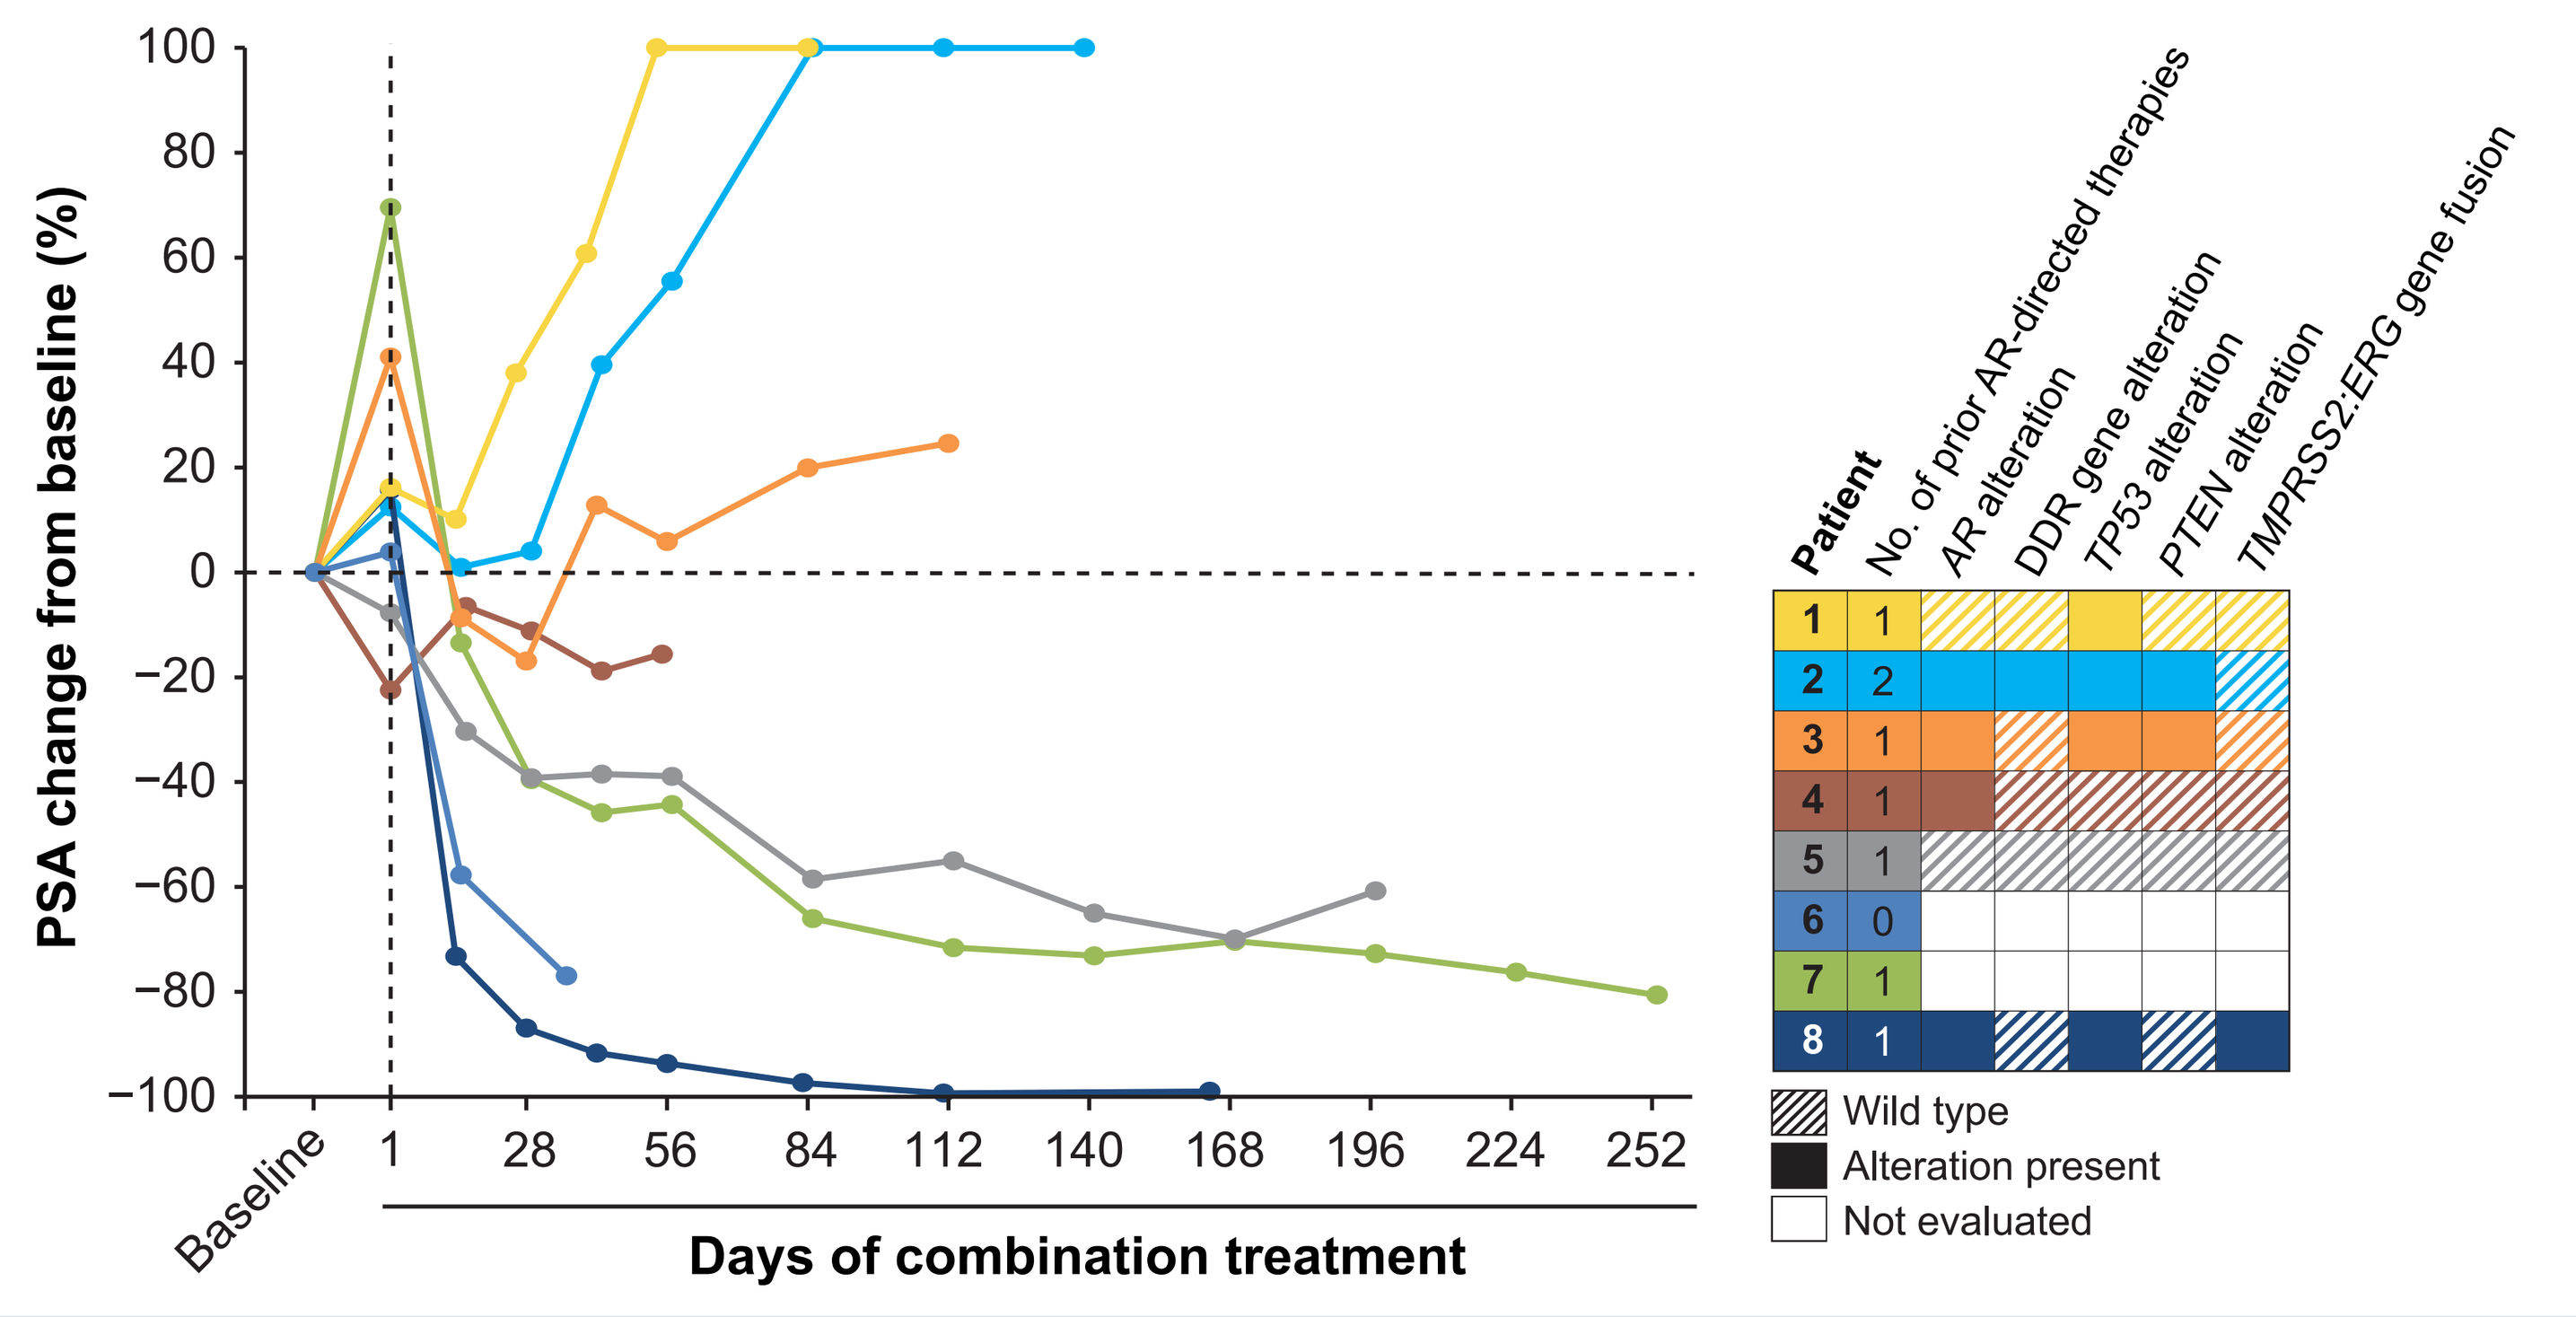
PSA change from baseline (%)
100
80
60
40
20
0
−20
−40
−60
−80
−100
Baseline
1
28
56
84
112
140
168
196
224
252
Days of combination treatment
Patient
No. of prior AR-directed therapies
AR alteration
DDR gene alteration
TP53 alteration
PTEN alteration
TMPRSS2:ERG gene fusion
1 1
2 2
3 1
4 1
5 1
6 0
7 1
8 1
Wild type
Alteration present
Not evaluated

Visit cutoff: December 2, 2020. PSA change from baseline were capped at 100% for visual clarity. The vertical dashed line indicates the start of the combination treatment following 1-week run-in with rucaparib monotherapy.
AR, androgen receptor; DDR, DNA damage repair; PSA, prostate-specific antigen.

