Abstract 3650

Preclinical evaluation of the PARP inhibitor rucaparib in combination with PD-1 and PD-L1 inhibition in a syngeneic *BRCA1* mutant ovarian cancer model

Liliane Robillard[1], Minh Nguyen[1], Andrea Loehr[1], Sandra Orsulic[2], Rebecca Kristeleit[3], Kevin Lin[1], Mitch Raponi[1], Thomas C. Harding[1], and Andrew D. Simmons[1]

[1]Clovis Oncology Inc., Boulder, CO, USA; [2]Cedars-Sinai Medical Center, Los Angeles, CA, USA; [3]University College London Cancer Institute, London, UK

BACKGROUND

- Rucaparib is an oral small molecule inhibitor of poly (ADP-ribose) polymerase (PARP) enzymes, including PARP-1, PARP-2, and PARP-3, and is approved for the treatment of ovarian cancer associated with germline or somatic alterations in *BRCA1* or *BRCA2*.[1]
- Monoclonal antibodies against programmed death receptor-1 (PD-1) and programmed death-ligand (PD-L1) have also shown efficacy in advanced ovarian cancer patients.[2]
- *BRCA1* and *BRCA2* mutated tumors have been reported to have a higher mutational load and increased CD8 T-cell infiltration, suggesting that the combination of rucaparib and immune checkpoint inhibition may be complementary.[3]
- PARP inhibition has also been reported to have an immunosuppressive effect in preclinical studies.[4]
- We investigated rucaparib in combination with PD-1 and PD-L1 inhibition in wild-type *BRCA* and *BRCA1* null syngeneic models.

Rucaparib has limited in vitro activity in wild-type *BRCA* syngeneic mouse models

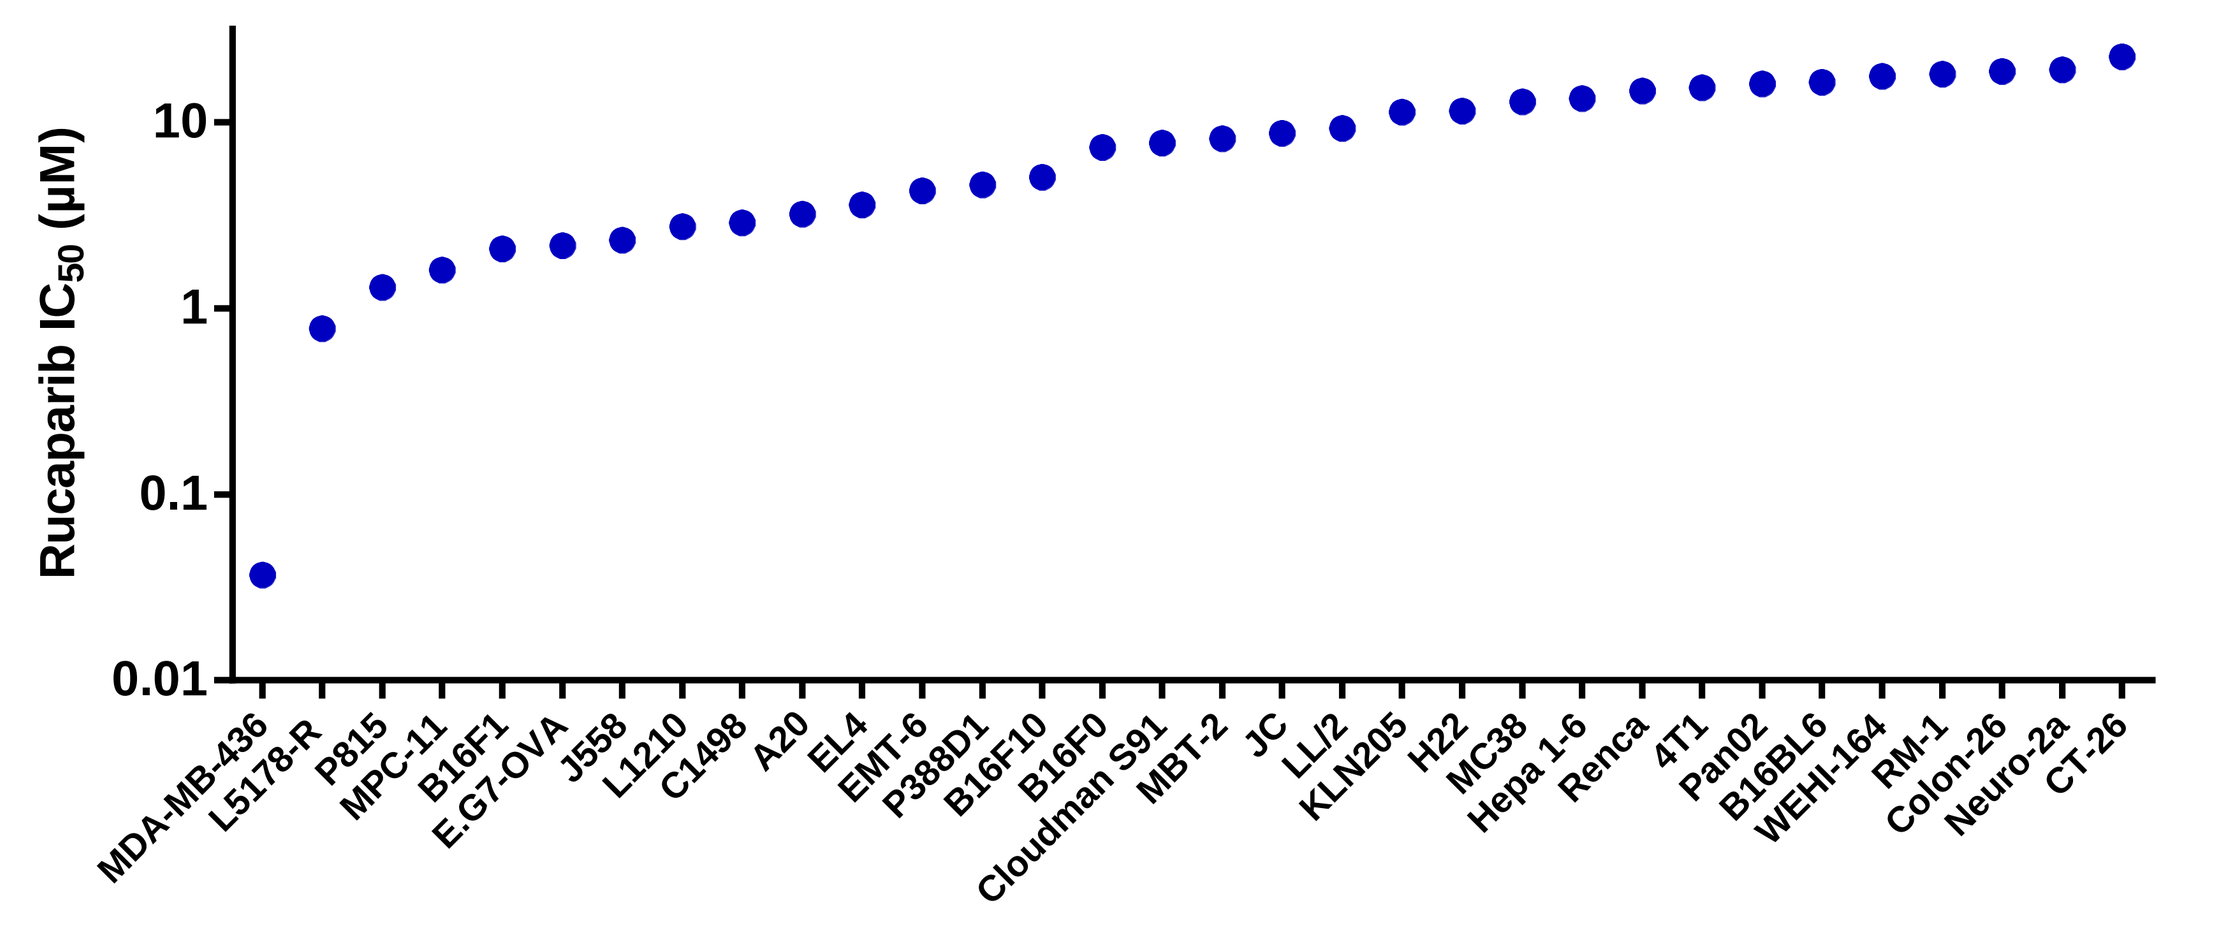
Rucaparib IC50 (µM)
MDA-MB-436 L5178-R P815 MPC-11 B16F1 E.G7-OVA J558 L1210 C1498 A20 EL4 EMT-6 P388D1 B16F10 B16F0 Cloudman S91 MBT-2 JC LL/2 KLN205 H22 MC38 Hepa 1-6 Renca 4T1 Pan02 B16BL6 WEHI-164 RM-1 Colon-26 Neuro-2a CT-26

- A panel of 31 tumorigenic syngeneic cancer cell lines was identified and screened for in vitro sensitivity to rucaparib in 6-day cell viability assays. No *BRCA* mutations have been reported in these cell lines. The human MDA-MB-436 (*BRCA1* mutant) cell line was also evaluated as a positive control.

Rucaparib demonstrated potent activity in a *BRCA1* null ovarian cancer syngeneic cell line

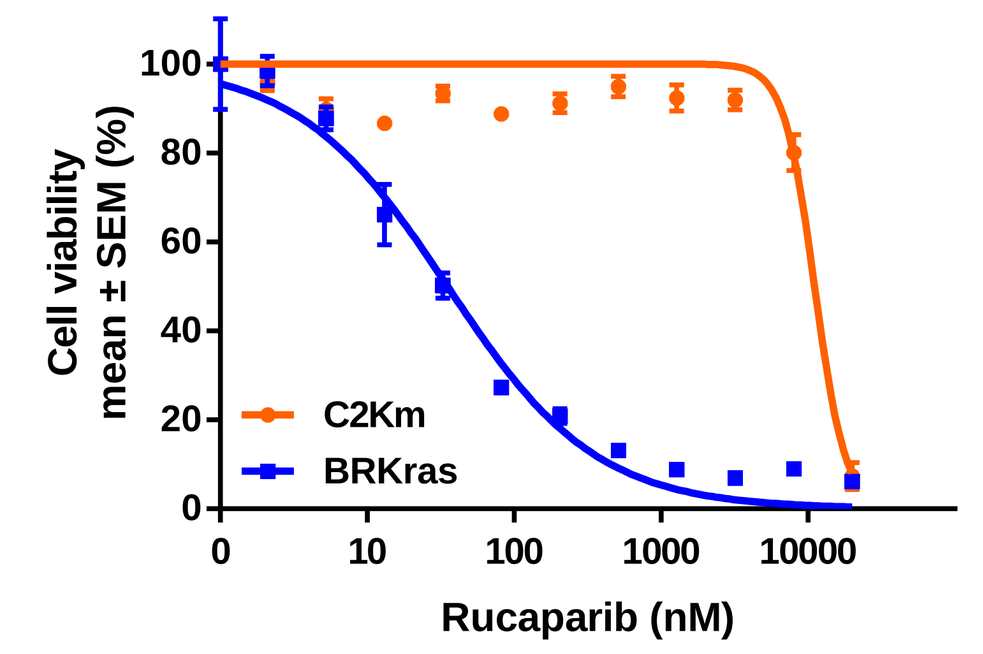
A
Cell viability mean ± SEM (%)
C2Km
BRKras
Rucaparib (nM)

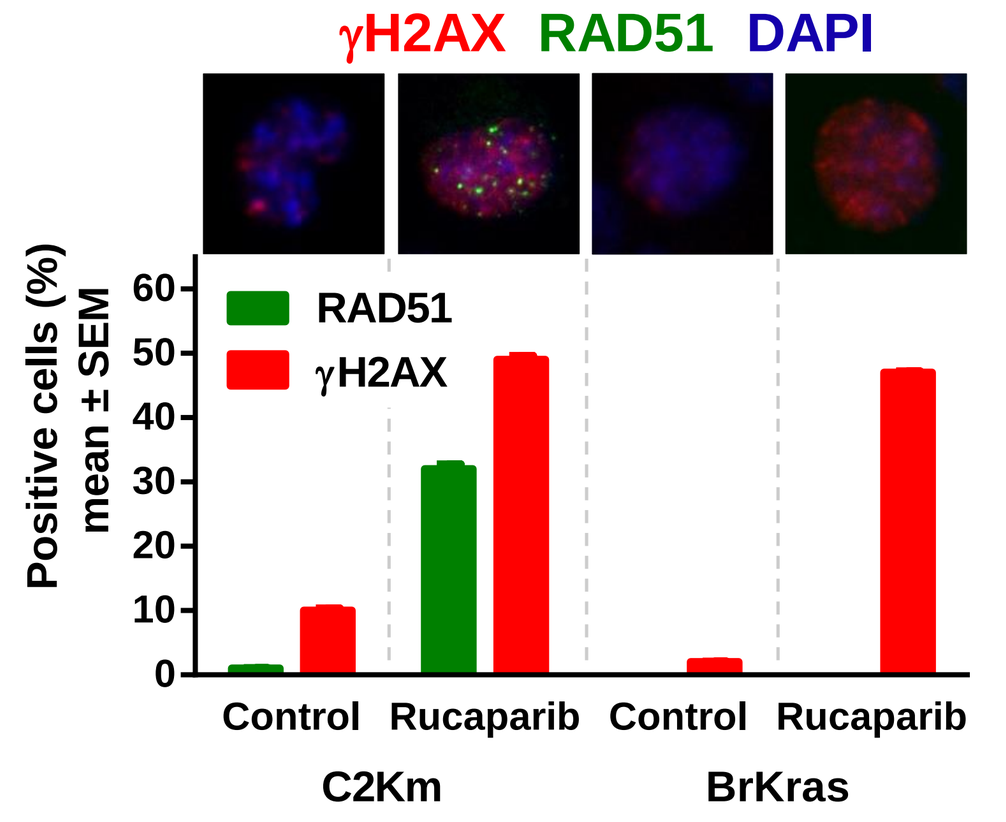
B
γH2AX RAD51 DAPI
Positive cells (%) mean ± SEM
RAD51
γH2AX
Control Rucaparib Control Rucaparib
C2Km BrKras

Cell Line	Genetic Modification	Rucaparib IC_{50} (nM) mean ± SD	RAD51 Foci
C2Km	p53-/-; myc; Kras-G12D; Akt-myr	12766 ± 1704	32-fold induction
BrKras	BRCA1-/-; p53-/-; myc; Kras-G12D; Akt-myr	83 ± 8	no induction

- Rucaparib sensitivity was evaluated in the C2Km (wild-type *BRCA1*) and BrKras (*BRCA1* null) syngeneic genetically engineered ovarian cancer cell lines[5] in a 6-day cell viability assay (A). Immunofluorescence staining confirmed reduced RAD51 foci formation, and thus homologous recombination (HR) deficiency, in the BrKras cell line as compared to the parental C2Km cell line (B).

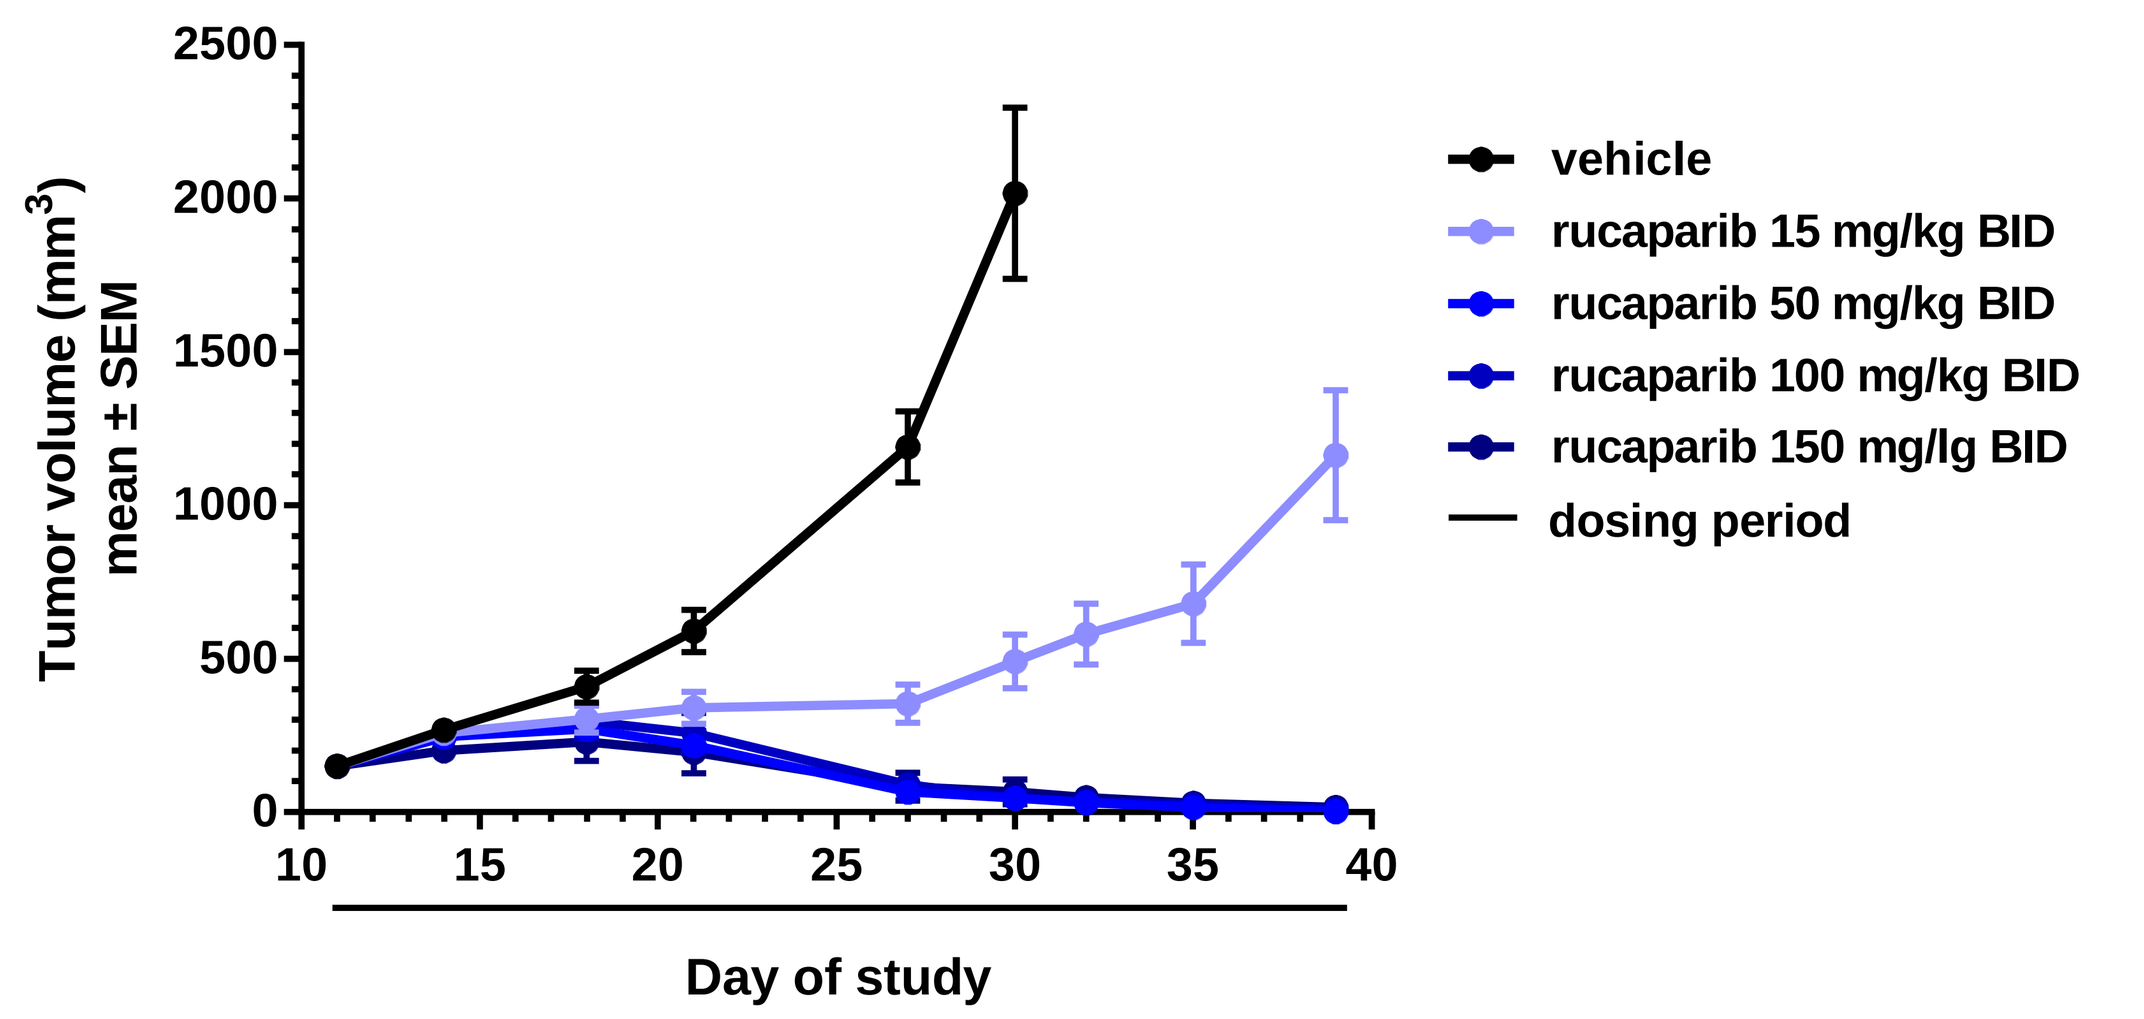
BrKras (BRCA1 null)
Tumor volume (mm3) mean ± SEM
vehicle
rucaparib 15 mg/kg BID
rucaparib 50 mg/kg BID
rucaparib 100 mg/kg BID
rucaparib 150 mg/lg BID
dosing period
Day of study

- The in vivo activity of rucaparib was assessed in the *BRCA1* null BrKras model. Animals were treated at the dose and schedules indicated.