Efficacy

- Overall, 6 (75%) of 8 patients reported a decline in PSA from baseline, including 4 (50%) with a confirmed PSA response (**Figure 2**)
- Patients previously treated with AR-directed therapies demonstrated reductions in PSA following the combination treatment even in the presence of multiple *AR* alterations and in the absence of DDR gene alterations (**Table 2**)
 - Patient 4 had 3 *AR* alterations including *AR* H875Y (variant allele frequency [VAF], 3.1%), *AR* L702H (3.8%), and rearrangement and reported a best PSA change from baseline of –22.5%
 - Patient 8, with a low allele frequency point mutation in *AR* T878A (VAF, <1%), had the greatest reduction in PSA from baseline (best PSA change of –99.3%) and achieved a confirmed complete radiographic response
 - The patient also had a *TP53* alteration (*TP53* R248Q) and *TMPRSS2:ERG* gene fusion
- At the time of data cutoff, 3 patients were still ongoing on study treatment, including 2 (25%) who remained on study for >6 cycles

Table 2. Genetic Alterations in *AR*

Patient	Prior AR therapies	AR alteration (variant allelic frequency)	Best PSA change from baseline, %
2	Abiraterone, enzalutamide	Amplification, rearrangement	0.9
3	Abiraterone	Amplification, rearrangement	−16.9
4	Abiraterone	H875Y (3.1%), L702H (3.8%), rearrangement	−22.5
8	Abiraterone	T878A (0.8%)	−99.3

Visit cutoff date: December 2, 2020.