Rucaparib did not hinder the in vivo activity of PD-1 and PD-L1 inhibition in wild-type *BRCA* syngeneic models

EMT-6

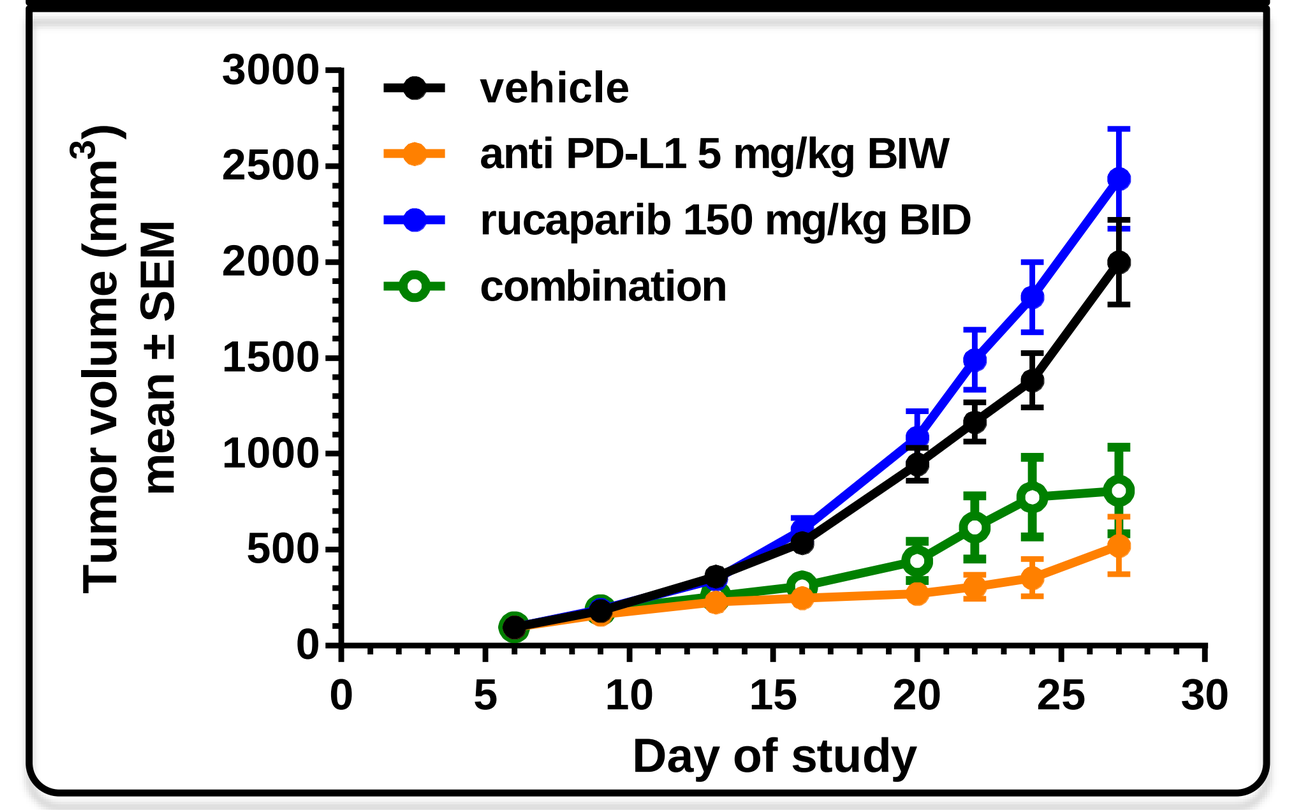
Tumor volume (mm3) mean ± SEM
vehicle
anti PD-L1 5 mg/kg BIW
rucaparib 150 mg/kg BID
combination
Day of study

Pan02

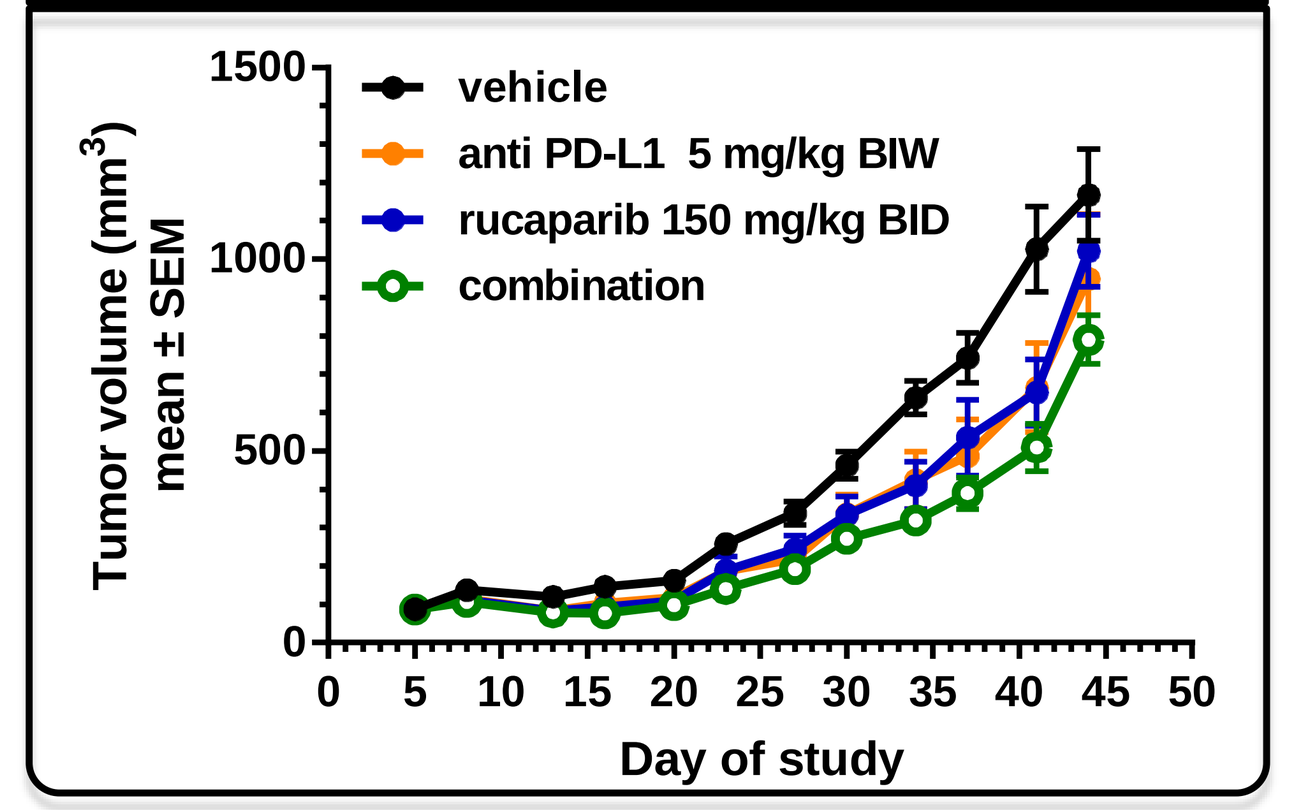
Tumor volume (mm3) mean ± SEM
vehicle
anti PD-L1 5 mg/kg BIW
rucaparib 150 mg/kg BID
combination
Day of study

MC38

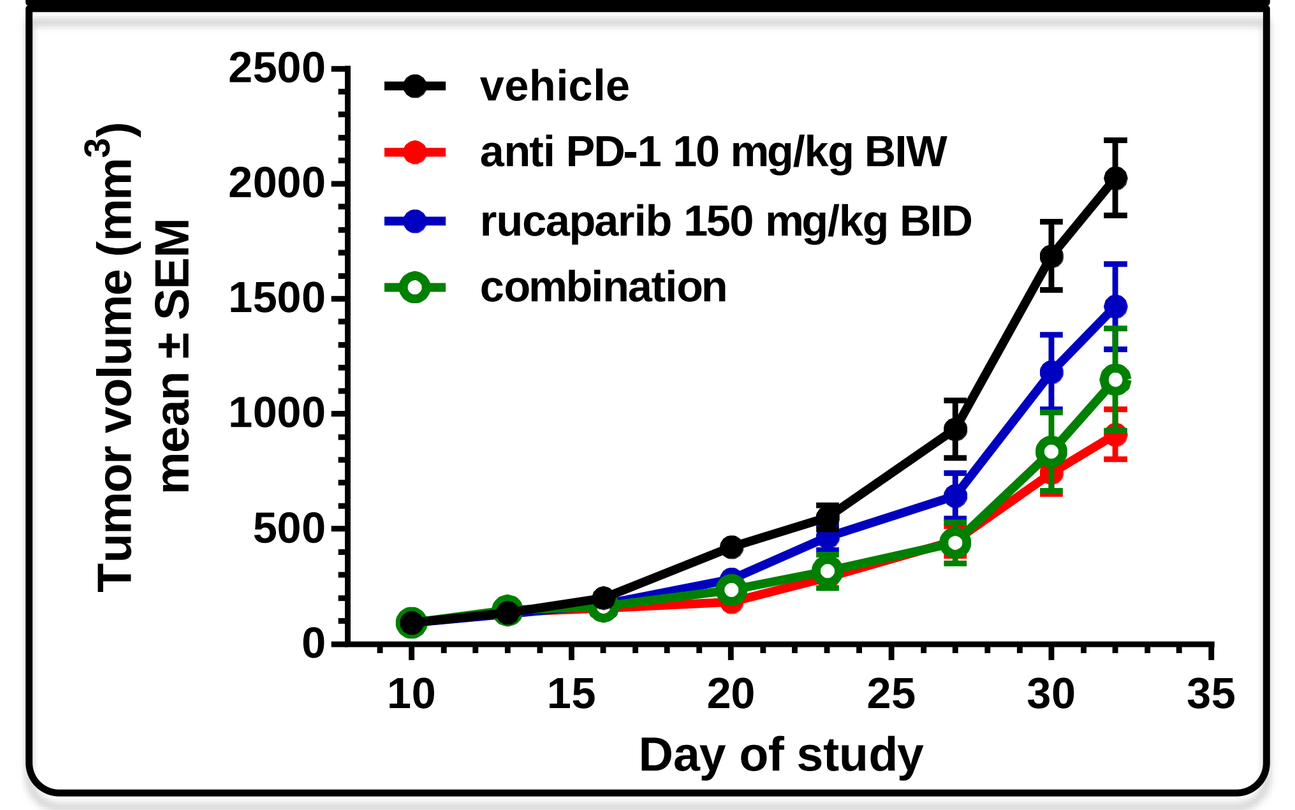
Tumor volume (mm3) mean ± SEM
vehicle
anti PD-1 10 mg/kg BIW
rucaparib 150 mg/kg BID
combination
Day of study

- Efficacy studies were performed using the syngeneic Pan02 (pancreas), MC38 (colon), and EMT-6 (breast) models. No *BRCA* mutations have been reported in these cell lines. The dose and schedule of the treatments were as indicated. No significant difference was observed between the anti-PD-1 or anti-PD-L1 single agent and combination treated groups in the 3 models evaluated.

Rucaparib and anti-PD-1/PD-L1 combinations had greater efficacy in *BRCA1* null syngeneic model

Anti-PD-1

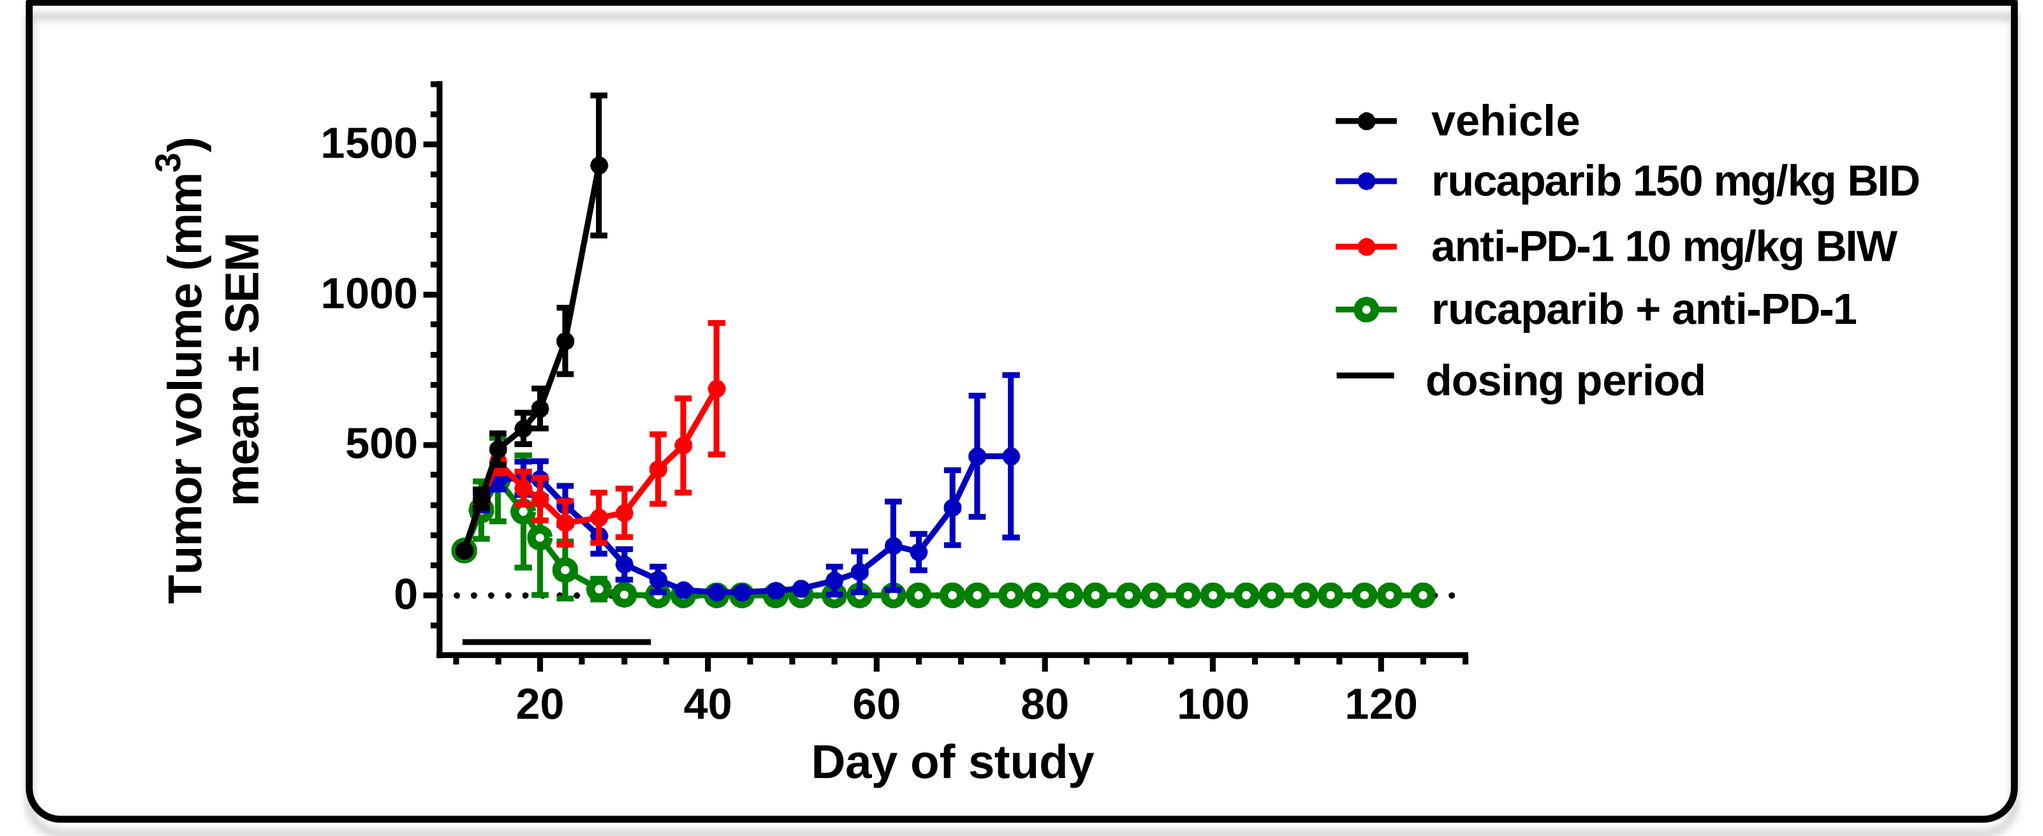
Tumor volume (mm3) mean ± SEM
vehicle
rucaparib 150 mg/kg BID
anti-PD-1 10 mg/kg BIW
rucaparib + anti-PD-1
dosing period
Day of study

Anti-PD-L1

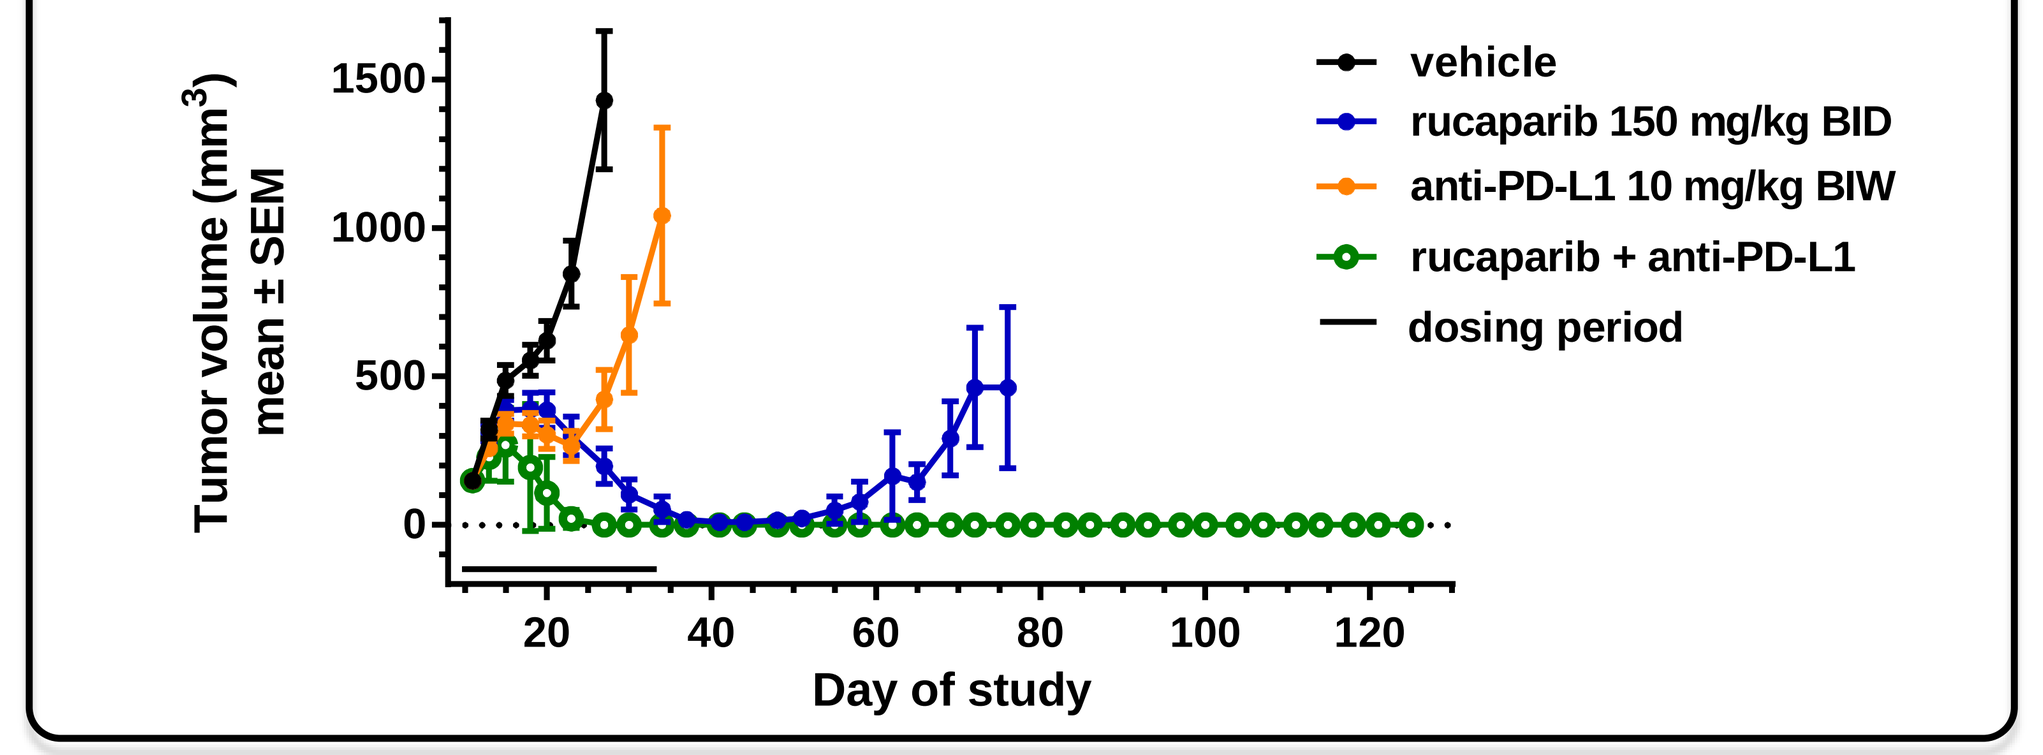
Tumor volume (mm3) mean ± SEM
vehicle
rucaparib 150 mg/kg BID
anti-PD-L1 10 mg/kg BIW
rucaparib + anti-PD-L1
dosing period
Day of study