AR, androgen receptor; PSA, prostate-specific antigen.

Figure 2. Best Percent Change in PSA From Baseline in Patients Treated With Rucaparib and Enzalutamide

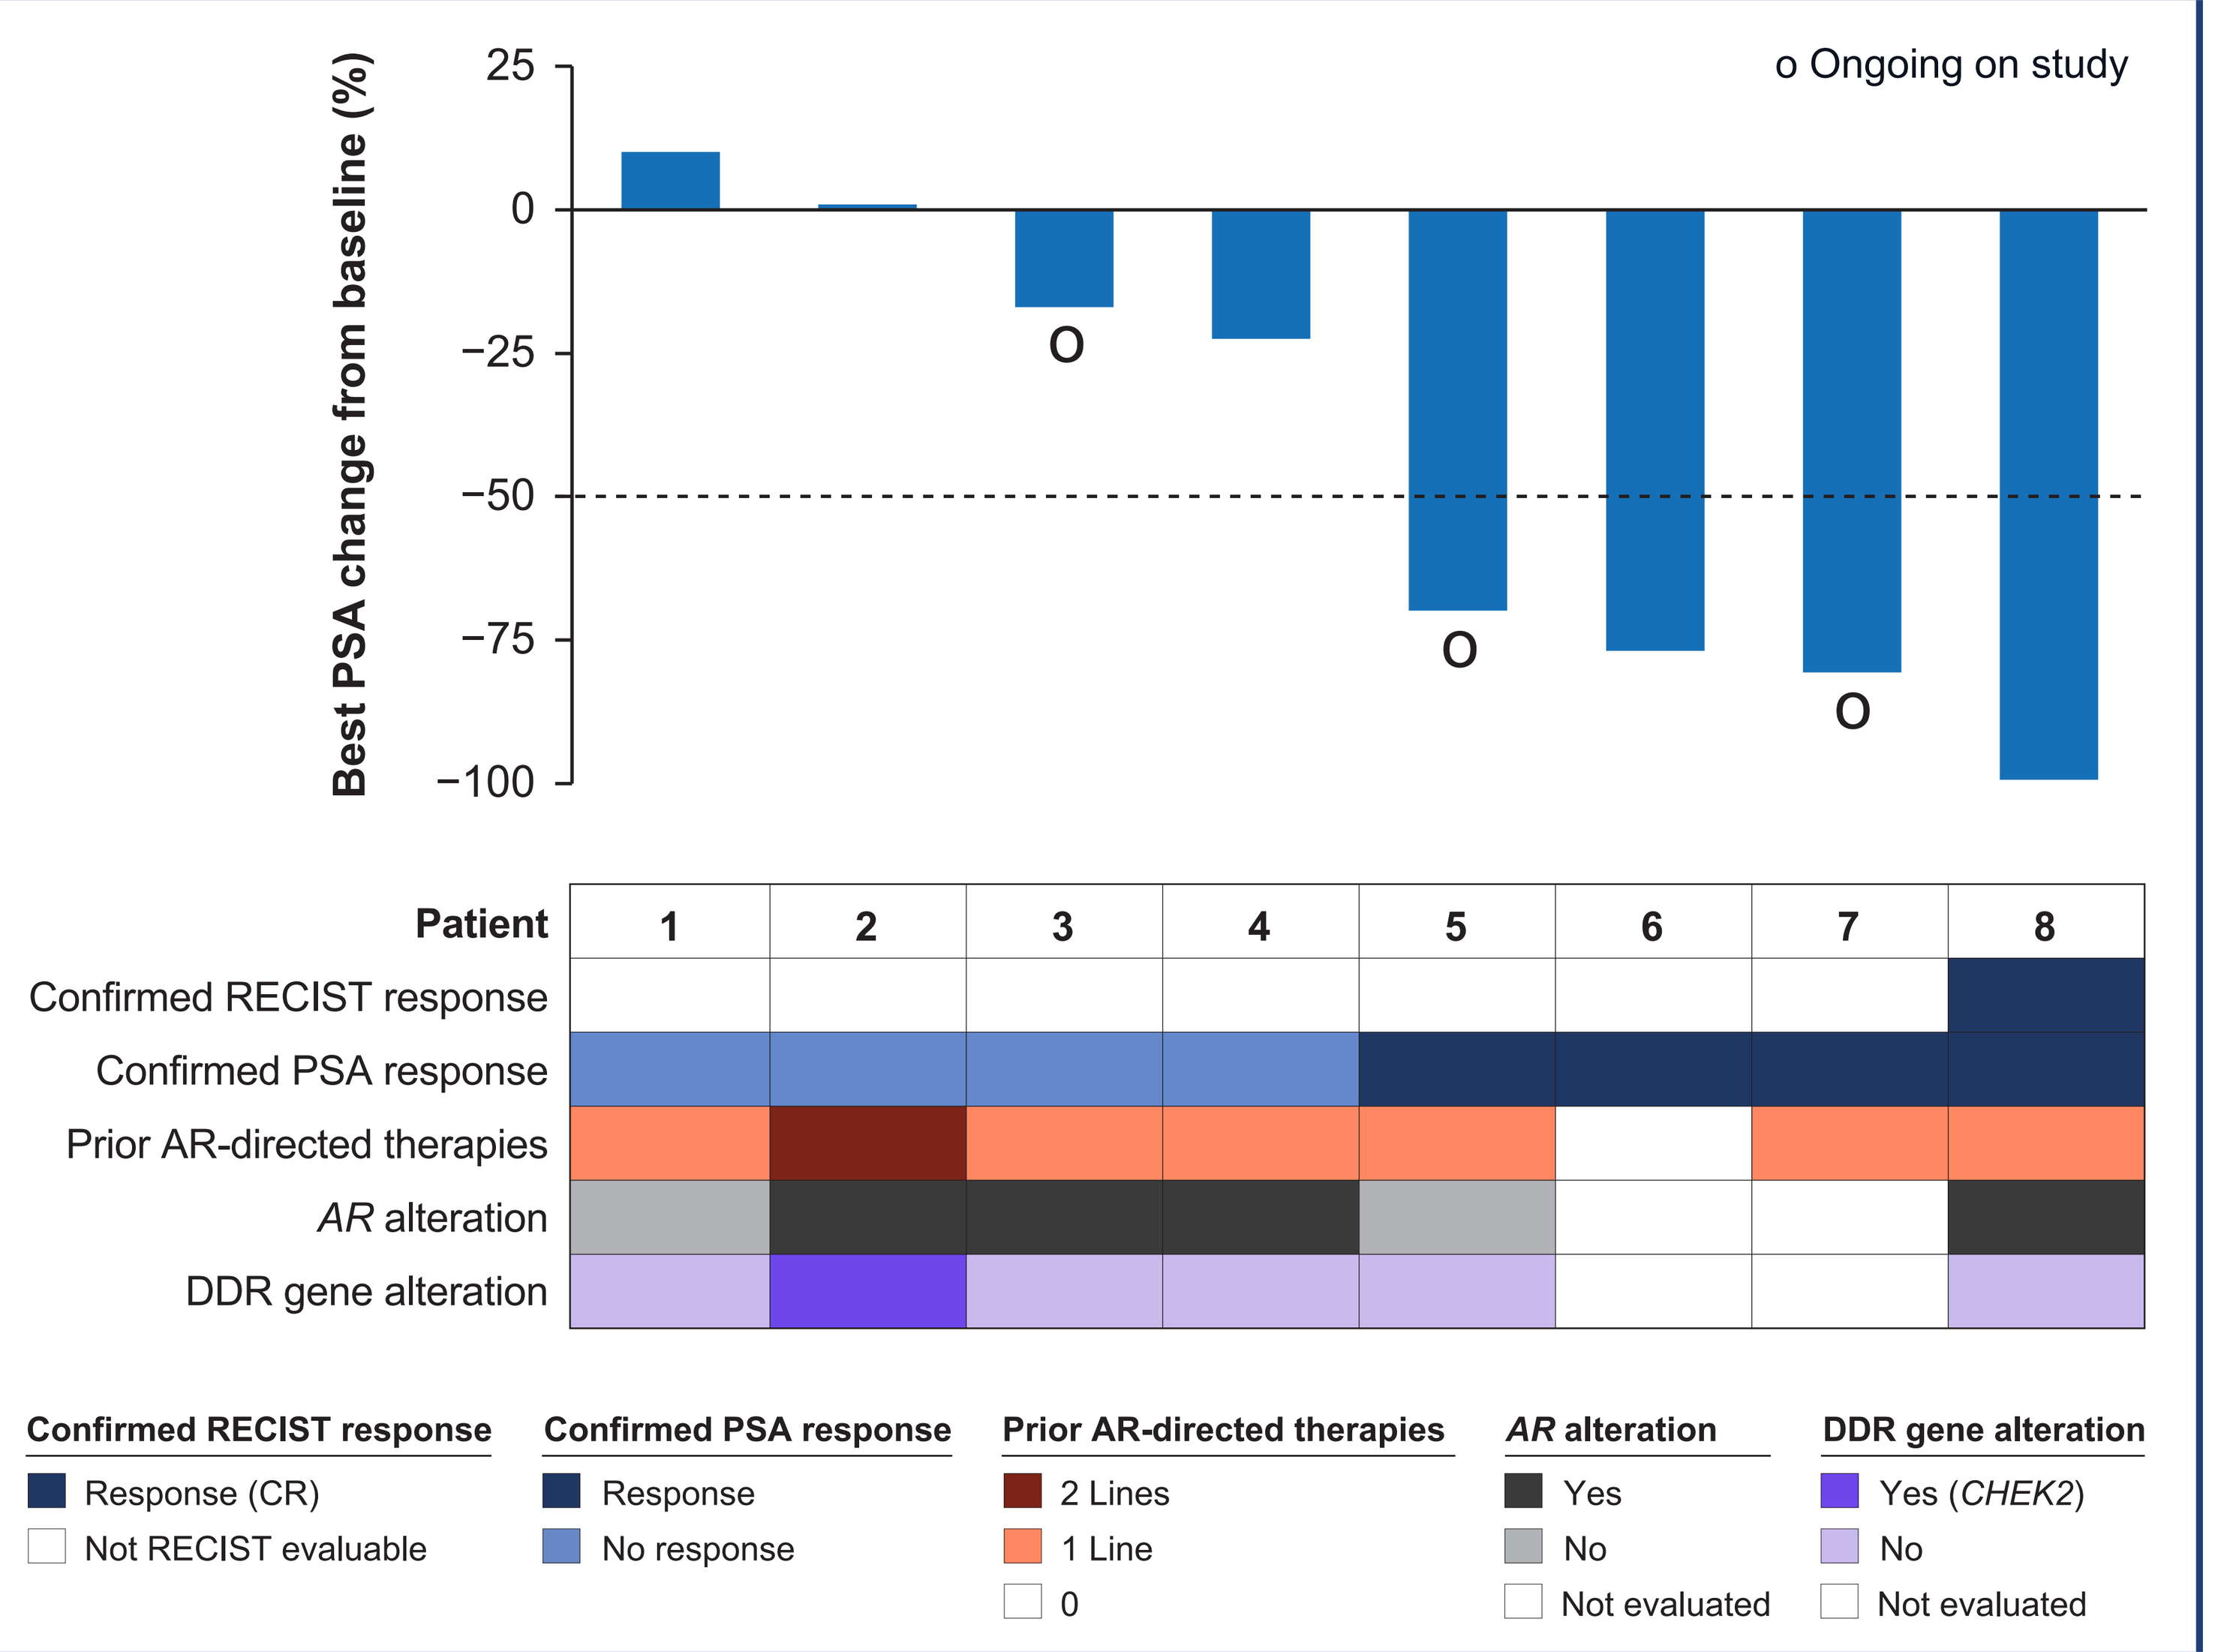
o Ongoing on study
Best PSA change from baseline (%)
25
0
−25
−50
−75
−100
Patient
1
2
3
4
5
6
7
8
Confirmed RECIST response
Confirmed PSA response
Prior AR-directed therapies
AR alteration
DDR gene alteration
Confirmed RECIST response
Response (CR)
Not RECIST evaluable
Confirmed PSA response
Response
No response
Prior AR-directed therapies
2 Lines
1 Line
0
AR alteration
Yes
No
Not evaluated
DDR gene alteration
Yes (CHEK2)
No
Not evaluated

Visit cutoff: December 2, 2020. The horizontal dashed line indicates the threshold for PSA response, a 50% decrease from baseline.
AR, androgen receptor; CR, complete response; DDR, DNA damage repair; PSA, prostate-specific antigen; RECIST, Response Evaluation Criteria In Solid Tumors version 1.1.

AACR Annual Meeting | April 10–15, 2021 | Virtual Meeting