Survival analysis

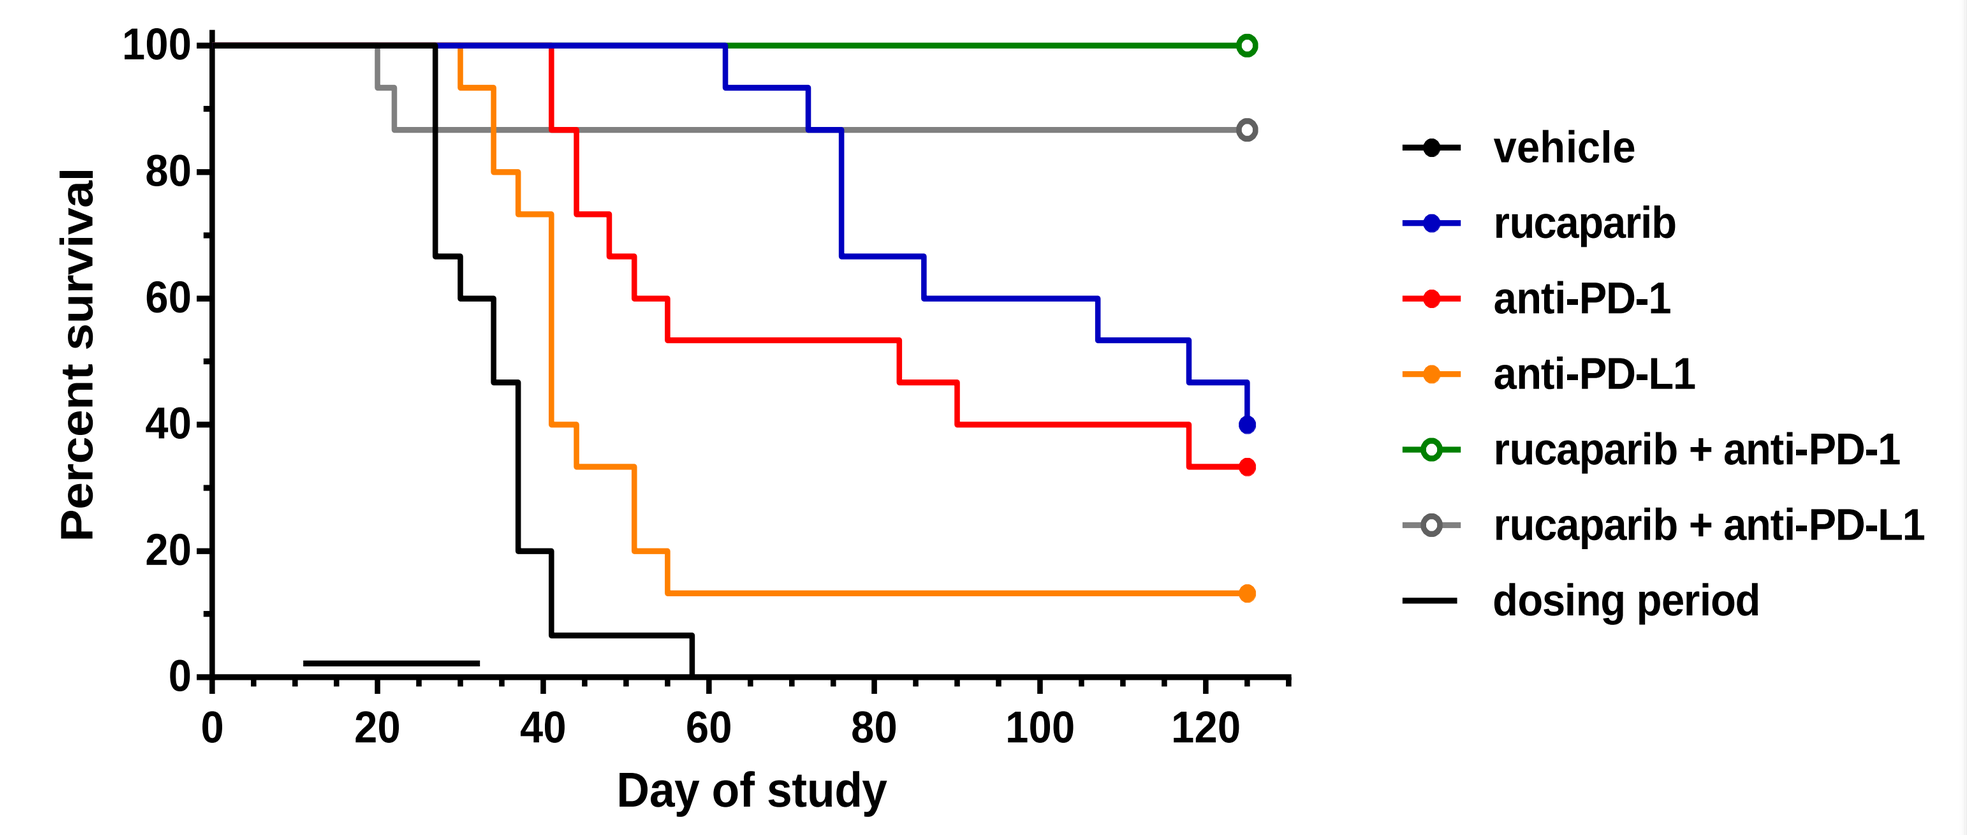
Percent survival
vehicle
rucaparib
anti-PD-1
anti-PD-L1
rucaparib + anti-PD-1
rucaparib + anti-PD-L1
dosing period
Day of study

- The BrKras (*BRCA1* null) syngeneic model was used to assess the impact of combining rucaparib with anti-PD-1 and anti-PD-L1 inhibition. The dosing and schedules are indicated; the combination groups were treated at the same dose and schedule as the monotherapy groups.

Re-challenge

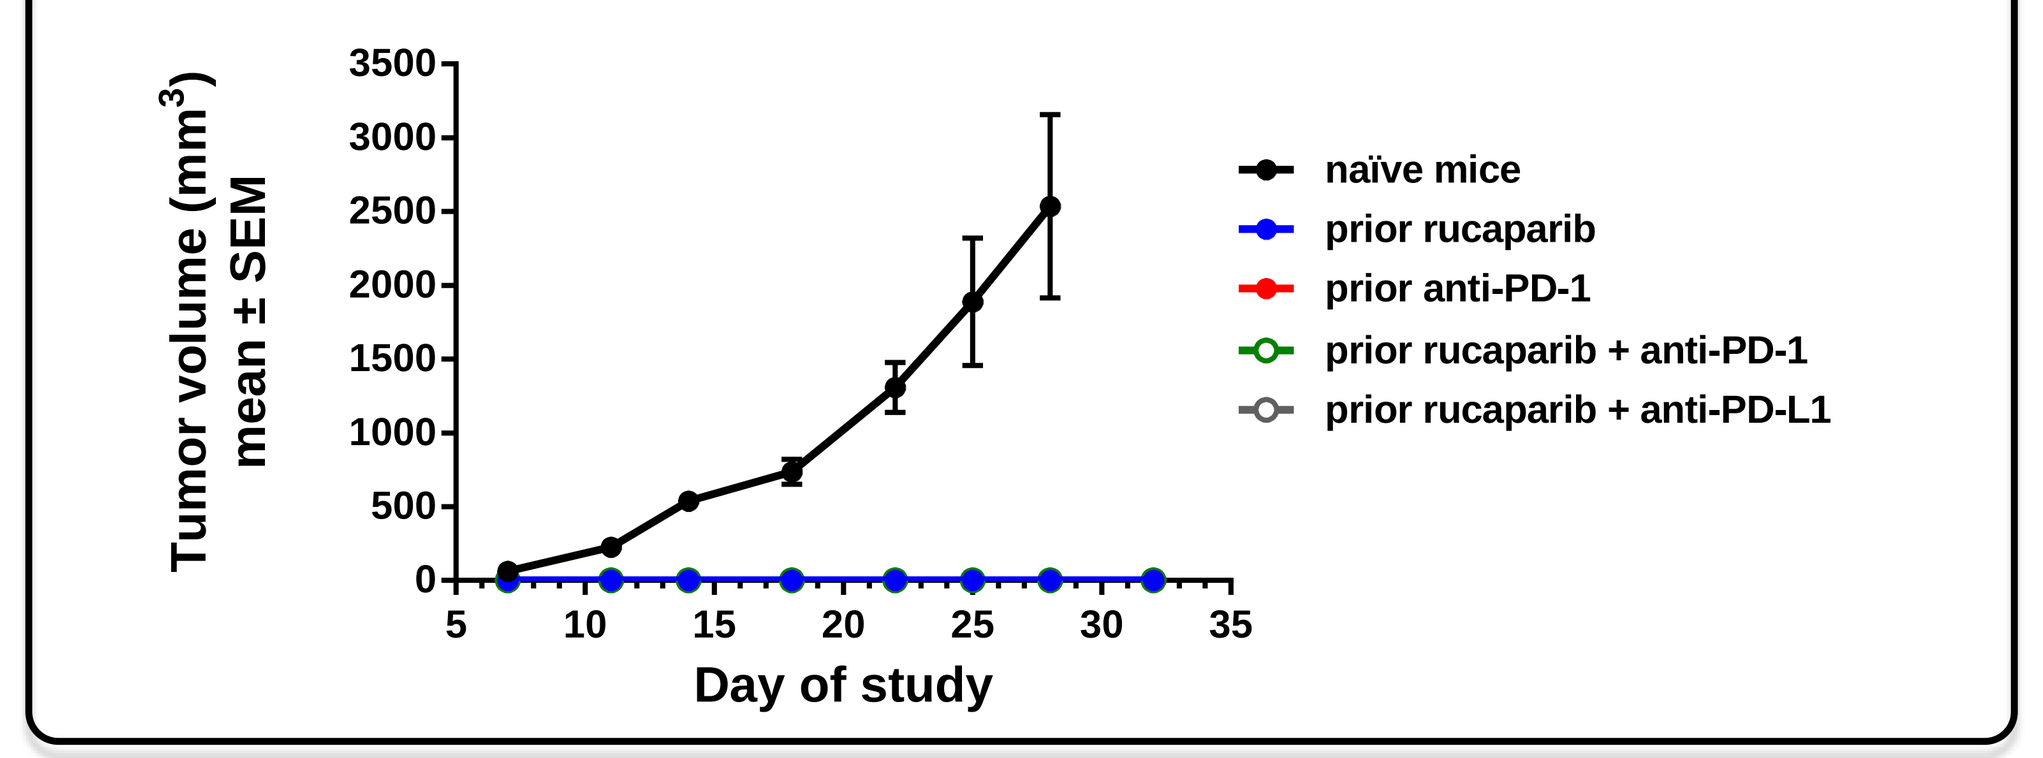
Tumor volume (mm3) mean ± SEM
naïve mice
prior rucaparib
prior anti-PD-1
prior rucaparib + anti-PD-1
prior rucaparib + anti-PD-L1
Day of study

- A re-challenge study was performed on day 125 with 5 surviving animals/group from the previously described efficacy study. Mice were re-challenged with BrKras cells and monitored for tumor growth. Tumor growth was observed in all naïve mice, whereas no tumor growth was observed in any of the other groups.

Rucaparib efficacy in *BRCA1* null syngeneic tumors is dependent on both CD4 and CD8 T-cells

Rucaparib

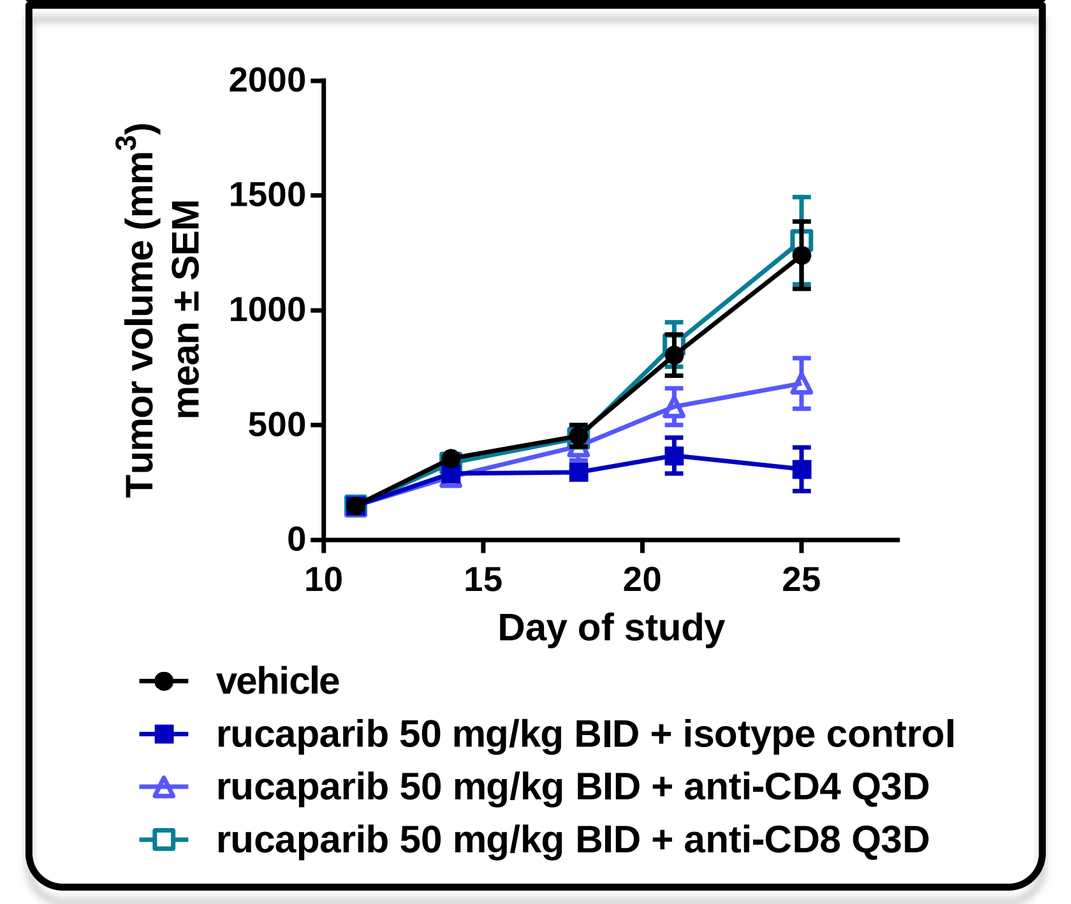
Tumor volume (mm3) mean ± SEM
Day of study
vehicle
rucaparib 50 mg/kg BID + isotype control
rucaparib 50 mg/kg BID + anti-CD4 Q3D
rucaparib 50 mg/kg BID + anti-CD8 Q3D

Anti-PD-L1

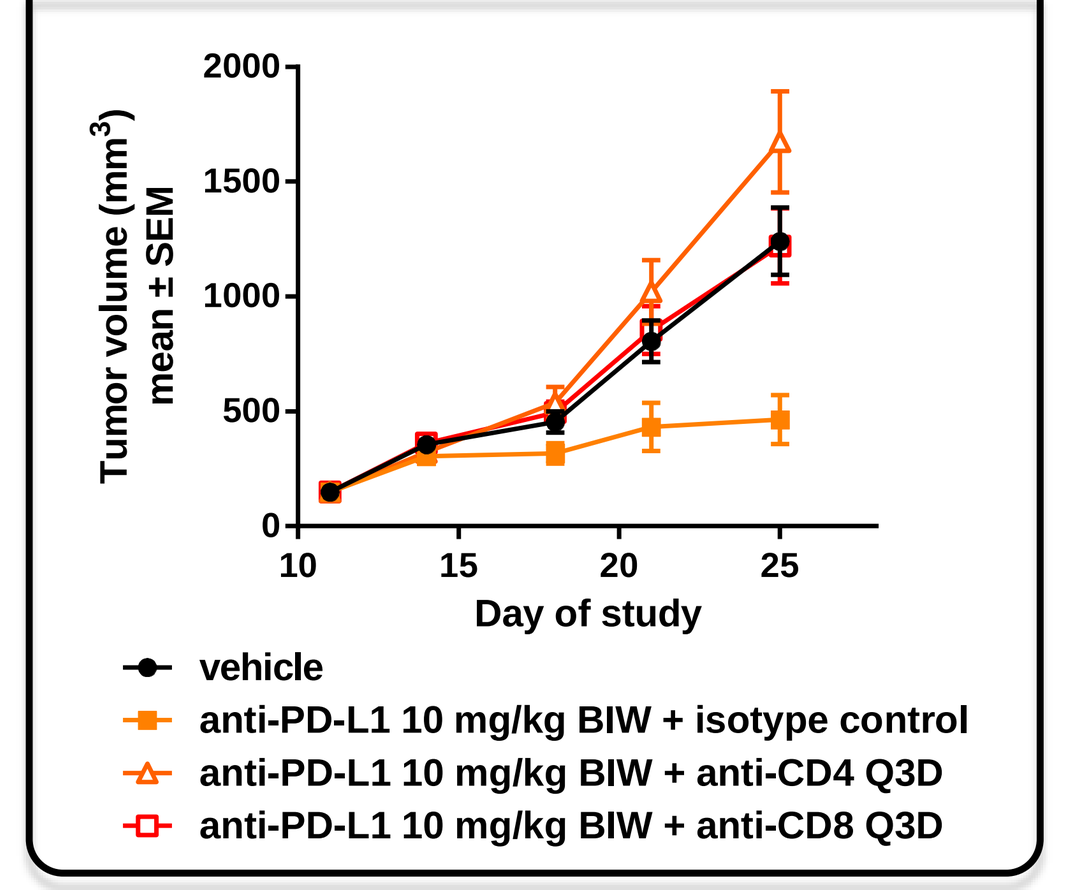
Tumor volume (mm3) mean ± SEM
Day of study
vehicle
anti-PD-L1 10 mg/kg BIW + isotype control
anti-PD-L1 10 mg/kg BIW + anti-CD4 Q3D
anti-PD-L1 10 mg/kg BIW + anti-CD8 Q3D

- The impact of CD4 and CD8 depletion on rucaparib activity was evaluated in the *BRCA1* null BrKras model. Rucaparib activity was abrogated with CD8 depletion and to a lesser extent CD4 depletion, while both CD4 and CD8 depletion decreased anti-PD-L1 activity.

Rucaparib increases the infiltration of CD8 T-cells and CD8 to CD4 T-cell ratio in the tumor

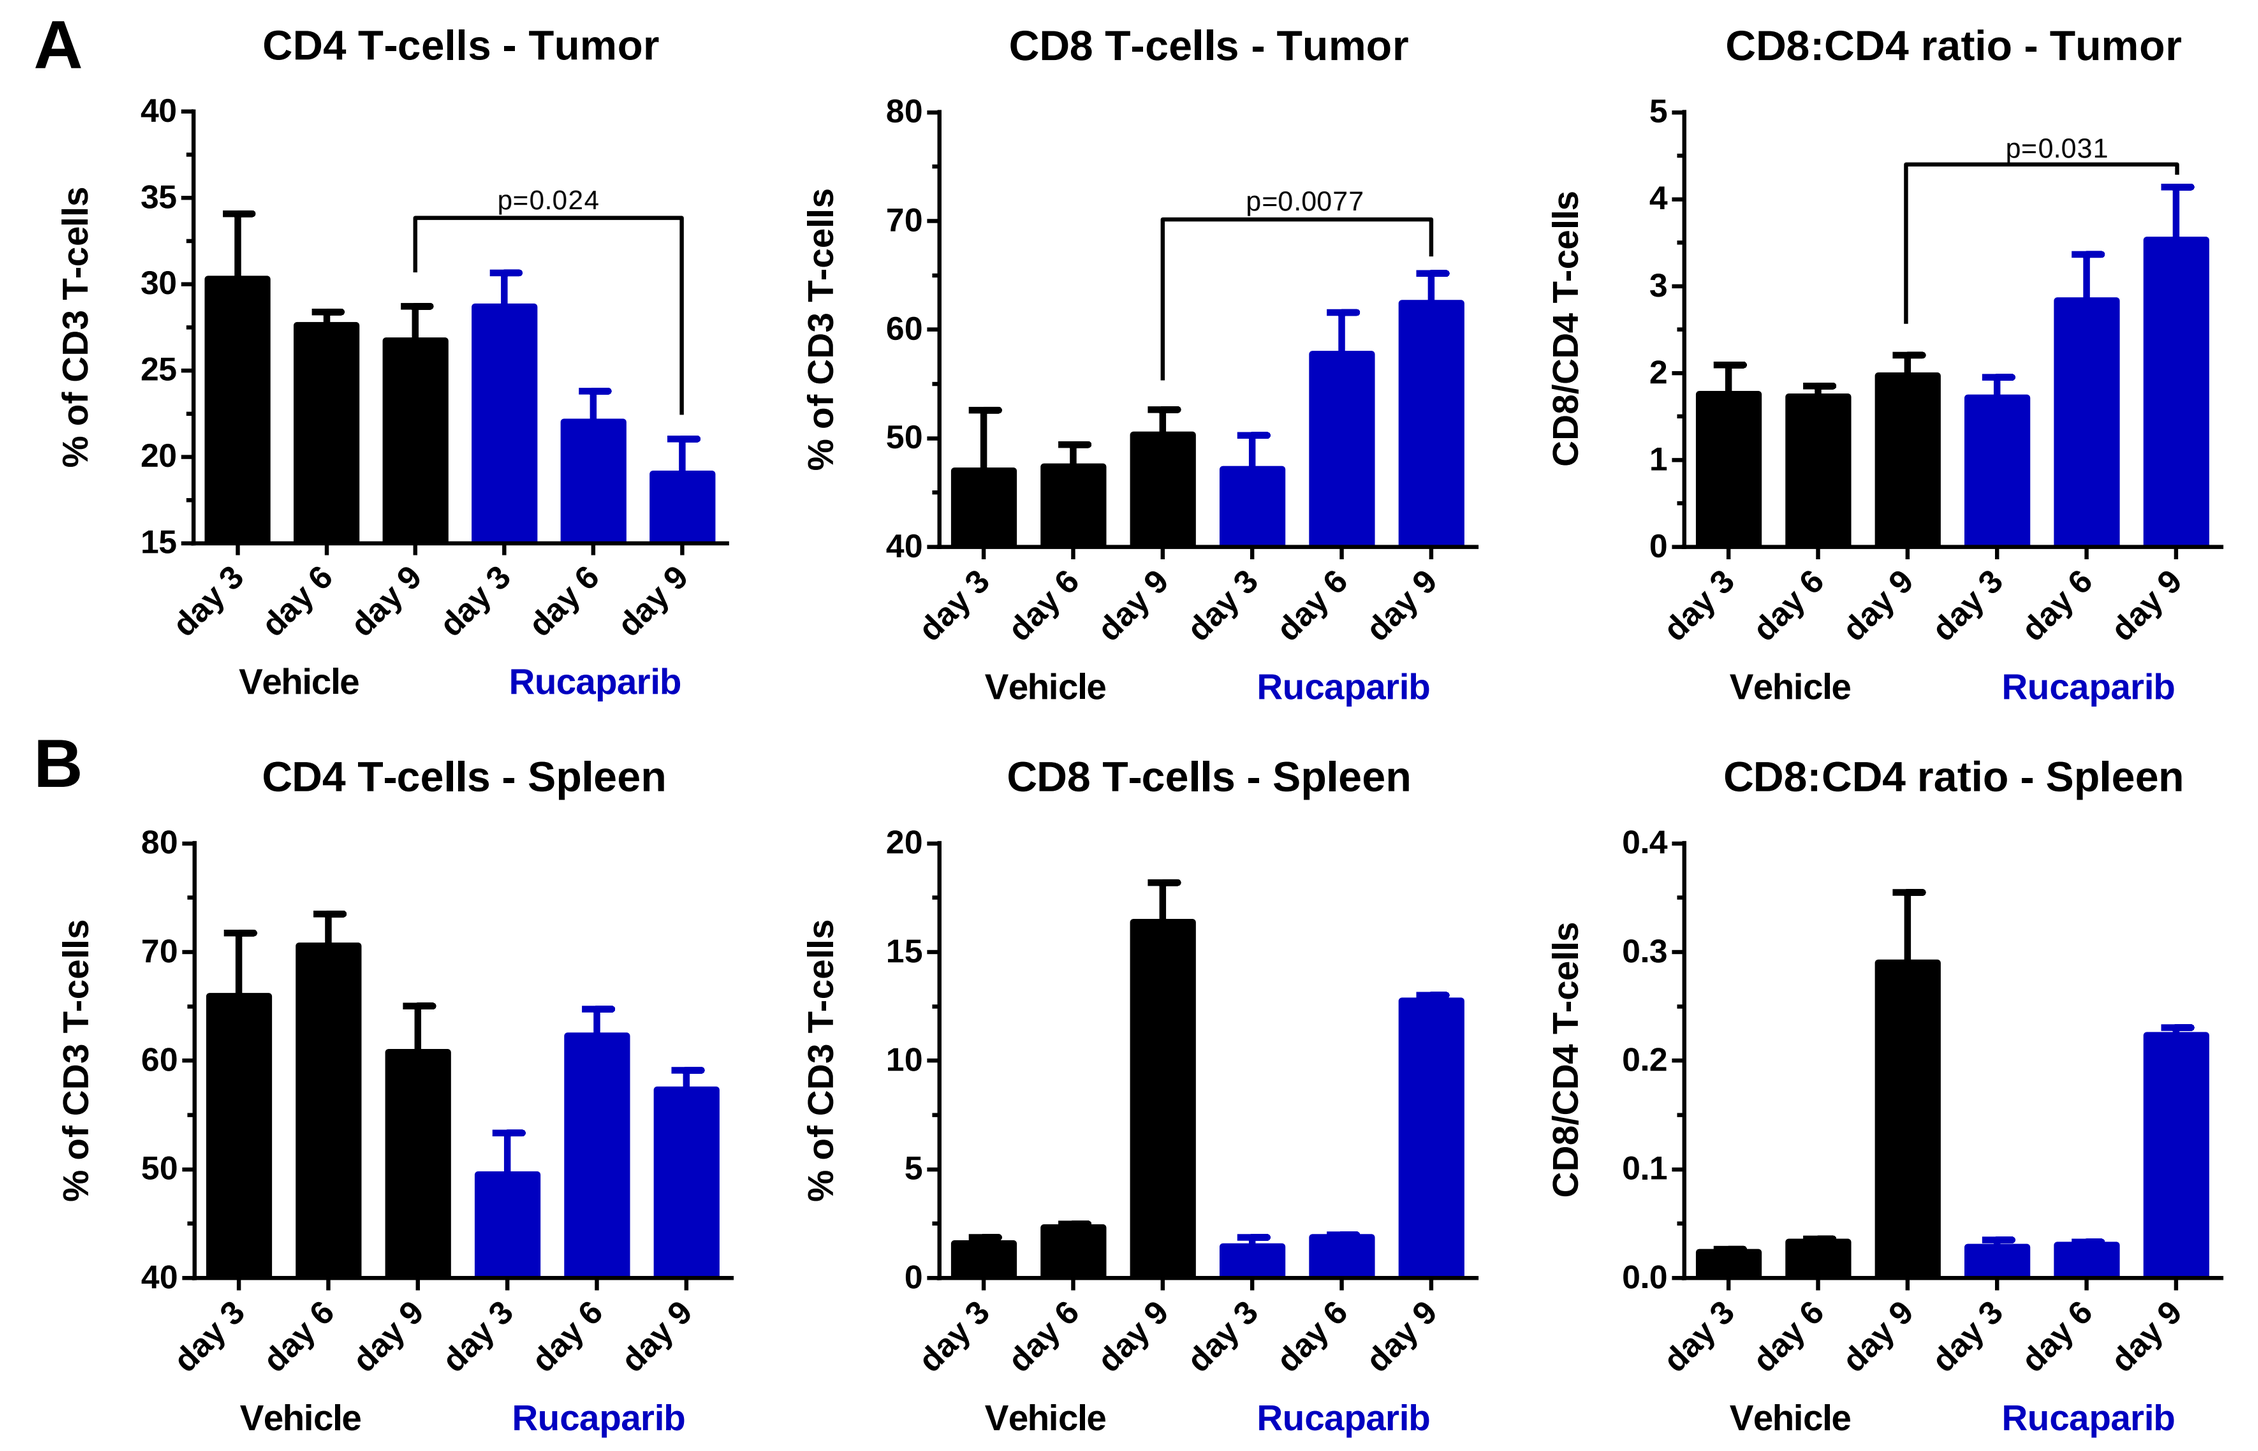
A
CD4 T-cells - Tumor
p=0.024
CD8 T-cells - Tumor
p=0.0077
CD8:CD4 ratio - Tumor
p=0.031
B
CD4 T-cells - Spleen
CD8 T-cells - Spleen
CD8:CD4 ratio - Spleen
% of CD3 T-cells
CD8/CD4 T-cells
day 3 day 6 day 9 day 3 day 6 day 9
Vehicle Rucaparib

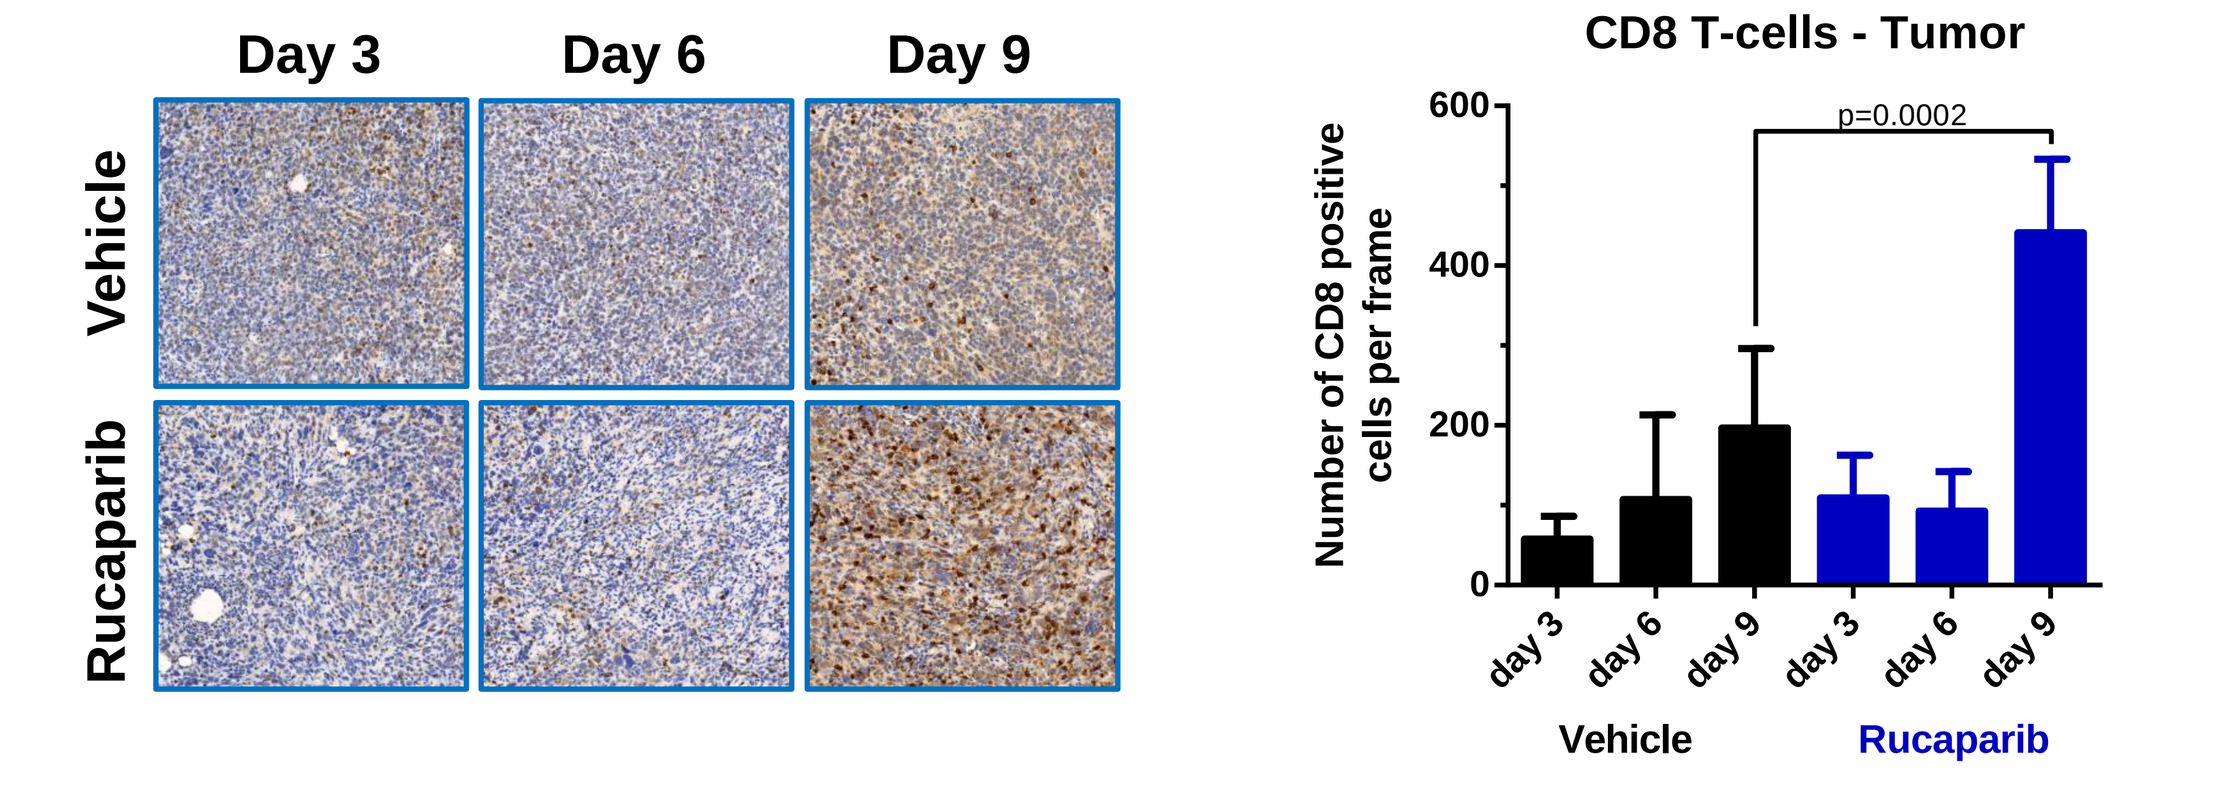
C
Day 3 Day 6 Day 9
Vehicle
Rucaparib
CD8 T-cells - Tumor
p=0.0002
Number of CD8 positive cells per frame
day 3 day 6 day 9 day 3 day 6 day 9
Vehicle Rucaparib

- BrKras tumors were collected from mice treated with vehicle or rucaparib at 150 mg/kg BID and analyzed by flow cytometry. On day 9, a significant decrease in CD4 T-cells, increase in CD8 T-cells, and increase in the CD8/CD4 ratio were observed in tumors treated with rucaparib as compared to vehicle (A). No significant changes in these T-cell populations were observed in the spleen on day 9 (B).
- Tumors were also analyzed for CD8 T-cells by immunohistochemistry, and on day 9 a significant increase in CD8 T-cells was observed in the rucaparib, as compared to vehicle, treated animals (C).

CONCLUSIONS

- **The addition of rucaparib does not impact the activity of PD-1 and PD-L1 inhibition in wild-type *BRCA* syngeneic models**
- **Rucaparib demonstrates potent in vitro and in vivo activity in the BrKras *BRCA1* null syngeneic ovarian cancer model**
- **The combination of rucaparib and PD-1/PD-L1 inhibition provides a greater survival benefit in the BrKras model as compared to monotherapy treatments**
- **Rucaparib monotherapy treatment generated immunological memory in the BrKras model**
- **CD8 T-cells potentially contribute to rucaparib efficacy in the BrKras model**

METHODS

- Cell viability assays: Cells (400-3000/well) were seeded in complete media in 96-well plates, allowed to adhere overnight, and treated with a dilution series of test compounds for 6 days. Cell viability was determined by CellTiter-Glo (Promega).
- Immunofluorescence: Cells were seeded on poly-D-lysine coated chamber slides and treated with 10 µM of rucaparib for 24 hours before methanol fixation and staining using RAD51 (Calbiochem), γH2AX (Millipore), and secondary Alexa Fluor antibodies (Life Technologies).[6]
- Murine syngeneic in vivo studies: Pan02 cells (3×10^6) or MC38 cells (1×10^6) were implanted in C57BL/6J mice (n=10) while EMT6 cells (5×10^5) were implanted in BALB/c mice (n=10). EMT-6 and MC38 tumor-bearing mice were dosed for 21 days, and Pan02 tumor-bearing mice were dosed for 39 days, with rucaparib, anti-PD-L1 (10F.9G2; BioXcell) or anti-PD-1 antibodies (RPMI1-14; BioXcell). All in vivo studies were performed at Crown Biosciences.
- BrKras in vivo studies: BrKras cells (10×10^6) were implanted in FVB mice (n=15). In the dose response study, mice were dosed as indicated with rucaparib for 28 days. In the combination studies of rucaparib and anti-PD-L1 (10F.9G2; BioXcell) or anti-PD-1 (RPMI1-14; BioXcell) mice were dosed for 21 days, and monitored until day 125. On day 125, remaining mice (5 mice per group) and naïve mice were re-implanted with 10×10^6 BrKras cells in the opposing flanks and tumor growth was monitored (with no drug administration).
- Immune depletion study: BrKras cells (10×10^6) were implanted in FVB mice (n=12), and mice were dosed for 21 days. Depletion of CD4 or CD8 was done using anti-CD4 (250 µg) or anti-CD8 (200 µg) antibodies (BioXcell), and initiated simultaneously with the rucaparib and anti-PD-L1 treatments.
- Flow cytometry and IHC: BrKras cells (10×10^6) were implanted in FVB mice (n=6). Mice were treated with vehicle or rucaparib at 150 mg/kg BID, and spleens and tumors were collected on days 3, 6 or 9. For flow cytometry analysis, cells were dissociated using a mouse dissociation kit (Miltenyi Biotec) and a GentleMACS dissociator, and stained with antibodies (BioLegend, eBiosciences or Invitrogen). For IHC, FFPE samples were stained with anti-CD4 (R&D Systems) and anti-CD8 antibodies (Abbiotec) and scanned using a NanoZoomer-HT 2.0 Image system (Hamamatsu).

REFERENCES

1. Rubraca® (rucaparib) tablets [prescribing information]. Boulder, CO: Clovis Oncology, Inc.; 2016.
2. Disis ML, et al. *J Clin Oncol*. 2015;33.
3. Strickland KC, et al. *Oncotarget.* 2016;7:13587-98.
4. Rosado MM et al. *Immunology*. 2013;139:428–37.
5. Xing D and Orsulic S. *Cancer Res.* 2006;66:8949-53.
6. Mukhopadhyay A et al. *Clin Cancer Res.* 2010;16:2344-51.

AACR Annual Meeting Washington DC, April 1-5, 2017